3078

Farnesyl transferase inhibitor darlifarnib in combination with adagrasib in *KRAS* G12C mutated advanced solid tumors: Preliminary results from FIT-001 phase 1 first-in-human trial

Andrea Zivi[1], David S. Hong[2], Manish R. Patel[3], Jacob Thomas[4], Justine Y. Bruce[5], Sanjay Goel[6], Glenn J. Hanna[7], Jacques Medioni[8], Stefania Salvagni[9], Susanna Ulahannan[10], Andrew Hendifar[11], Douglas E. Laux[12], Guzman Alonso[13], Aurore Vozy[14], Ramon Yarza[15], Sabrina Marini[1], Mini Manchanda[16], Tuan Anh Tran[16], Jenchun Kuan[16], Binaifer Balsara[16], Mollie Leoni[16], **Vanesa Gregorc**[17]

[1]Centro Ricerche Cliniche di Verona, Verona, Italy; [2]The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [3]Florida Cancer Specialists, Sarah Cannon Research Institute, Sarasota, FL, USA; [4]Division of Medical Oncology, Department of Medicine, University of Southern California Norris Comprehensive Cancer Center, Los Angeles, CA, USA; [5]University of Wisconsin Carbone Cancer Center, Madison, WI, USA; [6]Rutgers Cancer Institute, New Brunswick, NJ, USA; [7]Center for Head and Neck Oncology, Dana-Farber Cancer Institute, Boston, MA, USA; [8]Hôpital Européen Georges-Pompidou, Paris Cité University, Paris, France; [9]IRCCS Azienda Ospedaliero Universitaria di Bologna, Bologna, Italy; [10]The University of Oklahoma Stephenson Cancer Center/SCRI, Oklahoma City, OK, USA; [11]Cedars Sinai Medical Center, Los Angeles, CA, USA; [12]Division of Hematology, Oncology and Blood & Marrow Transplantation, University of Iowa Hospitals and Clinics, Iowa City, IA, USA; [13]Hospital Universitari Vall d'Hebron, Barcelona, Spain; [14]Hôpital Pitié-Salpêtrière, Paris, France; [15]Hospital Universitario HM Sanchinarro, START Madrid – CIOCC, Madrid, Spain; [16]Kura Oncology, Inc., San Diego, CA, USA; [17]Candiolo Cancer Institute FPO – IRCCS, Candiolo (TO), Italy

BACKGROUND

- Clinical activity of RAS inhibitors (RASi), including KRAS G12Ci, is limited by inherent and adaptive resistance associated with **persistent mTORC1 signaling**[1,2]
- RHEB, a farnesylation-dependent GTPase and key activator of mTORC1, represents a potentially targetable mediator of this resistance[3]
- Darlifarnib (KO-2806), a potent next-generation farnesyl transferase inhibitor (FTI), blocks RHEB farnesylation, resulting in selective inhibition of mTORC1 while sparing mTORC2[4] (**Figure 1**)
- Darlifarnib demonstrates **synergistic antitumor activity with RASi** in preclinical models, including with the KRAS G12Ci adagrasib, with enhanced suppression of resistance pathway signaling and tumor growth[4]
 - In *KRAS* G12C-mutated patients treated with adagrasib monotherapy in the KRYSTAL-1 study, ORRs were 43% in NSCLC (pretreated), 33% in PDAC, and 19% in metastatic CRC (pretreated)[5–7]
- These data support **RHEB-dependent** mTORC1 inhibition as a potential strategy to enhance and prolong responses and also resensitize to KRAS G12C inhibition in *KRAS*-mutant tumors

Figure 1. Darlifarnib + RASi mechanism of action

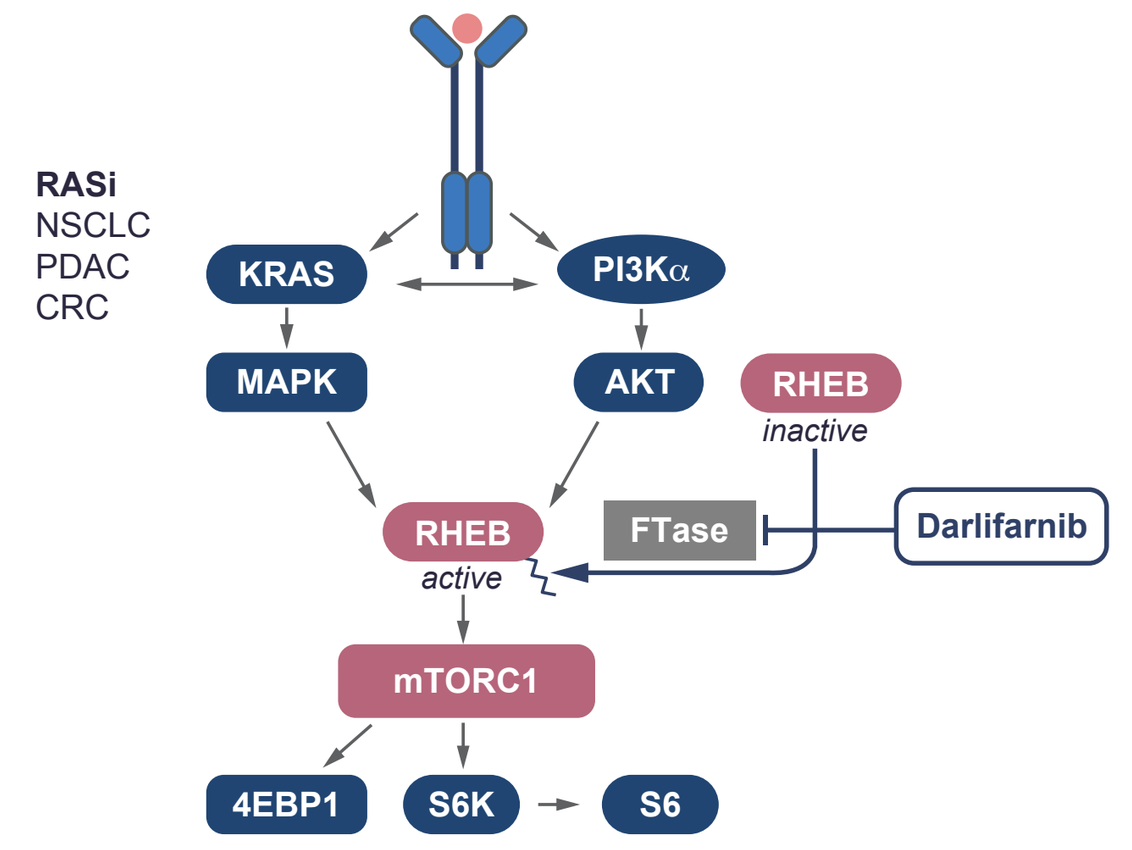

RASi
NSCLC
PDAC
CRC
KRAS
PI3Kα
MAPK
AKT
RHEB
inactive
RHEB
active
FTase
Darlifarnib
mTORC1
4EBP1
S6K
S6

4EBP1, 4E binding protein 1; AKT, protein kinase B; CRC, colorectal cancer; FTase, farnesyl transferase; KRAS, Kirsten rat sarcoma viral oncogene homolog; MAPK, mitogen-activated protein kinase; mTORC1, mechanistic target of rapamycin complex 1; NSCLC, non-small cell lung cancer; PDAC, pancreatic ductal adenocarcinoma; PI3K, phosphoinositide 3-kinase; RASi, RAS inhibitor RHEB, Ras homolog enriched in brain; S6, protein S6; S6K, protein S6 kinase.

AIM

- To assess safety, tolerability, and preliminary antitumor activity of darlifarnib in combination with adagrasib in **heavily pretreated patients with *KRAS* G12C-mutated NSCLC, PDAC, or CRC** (FIT-001 study; NCT06026410)

METHODS

- FIT-001 (NCT06026410) is an ongoing first-in-human, multicenter, open-label, phase 1 dose-escalation/-expansion study evaluating darlifarnib alone and in combination with targeted therapies in patients with advanced solid tumors (**Figure 2**)
- **Darlifarnib + adagrasib combination:**
 - Darlifarnib 3, 5, or 8 mg was administered QD on Days 1–7 and 15–21 of each 28-day cycle in combination with adagrasib 400 mg BID
 - Darlifarnib 8 mg + adagrasib 400 mg BID is not being advanced for further evaluation at this time, per protocol
 - Here we present data for the candidate doses: darlifarnib 3 and 5 mg + adagrasib 400 mg BID

Figure 2. FIT-001: Darlifarnib + adagrasib in *KRAS* G12C-mutated NSCLC, PDAC, and CRC

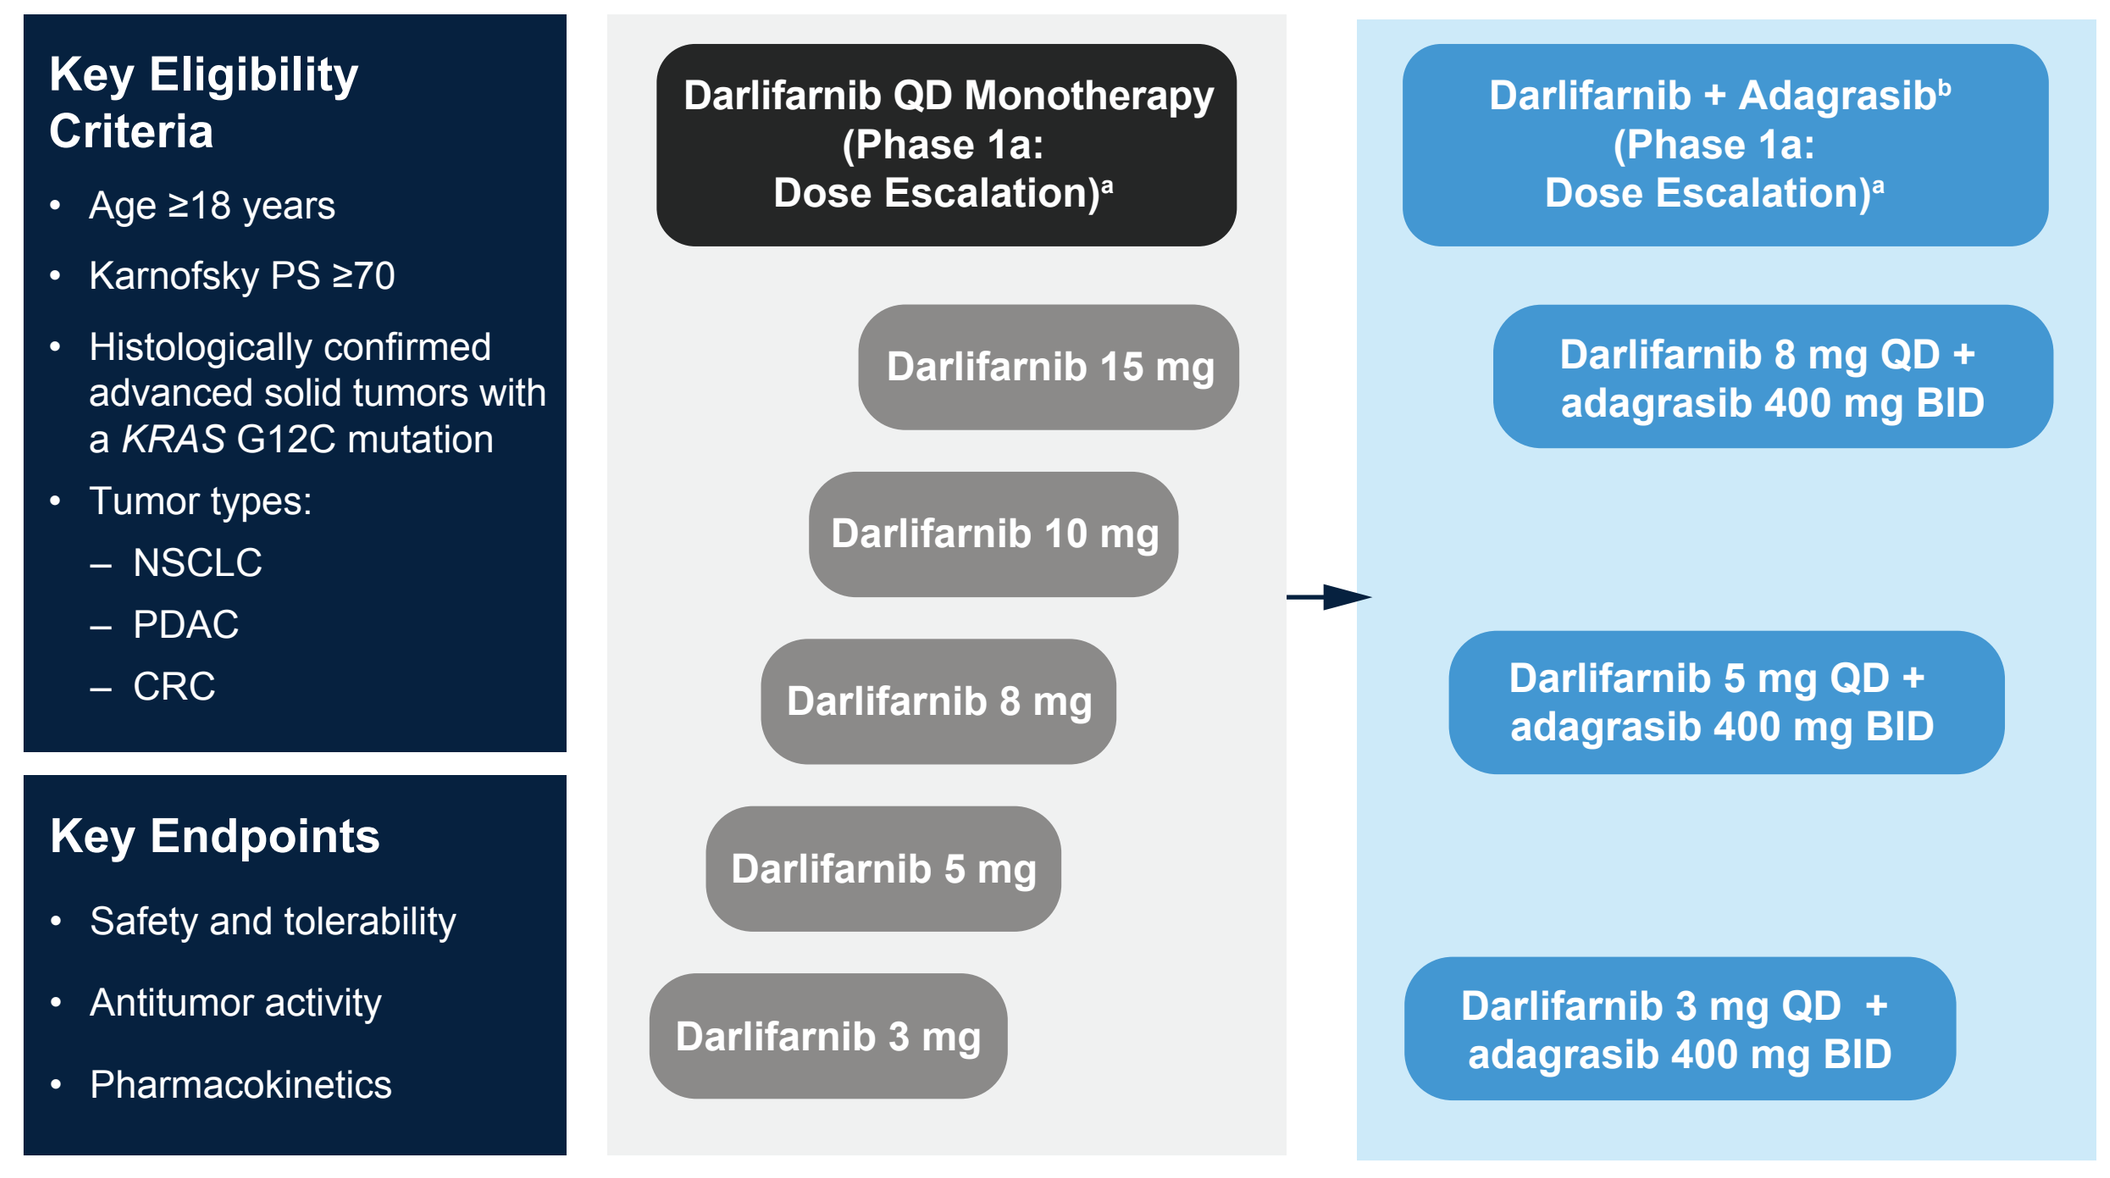

Key Eligibility Criteria
Age ≥18 years
Karnofsky PS ≥70
Histologically confirmed advanced solid tumors with a KRAS G12C mutation
Tumor types:
– NSCLC
– PDAC
– CRC
Key Endpoints
Safety and tolerability
Antitumor activity
Pharmacokinetics
Darlifarnib QD Monotherapy (Phase 1a: Dose Escalation)a
Darlifarnib 15 mg
Darlifarnib 10 mg
Darlifarnib 8 mg
Darlifarnib 5 mg
Darlifarnib 3 mg
Darlifarnib + Adagrasibb (Phase 1a: Dose Escalation)a
Darlifarnib 8 mg QD + adagrasib 400 mg BID
Darlifarnib 5 mg QD + adagrasib 400 mg BID
Darlifarnib 3 mg QD + adagrasib 400 mg BID

[a]Each individual patient will receive one of the planned dose levels of darlifarnib. [b]Adagrasib 400 mg BID; approved adagrasib monotherapy dose is 600 mg BID.[8] BID, twice daily; CRC, colorectal cancer; KRAS, Kirsten rat sarcoma viral oncogene homolog; NSCLC, non-small cell lung cancer; PDAC, pancreatic ductal adenocarcinoma; PS, performance status; QD, once daily. Data cutoff: Mar 25, 2026.

RESULTS

Patients and treatment

- As of March 25, 2026, 30 patients with *KRAS* G12C-mutated NSCLC, PDAC, or CRC were enrolled and treated with darlifarnib 3 or 5 mg + adagrasib across 18 sites in the US and EU (**Table 1**)
- Two-thirds of patients were heavily pretreated and received the darlifarnib + adagrasib combination as third line or higher treatment

Table 1. Summary of patient disposition, demographics, and baseline characteristics

n (%)	Darlifarnib 3 mg + adagrasib[a] (n=15)	Darlifarnib 5 mg + adagrasib[a] (n=15)
Patient disposition		
Treated with darlifarnib	15 (100)	15 (100)
Ongoing on study treatment	3 (20)	8 (53)
Median follow-up, months (range)	6.3 (0.3–13.2)	9.0 (0.5–11.8)
Discontinued	12 (80)	7 (47)
Progressive disease	8 (53)	3 (20)
Patient withdrawal[b]	2 (13)	1 (7)
Symptomatic deterioration	1 (7)	1 (7)
Adverse event[c]	0	1 (7)
Physician decision	0	1 (7)
Death[d]	1 (7)	0
Demographics and baseline characteristics		
Median age, years (range)	61 (45–79)	60 (22–74)
Sex		
Male	12 (80)	7 (47)
Female	3 (20)	8 (53)
Race		
White	12 (80)	13 (87)
Non-White[e]	3 (20)	2 (13)
Karnofsky PS		
80–100	14 (93)	14 (93)
50–70	1 (7)	1 (7)
Primary tumor type		
NSCLC	5 (33)	4 (27)
PDAC	3 (20)	3 (20)
CRC	7 (47)	8 (53)
Prior systemic therapy		
1	5 (33)	5 (33)
2	1 (7)	6 (40)
≥3	9 (60)	4 (27)
Prior KRAS G12Ci (any type)	8 (53)	4 (27)
KRAS G12Ci as immediate prior line	4 (27)	1 (7)
Prior KRASi[f]		
Sotorasib	5 (33)	2 (13)
Adagrasib	2 (13)	1 (7)
Daraxonrasib + elironrasib combination	1 (7)	1 (7)
Divarasib	1 (7)	0

[a]Adagrasib 400 mg BID. [b]Includes treatment discontinuation due to "Other: withdrawal by patient due to toxicity." [c]Grade 5 bronchopulmonary hemorrhage unrelated to darlifarnib or adagrasib. [d]Death due to cardiac arrest unrelated to darlifarnib or adagrasib; patient had prior medical history of chronic atrial fibrillation. [e]Includes Black or African American, Asian, American Indian or Alaska Native, Other, and Multiple. [f]A subset of patients received multiple KRASi. BID, twice daily; CRC, colorectal cancer; G12Ci, G12C inhibitor; KRAS, Kirsten rat sarcoma viral oncogene homolog; KRASi, KRAS inhibitor; NSCLC, non-small cell lung cancer; PDAC, pancreatic ductal adenocarcinoma; PS, performance status. Data cutoff: Mar 25, 2026.

- Patients with NCSLC or CRC were heavily pretreated; approximately half of these patients had received prior KRAS G12Ci therapy (**Table 2**)

Table 2. Prior therapy by tumor type

n (%)	PDAC (n=6)	NSCLC (n=9)	CRC (n=15)
Prior systemic therapy			
1	4 (67)	5 (56)	2 (13)
2	2 (33)	0	5 (33)
≥3	0	4 (44)	8 (53)
Prior KRAS G12Ci (any type)	0	4 (44)	8 (53)
KRAS G12Ci as immediate prior line	0	1 (7)	4 (27)
Prior KRASi			
Sotorasib	0	4 (44)	3 (20)
Adagrasib	0	0	3 (20)
Daraxonrasib + elironrasib combination	0	0	2 (13)
Divarasib	0	0	1 (7)

CRC, colorectal cancer; G12Ci, G12C inhibitor; KRAS, Kirsten rat sarcoma viral oncogene homolog; KRASi, KRAS inhibitor; NSCLC, non-small cell lung cancer; PDAC, pancreatic ductal adenocarcinoma. Data cutoff: Mar 25, 2026.

Safety and tolerability

- Darlifarnib 3 and 5 mg in combination with adagrasib demonstrated a well-tolerated safety profile, with safety data and adverse events consistent with expected on-target effects
- Most common any-grade TEAEs are presented in **Table 3**
 - TEAEs were predominantly grades 1–2, with low rates of treatment discontinuation
- Most common (≥30% of all patients) any-grade TRAEs:
 - Darlifarnib-related: Anemia (47%), diarrhea (47%), nausea (43%), neutropenia (43%), and thrombocytopenia (37%)
 - Adagrasib-related: Diarrhea (60%), nausea (57%), anemia (40%), vomiting (40%), thrombocytopenia (33%), asthenia (30%), and neutropenia (30%)
- Most common (≥15% of all patients) grade ≥3 TRAEs were neutropenia (33%) and anemia (20%) with darlifarnib, and neutropenia (20%) with adagrasib
- Hepatic and renal TEAEs were rare, and only 1 grade ≥3 event occurred
 - No hepatic or renal treatment-related grade ≥3 events, DLTs, or dose reductions or discontinuations were observed
- Darlifarnib dose reductions, interruptions, and discontinuations are presented in **Table 3**
 - Median length of dose interruptions due to darlifarnib-related adverse events was 7 days (range 1–29), primarily due to myelosuppression; 19 of 21 patients reinitiated treatment (3 at a reduced darlifarnib dose)
- Of 20 DLT-evaluable patients who received darlifarnib 3 or 5 mg + adagrasib, one DLT (grade 3 anemia) occurred in a patient treated with darlifarnib 5 mg + adagrasib, which resolved and the patient continued on study treatment

Table 3. Treatment-emergent adverse events[a]

n (%)	Darlifarnib 3 mg + adagrasib[b] (n=15)	Darlifarnib 5 mg + adagrasib[b] (n=15)
Any-grade TEAEs (≥30% of all patients)	15 (100)	15 (100)
Diarrhea	11 (73)	10 (67)
Nausea	10 (67)	10 (67)
Anemia	4 (27)	12 (80)
Vomiting	9 (60)	6 (40)
Neutropenia	4 (27)	9 (60)
Thrombocytopenia	2 (13)	10 (67)
Asthenia	2 (13)	8 (53)
Grade ≥3 TEAEs (≥15% of all patients)[c]	8 (53)	12 (80)
Neutropenia	3 (20)	7 (47)
Anemia	2 (13)	5 (33)
SAEs	2 (13)	7 (47)
Treatment-related SAEs	1 (7)	2 (13)
TEAEs leading to darlifarnib dose reduction	0	3 (20)
TRAEs leading to darlifarnib dose reduction	0	3 (20)
TEAEs leading to darlifarnib dose interruption	7 (47)	14 (93)
TRAEs leading to darlifarnib dose interruption	5 (33)	13 (87)
TEAEs leading to darlifarnib drug withdrawal	1 (7)	1 (7)
TRAEs leading to darlifarnib drug withdrawal	0	0

[a]Prophylaxis, including for myelosuppression, was not allowed during the DLT period (first 28 days in cycle 1). [b]Adagrasib 400 mg BID. [c]Of patients treated with darlifarnib 3 or 5 mg + adagrasib, one patient (1/30; 3%) had febrile neutropenia (grade 3). DLT, dose-limiting toxicity; SAE, serious adverse event; TEAE, treatment emergent adverse event; TRAE, treatment-related adverse event. Data cutoff: Mar 25, 2026.

Antitumor activity

- Among 26 response-evaluable patients, confirmed responses and clinical benefit were observed in each tumor type and at each dose level (**Table 4**, **Figure 3**)

Table 4. Antitumor activity in 26 response-evaluable[a] patients

% (95% CI)	n	ORR	CBR[b]	mDOR, mos
Tumor type				
PDAC	6	67 (22–96)	100 (54–100)	NE (3.8–NE)
NSCLC	6	50 (12–88)	50 (12–88)	NE (5.7–NE)
CRC	14	14 (2–43)	43 (18–71)	NE (4.4–NE)
KRASi-naive CRC	7	29 (3–60)	43 (8–70)	NE (4.4–NE)
Dose level				
Darlifarnib 3 mg + adagrasib[c]	12	25 (6–57)	42 (15–72)	NE (3.8–NE)
Darlifarnib 5 mg + adagrasib[c]	14	43 (18–71)	71 (42–92)	5.7 (4.4–NE)

[a]Response-evaluable patients had ≥1 darlifarnib dose and post-baseline scan. [b]CBR defined as the percentage of patients who achieved confirmed CR, confirmed PR, or SD duration ≥12 weeks. [c]Adagrasib 400 mg BID. BID, twice daily; CBR, clinical benefit rate; CI, confidence interval; CRC, colorectal cancer; KRASi, Kirsten rat sarcoma viral oncogene homolog inhibitor; mDOR, median duration of response; NE, not estimable; NSCLC, non-small cell lung cancer; ORR, objective response rate; PDAC, pancreatic ductal adenocarcinoma; PR, partial response; SD, stable disease. Data cutoff: Mar 25, 2026.

- Median follow-up time (range) was 6.9 (3.2–11.8), 8.9 (1.2–13.2), and 6.7 (4.0–10.4) months for NSCLC, CRC, and PDAC, respectively
- Tumor shrinkage was observed in 77% (20/26) of all response-evaluable patients, including in patients with prior KRASi treatment (**Figure 4**)
- 37% of patients remained on study treatment as of the data cutoff date (**Figure 5**)

Figure 3. Duration and response in evaluable[a] patients

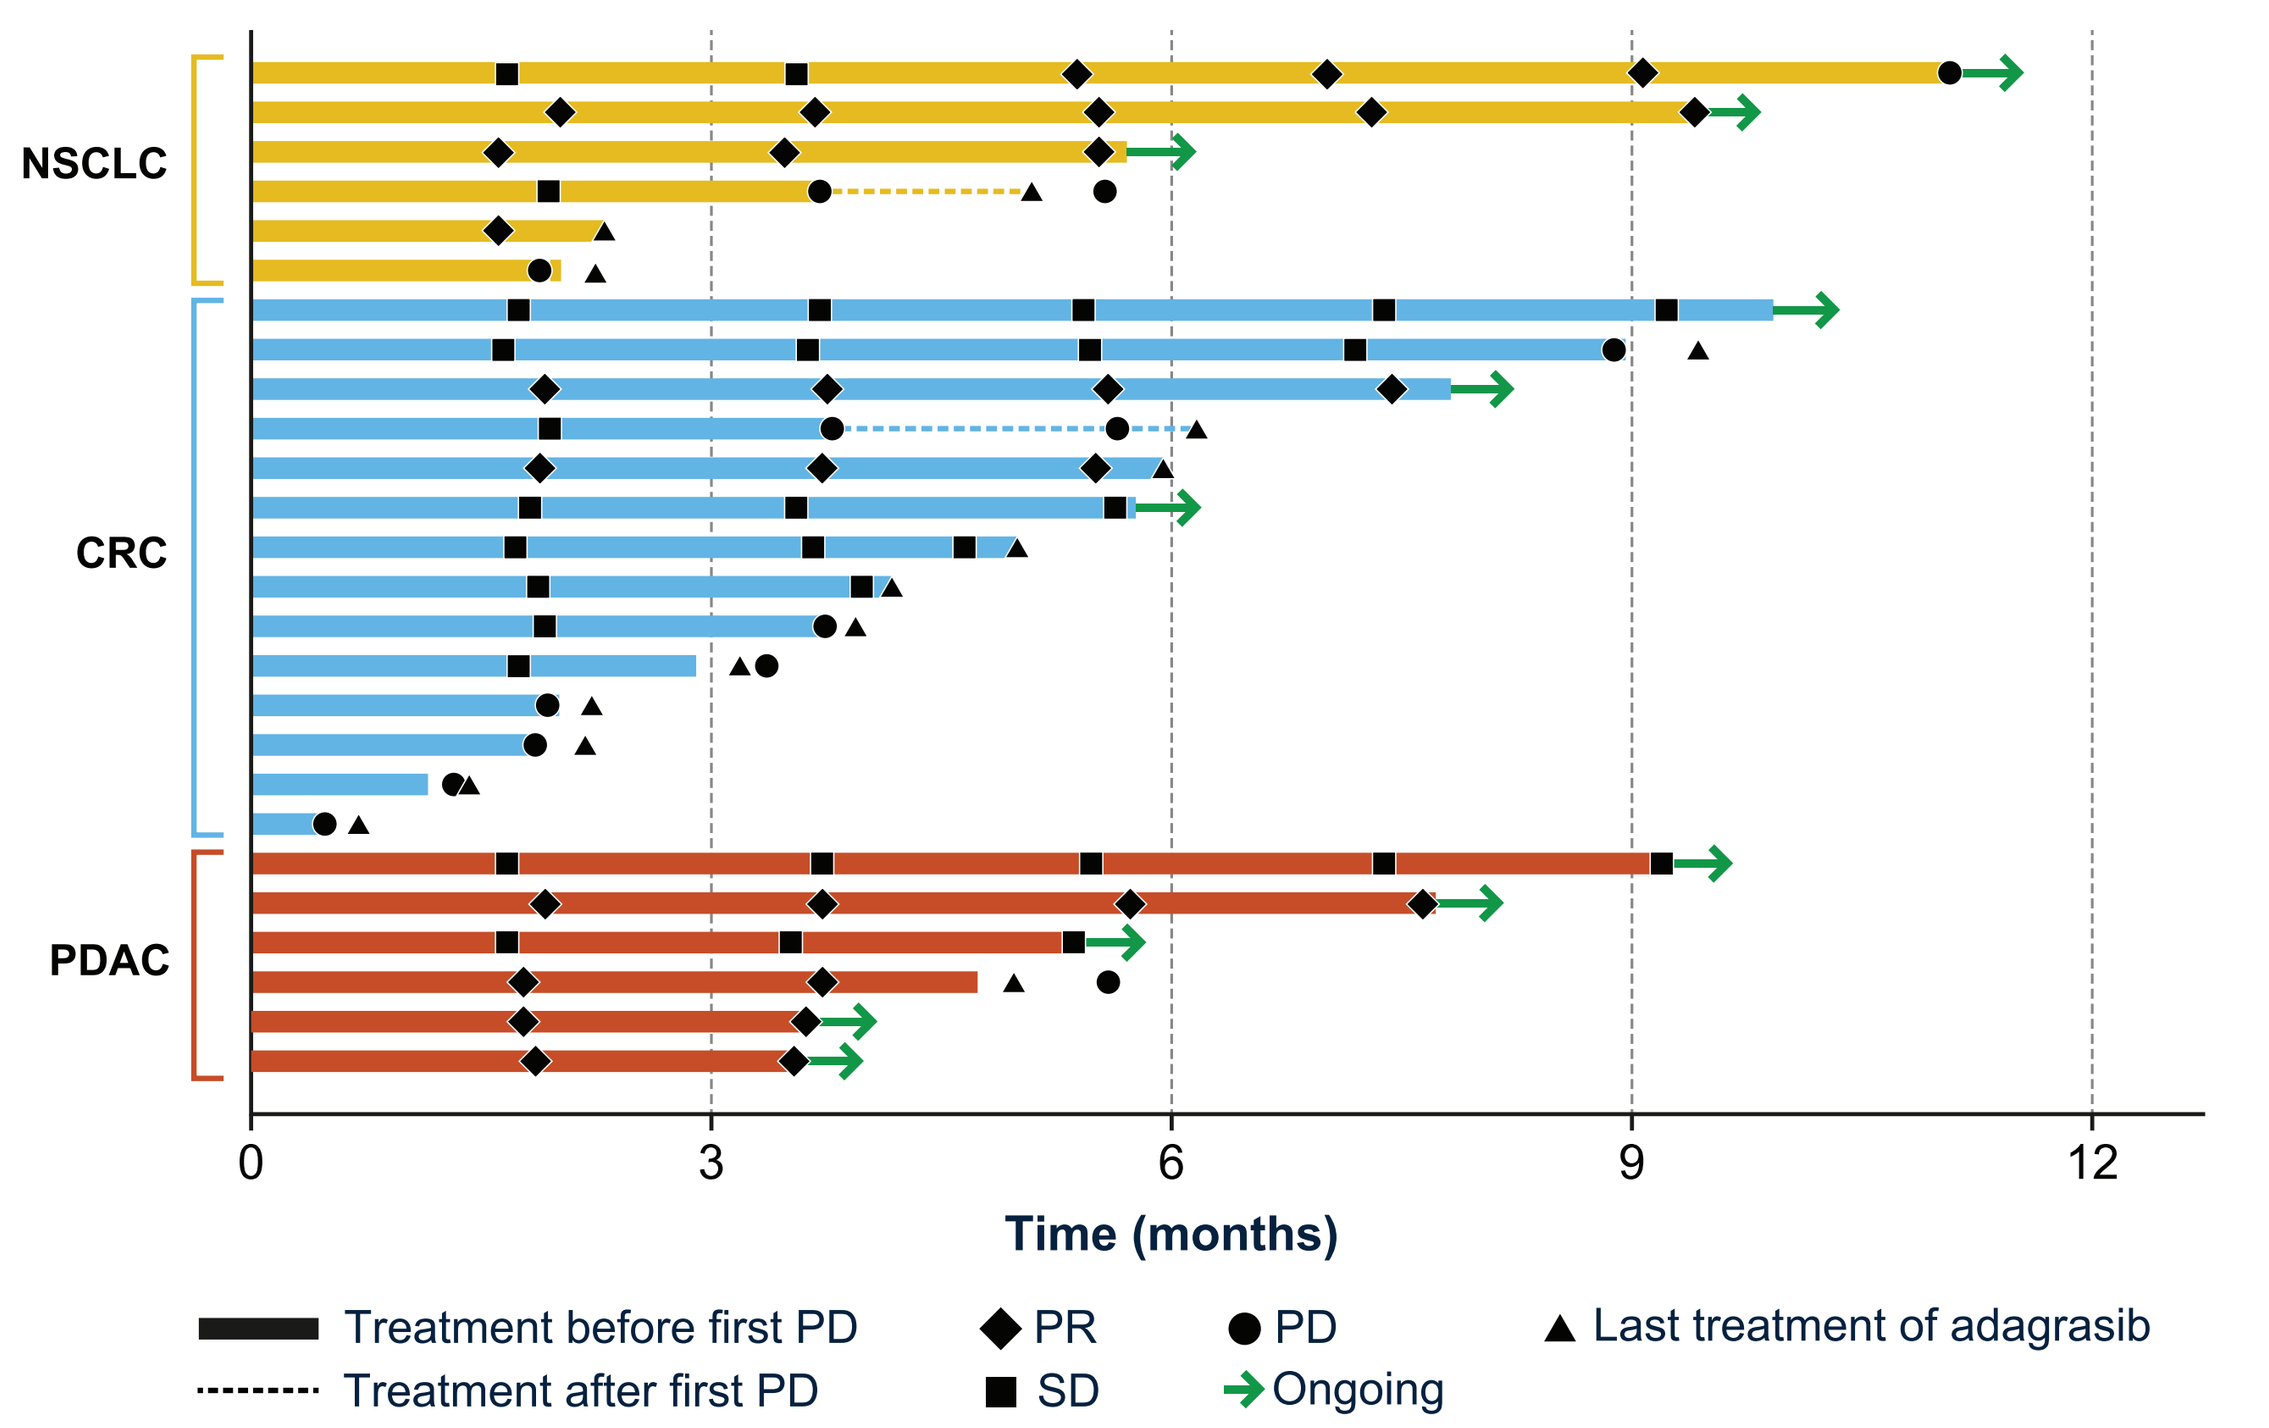

NSCLC
CRC
PDAC
0
3
6
9
12
Time (months)
Treatment before first PD
Treatment after first PD
PR
SD
PD
Ongoing
Last treatment of adagrasib

[a]Response-evaluable patients had ≥1 darlifarnib dose and post-baseline scan. CRC, colorectal cancer; NSCLC, non-small cell lung cancer; PD, progressive disease; PDAC, pancreatic ductal adenocarcinoma; PR, partial response; SD, stable disease. Data cutoff: Mar 25, 2026.

Figure 4. Best percent change from baseline in target lesion size

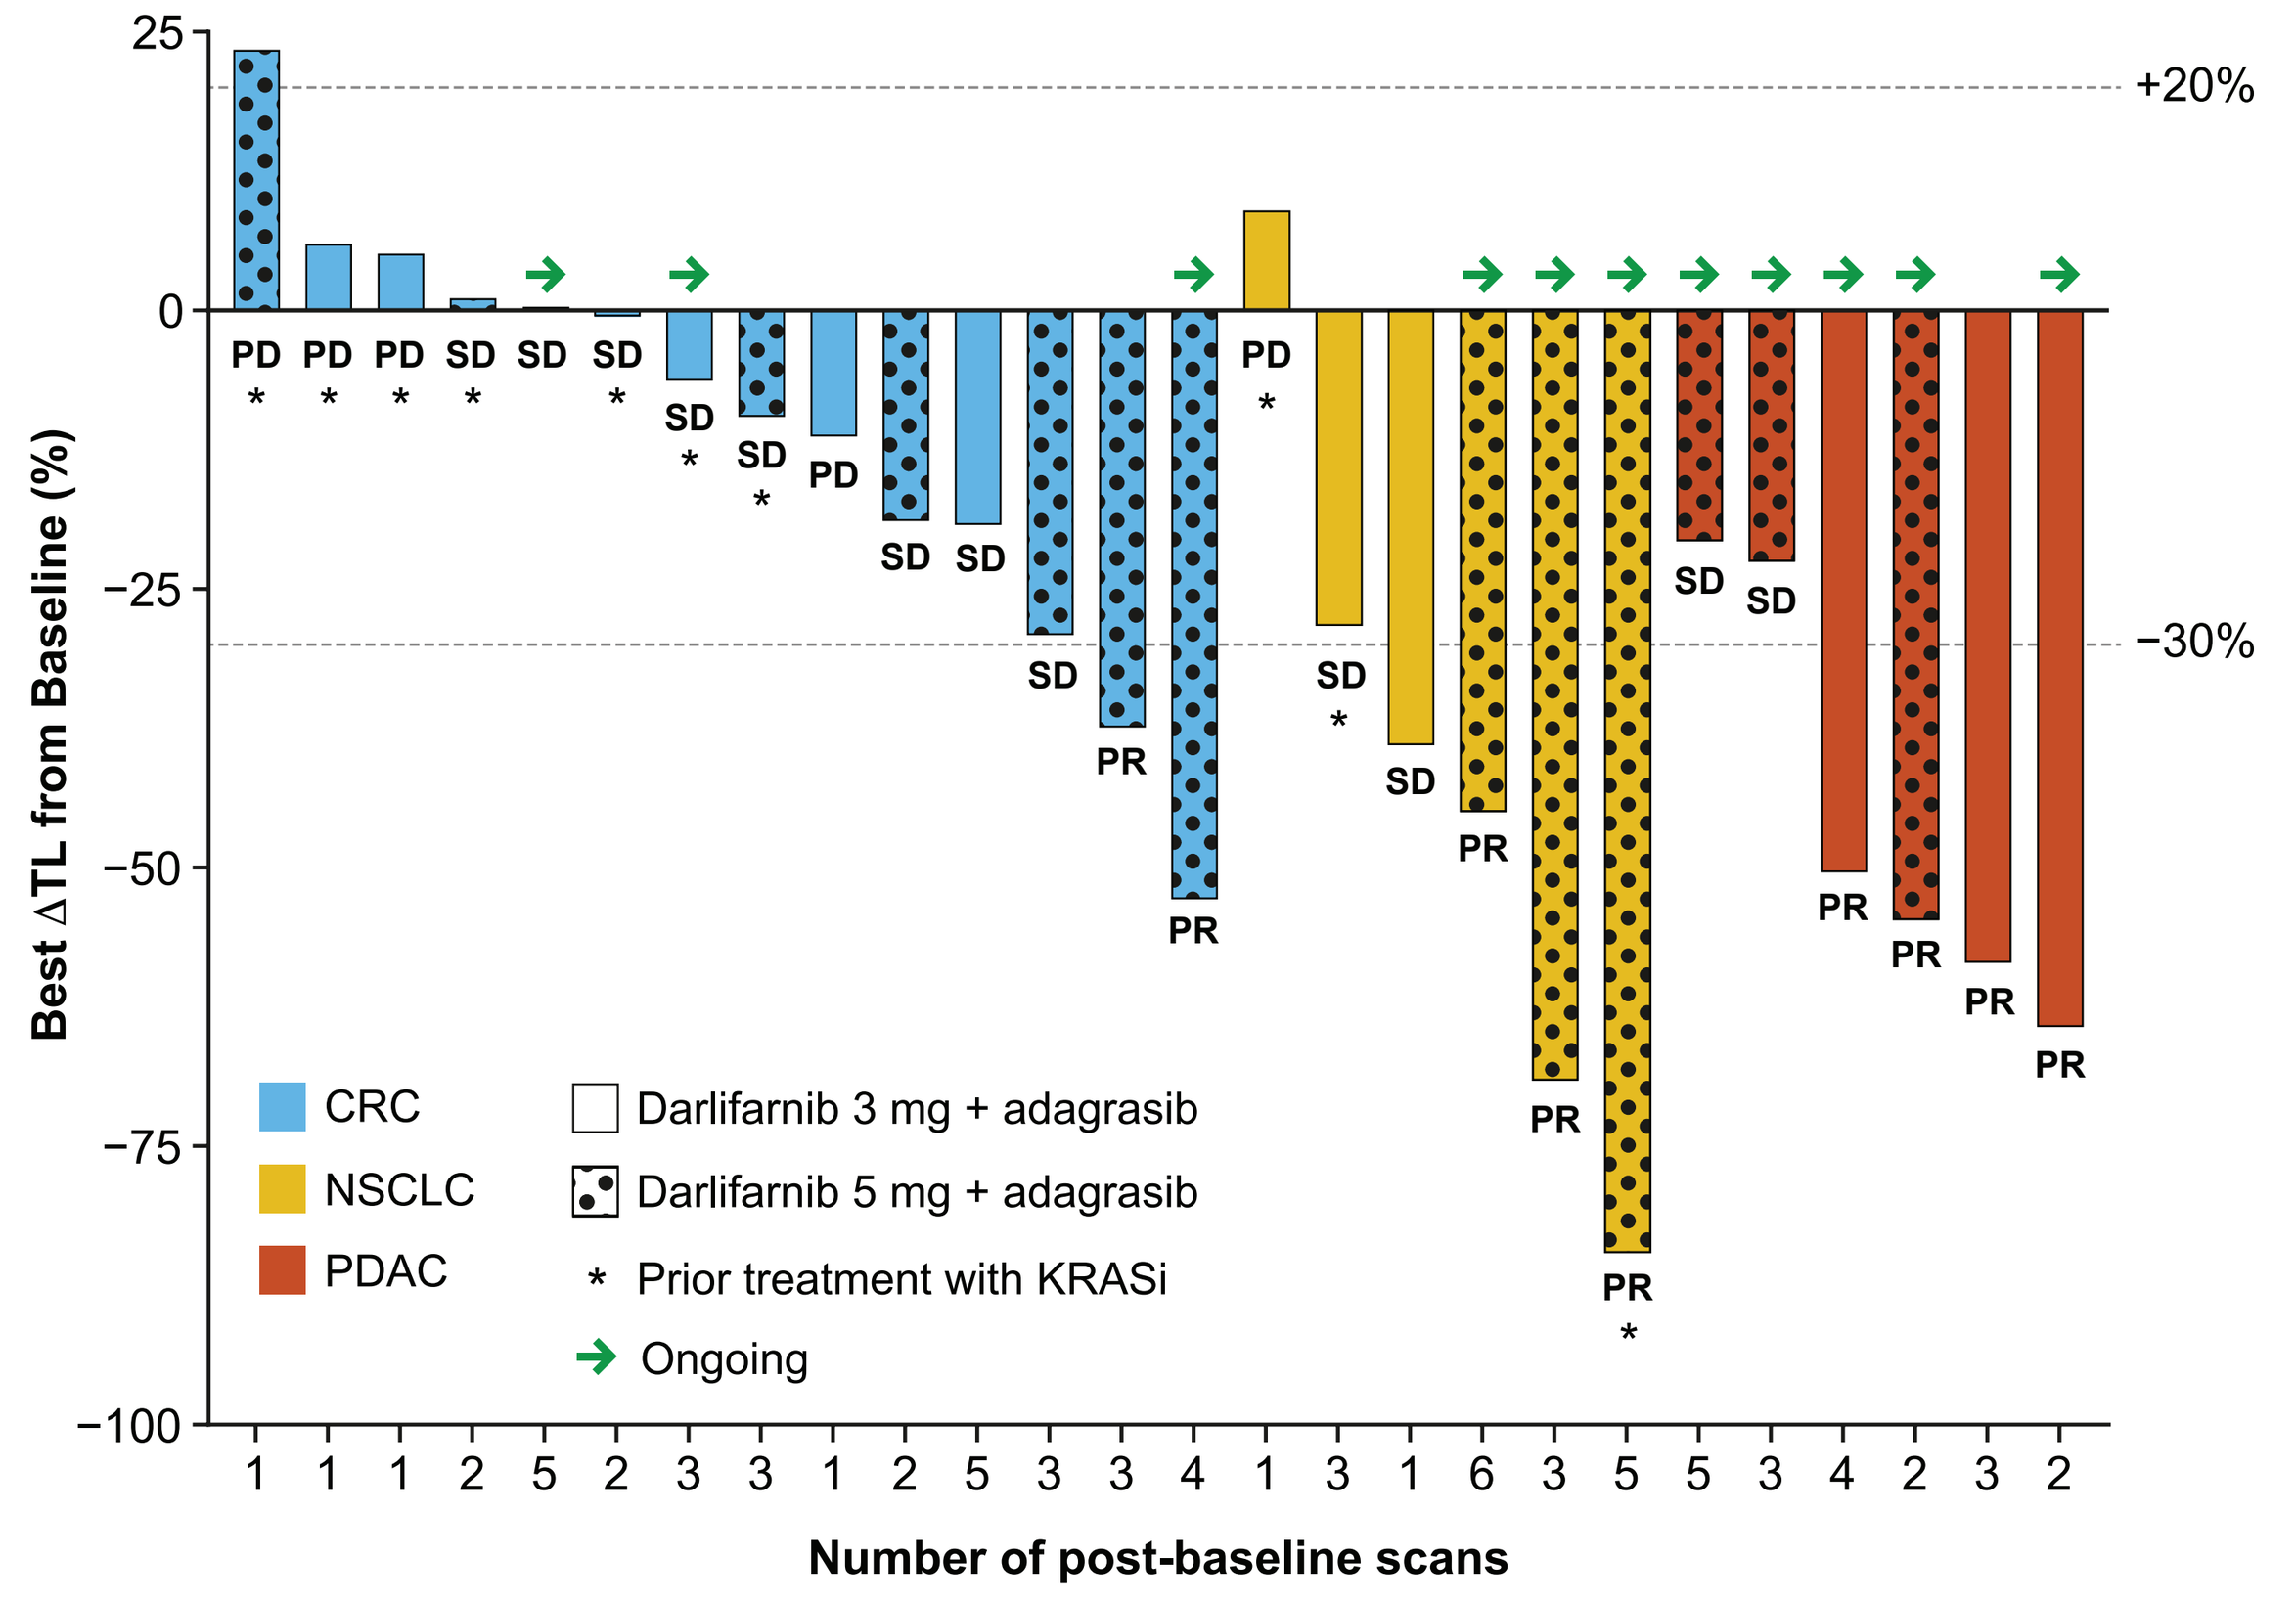

Best ΔTL from Baseline (%)
+20%
−30%
CRC
NSCLC
PDAC
Darlifarnib 3 mg + adagrasib
Darlifarnib 5 mg + adagrasib
* Prior treatment with KRASi
Ongoing
1 1 1 2 5 2 3 3 1 2 5 3 3 4 1 3 1 6 3 5 5 3 4 2 3 2
Number of post-baseline scans

Response-evaluable patients had ≥1 darlifarnib dose and post-baseline scan. Adagrasib 400 mg BID. CRC, colorectal cancer; KRASi, Kirsten rat sarcoma viral oncogene homolog inhibitor; NSCLC, non-small cell lung cancer; PD, progressive disease; PDAC, pancreatic ductal adenocarcinoma; PR, partial response; SD, stable disease; TL, target lesion. Data cutoff: Mar 25, 2026.

Figure 5. Change in target lesion SoD over time in evaluable[a] patients

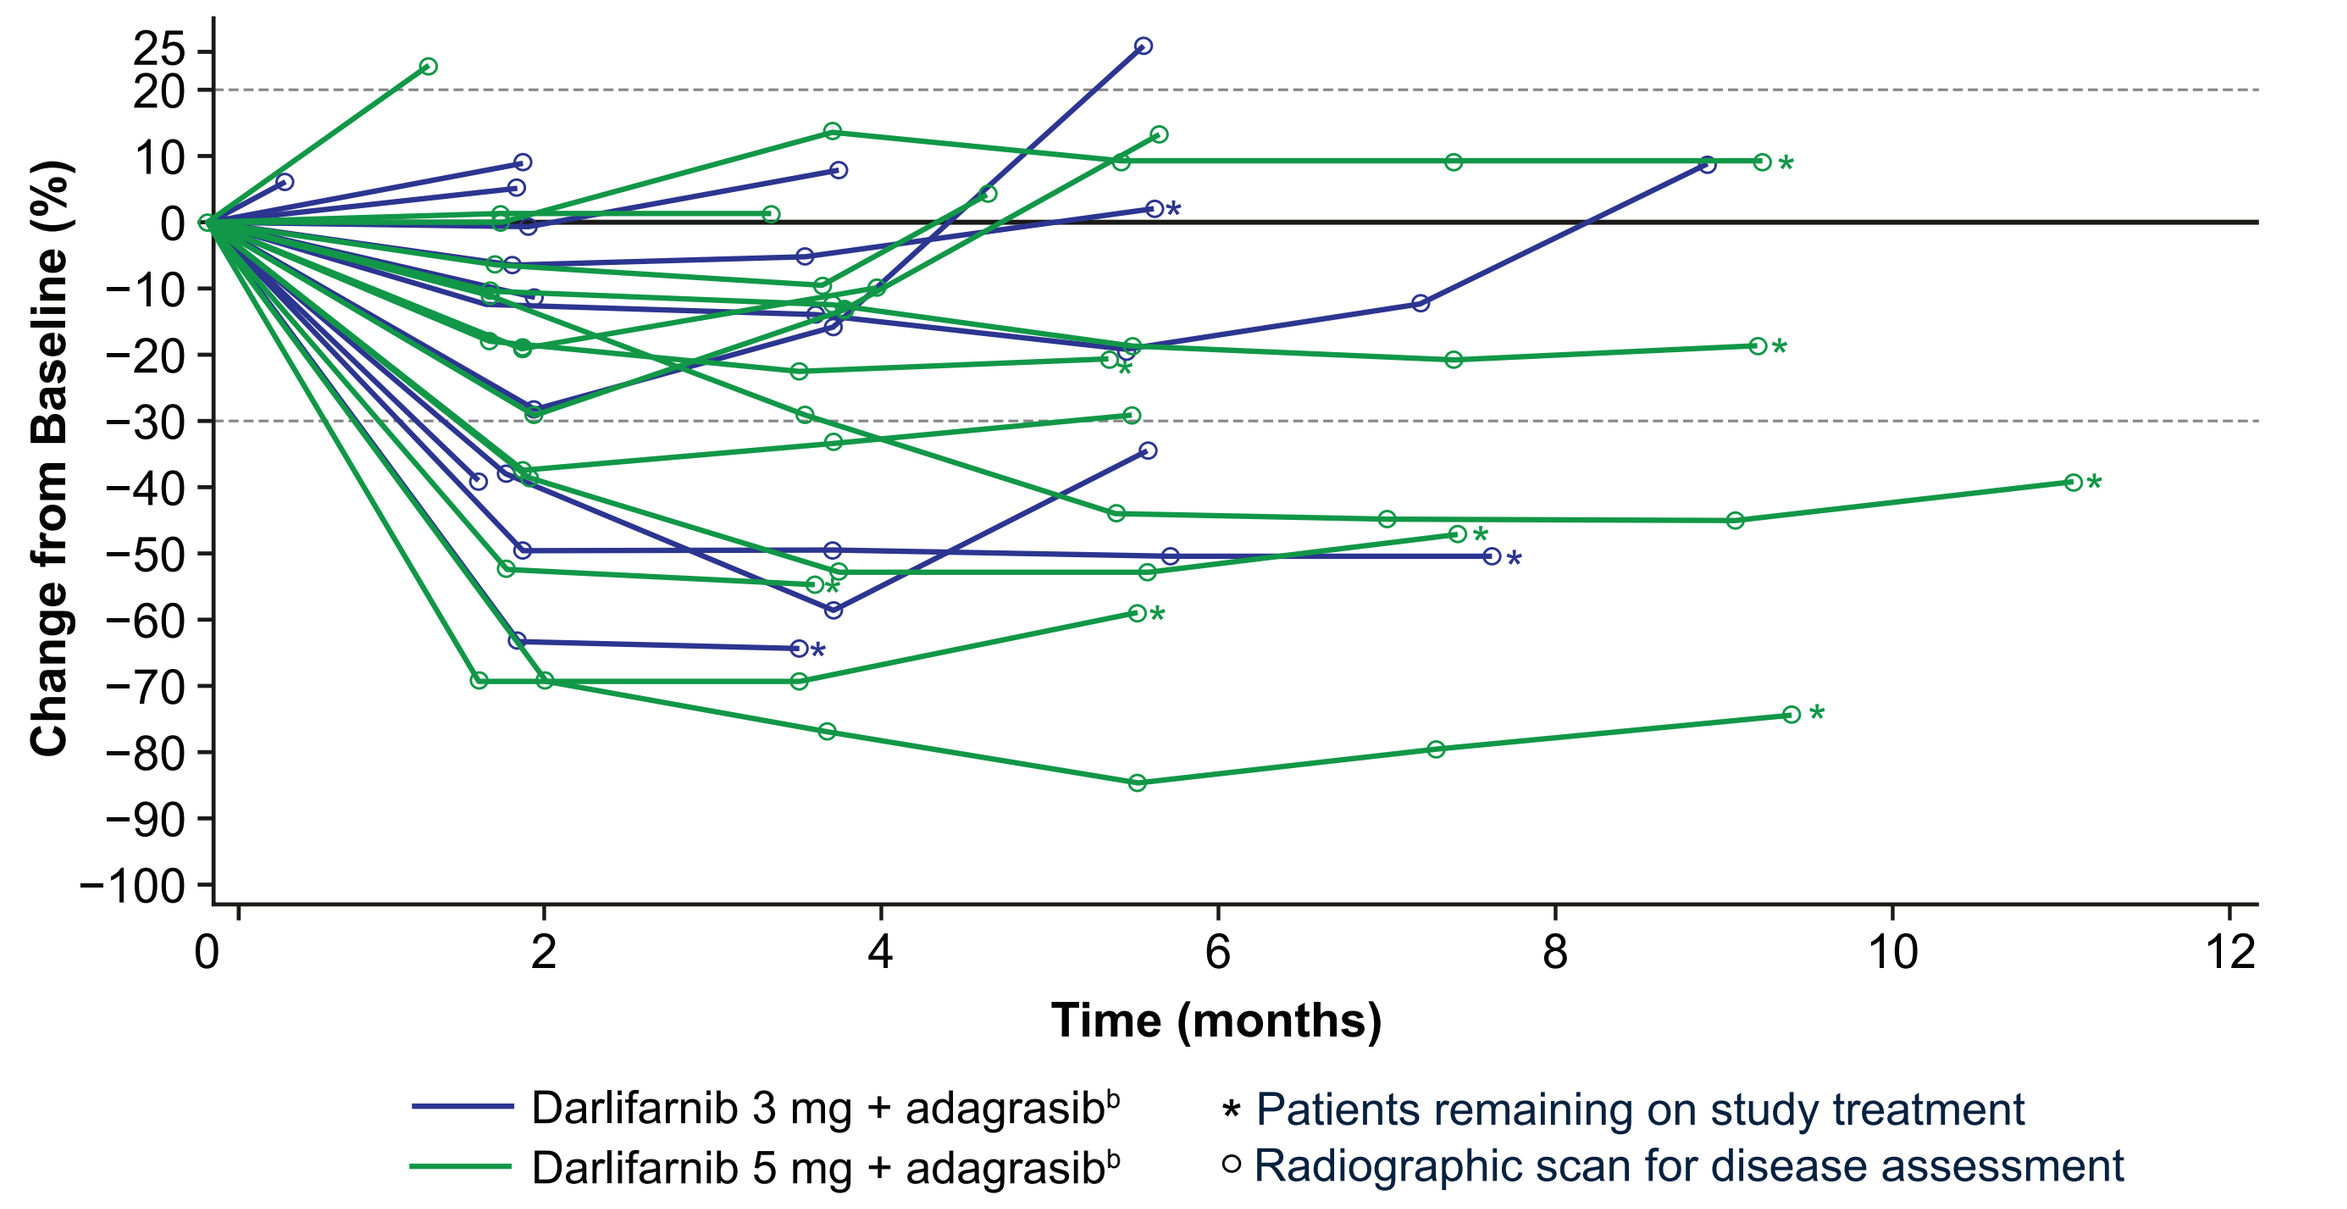

Change from Baseline (%)
Time (months)
Darlifarnib 3 mg + adagrasibb
Darlifarnib 5 mg + adagrasibb
* Patients remaining on study treatment
○ Radiographic scan for disease assessment

[a]Response-evaluable patients had ≥1 darlifarnib dose and post-baseline scan. [b]Adagrasib 400 mg BID. SoD, sum of diameters. Data cutoff: Mar 25, 2026.

- 51-year-old male, former smoker with stage IV *KRAS* G12C-mutated NSCLC (**Figure 6**)
 - Prior therapy:
 - Induction and consolidation: Cisplatin + gemcitabine, durvalumab
 - 1L: Docetaxel + nintedanib
 - 2L: Sotorasib (BOR: PR, discontinued due to PD)
 - 3L: Cisplatin + pemetrexed
 - 4L: Vinorelbine
 - Response to darlifarnib 5 mg + adagrasib:
 - BOR: PR (−84.6%)
 - **PR achieved at week 8 and maintained through week 40, the last disease assessment before data cutoff**

Key Considerations

- Deep, durable response, including resolution of 2/3 target lesions, in a heavily pretreated fifth-line *KRAS* G12C-mutated NSCLC patient
- Activity was observed despite prior progression on the KRAS G12Ci sotorasib, suggesting potential to overcome resistance mechanisms

Figure 6. Scans from a responder treated with darlifarnib 5 mg + adagrasib[a]

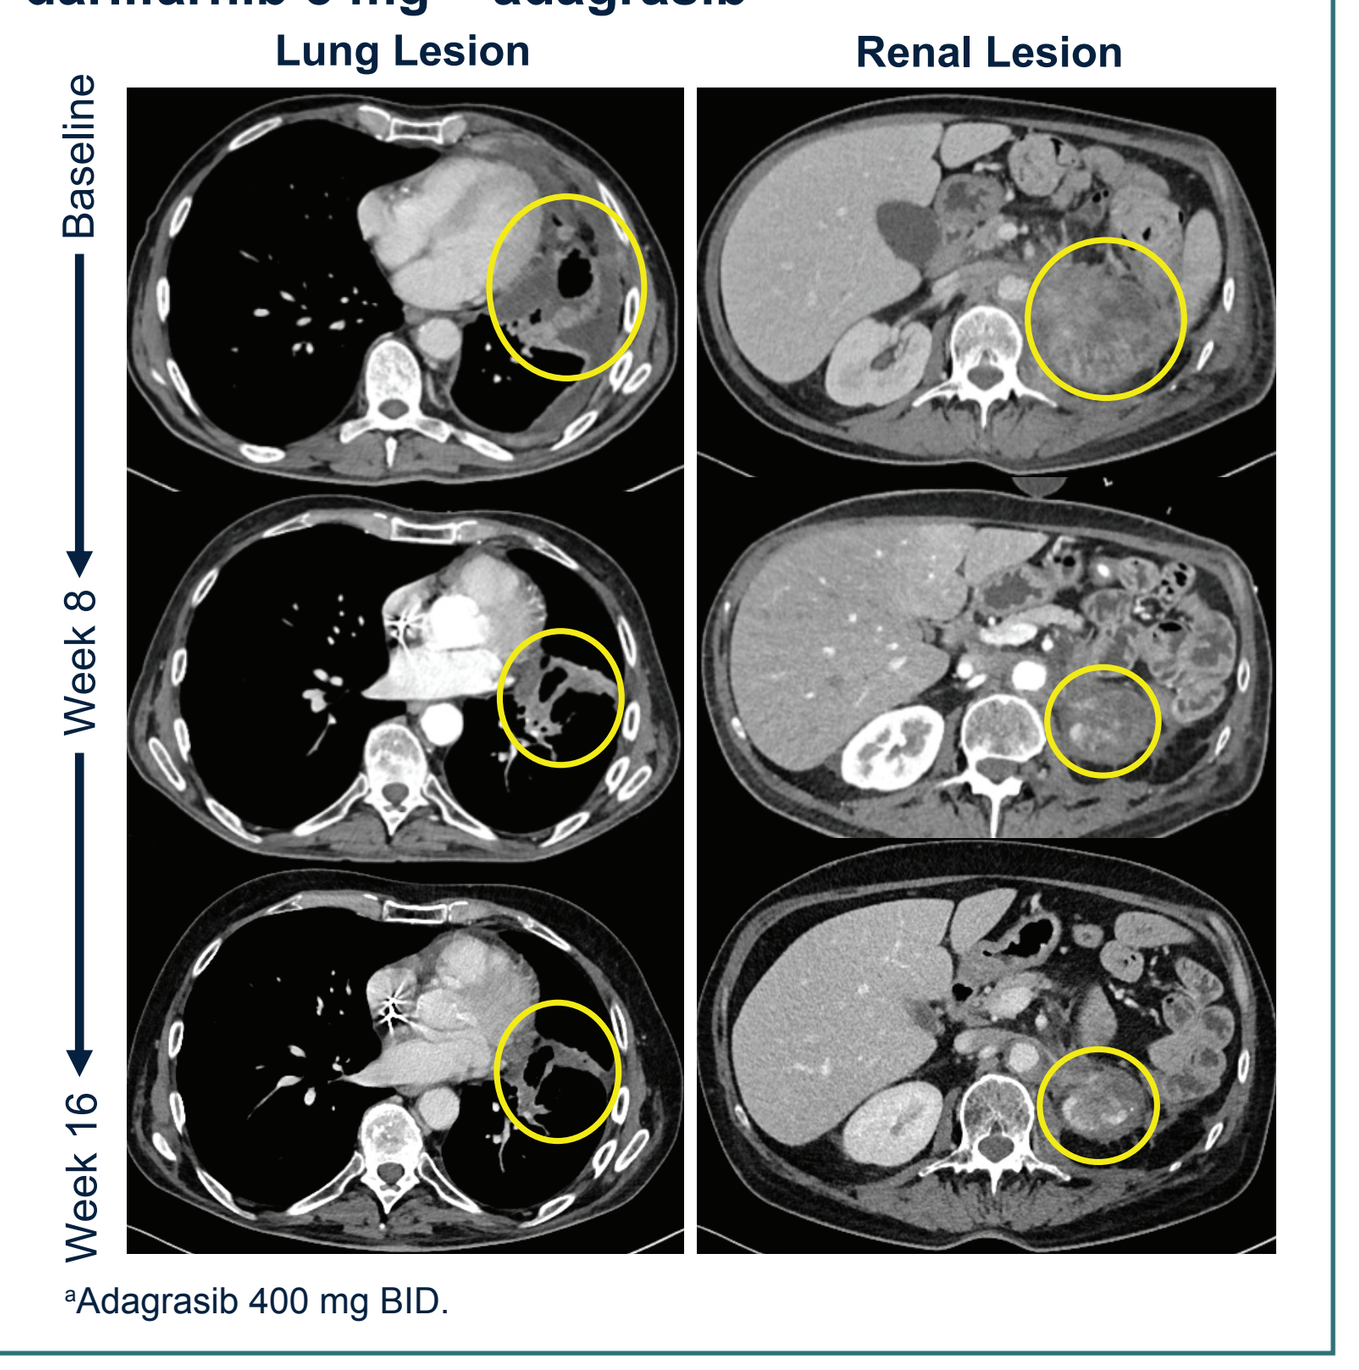

Lung Lesion
Renal Lesion
Baseline
Week 8
Week 16

[a]Adagrasib 400 mg BID.

- Response and early clinical benefit were reflected in *KRAS*m ctDNA changes by C1D21, preceding the first radiographic assessment (**Figure 7**)
 - Patients with PR showed larger *KRAS*m ctDNA reductions (>16-fold), while patients with SD or PD had smaller (≤4-fold) decreases or increases
 - *KRAS*m ctDNA decreased in 82% (9/11) of patients with detectable baseline levels, with reductions ranging from 2 to >100-fold
 - ctDNA became undetectable in 3 NSCLC patients, and dropped markedly in 2 PDAC patients (37- and 114-fold)

Figure 7. *KRAS*m ctDNA dynamics for FIT-001[a,b]

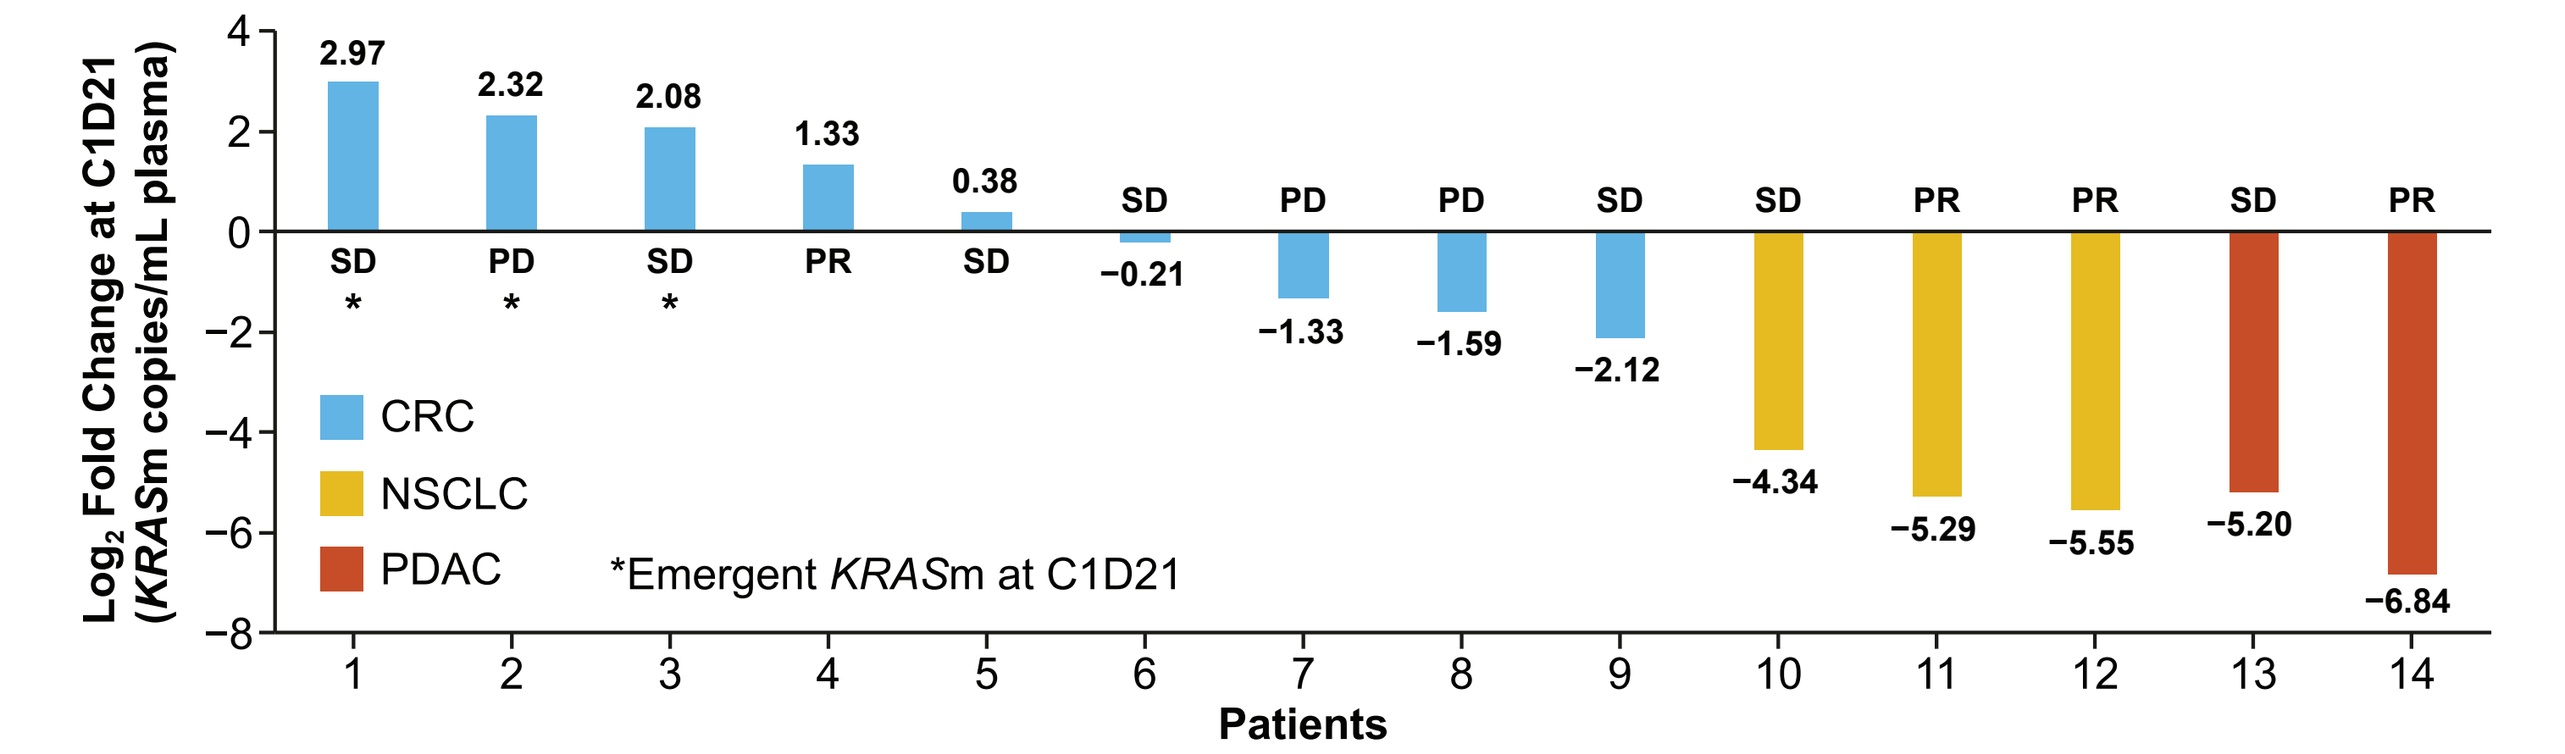

Log2 Fold Change at C1D21 (KRASm copies/mL plasma)
2.97 SD *
2.32 PD *
2.08 SD *
1.33 PR
0.38 SD
−0.21 SD
−1.33 PD
−1.59 PD
−2.12 SD
−4.34 SD
−5.29 PR
−5.55 PR
−5.20 SD
−6.84 PR
CRC
NSCLC
PDAC
*Emergent KRASm at C1D21
Patients

[a]Fold change in *KRAS*m ctDNA was measured by digital PCR in 14 patients with paired samples (pre-treatment and C1D21). [b]*KRAS*m ctDNA levels at C1D21 increased from 1.3- to 8-fold in 5/14 patients (35%); *KRAS*m ctDNA increased by 1.3, 2.5, and 8-fold at C1D21 in 3 CRC patients where *KRAS*m ctDNA was not detected prior to treatment. C1D21, Cycle 1, Day 21; CRC, colorectal cancer; ctDNA, circulating tumor DNA, *KRAS*m, *KRAS* mutated; NSCLC, non-small cell lung cancer; PCR, polymerase chain reaction; PD, progressive disease; PDAC, pancreatic ductal adenocarcinoma; PR, partial response; SD, stable disease.

CONCLUSIONS

- In the ongoing FIT-001 trial, darlifarnib 3 mg and 5 mg + adagrasib regimens were well tolerated, with a manageable safety profile supporting the clinical feasibility of this combination
- **Encouraging antitumor activity** was observed across *KRAS* G12C-mutated NSCLC, PDAC, and CRC, including among **heavily pretreated patients**:
 - In *KRAS* G12C-mutated patients from KRYSTAL-1, adagrasib monotherapy ORR was 19% in metastatic CRC (pretreated), 43% in NSCLC (pretreated), and 33% in PDAC[5–7]
 - ORR of darlifarnib + adagrasib: 29% in CRC (KRASi naive), 50% in NSCLC, and 67% in PDAC
- Notably, clinical activity was observed in patients with **prior KRASi exposure**, consistent with potential targeting of acquired resistance mechanisms
- Together, these data support further clinical evaluation of darlifarnib in combination with KRASi as a potential strategy to **improve depth and durability of response** in patients with *KRAS*-mutant cancers

References

1. Liu J, et al. *Cancer Gene Ther.* 2022 Jul;29(7):875-878. 2. Dilly J, et al. *Cancer Discov.* 2024;14(11):2135-2161. 3. Smith AE, et al. *Cancer Res.* 2023;83(19):3252-3263. 4. Patel HV, et al. *Cancer Res.* Published online February 9, 2026. 5. Bekaii-Saab TS, et al. *J Clin Oncol.* 2023;41(25):4097-4106. 6. Jänne PA, et al. *N Engl J Med.* 2022;387:120-131. 7. Yaeger R, et al. *N Engl J Med.* 2023;388:44-54. 8. U.S. Food and Drug Administration. KRAZATI® (adagrasib) tablets: prescribing information. Mirati Therapeutics, Inc; 2026.

Acknowledgments

The authors would like to thank the patients, their families, and their caregivers, as well as the FIT-001 study investigators and their study teams.

Disclosures

This study was sponsored by Kura Oncology, Inc. Adagrasib supply was provided by Bristol Myers Squibb. Medical writing support was provided by Shawn Jaramillo, PharmD, of Oxford PharmaGenesis Inc., Wilmington, DE, USA, and was funded by Kura Oncology, Inc.

SM: no disclosures. **MM** and **ML:** employment/stock ownership - Kura Oncology Inc. **VG:** consulting/advisory role - BMS, Pierre Fabre, Regeneron, Daiichi Sankyo; travel/accommodations/expenses - GenFleet, MSD, Pierre Fabre. Please refer to ASCO portal for remaining authors' disclosures.

Abbreviations

BID, twice daily; BOR, best overall response; C1D21, cycle 1, day 21; CBR, clinical benefit rate; CI, confidence interval; CRC, colorectal cancer; ctDNA, circulating tumor DNA; DLT, dose-limiting toxicity; FTase, farnesyl transferase; FTI, farnesyl transferase inhibitor; G12Ci, G12C inhibitor; KRAS, Kirsten rat sarcoma viral oncogene homolog; KRASi, KRAS inhibitor; *KRAS*m, *KRAS* mutated; mDOR, median duration of response; mTORC1, mechanistic target of rapamycin complex 1; mTORC2, mechanistic target of rapamycin complex 2; NSCLC, non-small cell lung cancer; ORR, objective response rate; PCR, polymerase chain reaction; PDAC, pancreatic ductal adenocarcinoma; PD, progressive disease; PR, partial response; PS, performance status; QD, once daily; RASi, RAS inhibitor; RHEB, Ras homolog enriched in brain; SAE, serious adverse event; SD, stable disease; SoD, sum of diameters; TEAE, treatment-emergent adverse event; TL, target lesion; TRAE, treatment-related adverse event.

Poster PDF can be accessed through this Quick Response (QR) code.
Copies of this poster obtained through QR Code are for personal use only and may not be reproduced without permission from ASCO® or the author.

Additional information on the FIT-001 study can be accessed here

***Poster presented at:** the American Society of Clinical Oncology (ASCO) Annual Meeting; May 29–June 2, 2026; Chicago, IL, USA*