Pre-clinical Activity of Tipifarnib in Cutaneous T-cell Lymphoma

Kohnken, R.[1], McNeil, B.[2], Wen, J.[2], McConnell, K.[2], Burrows, F.[4], Porcu, P, and A. Mishra[2,3]
[1]Department of Veterinary Biosciences; [2]Comprehensive Cancer Center; [3]Department of Internal Medicine, Division of Dermatology, Ohio State University, Columbus OH USA, [4]Kura Oncology, San Diego CA USA

The James
THE OHIO STATE UNIVERSITY
COMPREHENSIVE CANCER CENTER

Introduction

Cutaneous T-cell lymphoma (CTCL) is a rare form of non-Hodgkin lymphoma characterized by malignant infiltration of skin-homing CD4+ T-cells, manifesting clinically as patches and plaques on the skin. The disease will often progress from an indolent skin-limited form, Mycosis Fungoides (MF), to an aggressive leukemic variant, Sezary Syndrome (SS).

As there are few effective therapies available for advanced-stage CTCL, we sought to explore potential novel therapeutic modalities. Farnesyl transferase inhibitors (FTIs) are a relatively new class of anticancer drugs, the exact mechanism of which is unknown. Tipifarnib is one such FTI, which has demonstrated clinical activity as a single agent in patients with relapsed and refractory lymphomas. Despite encouraging clinical and preclinical data, the precise molecular mechanisms of tipifarnib's activity in lymphoma remain to be fully elucidated.

Methods and Results

We utilized CTCL patient-derived cell lines to evaluate the potential efficacy of tipifarnib *in vitro*. Our data on patient-derived cell lines show that half maximal effective concentration (EC_{50}) ranged from 7.5nM for HuT78 to 50nM for SeAx (**Figure 1A**). This cytotoxicity was correlated to increases in apoptosis in these lines (**Figure 1B**). Importantly, there was no cytotoxic effect on normal donor CD4+ T-cells *in vitro* (data not shown). Based on these promising findings, we evaluated tipifarnib *in vivo* using a recently described interleukin-15 (IL-15) transgenic mouse model of CTCL (Mishra A, et al. Can Discov 2016). Following 2 weeks of dosing in 5 week old mice, we found a significant decrease in gross lesion severity in tipifarnib-treated mice (0.5±1.22 out of a possible score of 5) compared to vehicle-treated mice (3.33±0.57, p=0.0076) (**Figure 2A, 2B**). Histologically, IL-15 transgenic mice develop characteristic malignant CD4+ T-cell infiltrates in the dermis and epidermis (**Figure 3A**). Tipifarnib-treated mice, however, have minimal lesions in the skin (**Figure 3A**, bottom). These dramatic changes are reflected in a significantly decreased histologic severity score in tipifarnib-treated mice (3.8±0.75 out of a possible score of 7 (compared to vehicle-treated mice (7±0, p=0.0002) (**Figure 3B**).

Results

Figure 1. Tipifarnib, a farnesyl transferase inhibitor, decreases cell viability and increases apoptosis in CTCL lines *in vitro*

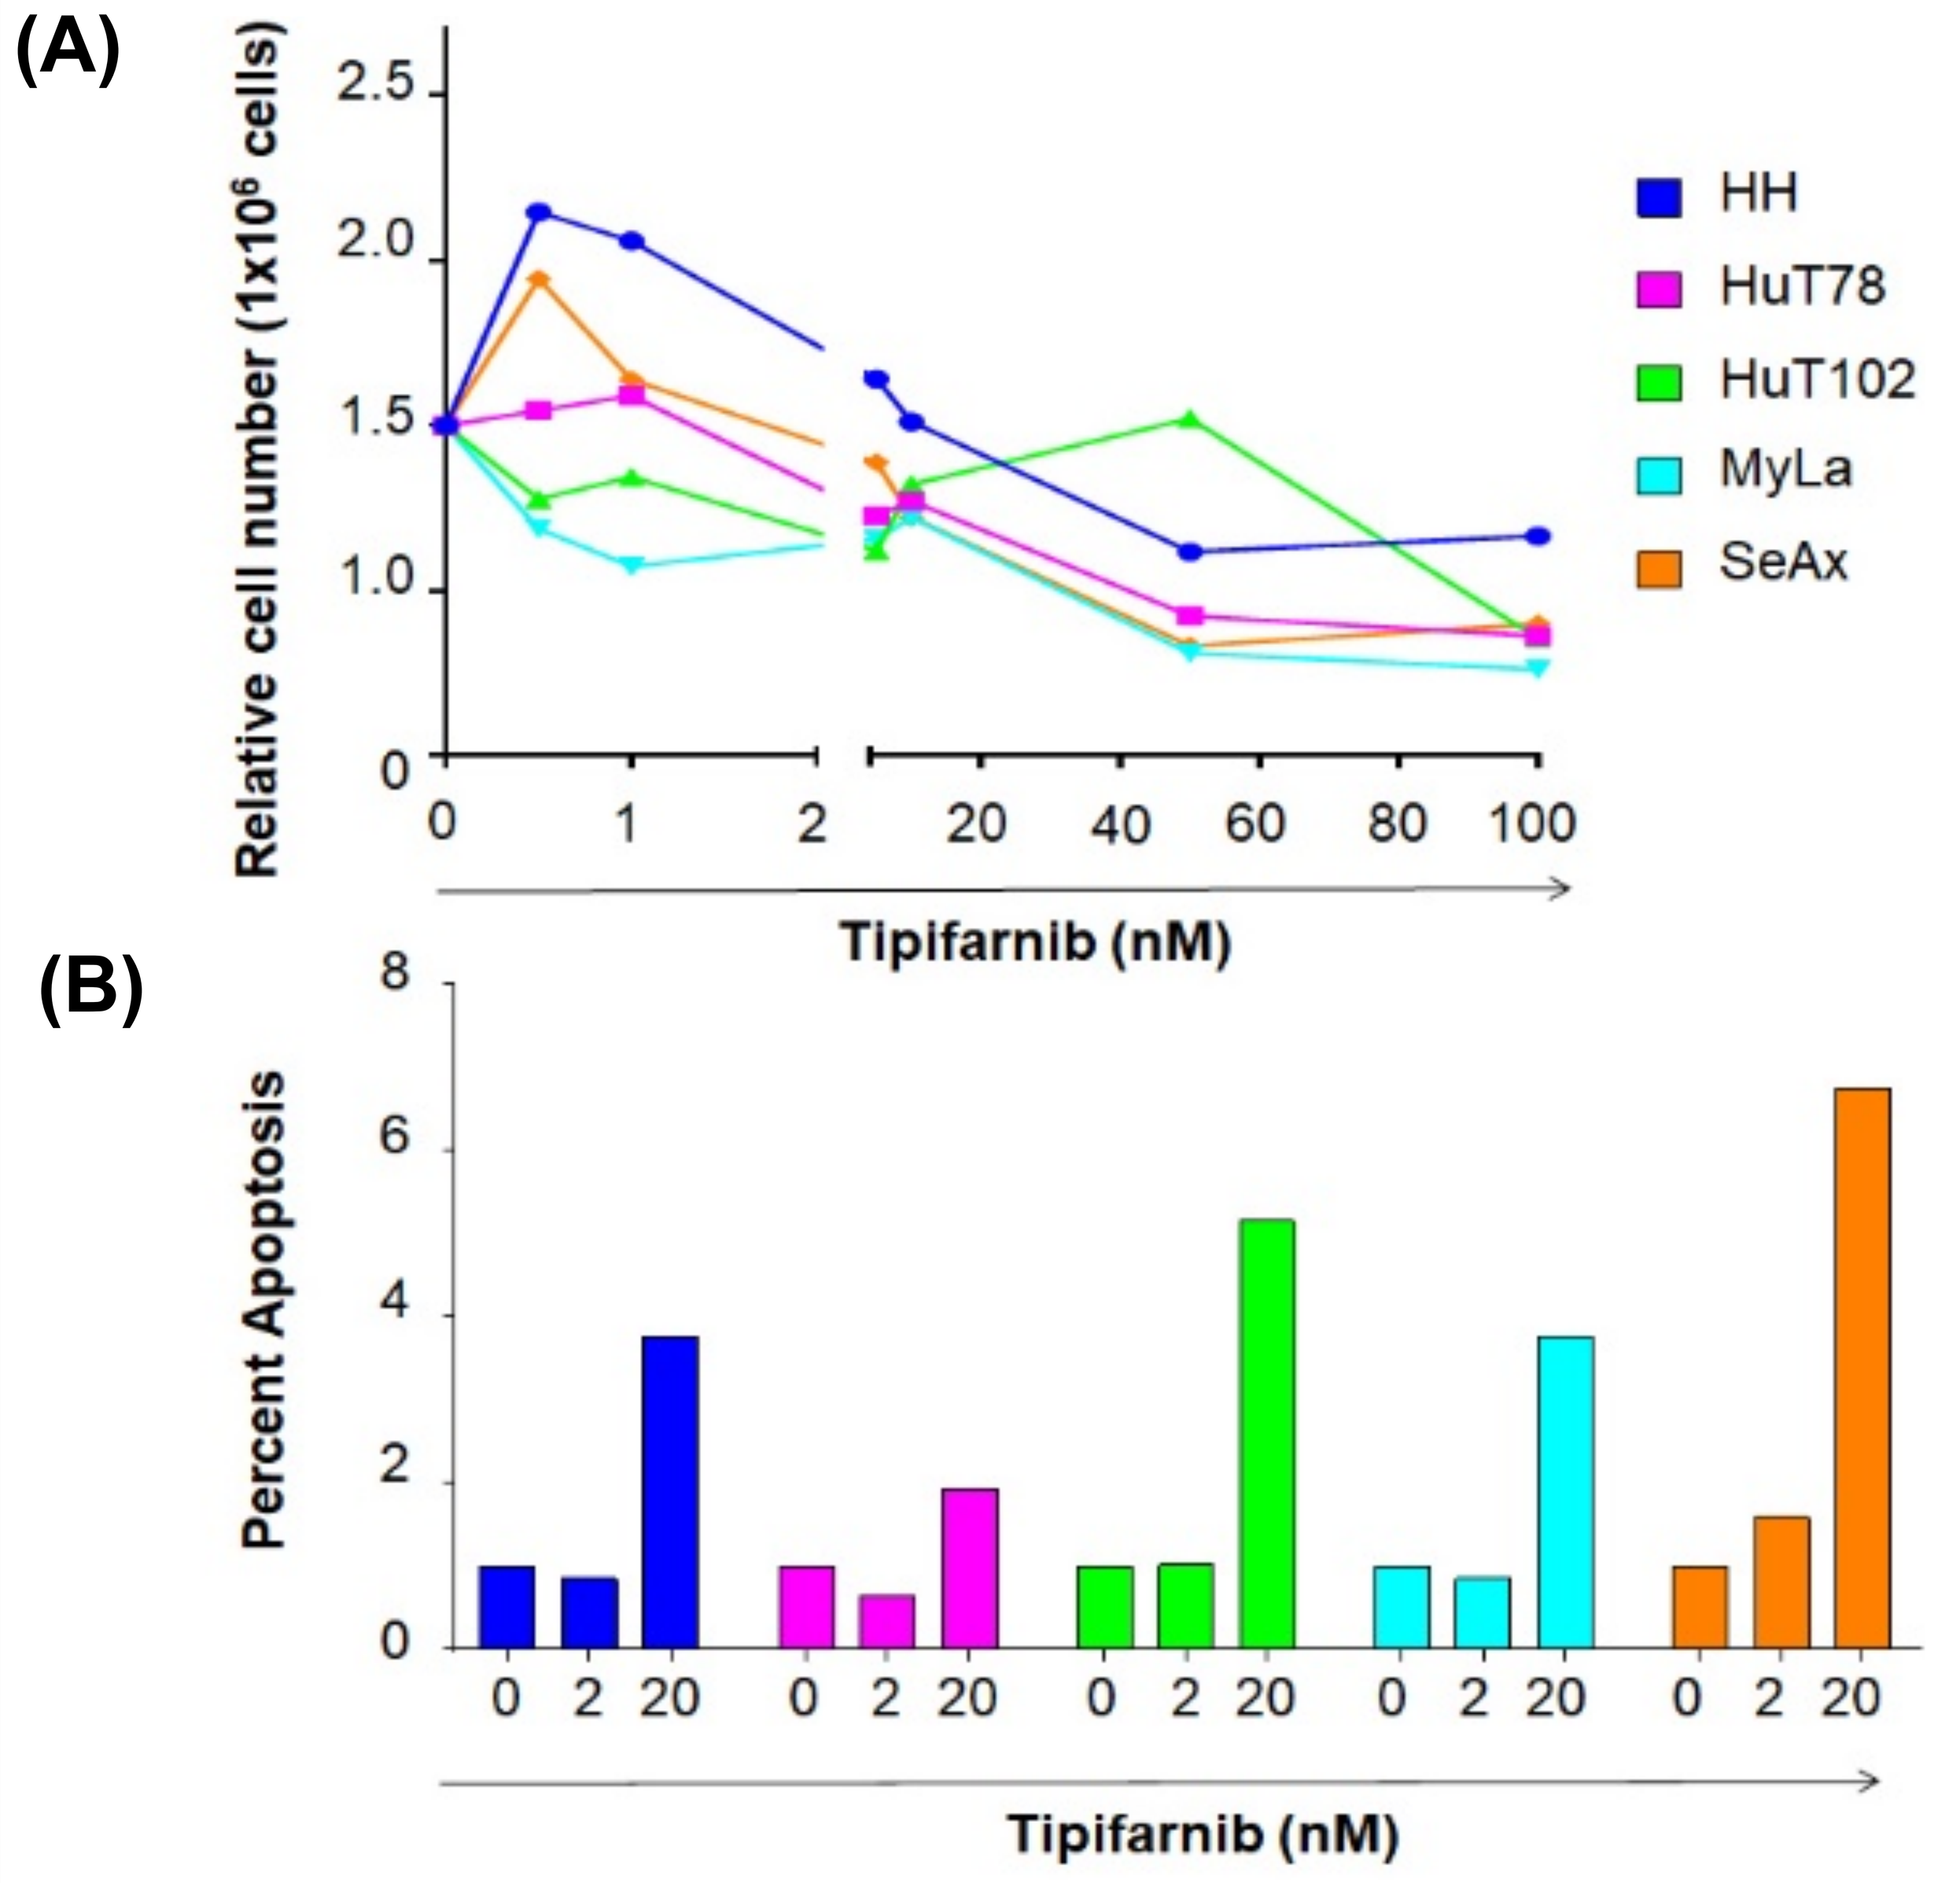
(A)
Relative cell number (1x10⁶ cells)
2.5
2.0
1.5
1.0
0
0 1 2 20 40 60 80 100
Tipifarnib (nM)
HH
HuT78
HuT102
MyLa
SeAx
(B)
Percent Apoptosis
8
6
4
2
0
0 2 20 0 2 20 0 2 20 0 2 20 0 2 20
Tipifarnib (nM)

(A) Dose escalation studies of tipifarninb *in vitro,* demonstrate decreased cell viability in CTCL parent lines.

(B) HH, HuT78. HuT102, MyLa and SeAx CTCL lines demonstrate an increase in apoptosis at 20nM tipifarnib.

Figure 2. Tipifarnib treatment *in vivo* decreases CTCL disease severity in IL-15 transgenic mice

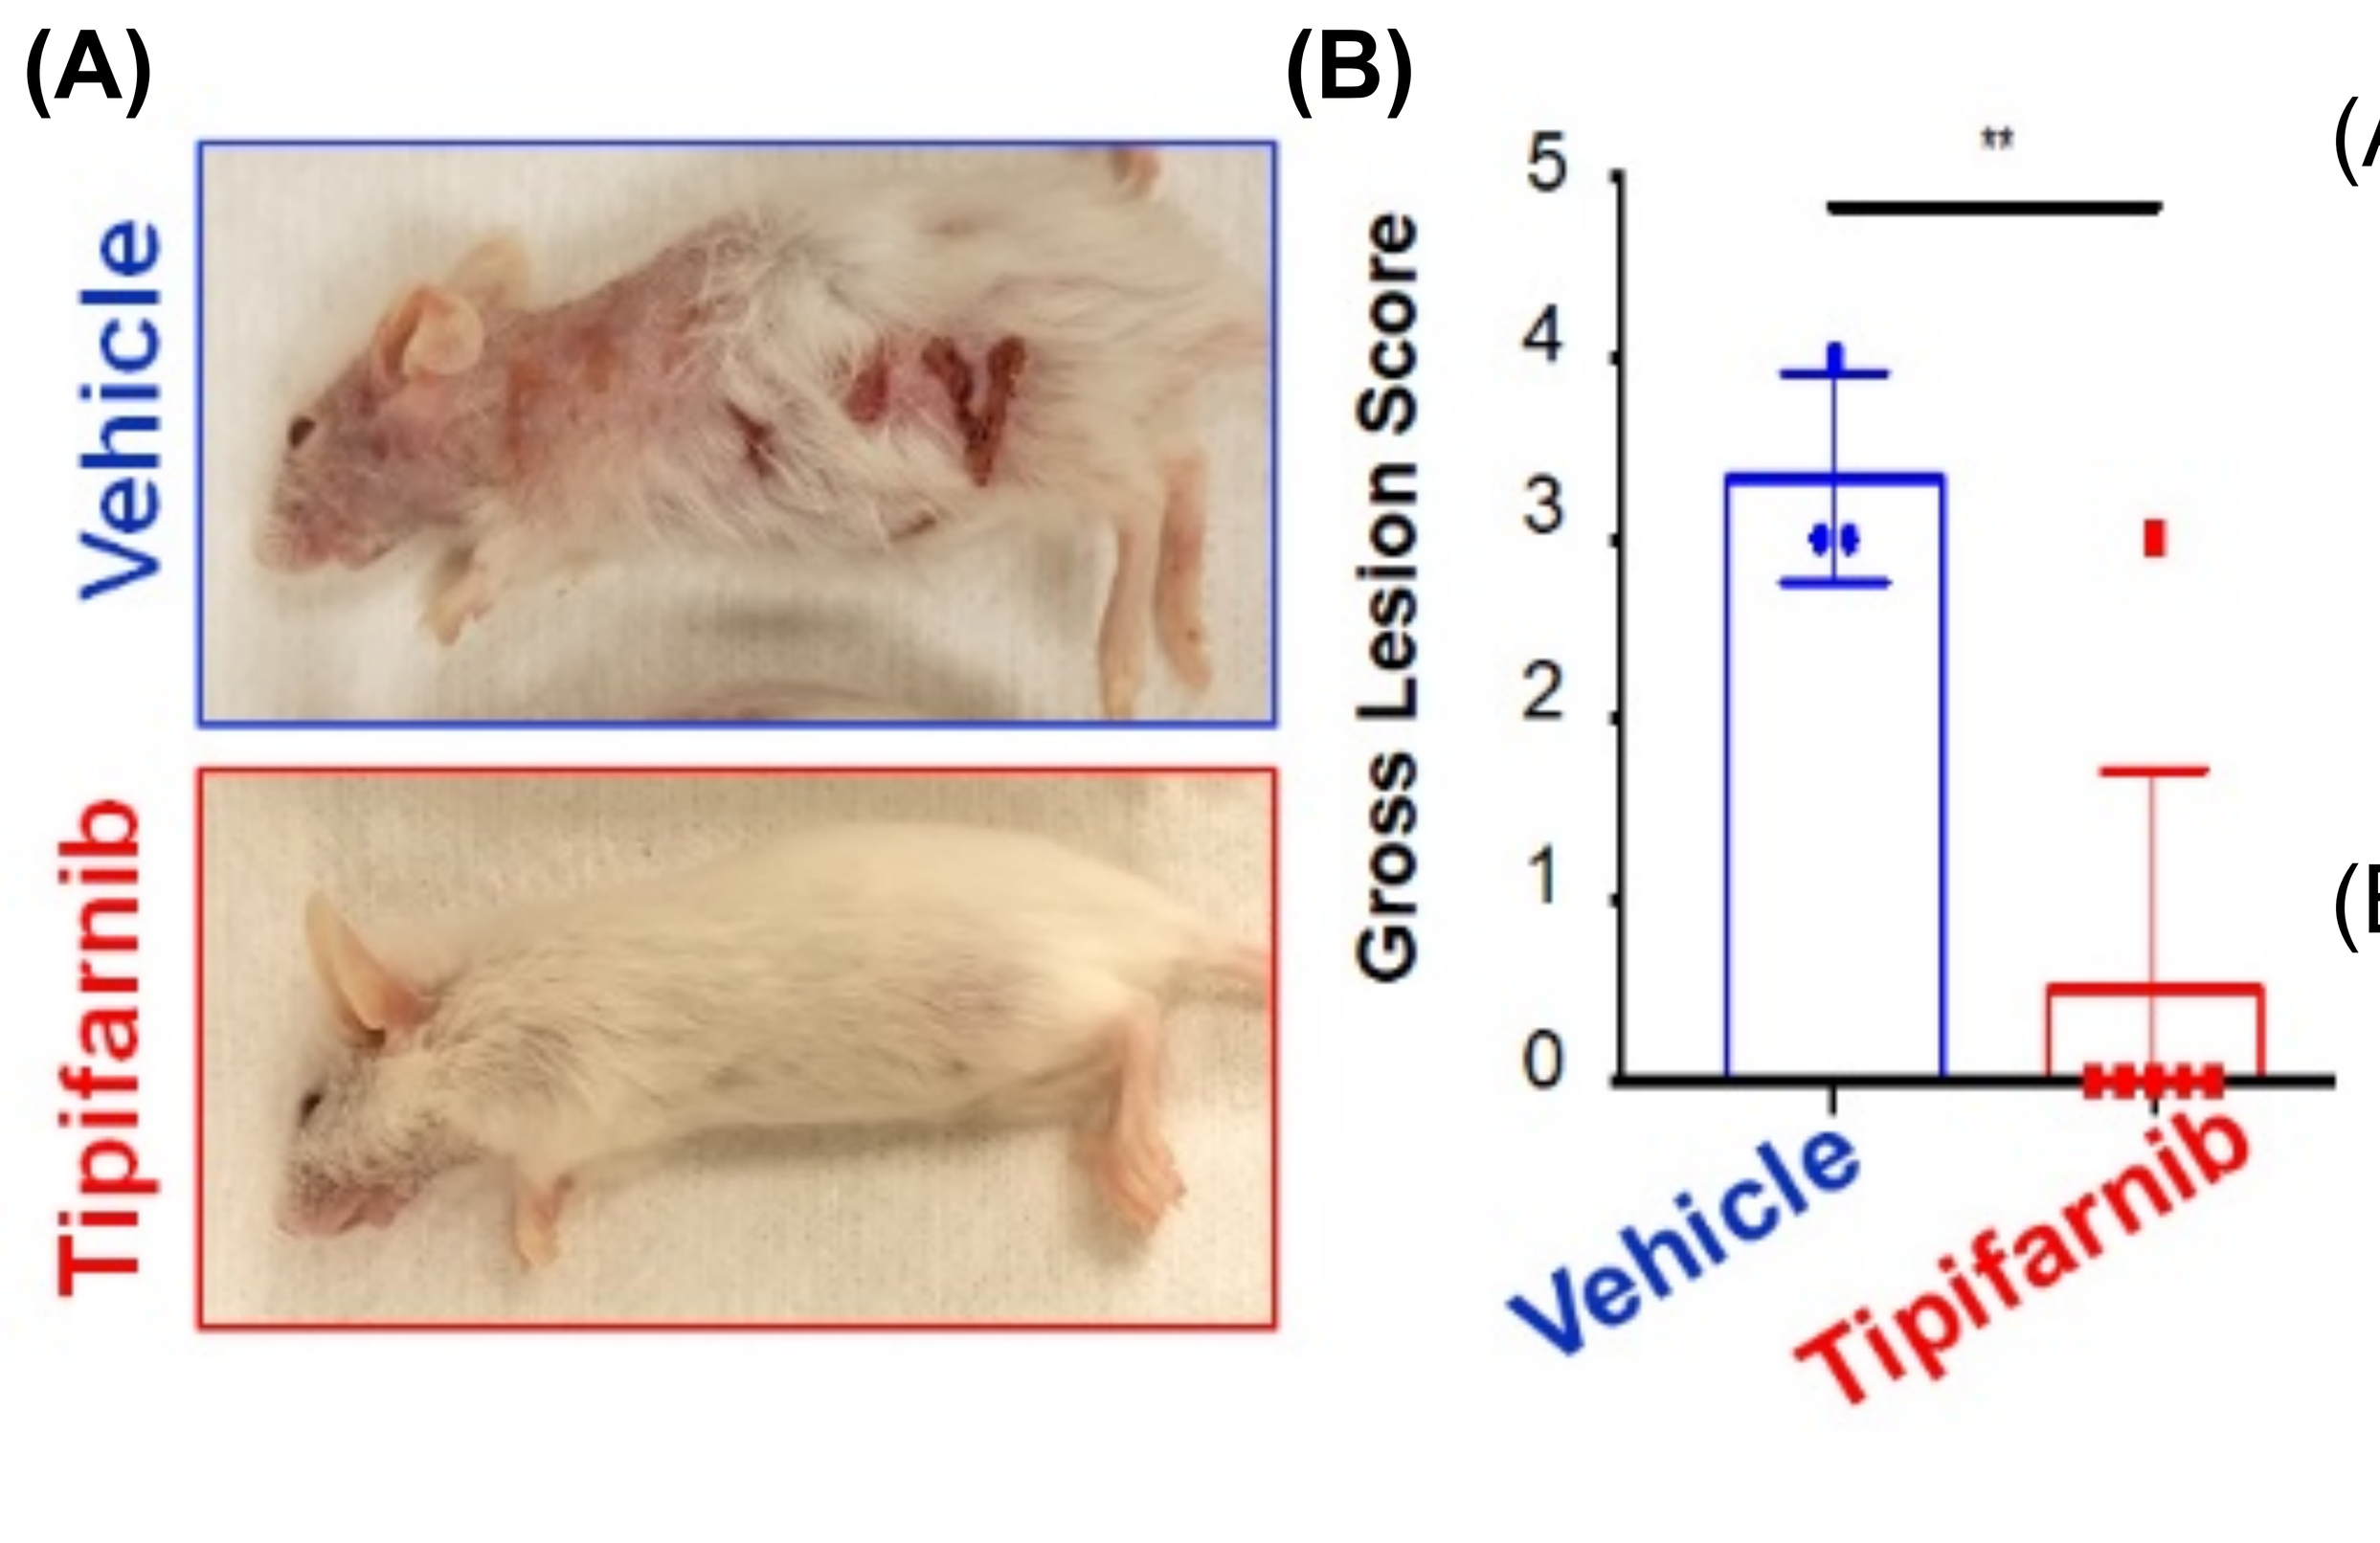
(A)
Vehicle
Tipifarnib
(B)
Gross Lesion Score
5
4
3
2
1
0
**
Vehicle
Tipifarnib

(A) IL-15 transgenic mice treated with tipifarnib have less extensive cutaneous disease.

(B) Gross lesion severity in tipifarnib-treated mice, as compared to vehicle-treated mice.

Results

Figure 3. Tipifarnib treatment *in vivo* decreases histologic lesion severity in IL-15 transgenic mice

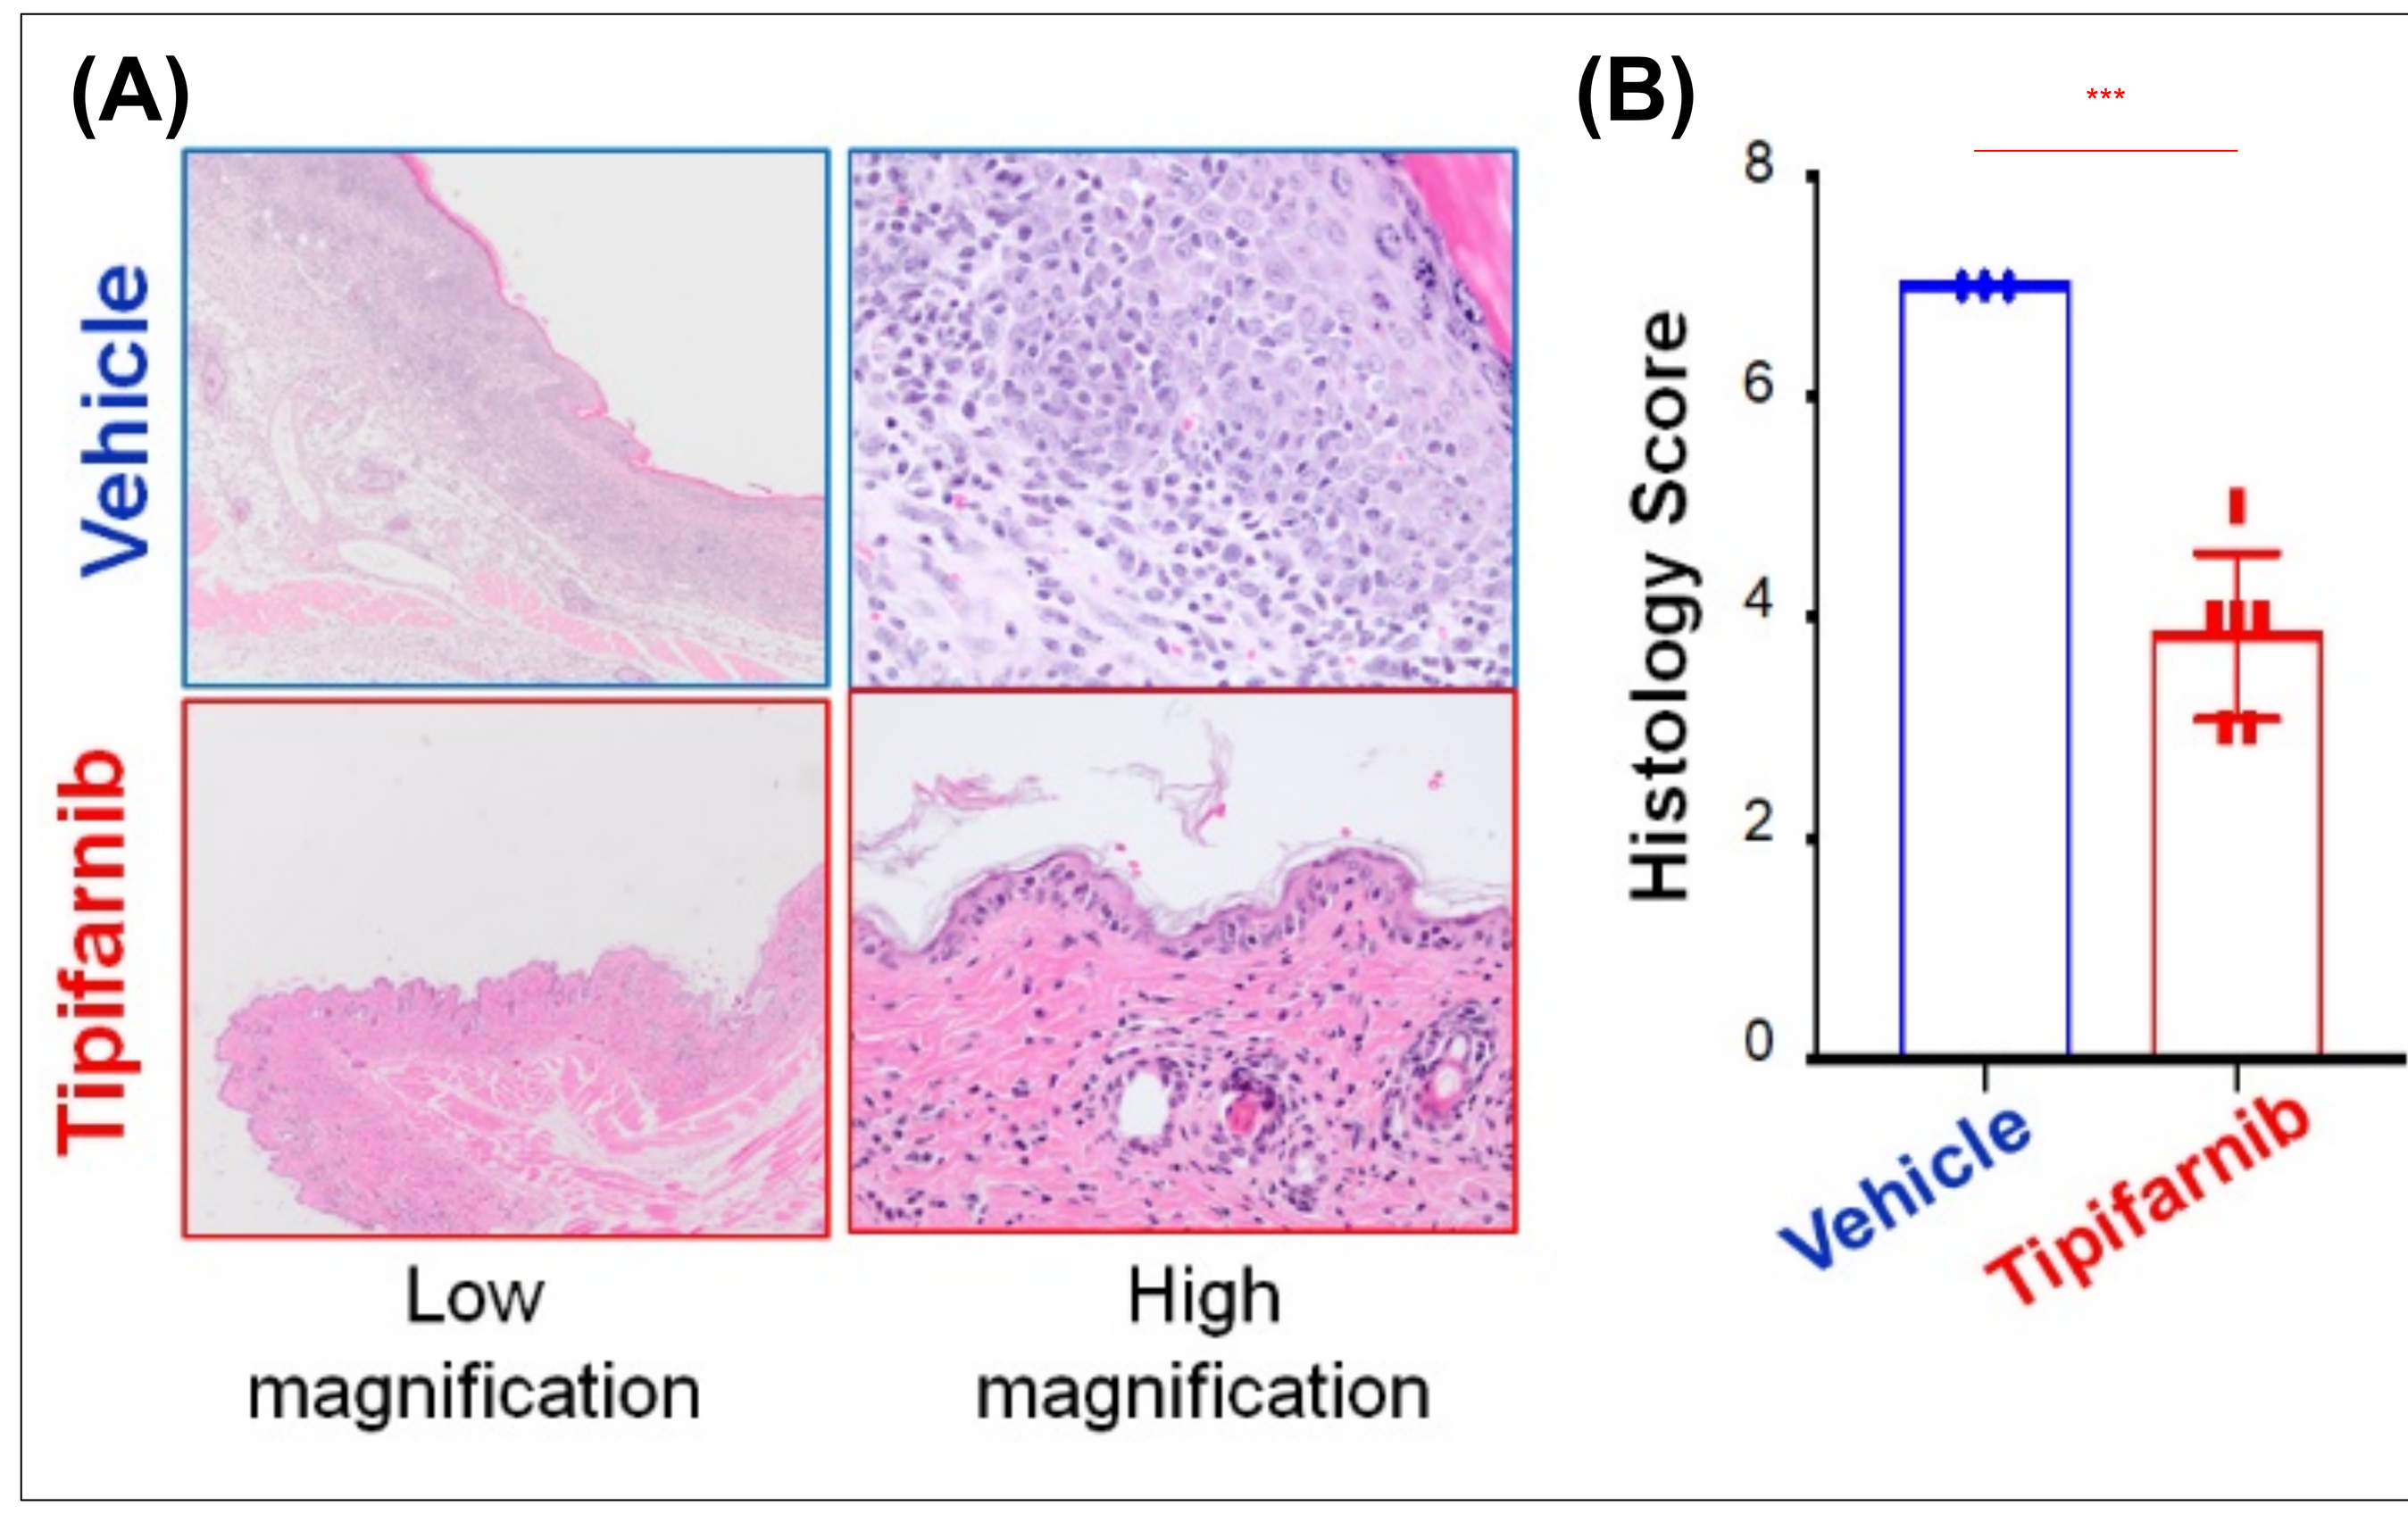
(A)
Vehicle
Tipifarnib
Low magnification
High magnification
(B)
Histology Score
8
6
4
2
0

Vehicle
Tipifarnib

(A) Tipifarnib-treated mice have minimal lesions of CTCL compared to vehicle-treated mice.

(B) Histologic lesion severity is significantly reduced in tipifarnib-treated mice.

Conclusions

Our findings demonstrate that tipifarnib may prove beneficial for the treatment of CTCL as shown in efficacy observed in patient derived cell lines and mouse models of cutaneous T-cell lymphoma. Further studies to elucidate the mechanism of antitumor activity of farnesyl transferase inhibition in malignant T-cells are in progress.

Authors declare no conflicts of interest.

De-identified patient materials provided by OSUCC Leukemia Tissue Bank Shared Resource (P30CA016058).

Funding for this project by Kura Oncology to AM.

References:

1. Mishra A, La Perle K, Kwiatkowski S, Sullivan L, Sams GH, Johns J, Curphey DP, Wen J, McConnell K, Qi J, Wong H, Russo G, Zhang J, Marcucci G, Bradner JE, Porcu P, Caligiuri MA. Mechanism, consequences and therapeutic targeting of abnormal IL-15 signaling in cutaneous T-cell lymphoma. ***Cancer Discovery 2016*** Sep;6(9):986-1005.
2. Witzig T, Tang H, Micallef I, Ansell S, Link B, Inwards D, Porrat L, Johnston P, Colgan J, Markovic S, Nowakowski G, Thompson C, Allner C, Maurer M, Gupta M, Weiner G, Hohl R, Kurtin P, Ding H, Loegering D, Schneider P, Peterson K, Habermann T, Kaufmann S. Multi-institutional phase 2 study of the farnesyl transferase inhibitor tipifarnib (R115777) in patients with relapsed and refractory lymphomas. ***Blood 2011*** Nov;118(18): 4882-4889.
3. Sparano J, Moulder S, Kazi A, Coppola D, Negassa A, Vahdat L, Li T, Pellegrino C, Fineberg S, Munster P, Malafa M, Lee D, Hoschander S, Hopkins U, Hershman D, Wright J, Kleer C, Merajver S, Sebti S. Phase II trial of the farnesyl transferase inhibitor tipifarnib plus neoadjuvant doxorubicin-cyclophosphamide in patients with clinical stage IIB-IIIC breast cancer. ***Clinical Cancer Research 2009*** Apr; 15(8): 2942-2948.