Tipifarnib is Highly Active in Combination with Alpelisib in Models of HRAS/PI3K-dysregulated HNSCC

Abstract Number: 159

Shivani Malik[1], Stacia Chan[1], Mara Gilardi[2], Zhiyong Wang[2], J. Silvio Gutkind[2] and Francis Burrows[1]
[1]Kura Oncology, Inc., San Diego, CA, USA, [2]University of California, San Diego

BACKGROUND AND RATIONALE

Squamous cell carcinoma of the head and neck (HNSCC) is the seventh most common cancer worldwide, but few targeted therapy options exist for advanced disease. The farnesyltransferase inhibitor tipifarnib is active as a single agent in HRAS-mutant HNSCC, which represents 4-8% of patients. Several independent studies cluster HRAS mutant HNSCCs as part of a larger subset[1-3] and HRAS is overexpressed in around a quarter of HNSCC in TCGA[4], raising the possibility that wild-type (WT) HRAS could support malignant progression in a significant subset of HNSCC patients. PIK3CA is the most commonly dysregulated oncogene in HNSCC and the HRAS-MAPK and PI3K pathways are interdependent in SCCs. HRAS requires PI3Kα to transform squamous epithelial cells[5] and helicase domain PIK3CA mutants must bind RAS to cause cancer[6]. HRAS and PIK3CA gene expression are negatively correlated in the TCGA cohort[4] (Spearman -0.50, p = 2.5e-32). Overexpression of mutant or WT HRAS drives resistance to PI3K inhibition in PIK3CA-mutant HNSCC cells[7]. Alpelisib (Piqray®) is a potent and selective inhibitor of PI3Kα that was recently approved for treatment of PIK3CA-mutant, HR-positive breast cancer in combination with fulvestrant and alpelisib is in clinical development in HNSCC.

HRAS Mutations Define a Unique Molecular Subset of HNSCC

HRAS mutant HNSCC (~4-8% at initial diagnosis) is characterized by frequent Caspase 8 mutations and an uncharacteristically low rate of TP53 mutation (below)

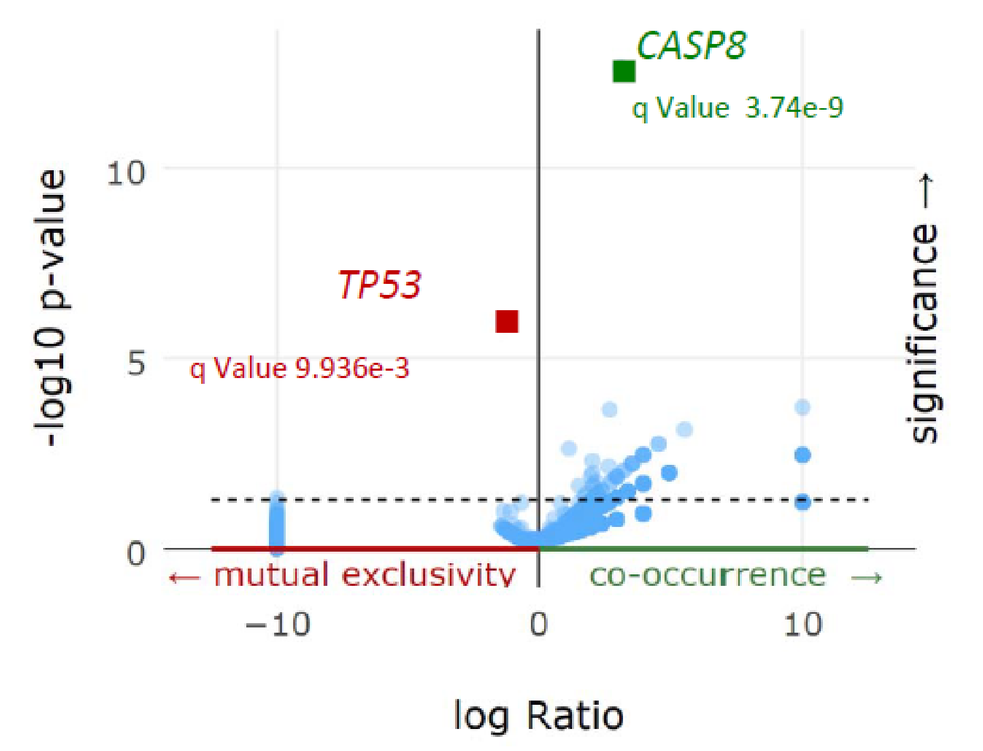
CASP8
q Value 3.74e-9
TP53
q Value 9.936e-3
-log10 p-value
significance
← mutual exclusivity
co-occurrence →
log Ratio

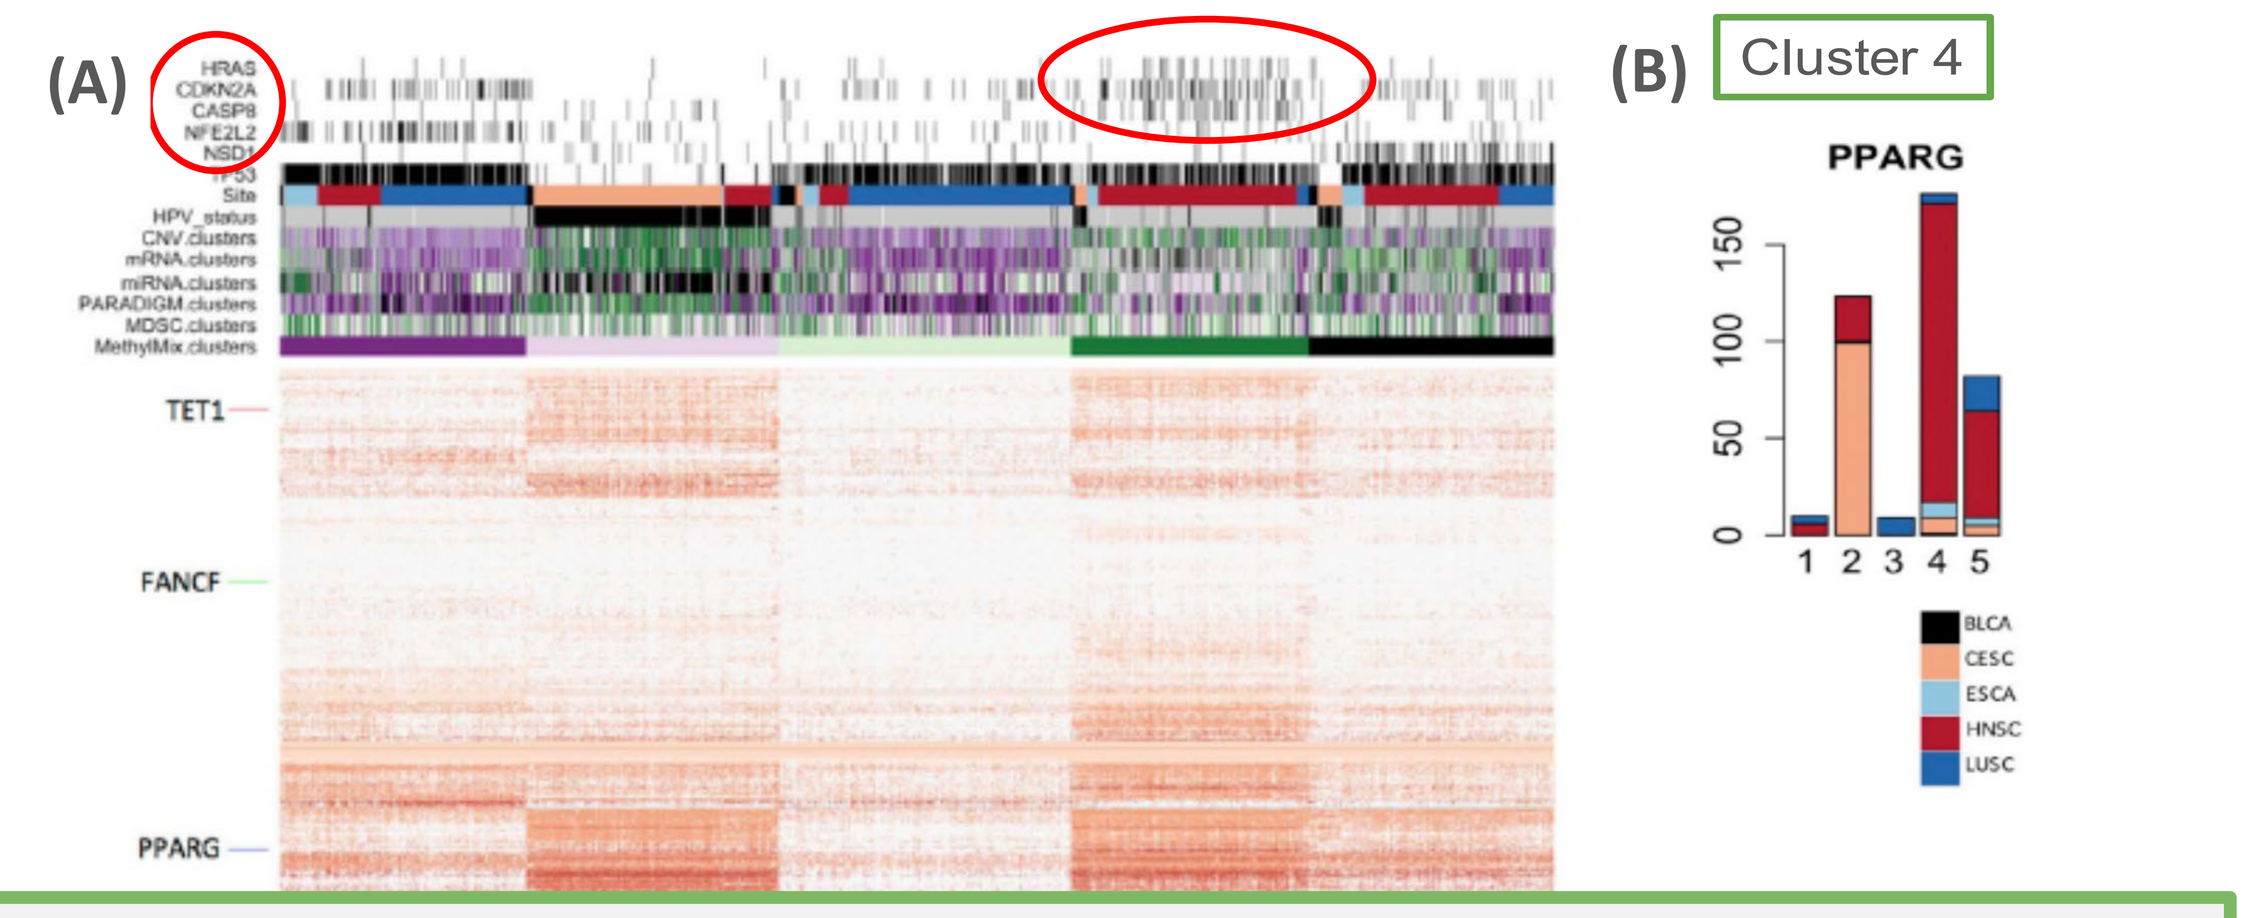
(A)
HRAS
CDKN2A
CASP8
NFE2L2
NSD1
TET1
FANCF
PPARG
(B)
Cluster 4
PPARG
BLCA
CESC
ESCA
HNSC
LUSC

- Campbell *et al* performed unbiased clustering of TCGA SCCs by methylation status[3]
- The HRAS/CASP8 subset is in Methylation Cluster 4 **(A)**
- Numerous HRAS-WT tumors are also found in Cluster 4
- Cluster 4 is defined by methylation silencing of PPARγ in HNSCC **(B)**
- PPAR silencing may allow SCC cells to tolerate higher levels of HRAS activity[8]

Tipifarnib is Highly Active in HRAS-mutant HNSCC Models

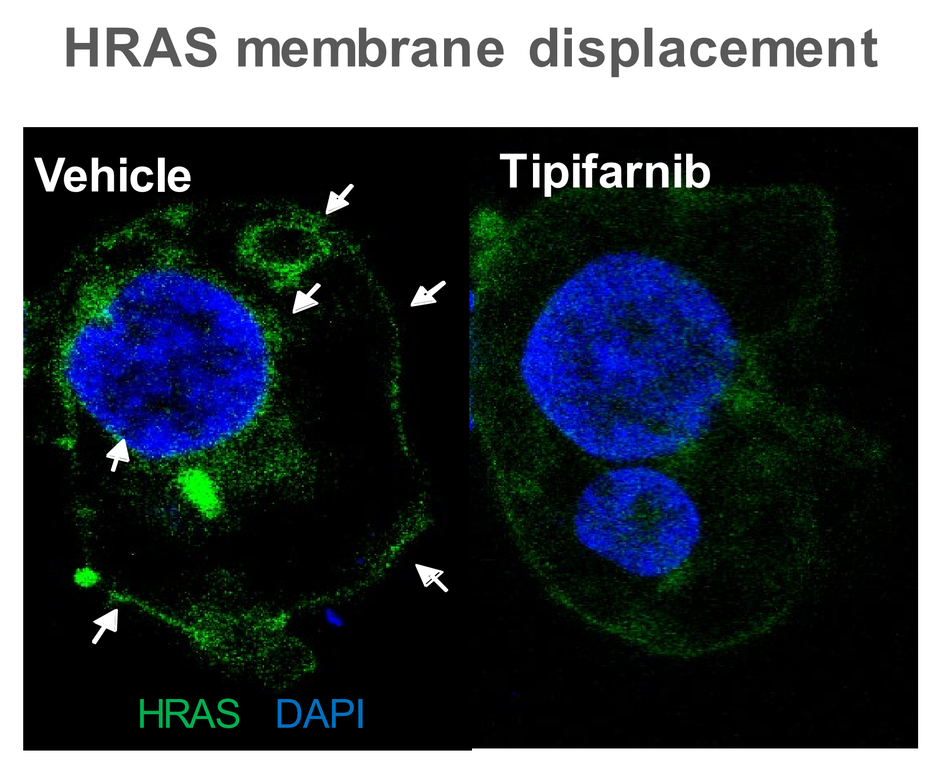
HRAS membrane displacement
Vehicle
Tipifarnib
HRAS
DAPI

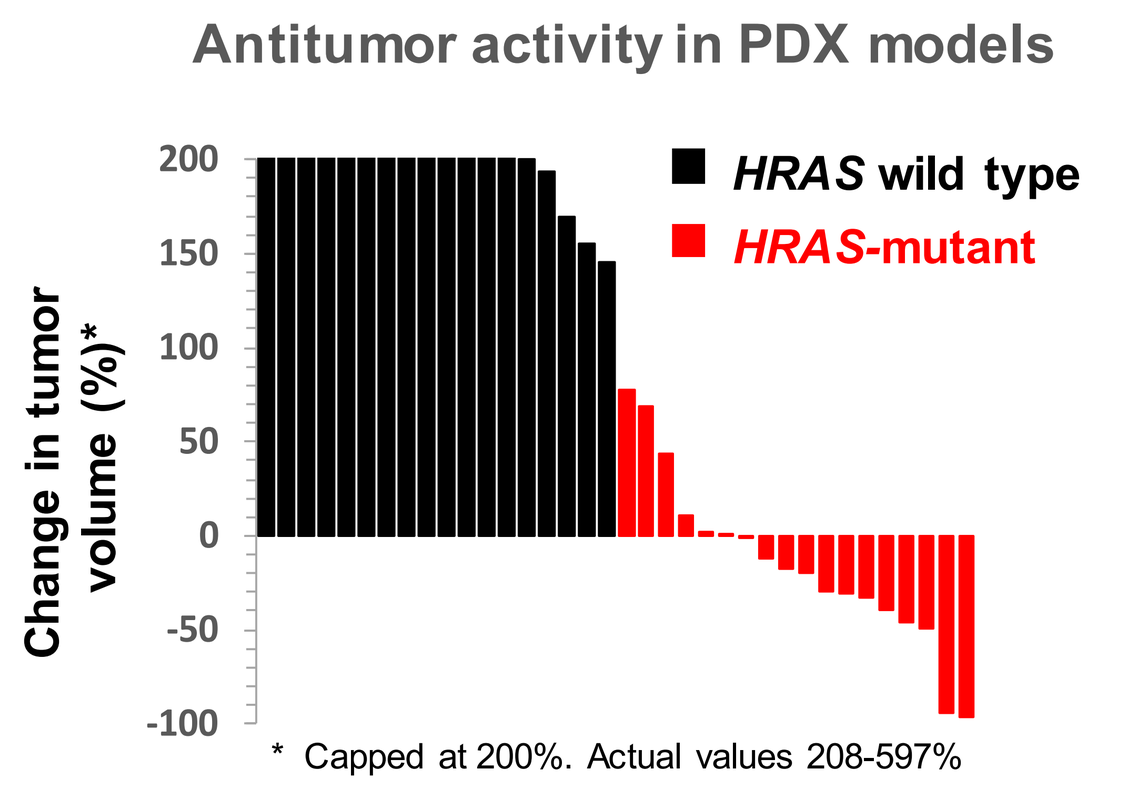
Antitumor activity in PDX models
HRAS wild type
HRAS-mutant
Change in tumor volume (%)*
* Capped at 200%. Actual values 208-597%

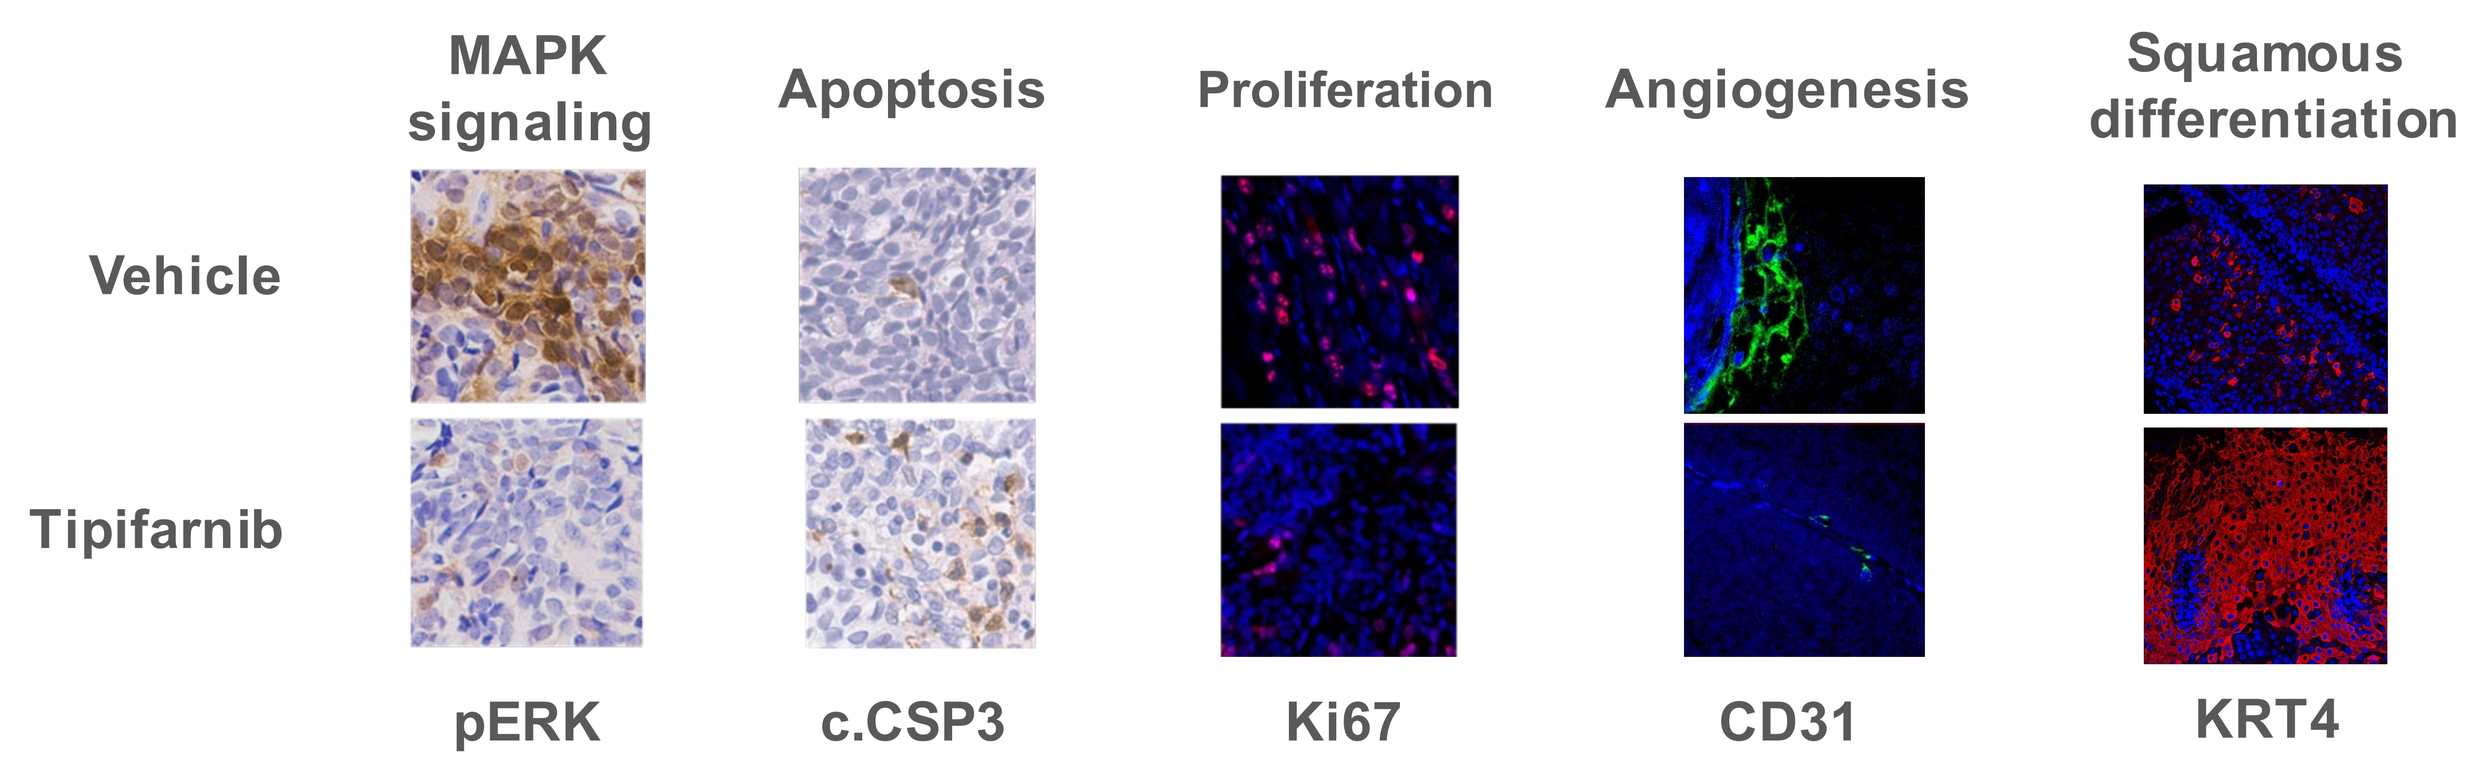
MAPK signaling
Apoptosis
Proliferation
Angiogenesis
Squamous differentiation
Vehicle
Tipifarnib
pERK
c.CSP3
Ki67
CD31
KRT4

Tipifarnib displayed robust single agent activity in preclinical models of HRAS mutant HNSCC and an ~ 50% ORR in a Phase 2 clinical trial (RUN-HN) in patients with R/R HRAS mutant HNSCC, leading to:

AIM-HN: Registration-directed trial of tipifarnib in *HRAS* mutant HNSCC (NCT03719690)

- Recurrent or metastatic patients after at least one prior platinum-containing line of therapy
- Now open in ~90 clinical sites in the U.S., Europe and Asia
- Intended to support an NDA seeking accelerated approval

A Subset of HNSCCs Overexpress HRAS in TCGA PanCancer Atlas

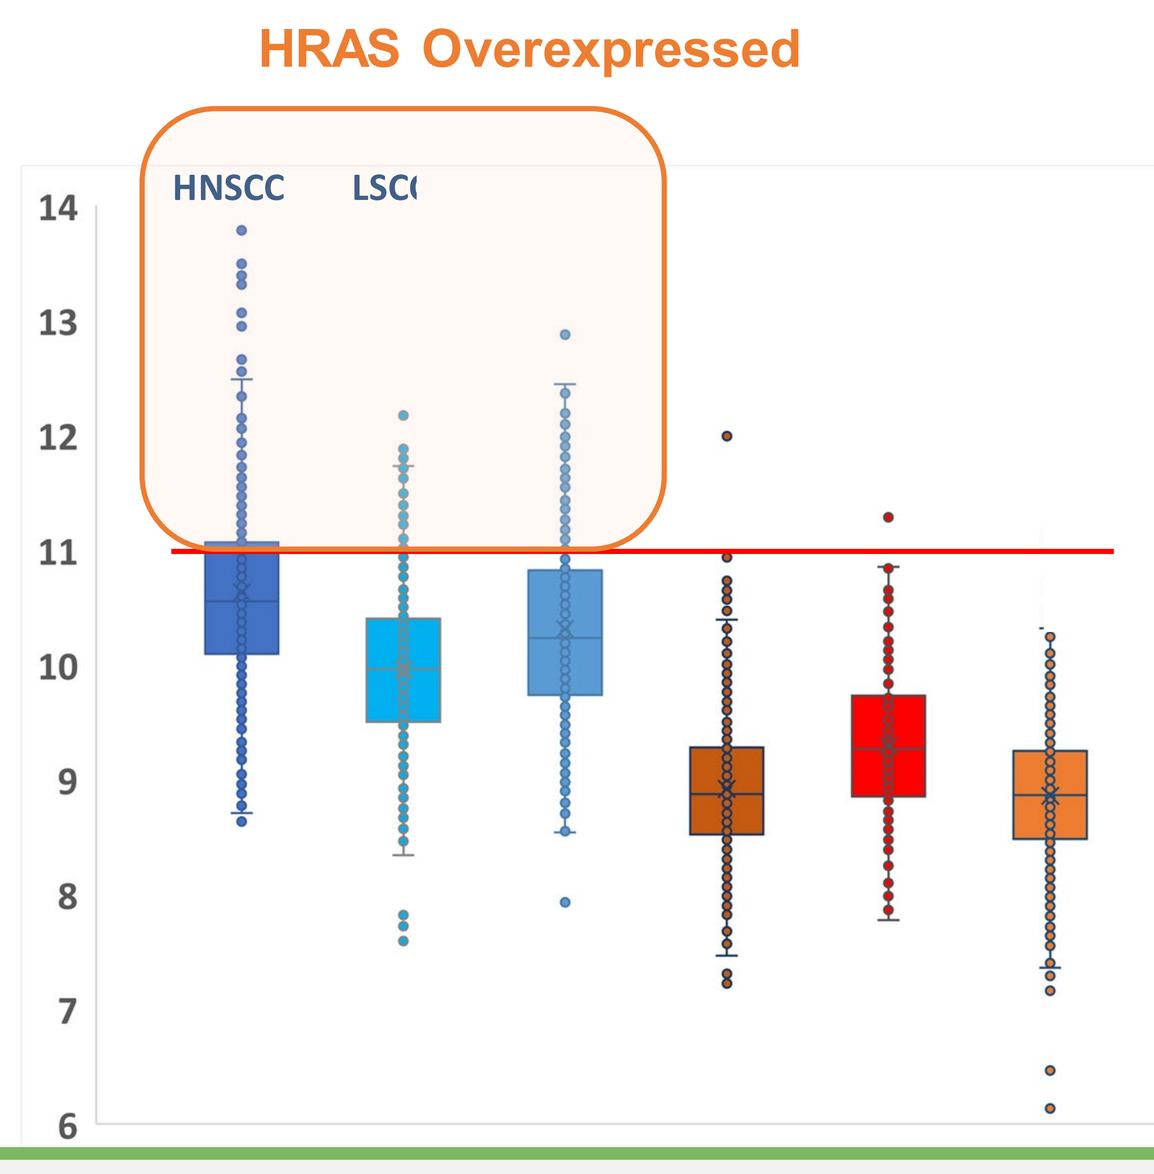
HRAS Overexpressed
HNSCC
LSCC

	# > CUTOFF	TOTAL	% HIGH
HNSCC	146	482	30.3
LSCC	39	461	8.5
UC	78	306	25.5
CRC	1	520	0.2
PDAC	2	163	1.2
LUAD	1	498	0.2

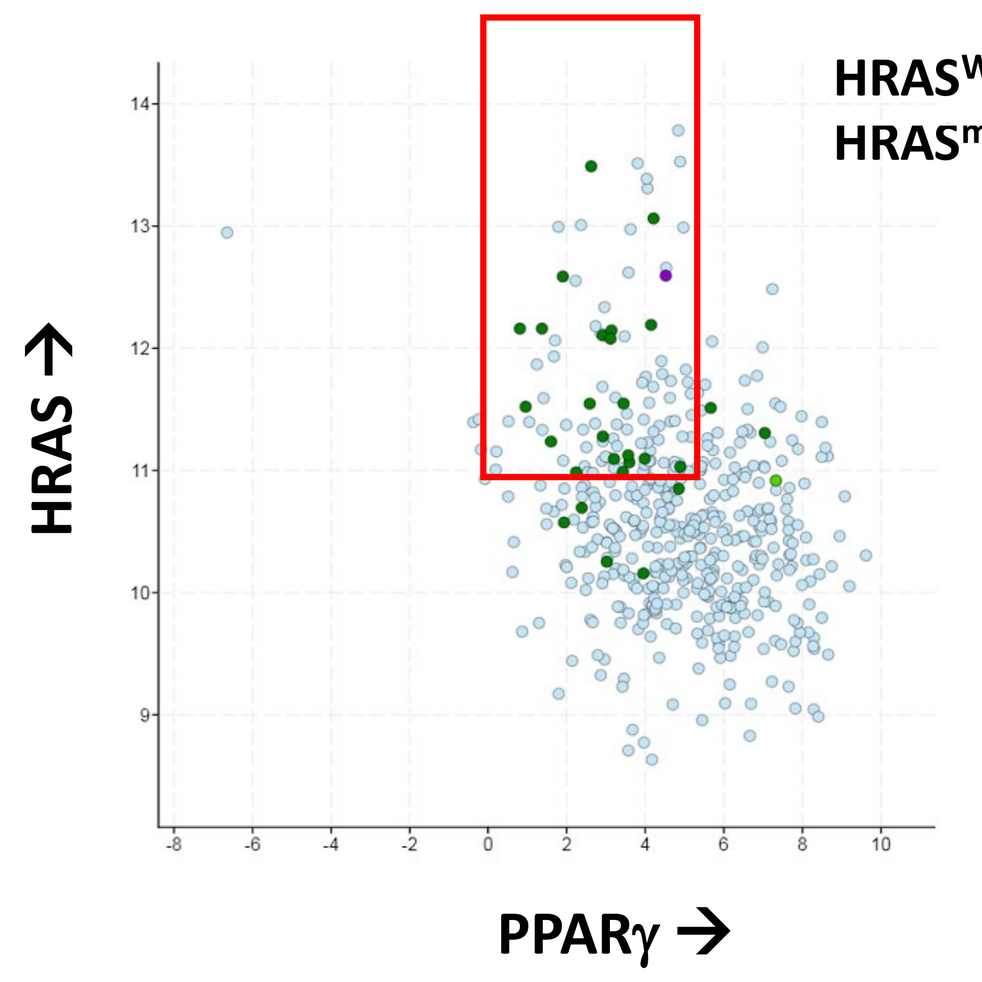
HRAS[WT]
HRAS[mut]
HRAS →
PPARγ →

- Average HRAS expression in HNSCC is 5-10x higher than other (colorectal, pancreatic, lung adenocarcinoma) tumor types
- HRAS-overexpressing HNSCCs are also commonly PPARγ-low
- Together with HRAS mutant tumors, HRAS-overexpressing HNSCC may represent a significant subset of **HRAS dependent tumors** with distinct biology that may be targeted by tipifarnib

HRAS[HIGH], PPAR-silenced HNSCC PDX are Sensitive to Tipifarnib

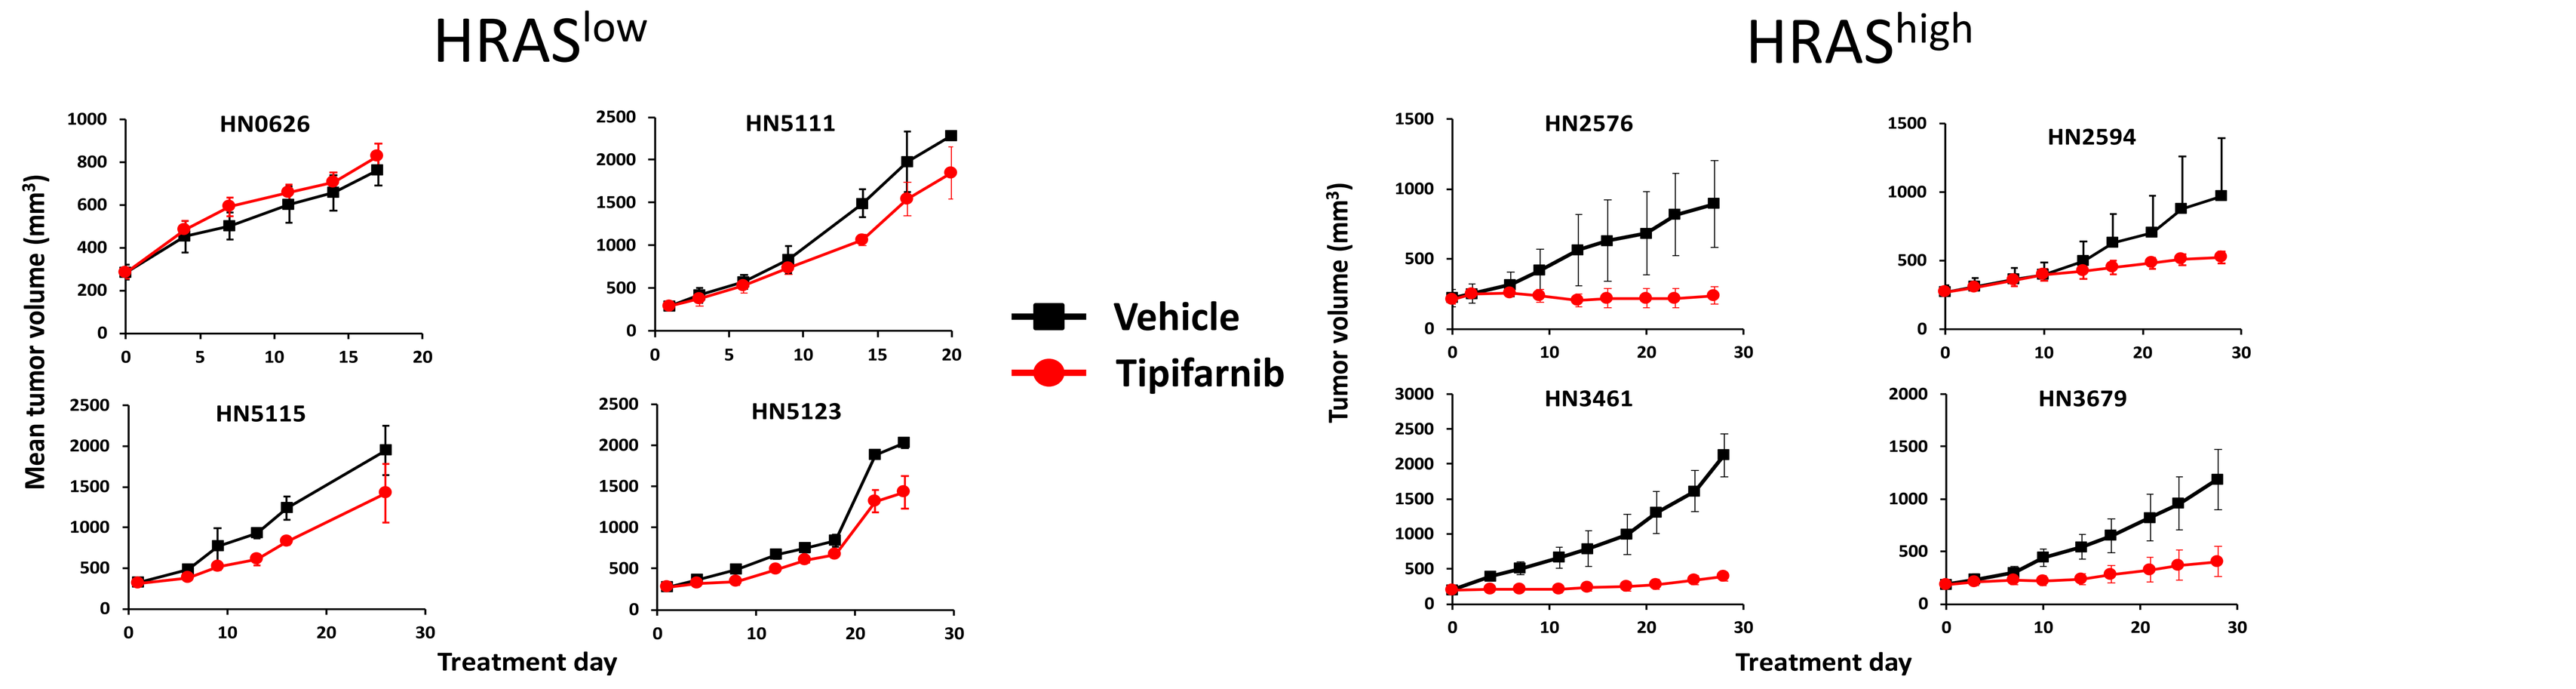
HRAS[low]
HRAS[high]
HN0626
HN5111
HN5115
HN5123
HN2576
HN2594
HN3461
HN3679
Mean tumor volume (mm³)
Tumor volume (mm³)
Vehicle
Tipifarnib
Treatment day

- Tipifarnib displayed robust inhibition of tumor growth in a subset of PDX models overexpressing wild-type HRAS
- In preliminary combination studies tipifarnib sensitized HRAS-WT-high HNSCC PDX to a variety of drugs, including cisplatin, cetuximab, palbociclib and the PI3K-pathway inhibitors alpelisib (PI3Kα), GSK2141795 (AKT) and TAK-228 (mTORK)
- These findings suggest that inhibition of overexpressed wild-type HRAS by tipifarnib could serve as an anchor for combination therapies. Alpelisib was chosen for further studies because of the close association of PI3Kα with HRAS in SCCs

Both HRAS and PI3K contribute to growth of transformed SCCs

Immortalized squamous epithelial cells + dox-inducible HRAS[G12D] (H) +/- constitutive PIK3CA[E545K] (HP)

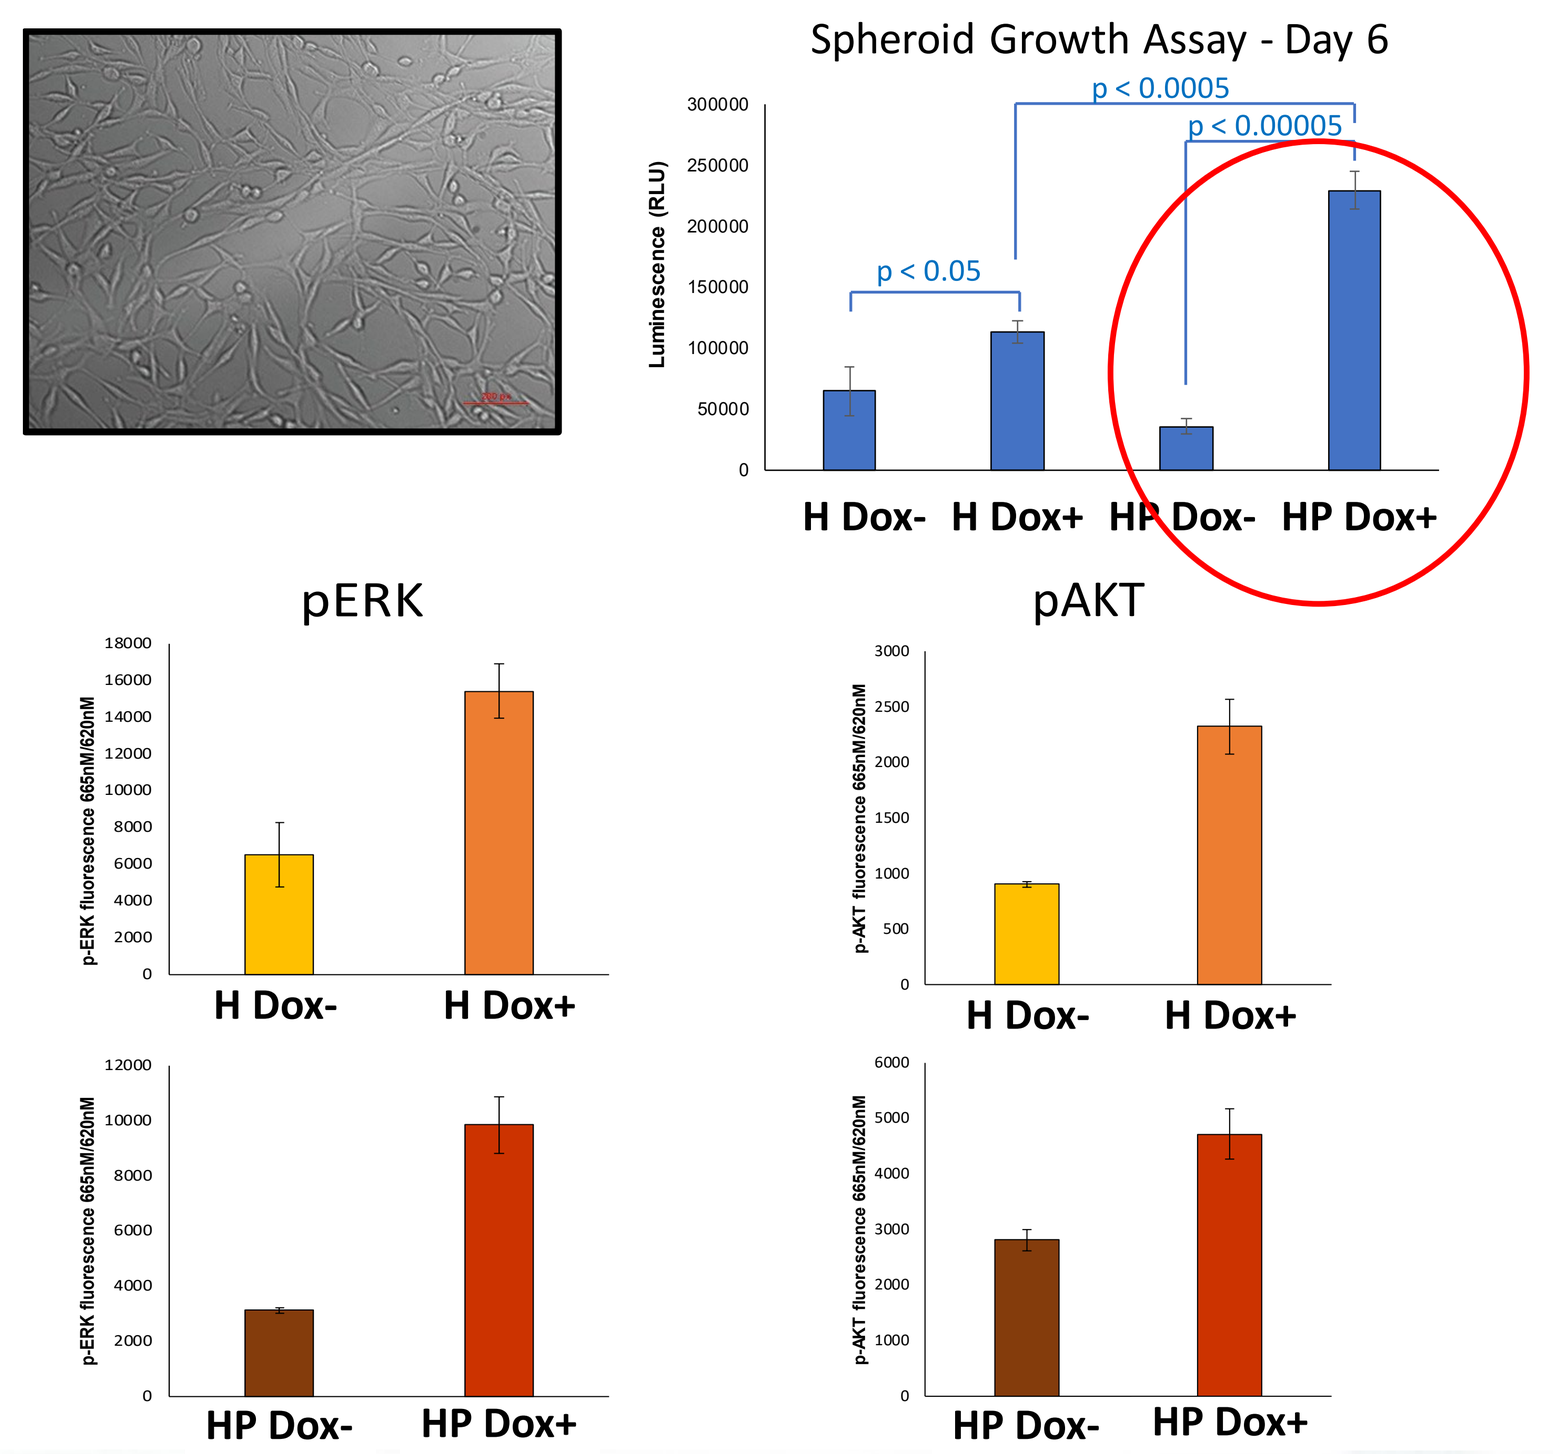
Spheroid Growth Assay - Day 6
p < 0.05
p < 0.0005
p < 0.00005
H Dox-
H Dox+
HP Dox-
HP Dox+
pERK
pAKT
H
HP

Mutant HRAS and PIK3CA both drive anchorage-independent growth and AKT signaling in transformed SCCs, but only HRAS activates MAPK. E545K mutant-driven growth is dependent on HRAS

Tipifarnib inhibits MAPK signaling and alpelisib blocks PI3K signaling in double mutants, but the combination is more effective against pAKT and spheroid formation

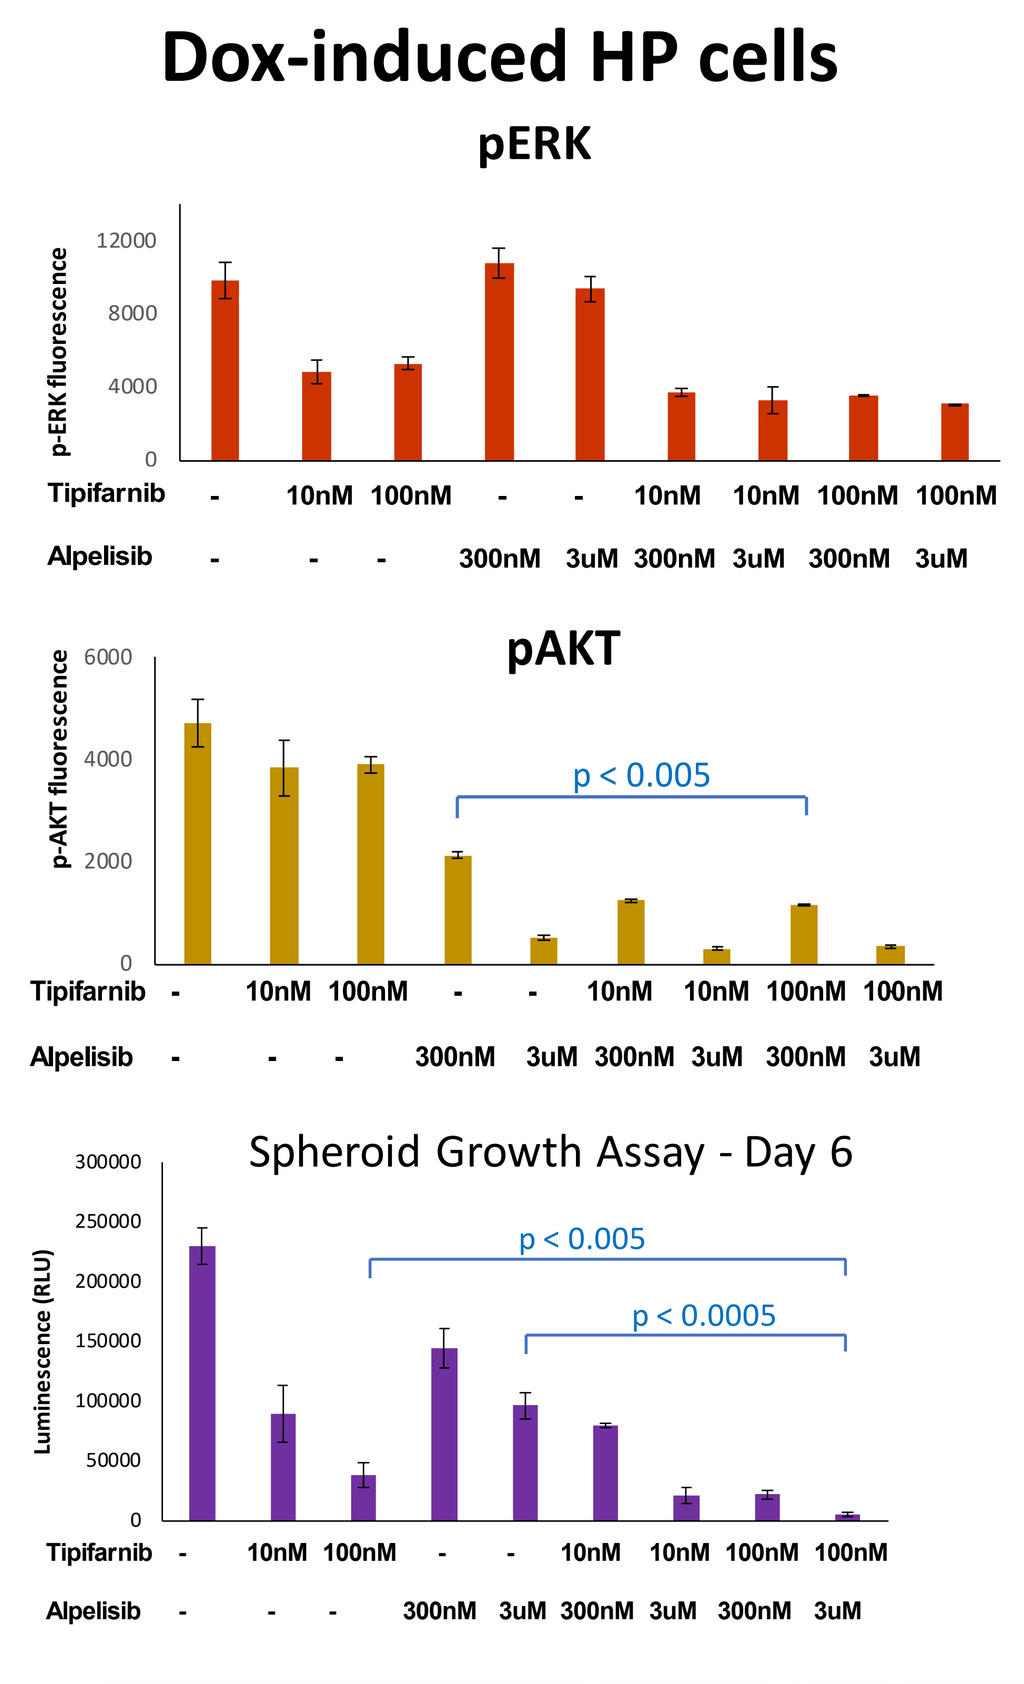
Dox-induced HP cells
pERK
pAKT
p < 0.005
Spheroid Growth Assay - Day 6
p < 0.0005
Tipifarnib
Alpelisib
10nM
100nM
300nM
3uM

Tipifarnib Enhances the activity of Alpelisib in PIK3CA-mutant and HRAS-mutant HNSCC PDX Models

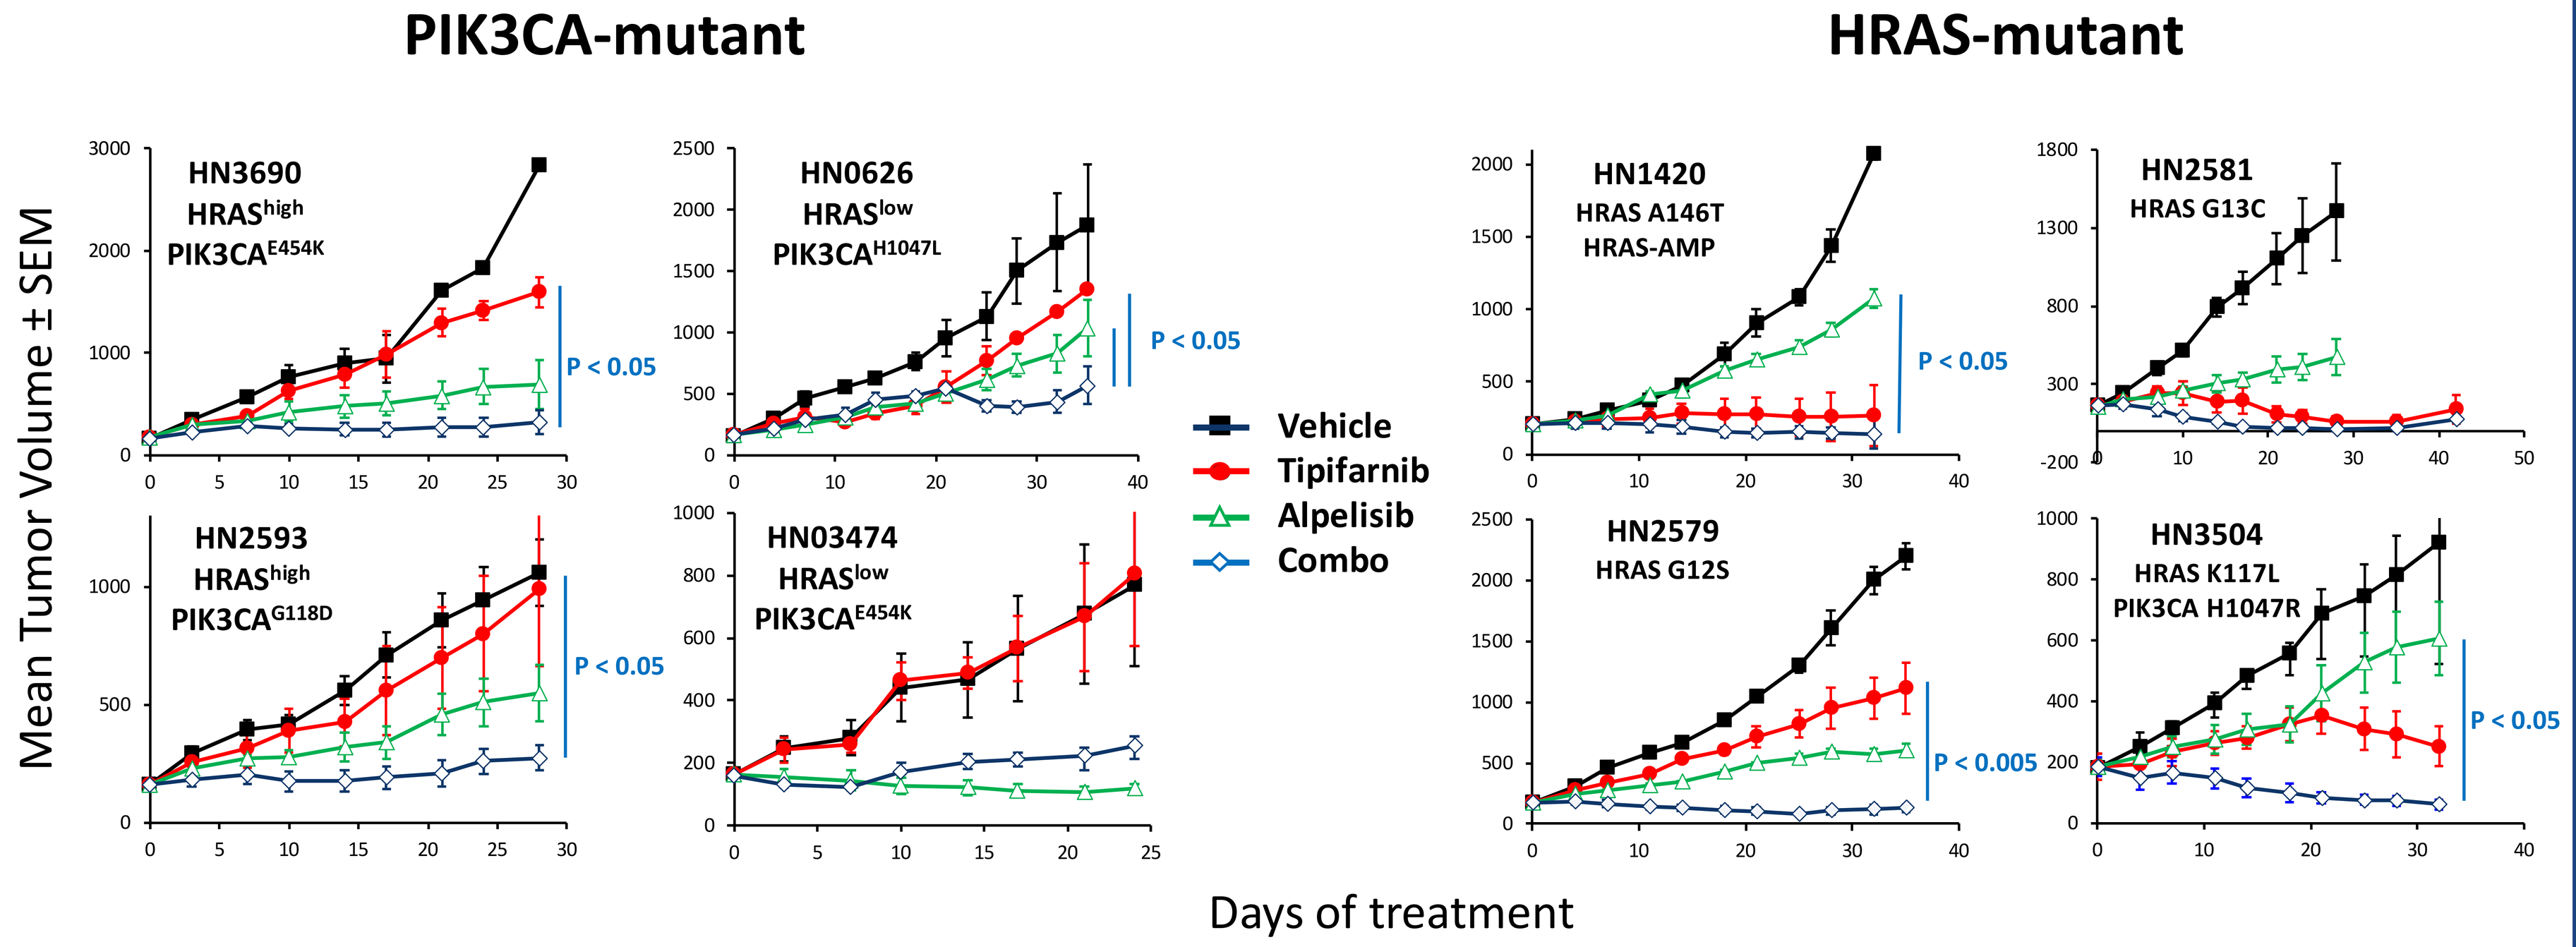
PIK3CA-mutant
HRAS-mutant
HN3690
HRAS[high]
PIK3CA[E454K]
HN0626
HRAS[low]
PIK3CA[H1047L]
HN2593
HRAS[high]
PIK3CA[G118D]
HN03474
HRAS[low]
PIK3CA[E454K]
HN1420
HRAS A146T
HRAS-AMP
HN2581
HRAS G13C
HN2579
HRAS G12S
HN3504
HRAS K117L
PIK3CA H1047R
Vehicle
Tipifarnib
Alpelisib
Combo
Mean Tumor Volume ± SEM
P < 0.05
P < 0.005
Days of treatment

Tipifarnib (80 → 60mg/kg BID) and alpelisib (60 → 40mg/kg QD) were used at reduced dose to reflect clinical practice

Tipifarnib Enhances the activity of Alpelisib in PIK3CA-amplified and HRAS-WT-overexpressing HNSCC PDX Models

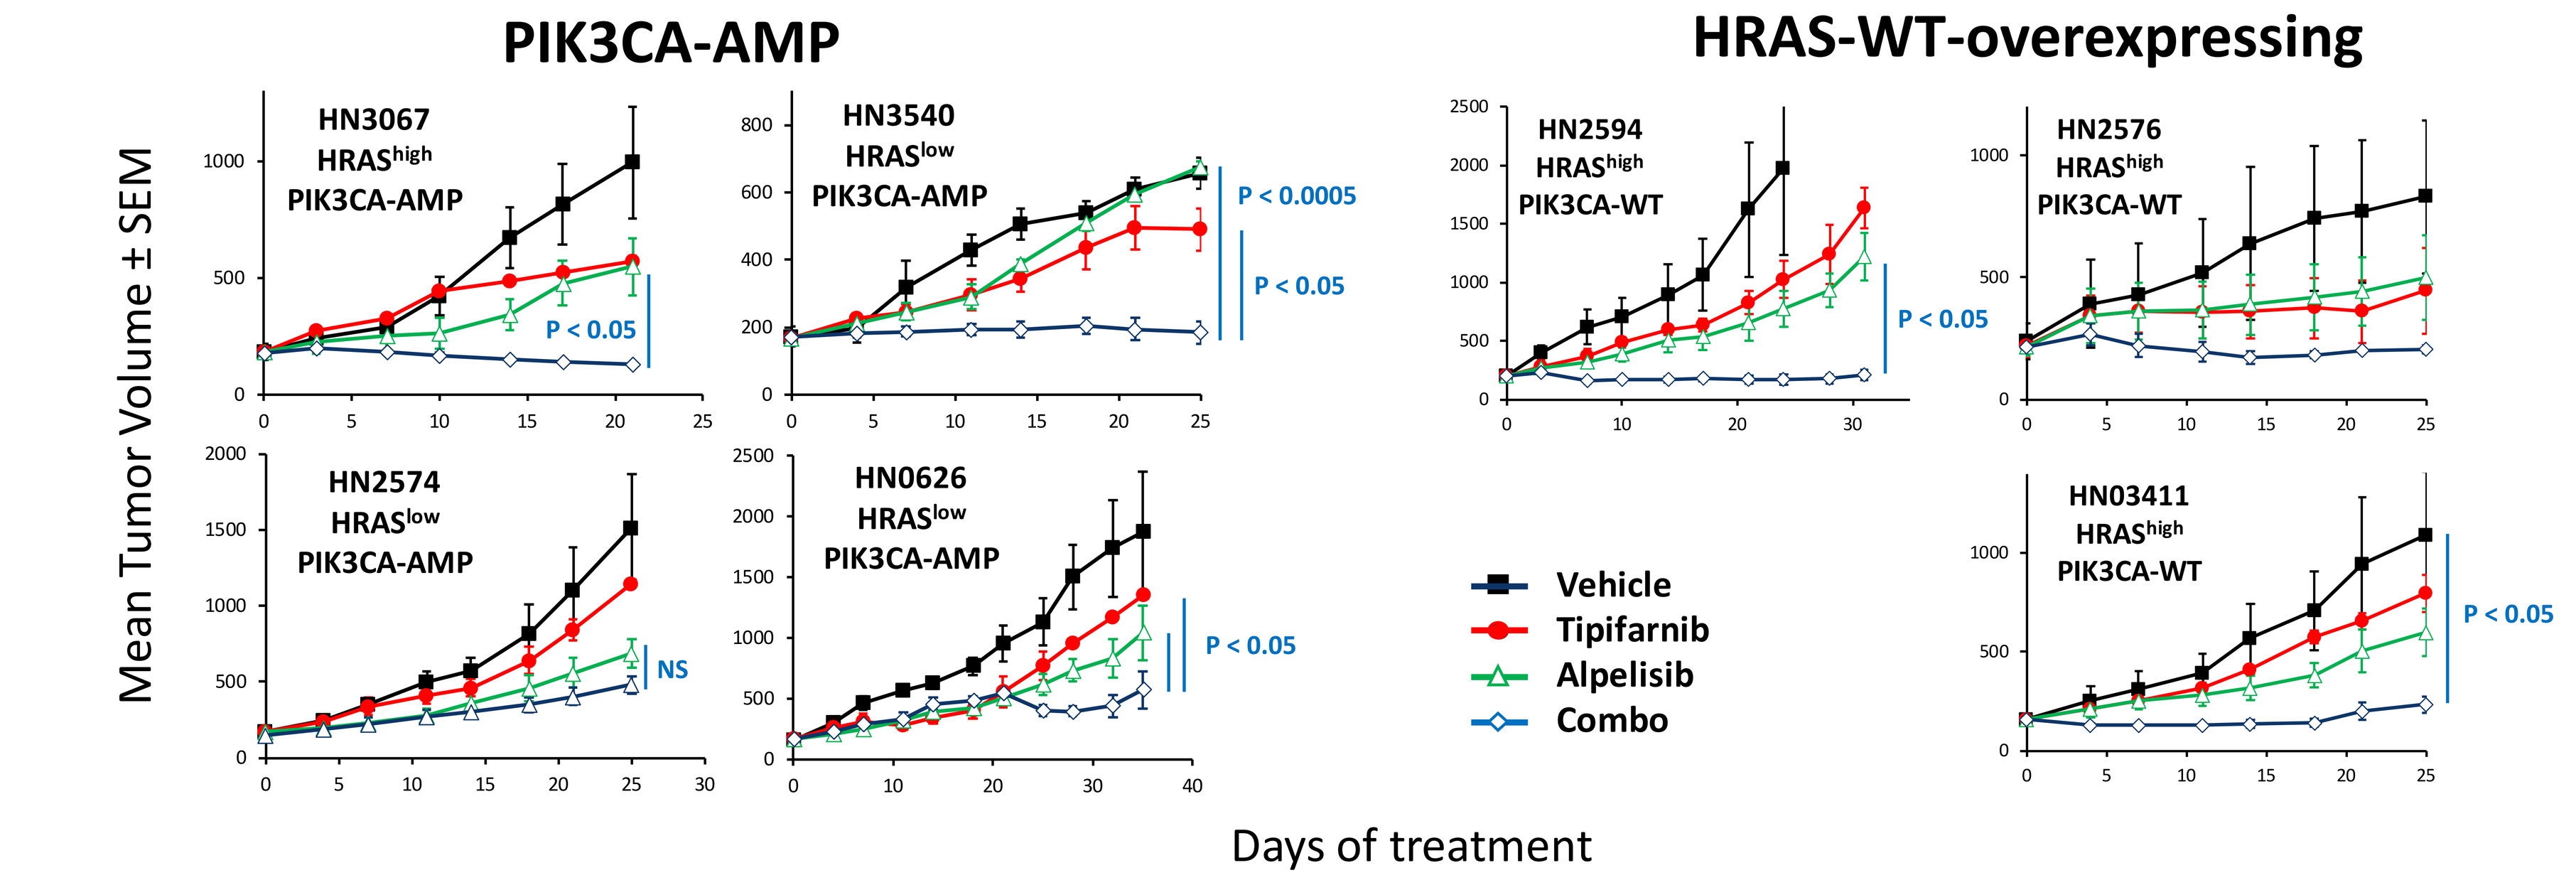
PIK3CA-AMP
HRAS-WT-overexpressing
HN3067
HRAS[high]
PIK3CA-AMP
HN3540
HRAS[low]
PIK3CA-AMP
HN2574
HRAS[low]
PIK3CA-AMP
HN0626
HRAS[low]
PIK3CA-AMP
HN2594
HRAS[high]
PIK3CA-WT
HN2576
HRAS[high]
PIK3CA-WT
HN03411
HRAS[high]
PIK3CA-WT
Vehicle
Tipifarnib
Alpelisib
Combo
Mean Tumor Volume ± SEM
P < 0.05
P < 0.0005
NS
Days of treatment

Tipifarnib (80→60mg/kg BID) and alpelisib (60→40mg/kg QD) were used at reduced dose to reflect clinical practice

CONCLUSIONS

- Kura is seeking to build upon clinical anti-tumor activity of tipifarnib demonstrated in patients with relapsed/refractory HRAS mutant HNSCC
- HRAS, both in the mutant and overexpressed form, acts as a key node at the center of HNSCC tumor biology for a significant subset of patients
- HRAS-MAPK and PI3K-AKT-mTOR are complementary pathways in HNSCC, each providing compensatory mechanisms of resistance to single agent inhibition of the other
- Combinations of tipifarnib and PI3Kα inhibitors have demonstrated compelling activity in PDX models of HRAS and PI3K dependent tumors
- Based on TCGA, ~50% of patients have HRAS or PI3K dependent HNSCC (HRAS overexpression or mutation, PI3K mutation or amplification)
- Kura is planning to conduct a Phase 1/2 proof-of-concept combination study of tipifarnib and a PI3Kα inhibitor in advanced or unresectable relapsed/refractory HNSCC harboring PIK3CA mutations or amplifications and/or HRAS overexpression

References: 1. Gilardi, M (2020) Mol Cancer Ther. 19:1784; 2. Su, S (2017) Theranostics 7:1088; 3. Campbell, J (2018) Cell Rep. 23:194; 4. International Cancer Genome Consortium (2013) Nat. Commun. 4:2873; 5. Gupta S (2007) Cell 129:957; 6. Zhao L (2008) PNAS 105:2652; 7. Ruicci K (2018) Oral Oncology 84:95; 8. Zhu B (2014) Oncogene 33:5348