Tipifarnib is Highly Active in HRAS-mutant Lung Squamous Cell Carcinoma Tumor Models

Linda Kessler[1], Catherine Scholz[2], Antonio Gualberto[2], Yi Liu[1] and Francis Burrows[1]

[1]Kura Oncology, Inc., San Diego, CA, USA, [2]Kura Oncology, Inc., Cambridge, MA, USA

BACKGROUND

- Tipifarnib is a potent and selective inhibitor of farnesyltransferase (FT) that catalyzes the post-translational attachment of farnesyl groups to proteins that require localization to the inner cell membrane. Although all RAS isoforms (KRAS/NRAS/HRAS) are FT substrates, HRAS is exclusively dependent upon farnesylation for membrane localization and signaling activation, making HRAS mutant tumors uniquely susceptible to tipifarnib-mediated inhibition of FT.
- HRAS is the most commonly mutated RAS species in both lung squamous cell carcinoma (LSCC) and head and neck squamous cell carcinoma (HNSCC), observed in approximately 2% and 5% of cases, respectively (TCGA, Nature 2012 and 2015).
- Recent evidence supports the clinical utility of tipifarnib for treatment of patients with HRAS-mutant HNSCC, and we present herein data supporting a potential utility of tipifarnib in the treatment of HRAS-mutant LSCC.

Genomic profiling of squamous tumors has revealed important similarities between lung squamous cell carcinoma (LSCC) and head and neck squamous cell carcinoma (HNSCC).

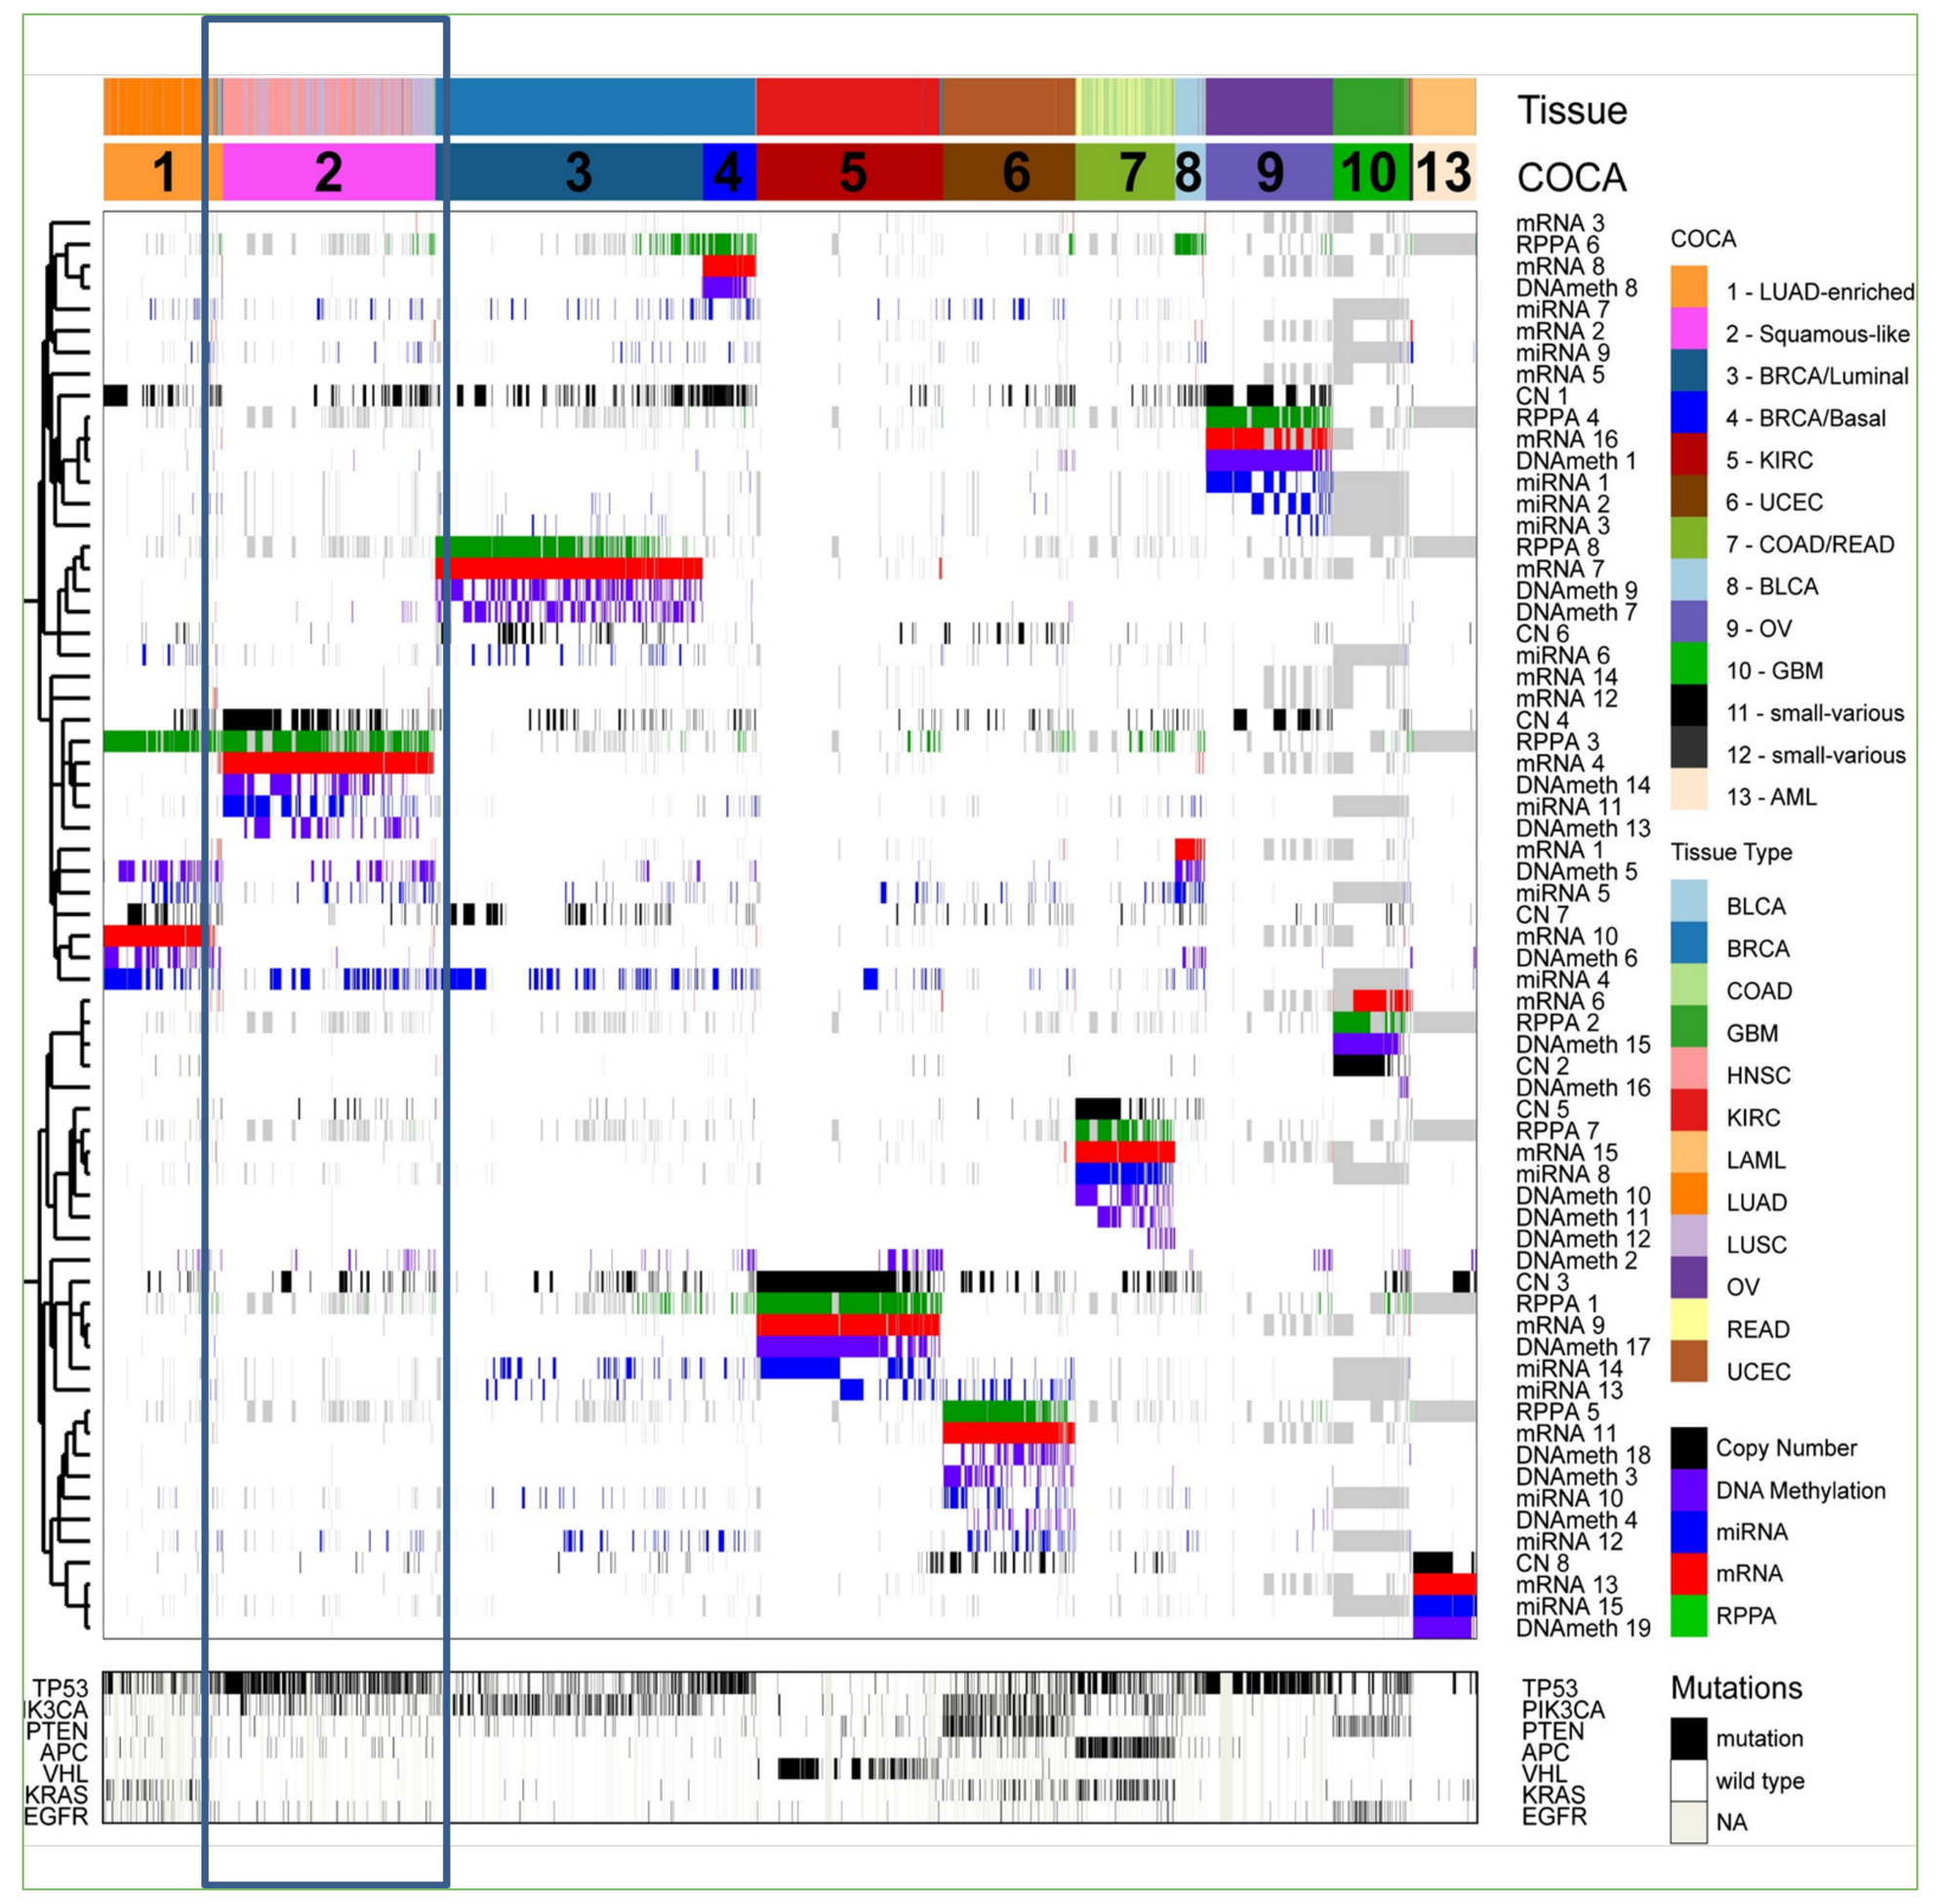
Tissue
COCA
1 2 3 4 5 6 7 8 9 10 13
1 - LUAD-enriched
2 - Squamous-like
3 - BRCA/Luminal
4 - BRCA/Basal
5 - KIRC
6 - UCEC
7 - COAD/READ
8 - BLCA
9 - OV
10 - GBM
11 - small-various
12 - small-various
13 - AML
Tissue Type
BLCA
BRCA
COAD
GBM
HNSC
KIRC
LAML
LUAD
LUSC
OV
READ
UCEC
Copy Number
DNA Methylation
miRNA
mRNA
RPPA
Mutations
mutation
wild type
NA
TP53
PIK3CA
PTEN
APC
VHL
KRAS
EGFR

Reprinted from Hoadley KA, et al. *Cell* 2014;58(4):929-944, with permission from Elsevier.

- Gene signatures have been described that correlate common molecular characteristics across squamous cell carcinomas including head and neck, lung, cutaneous, gastrointestinal and gynecologic cancers.[3]

[3] Schwaederle M, et al. Cell Cycle 2015;14(14):2355-2361

Cluster analysis of 12 pathologically defined cancer types using DNA methylation, RNA expression, copy number, RPPA and miRNA shows **strong correlation between LSCC and HNSCC tissue types**

Tipifarnib displays robust antitumor activity in PDX models of LSCC with HRAS mutations

Tipifarnib induces deep, sustained regression

Tipifarnib is active in model resistant to cetuximab

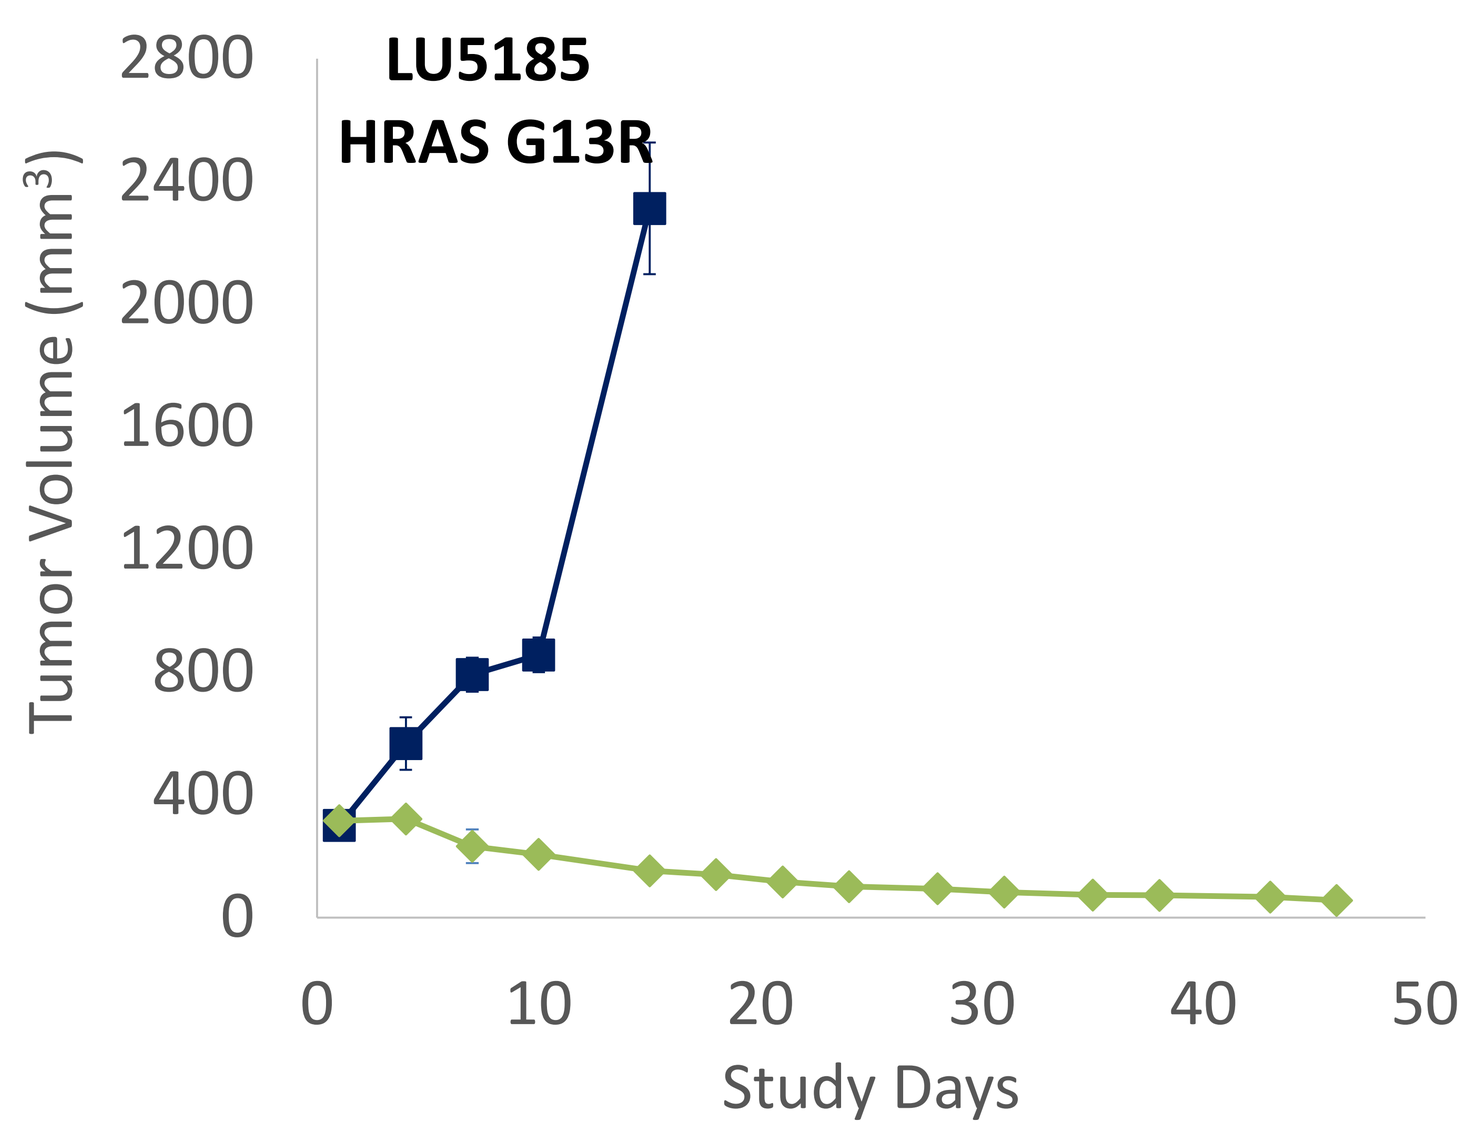
LU5185
HRAS G13R
Tumor Volume (mm³)
Study Days

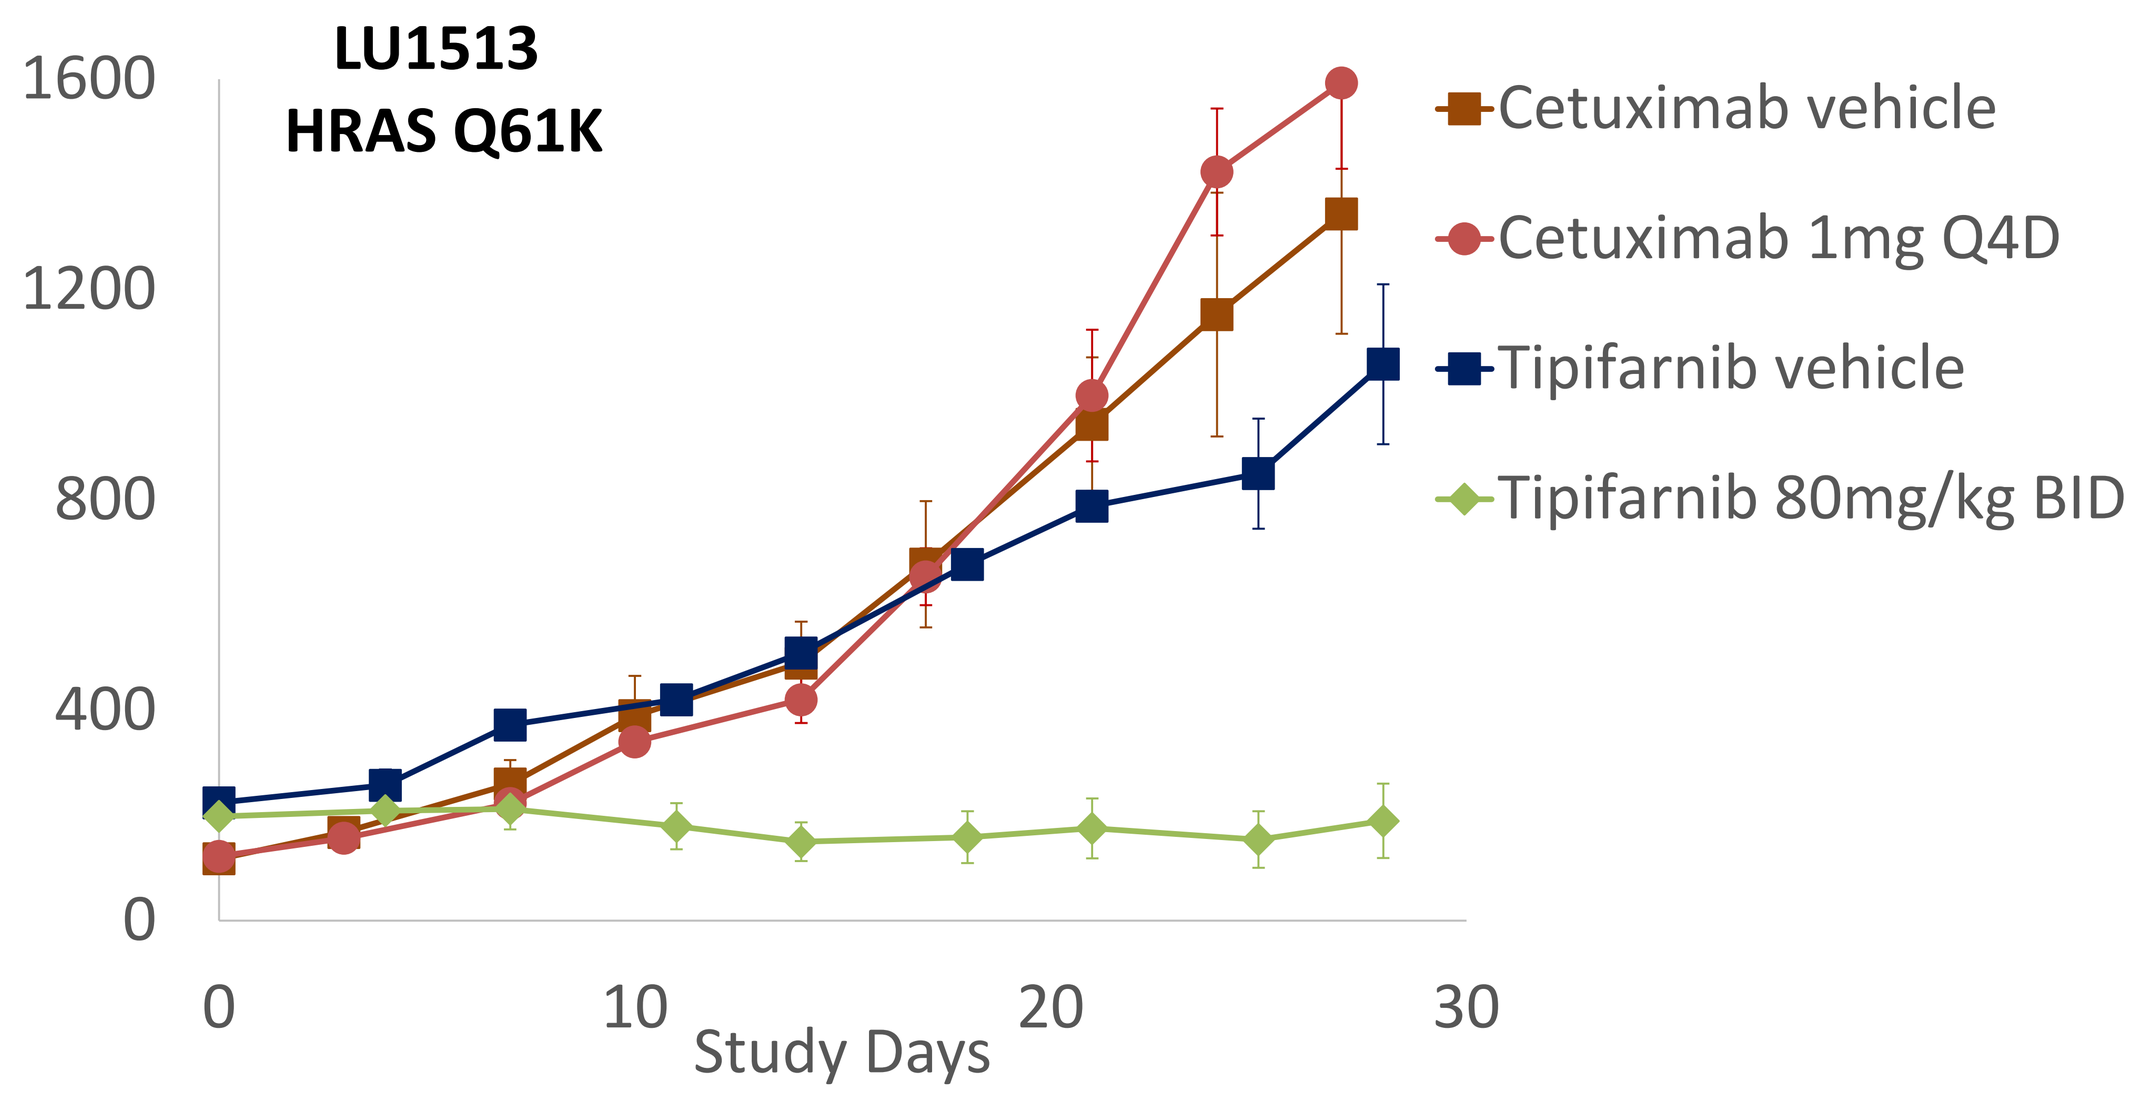
LU1513
HRAS Q61K
Cetuximab vehicle
Cetuximab 1mg Q4D
Tipifarnib vehicle
Tipifarnib 80mg/kg BID
Study Days

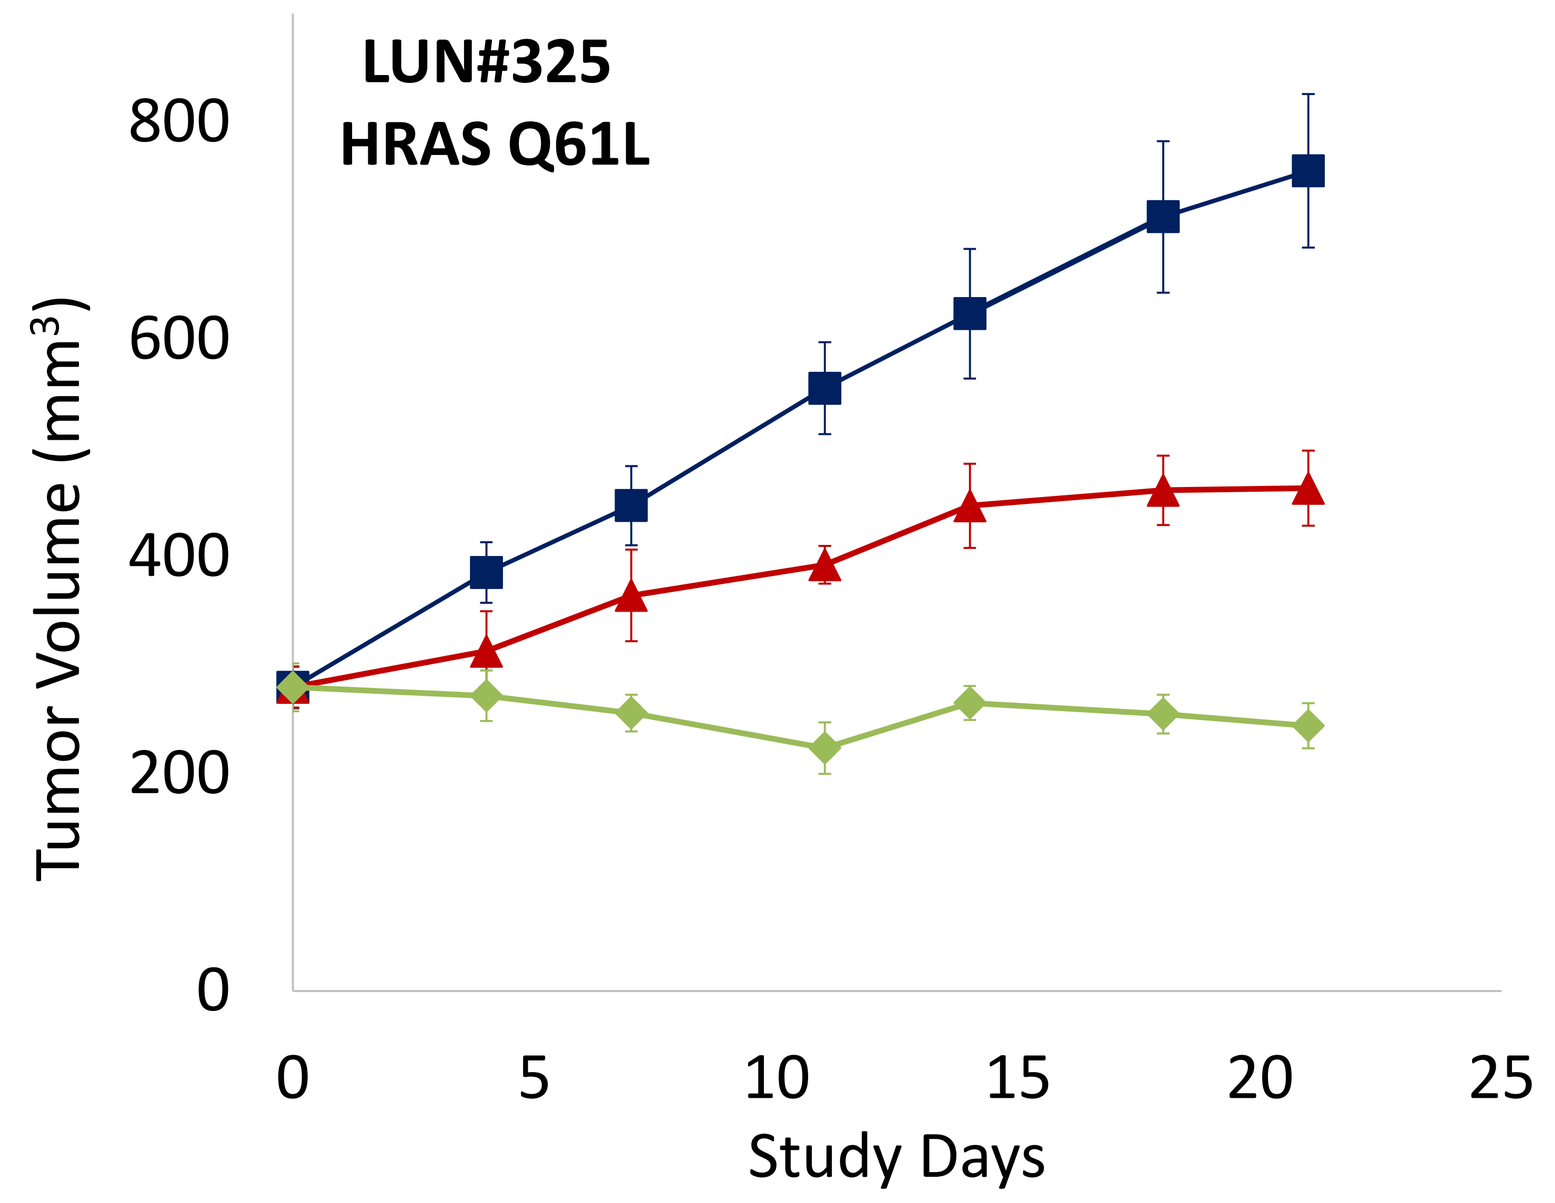
LUN#325
HRAS Q61L
Tumor Volume (mm³)
Study Days

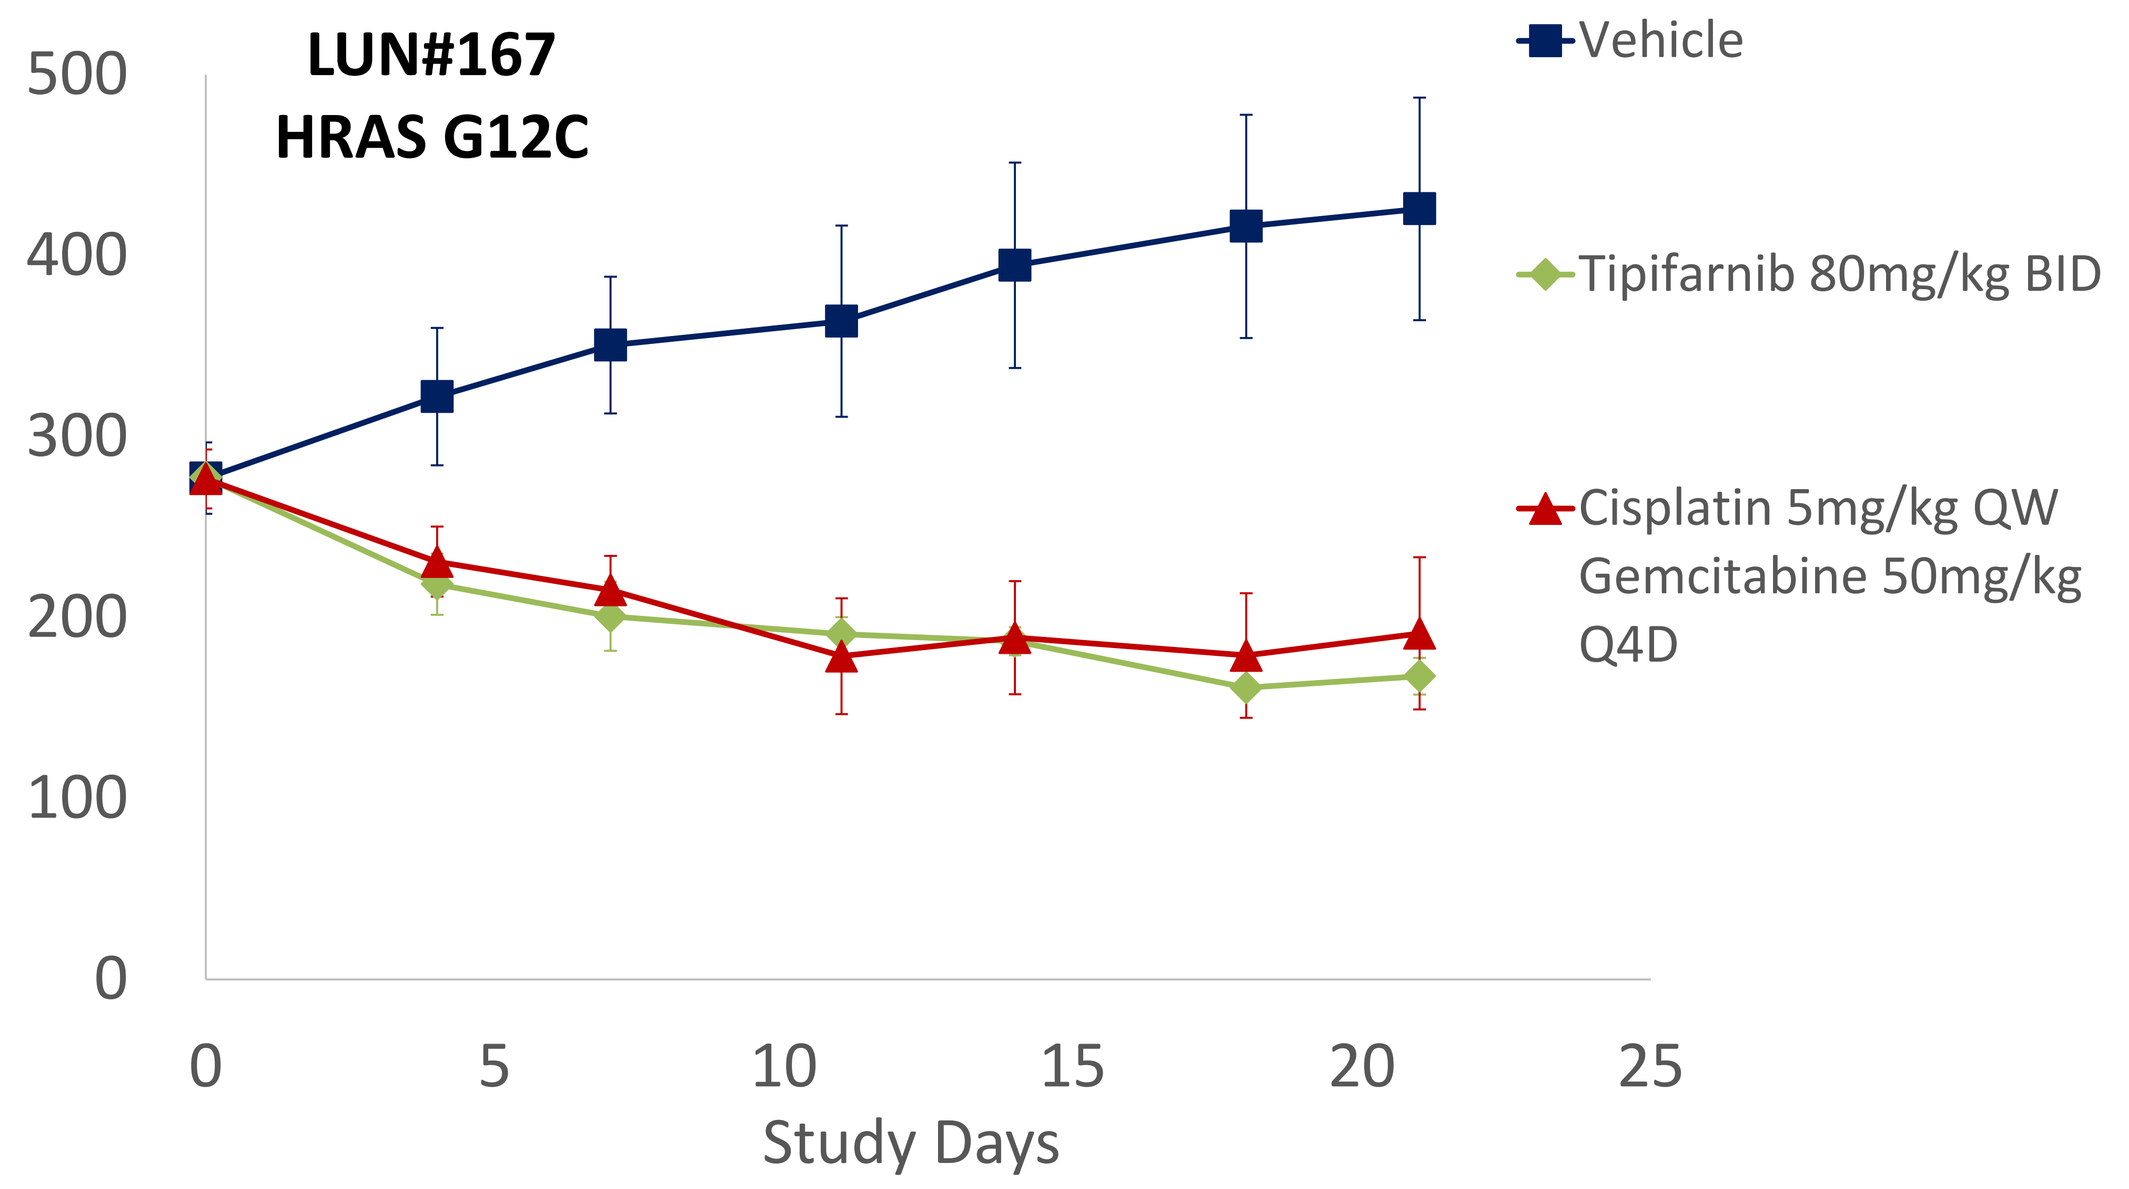
LUN#167
HRAS G12C
Vehicle
Tipifarnib 80mg/kg BID
Cisplatin 5mg/kg QW Gemcitabine 50mg/kg Q4D
Study Days

Comparison TCGA data for most frequently mutated genes in LSCC and HNSCC shows correlation in mutation frequency

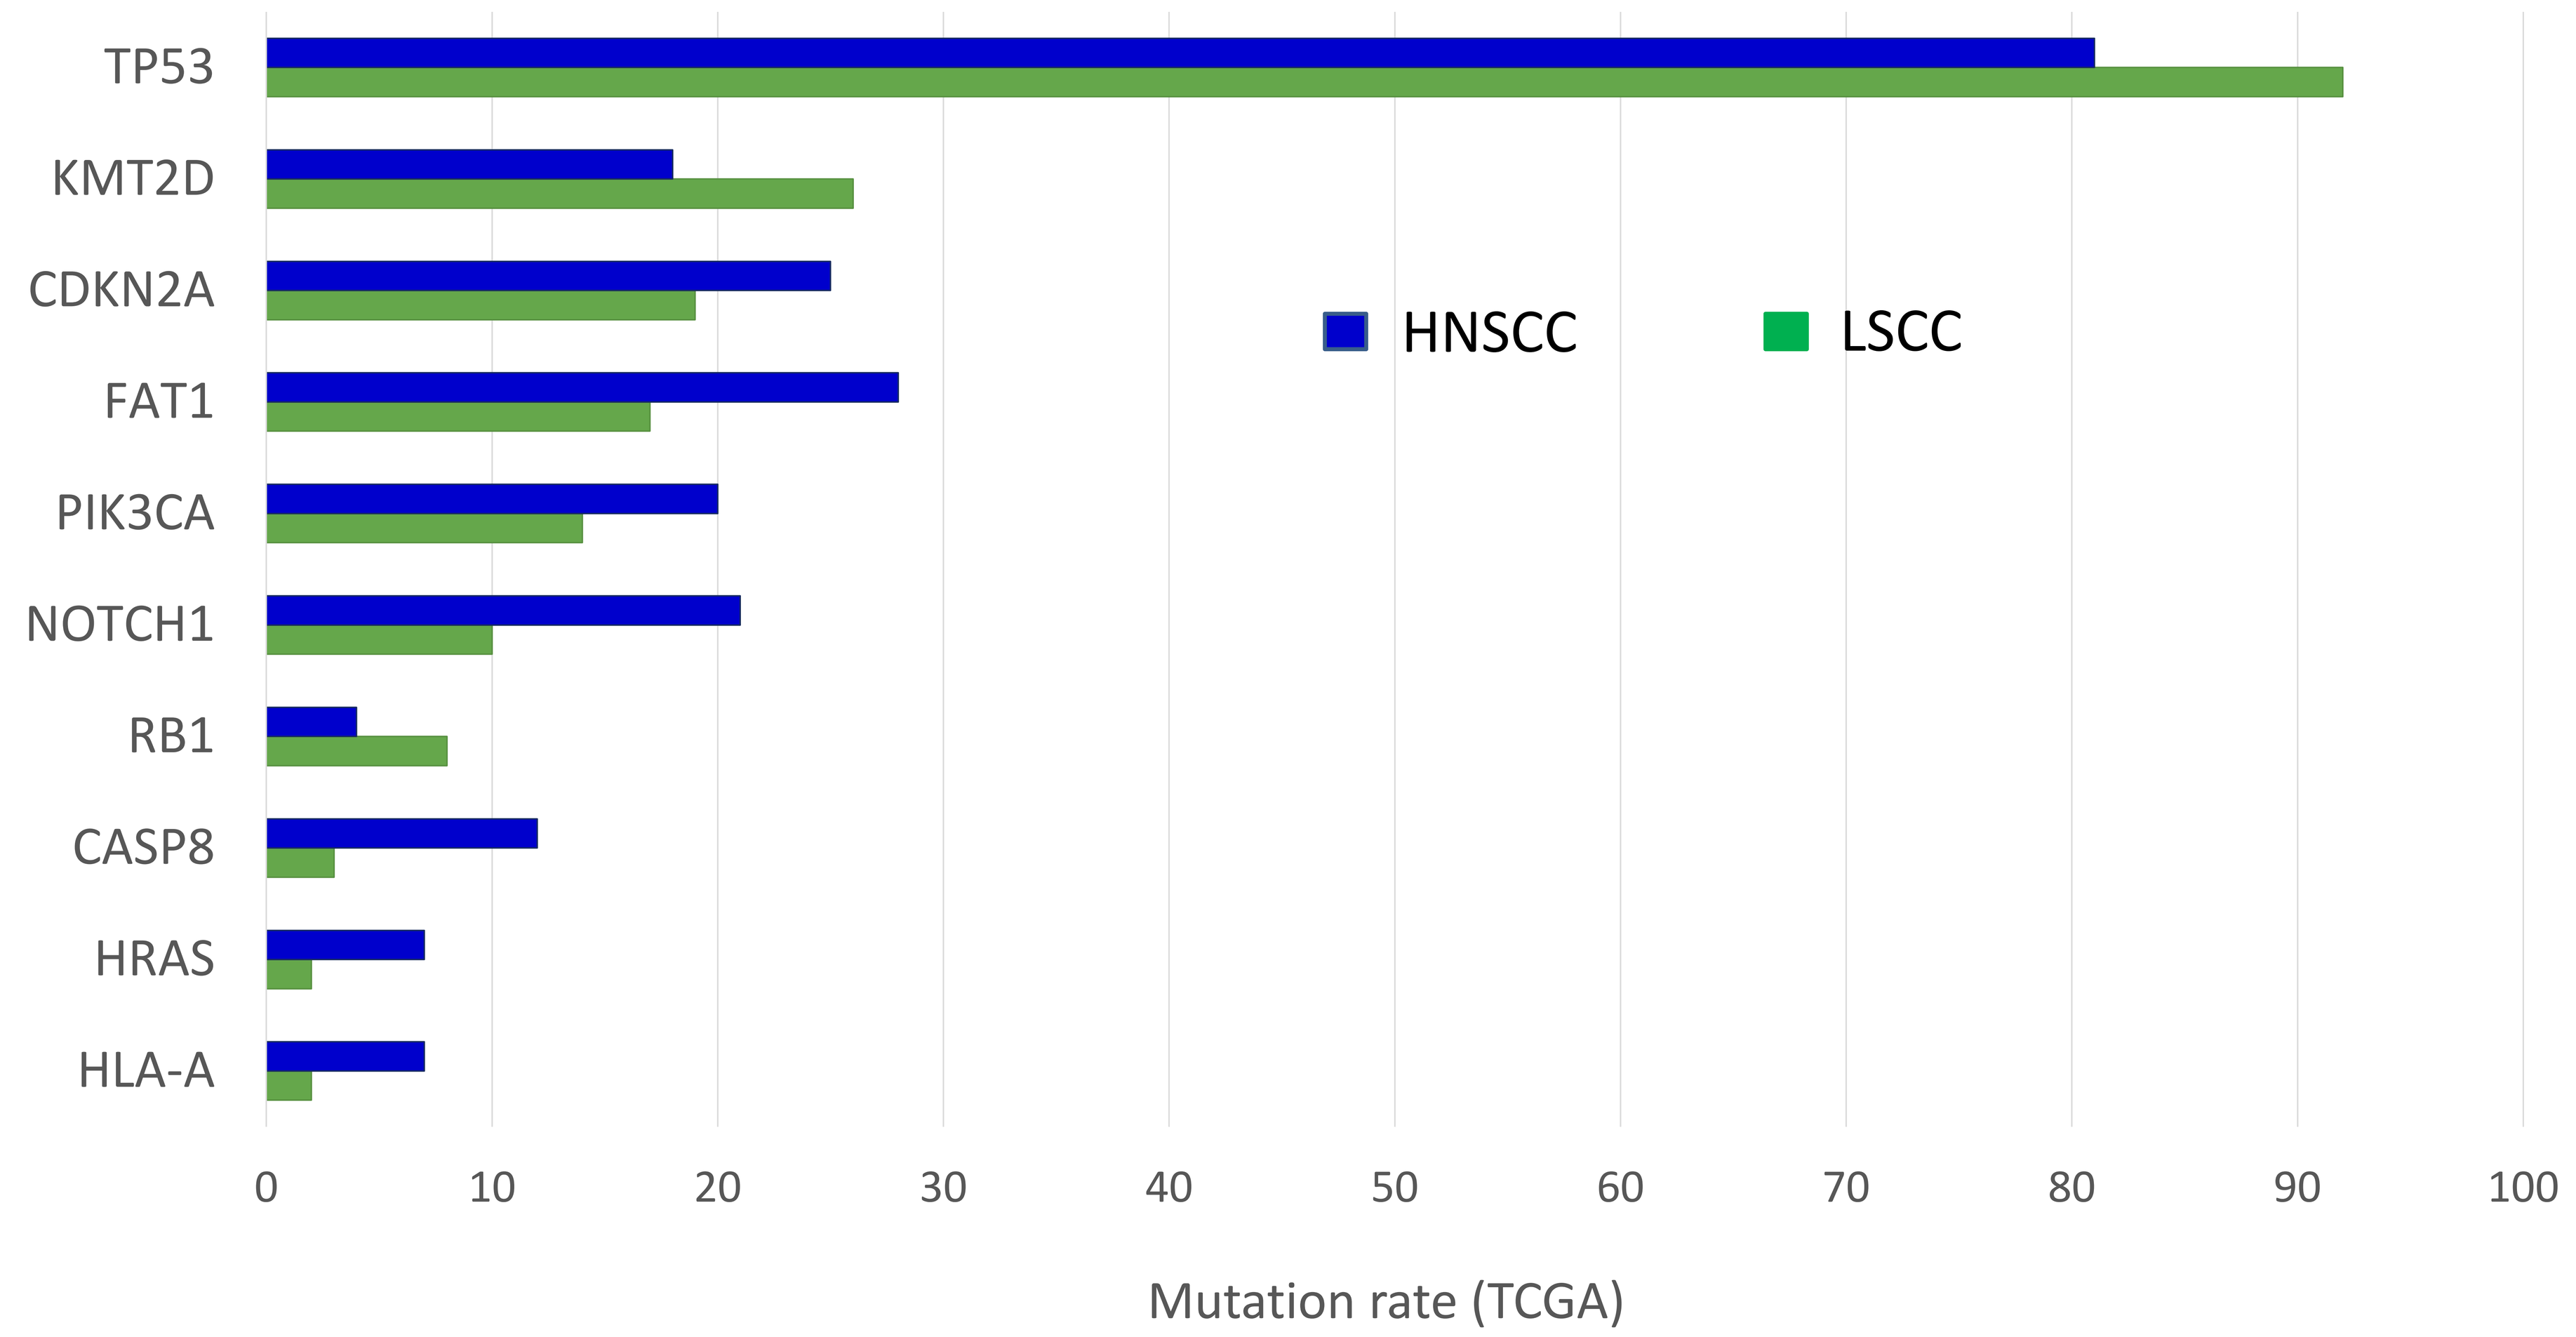
TP53
KMT2D
CDKN2A
FAT1
PIK3CA
NOTCH1
RB1
CASP8
HRAS
HLA-A
HNSCC
LSCC
Mutation rate (TCGA)

Robust activity of tipifarnib in HRAS-mutant HNSCC PDX models is consistent with responses observed in patients enrolled in the ongoing Phase 2 RUN-HN trial in HRAS-mutant HNSCC

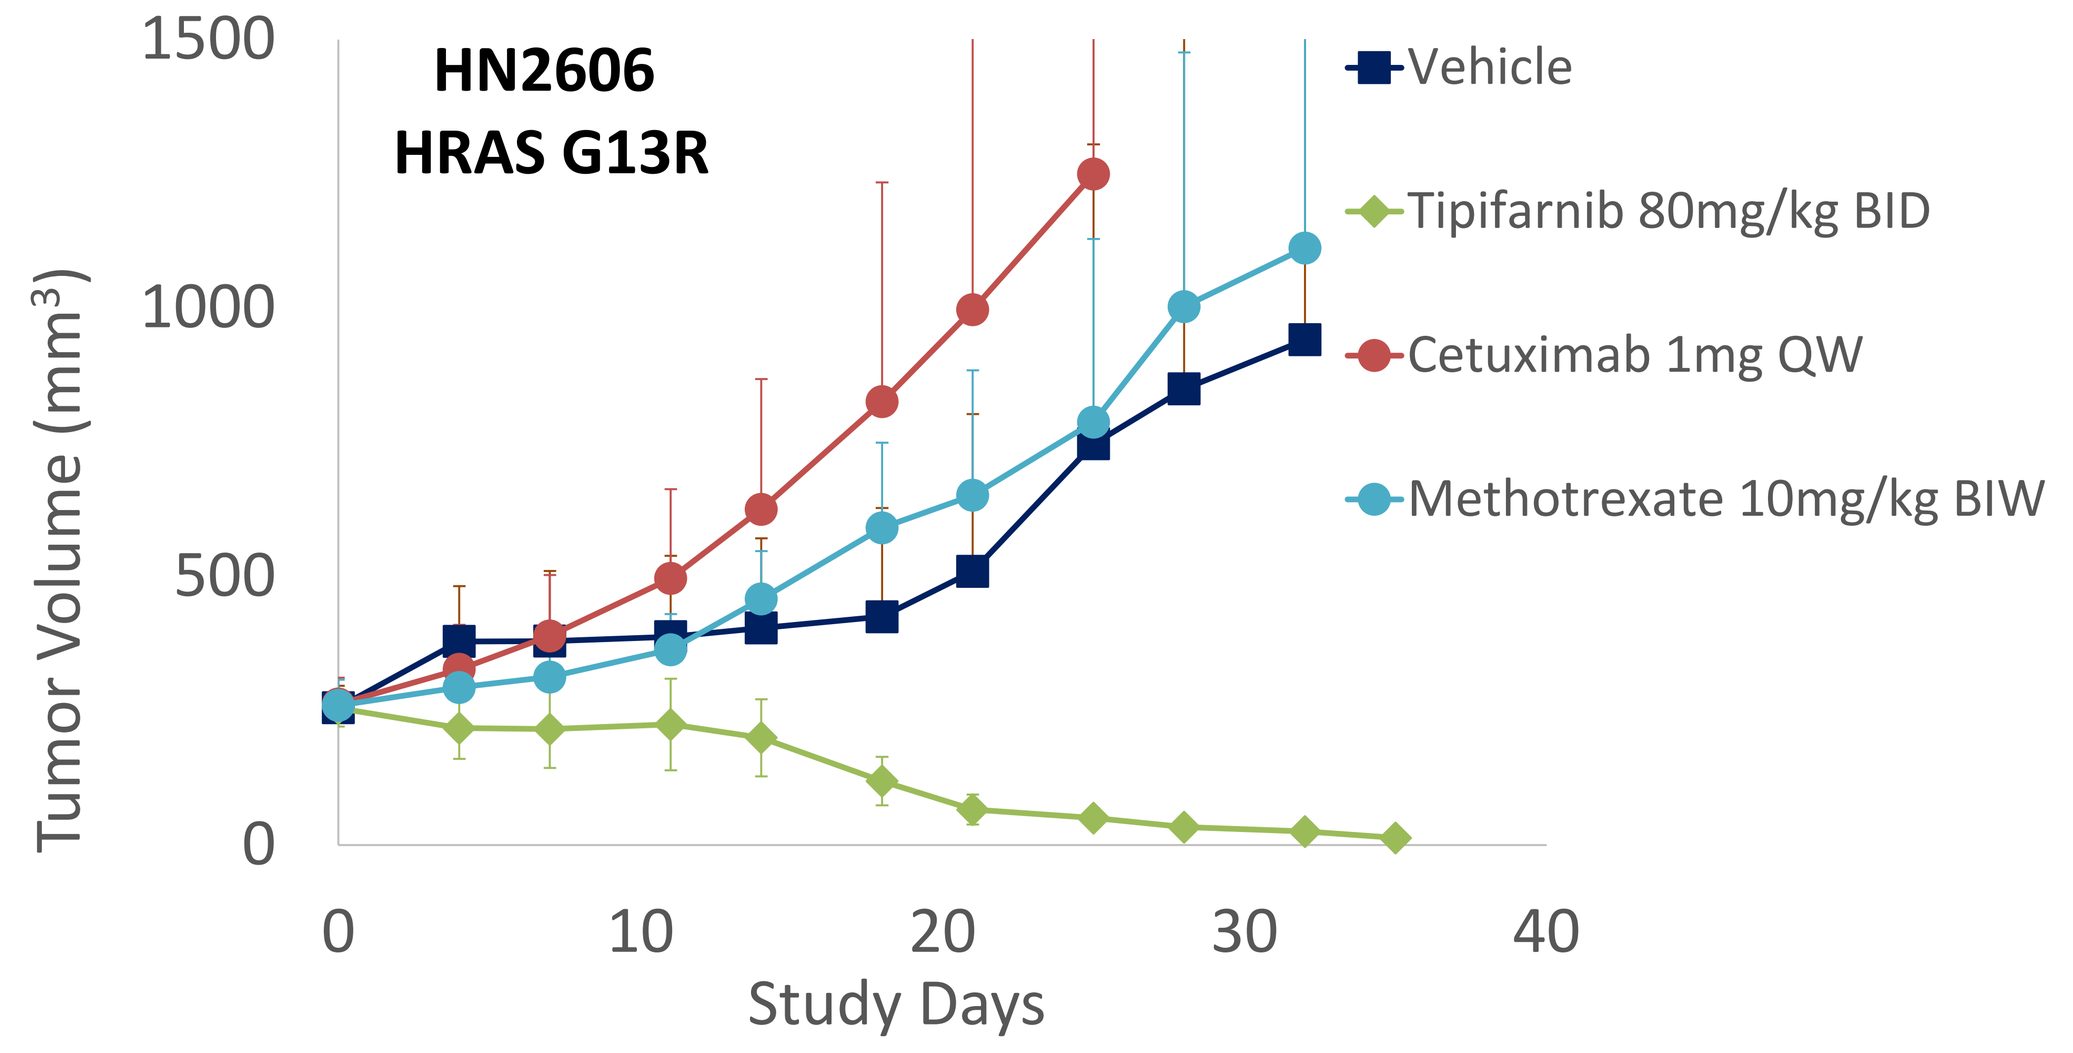
HN2606
HRAS G13R
Vehicle
Tipifarnib 80mg/kg BID
Cetuximab 1mg QW
Methotrexate 10mg/kg BIW
Tumor Volume (mm³)
Study Days

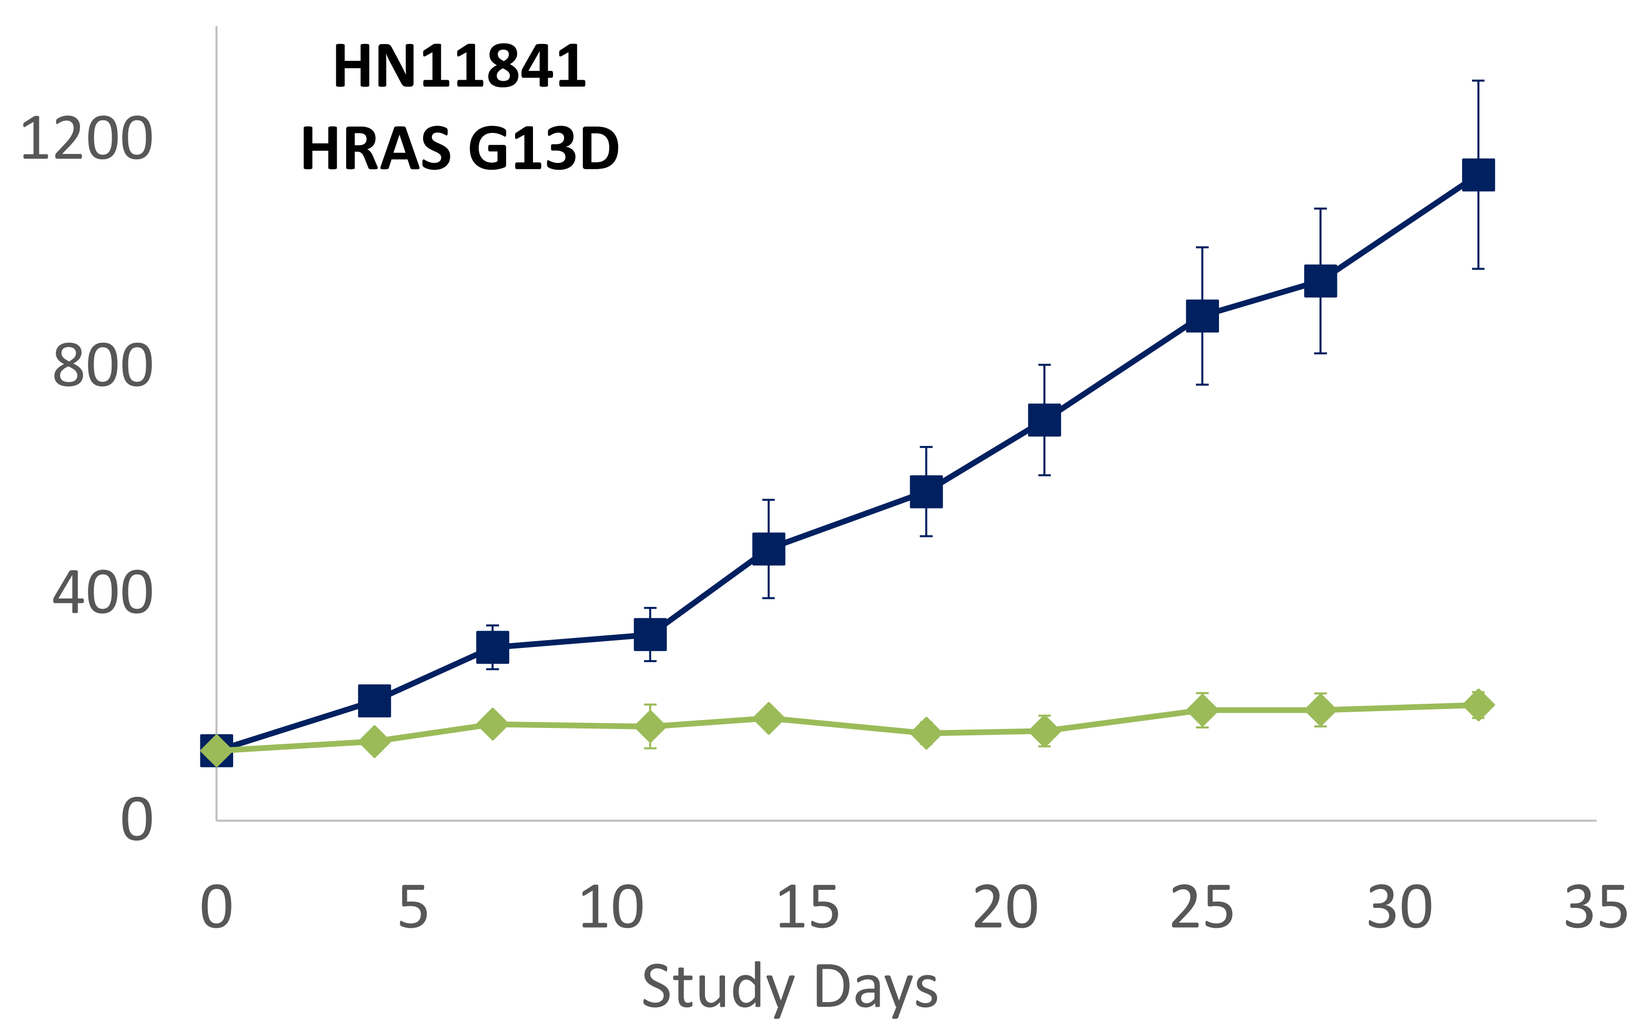
HN11841
HRAS G13D
Study Days

RUN-HN
KURA KO-TIP-001
NCT02383927

- Clinical proof-of-concept has been achieved in recurrent/metastatic HRAS-mutant HNSCC
- Confirmed PRs in 5 of 6 evaluable patients (83%, 36-99.6% 95% CI) as of February 08, 2018 data cutoff
- Durable responses observed, including 2 responses > 18 months in duration

Solid Tumors with HRAS mutations

Cohort 1: **Thyroid Cancer**

Cohort 2: **Other / HNSCC**

Cohort 3: SCC (excluding HNSCC) *Enrollment ongoing*

HNSCC Extension *Enrollment ongoing*

- Ongoing P2 study has been expanded to include a cohort of HRAS-mutant SCC, excluding patients with HRAS-mutant HNSCC.
- The Spanish Lung Cancer Group plans to initiate a Phase 2 IST of tipifarnib in HRASm LSCC

CONCLUSIONS

- **Treatment of HRAS-mutant LSCC PDX models induces tumor regressions, which is similar to the activity of tipifarnib observed in HRAS-mutant HNSCC PDX models.**
- **Clinical activity, including durable partial responses have been observed in patients with HRAS-mutant HNSCC in the ongoing RUN-HN trial**
- **These data demonstrate that HRAS is a targetable mutation in LSCC as well as in HNSCC and illustrate the potential for tipifarnib in the treatment of additional HRAS-mutant squamous cell carcinomas.**

Poster #4917, AACR Annual Meeting April 14-18th, 2018