Preliminary Results from a Phase 2 Trial of Tipifarnib in HRAS mutant Head & Neck Squamous Cell Carcinomas (HNSCC).

Alan Ho[1], Nicole Chau[2], Irene Brana[3], Charles Ferte[4], Caroline Even[4], Francis Burrows[5], Linda Kessler[5], Vishnu Mishra[5], Kelly Magnuson[6], Catherine Scholz[6] and Antonio Gualberto[6]

[1]Memorial Sloan Kettering Cancer Center, New York, NY USA, hoa@mskcc.org, [2]Dana Farber Cancer Institute, Boston, MA USA, [3]Vall D'Hebron Institute of Oncology, Barcelona, Spain, [4]Gustave Roussy, Villejuif, France, [5]Kura Oncology, Inc., San Diego, CA, USA, [6]Kura Oncology, Inc., Cambridge, MA, USA

BACKGROUND

Tipifarnib: Potent and highly selective inhibitor of farnesyl transferase (FT)

- All RAS isoforms (KRAS/NRAS/HRAS) are FT substrates.
- **HRAS is uniquely dependent upon farnesylation alone.**
- NRAS and KRAS are susceptible to redundant forms of prenylation and may lead to resistance to FT inhibition.

HRAS Mutations are Observed in an Unique Molecular Subset of HNSCC

HRAS mutations (~5% of tumors at diagnosis) are present in a distinctive molecular subset of HNSCC, characterized by: 1) low rate of genetic alterations, 2) less frequent TP53 mutation and 3) frequent CASP8 inactivation.

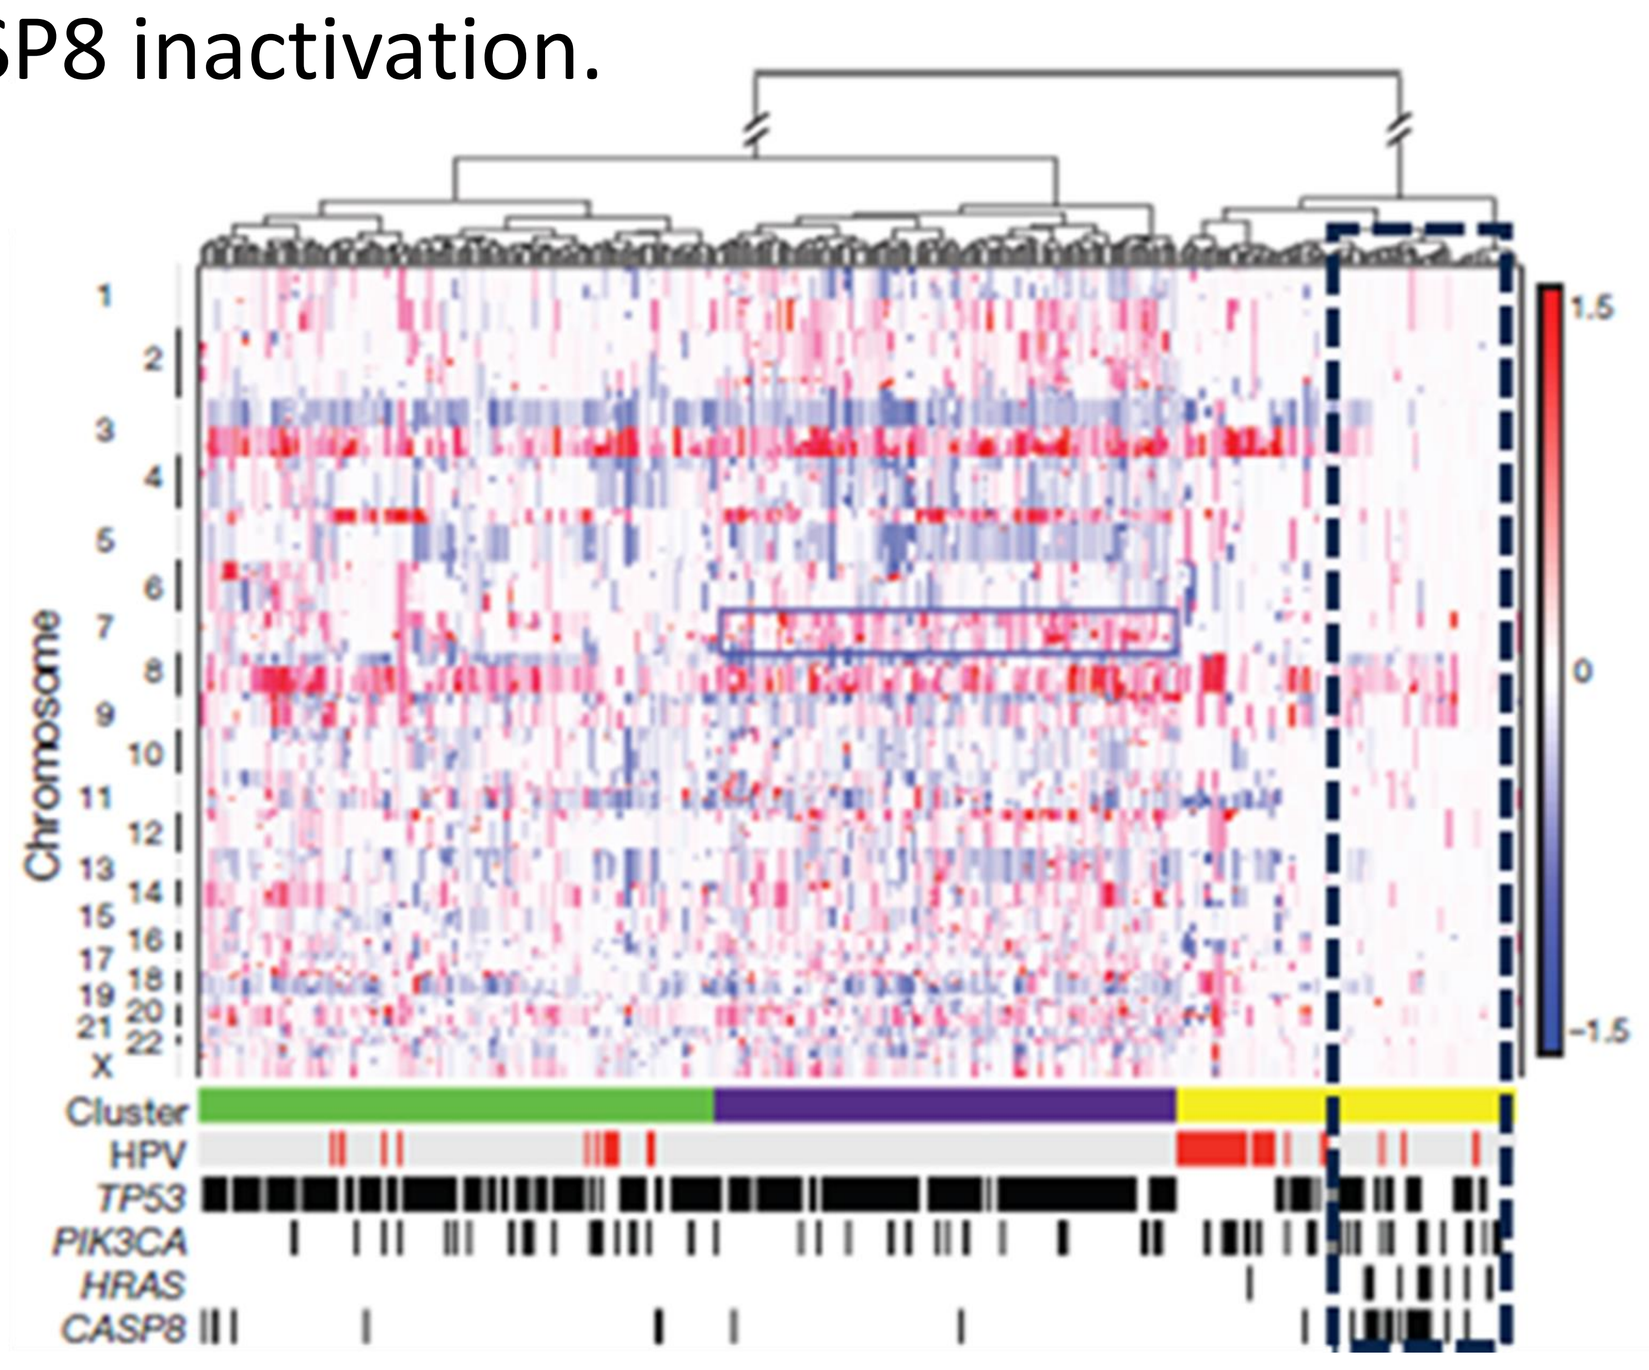

Chromosome
Cluster
HPV
TP53
PIK3CA
HRAS
CASP8
1.5
0
−1.5

Reprinted by permission from Macmillan Publishers Ltd: Nature 517:576-82, copyright 2015

Acquired HRAS Mutations Develop during 1st Line Therapy[1]

- Results suggest discovery of a ~15% rate of de novo HRAS mutation following cetuximab

[1] Braig *et al.*, *Oncotarget* 7:42988-42995, 2016

METHODS

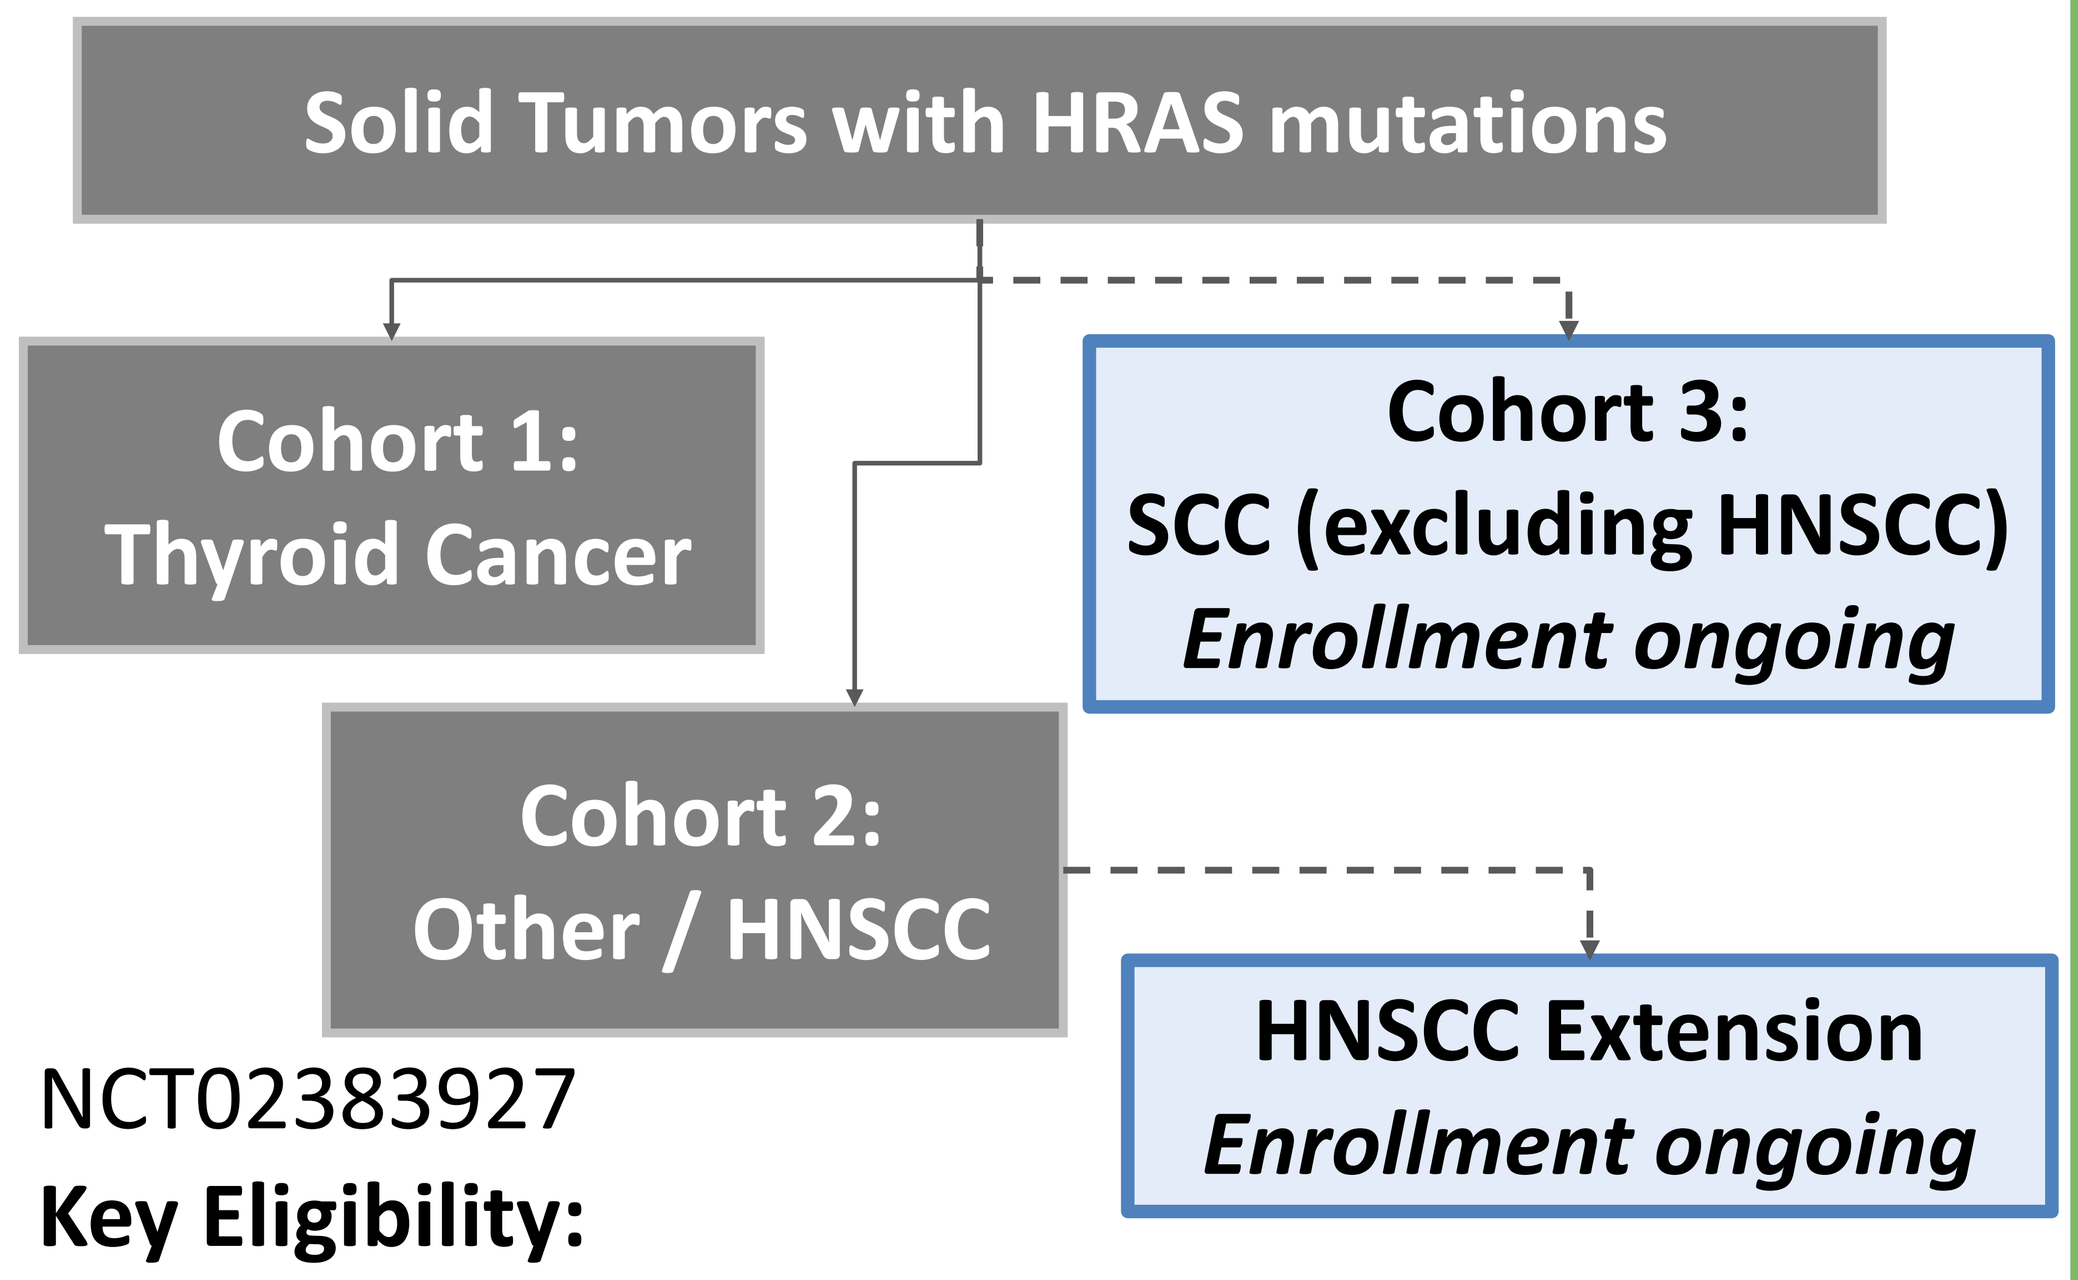

Solid Tumors with HRAS mutations
Cohort 1: Thyroid Cancer
Cohort 3: SCC (excluding HNSCC) Enrollment ongoing
Cohort 2: Other / HNSCC
HNSCC Extension Enrollment ongoing

NCT02383927

Key Eligibility:

- No curative therapy available
- HRAS mutation
- Measurable disease (RECIST v1.1)
- ECOG PS 0 – 1

Primary Objective: ORR

Design: Simon 2-stage (11+7)

Hypothesis: 10% (H0) vs 30% (H1) ORR, a=0.05, 80% power (4 responses needed)

Tipifarnib: 900 mg po bid on Days 1 – 7 and Days 15 – 21 of 28-day treatment cycles

For patient referral or clinical trial information: MEDICALAFFAIRS@KURAONCOLOGY.COM

RESULTS

HRAS mutant HNSCC patients received limited/no benefit from prior therapies, including chemotherapy, cetuximab and immunotherapy

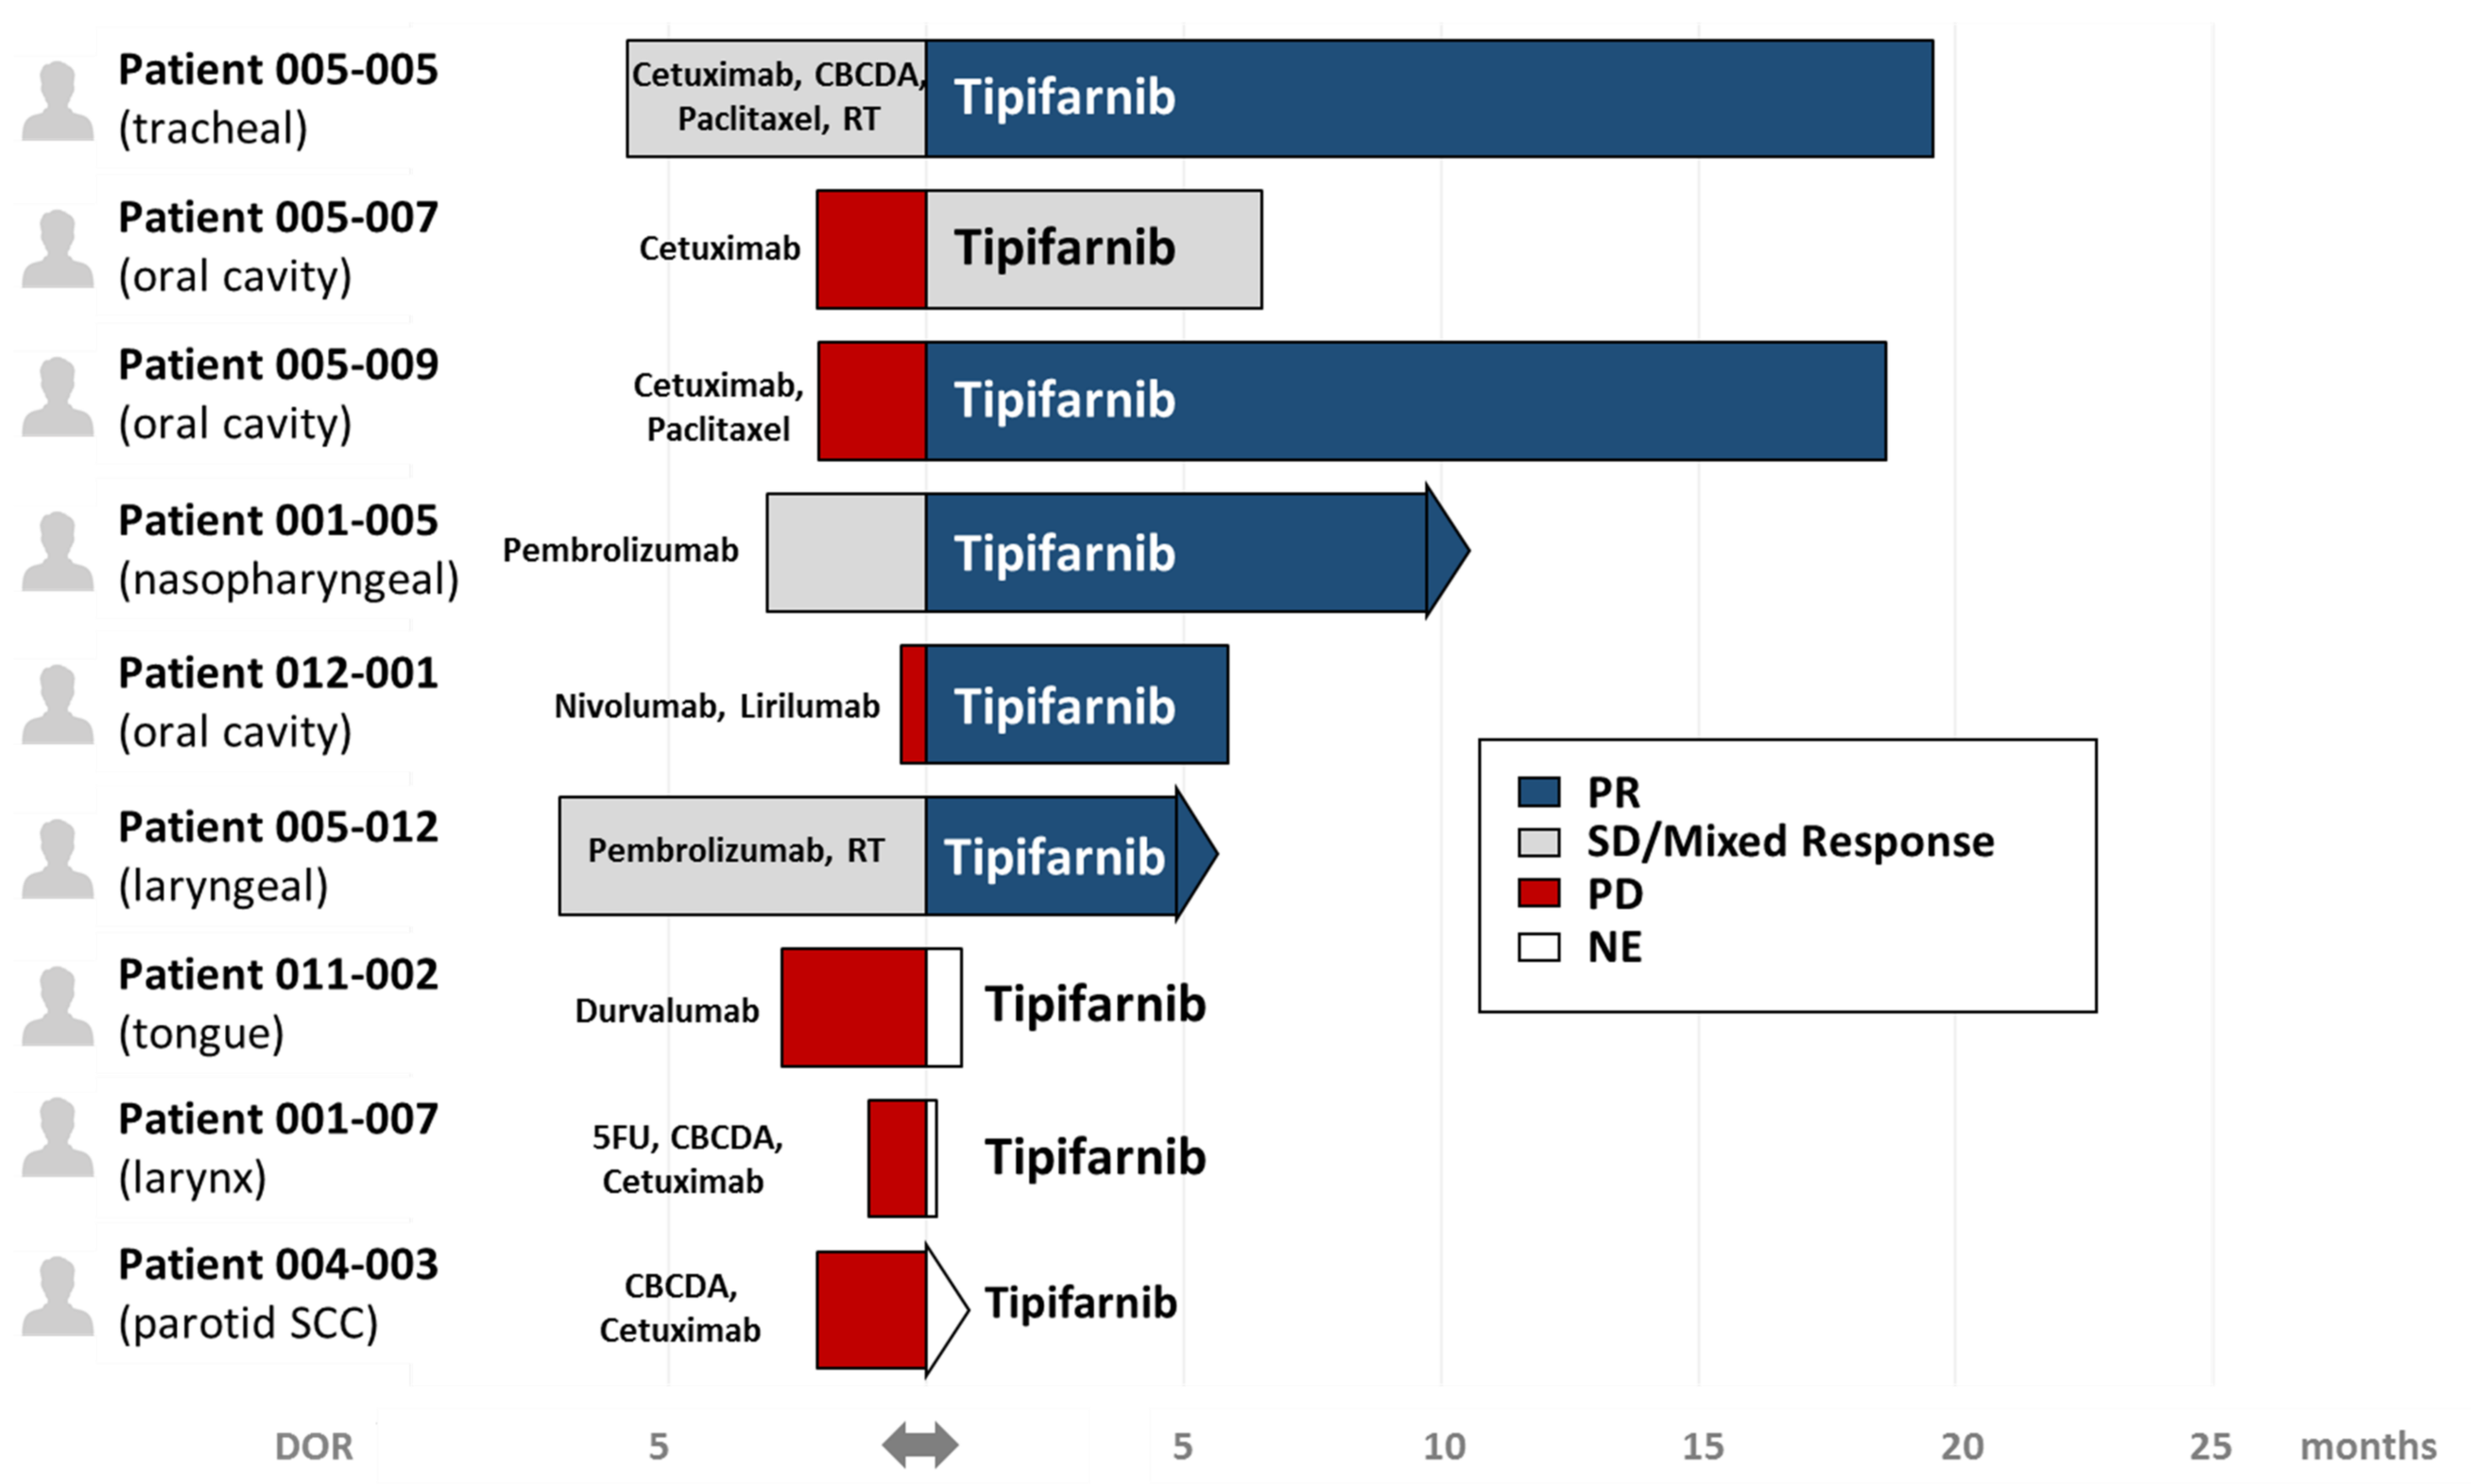

Patient 005-005 (tracheal)
Cetuximab, CBCDA, Paclitaxel, RT
Tipifarnib
Patient 005-007 (oral cavity)
Cetuximab
Tipifarnib
Patient 005-009 (oral cavity)
Cetuximab, Paclitaxel
Tipifarnib
Patient 001-005 (nasopharyngeal)
Pembrolizumab
Tipifarnib
Patient 012-001 (oral cavity)
Nivolumab, Lirilumab
Tipifarnib
Patient 005-012 (laryngeal)
Pembrolizumab, RT
Tipifarnib
Patient 011-002 (tongue)
Durvalumab
Tipifarnib
Patient 001-007 (larynx)
5FU, CBCDA, Cetuximab
Tipifarnib
Patient 004-003 (parotid SCC)
CBCDA, Cetuximab
Tipifarnib
PR
SD/Mixed Response
PD
NE
DOR
5
5
10
15
20
25
months

PR: partial response, SD: stable disease, PD: progressive disease, NE: not evaluable, CBCDA: carboplatin, RT: radiotherapy, 5FU: fluorouracil

Results based on preliminary data as of 08 February 2018.

Patient Disposition

Total HRASm HNSCC Treated	n (%)	9 (100)
Larynx	n	2
Oral Cavity	n	4
Pharynx (Nasopharyngeal)	n	1
Other: Trachea, Parotid	n	2
Prior Lines of Therapy	Median (Range)	2 (1 – 4)
Age, yrs	Median (Range)	58 (20 – 76)
Total Discontinuations	n (%)	6 (67)
Progressive Disease	n (%)	4 (44)
Withdrawal of Consent	n (%)	1 (11)
Early Death, unrelated	n (%)	1 (11)
Total Efficacy Evaluable	n	6
Not evaluable by RECIST†	n	3

†011-002 (early death, unrelated); 001-007 (consent withdrawal); 004-003 (too early for disease assessment)

Overall Response Rate (ORR)		83% (36-99.6%, 95%CI)
Overall Best Response		
Confirmed Partial Response	**n (%)**	**5 (83)**
Stable Disease	**n (%)**	**1 (17)**
Progressive Disease	**n (%)**	**0 (0)**

Safety & Tolerability

Tipifarnib was generally well-tolerated.

- AEs observed are consistent with the known safety profile of tipifarnib
- Most common treatment-related AEs (grade ≥ 3) were anemia (33.3%), nausea (22.2%), neutropenia, vomiting and decreased appetite (11.1% each).
- 2 HNSCC patients required dose reductions for Gr 2 peripheral neuropathy at Cycle 1 and Cycle 10. 1 HNSCC patient initiated treatment at 600 mg.

Reduction in target lesions observed in HRASm HNSCC pts treated with Tipifarnib

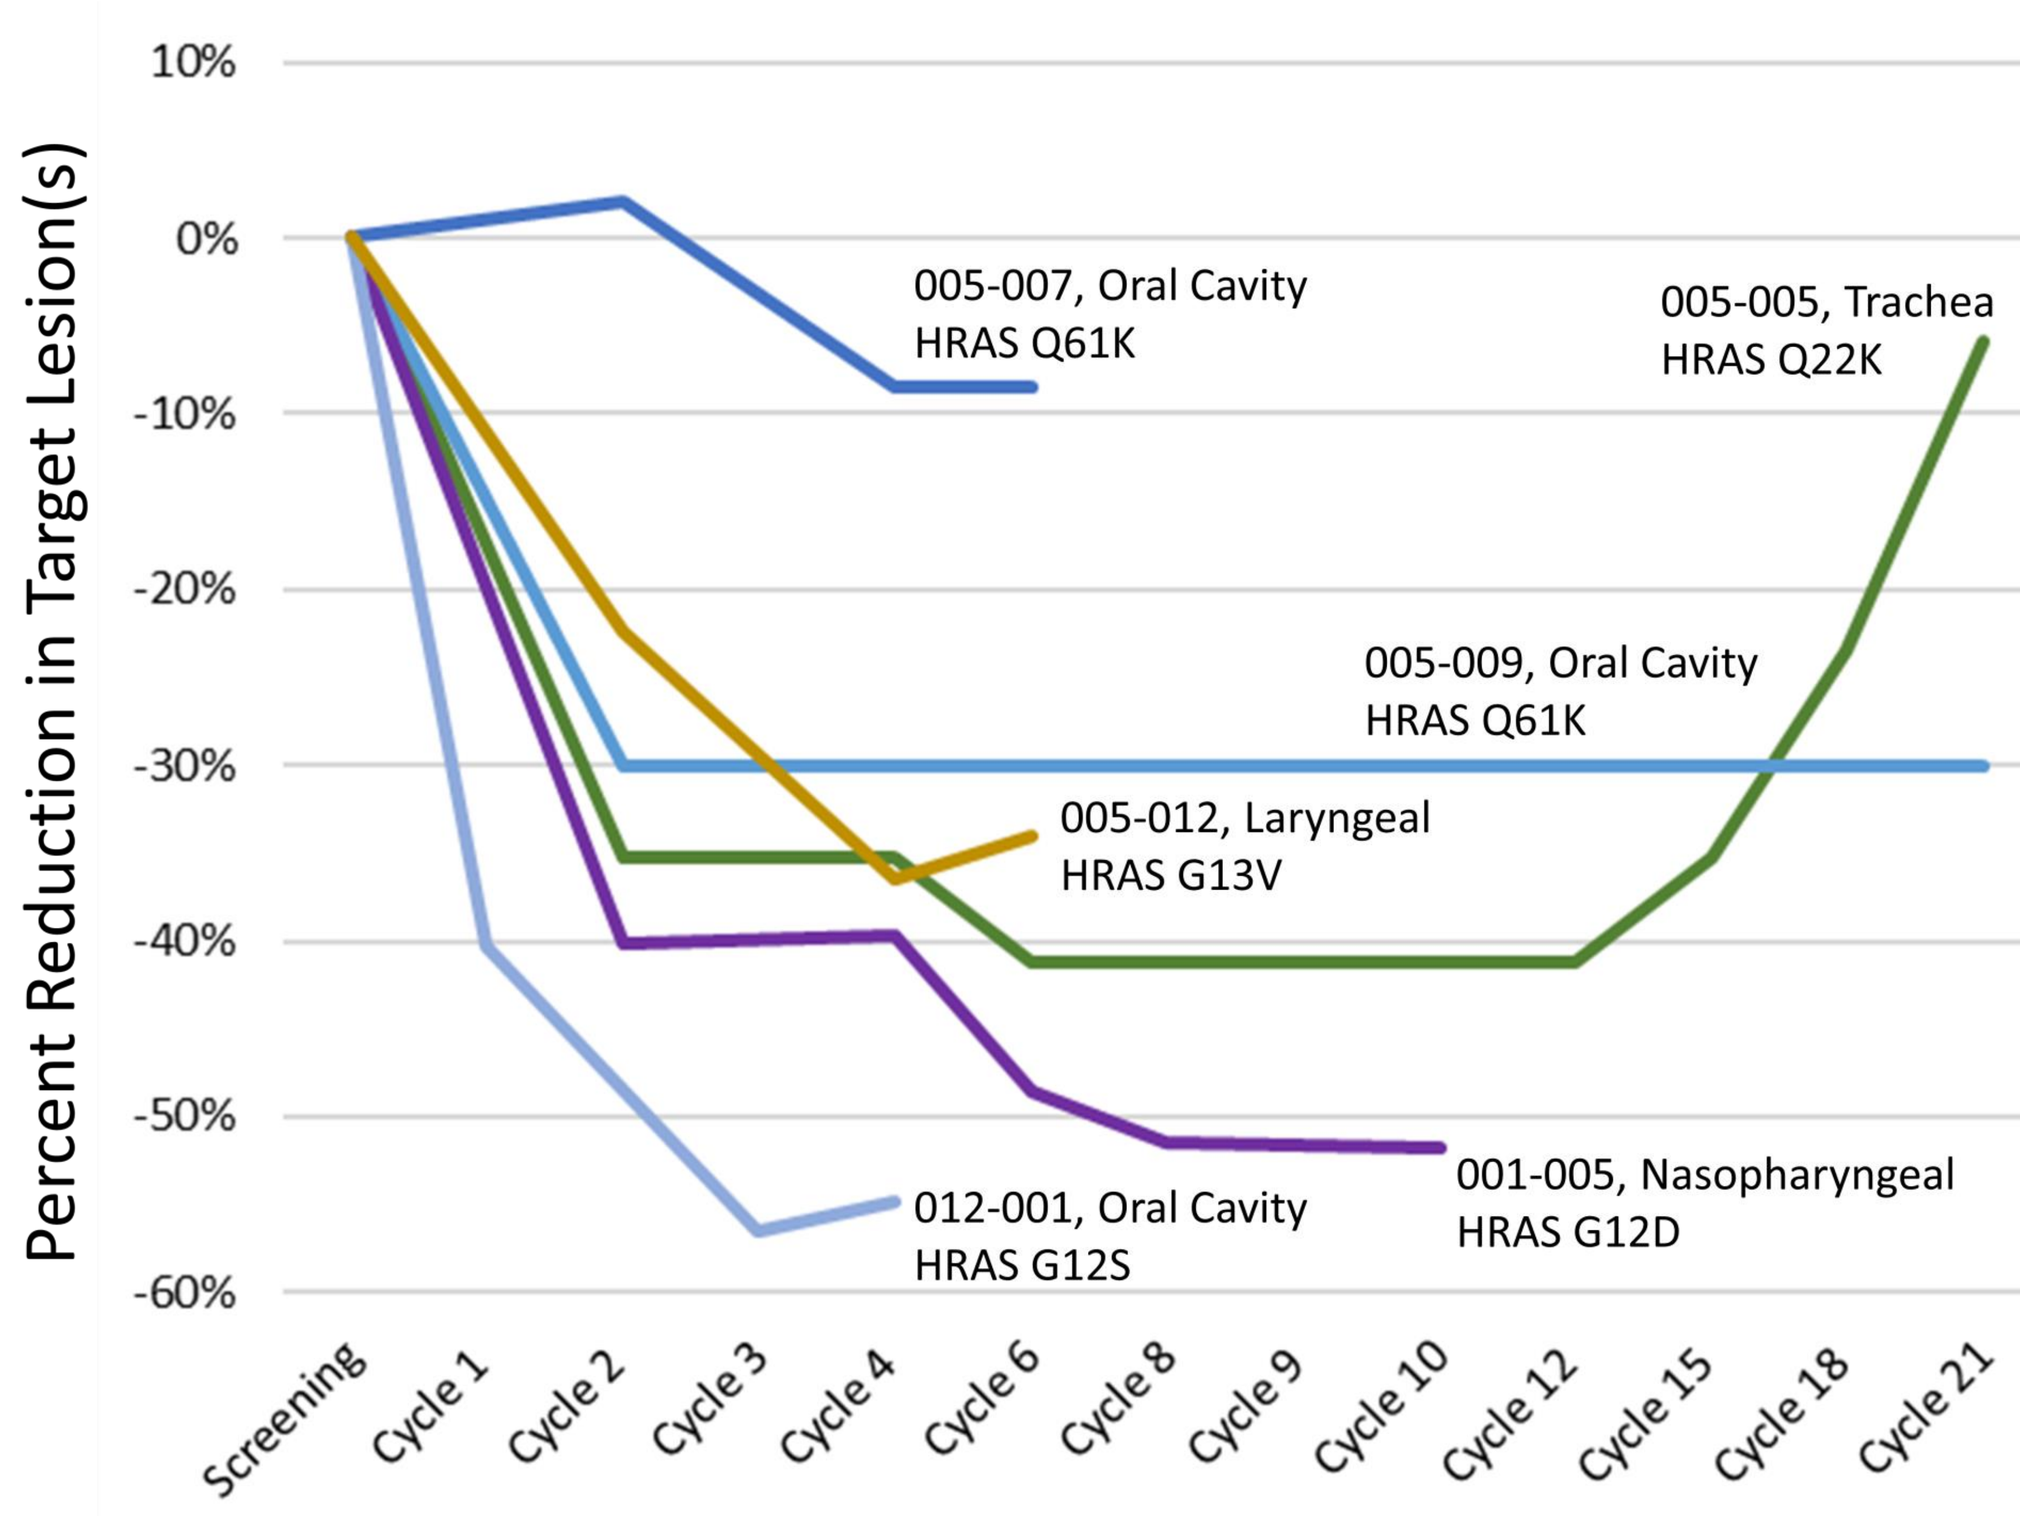

Percent Reduction in Target Lesion(s)
10%
0%
-10%
-20%
-30%
-40%
-50%
-60%
005-007, Oral Cavity HRAS Q61K
005-005, Trachea HRAS Q22K
005-009, Oral Cavity HRAS Q61K
005-012, Laryngeal HRAS G13V
012-001, Oral Cavity HRAS G12S
001-005, Nasopharyngeal HRAS G12D
Screening
Cycle 1
Cycle 2
Cycle 3
Cycle 4
Cycle 6
Cycle 8
Cycle 9
Cycle 10
Cycle 12
Cycle 15
Cycle 18
Cycle 21

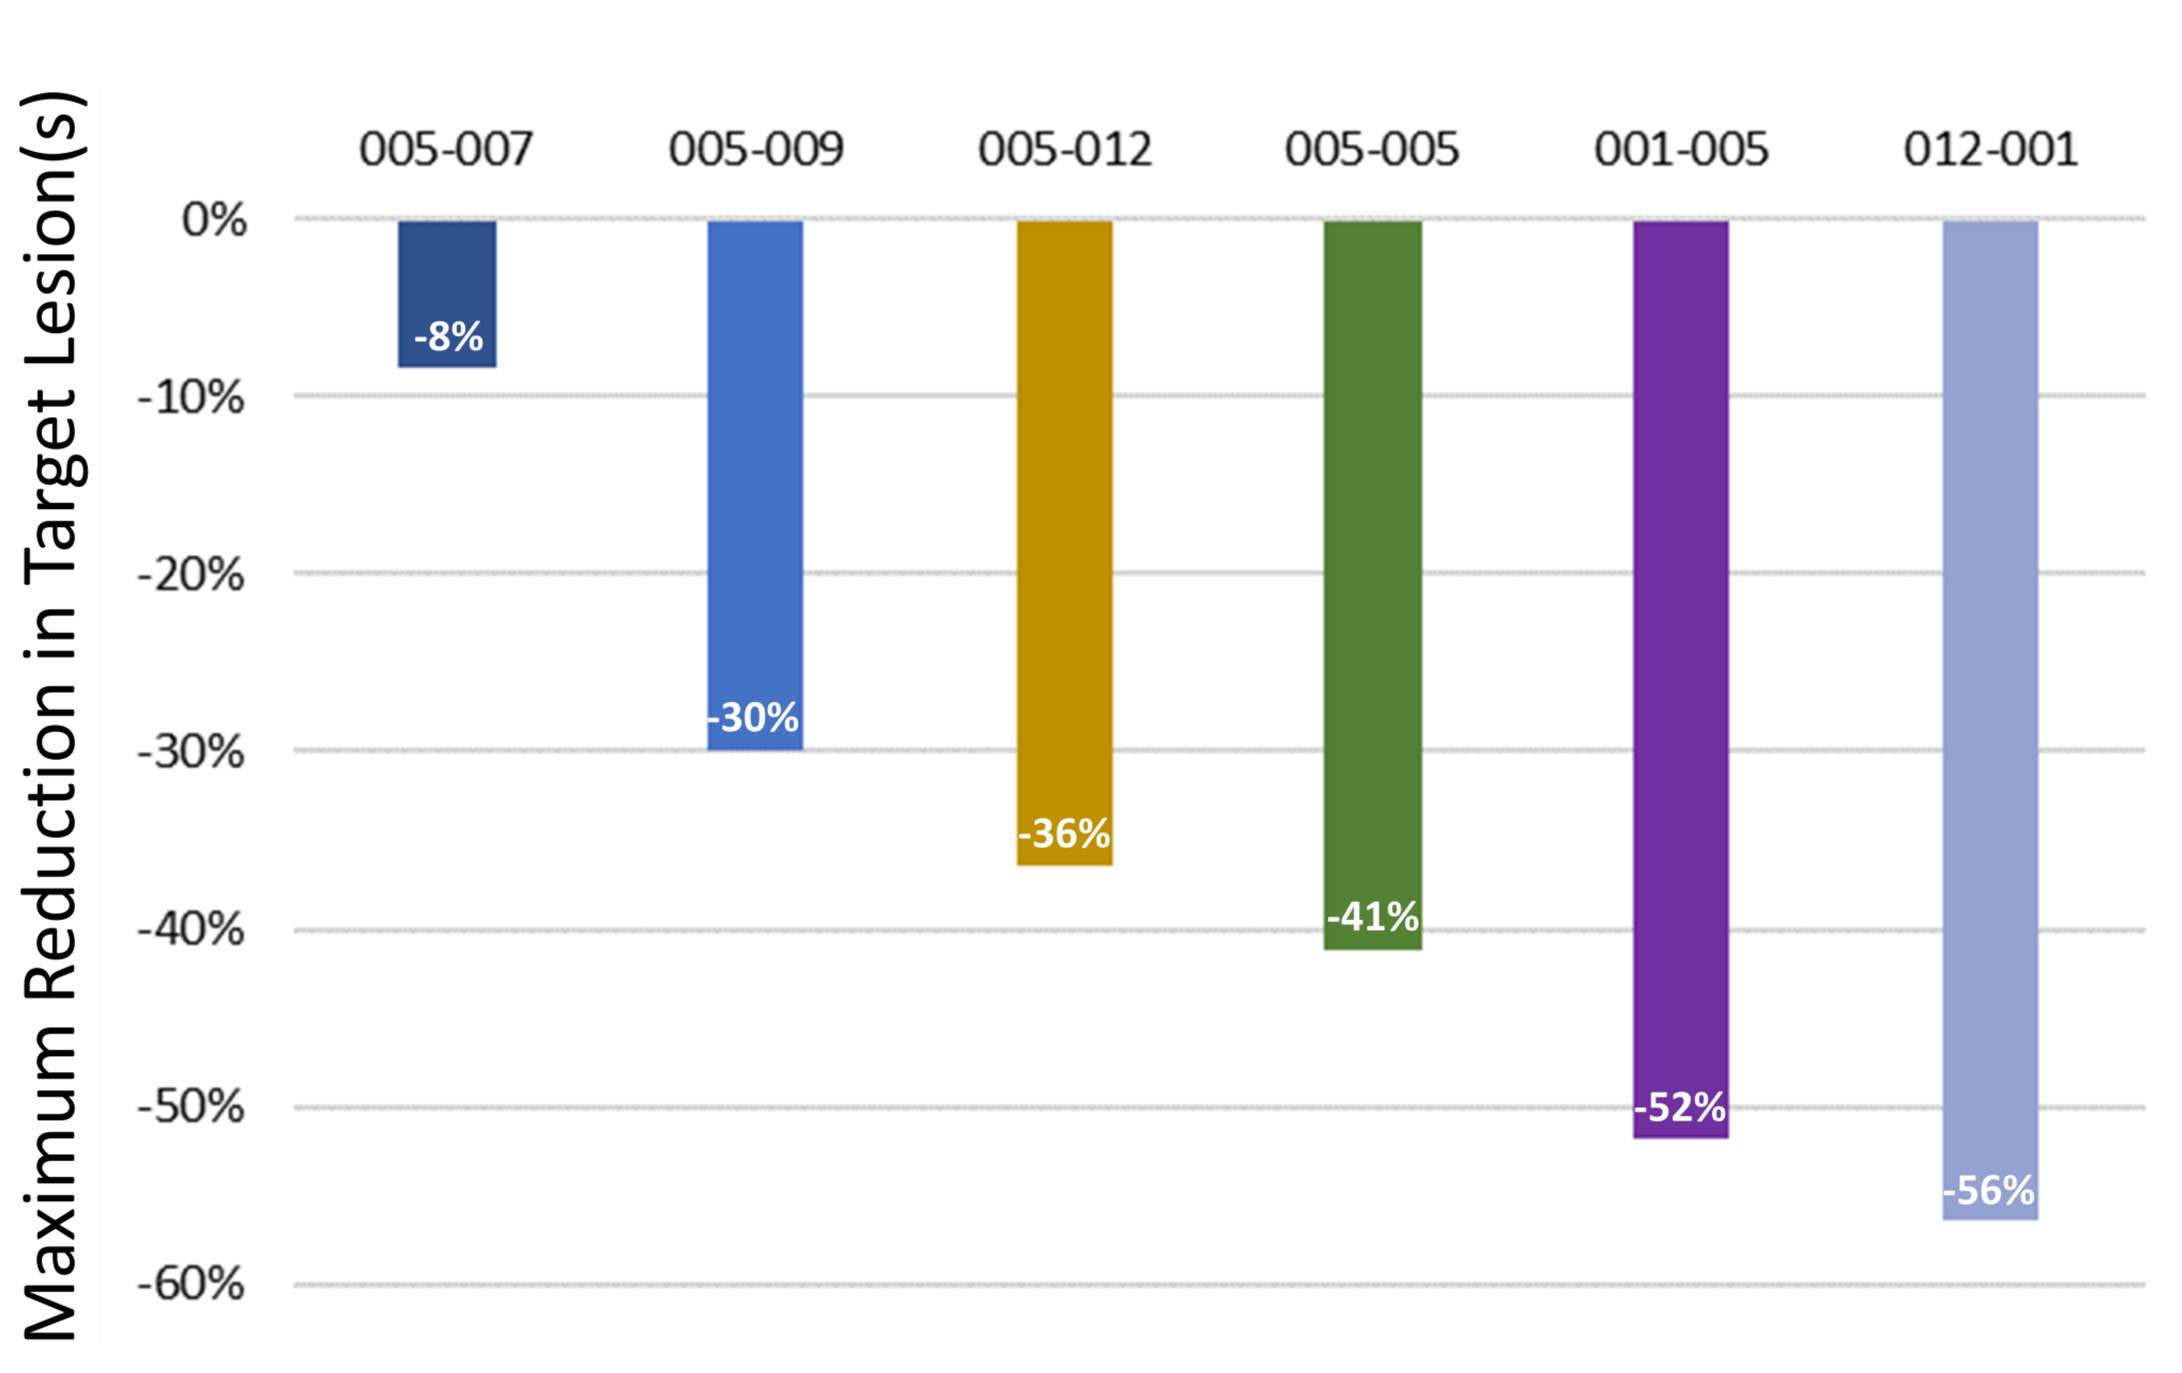

Maximum Reduction in Target Lesion(s)
005-007
005-009
005-012
005-005
001-005
012-001
0%
-10%
-20%
-30%
-40%
-50%
-60%
-8%
-30%
-36%
-41%
-52%
-56%

Tipifarnib Regression of Widely Metastatic Laryngeal SCC

- 55 yo male with metastatic laryngeal SCC (005-012, mediastinal LNs, muscle, adrenal gland, lung, bone)
- Prior therapies: cetuximab + carboplatin + paclitaxel and pembrolizumab + RT
- Molecular status: HRAS G13V, TP53 R248Q

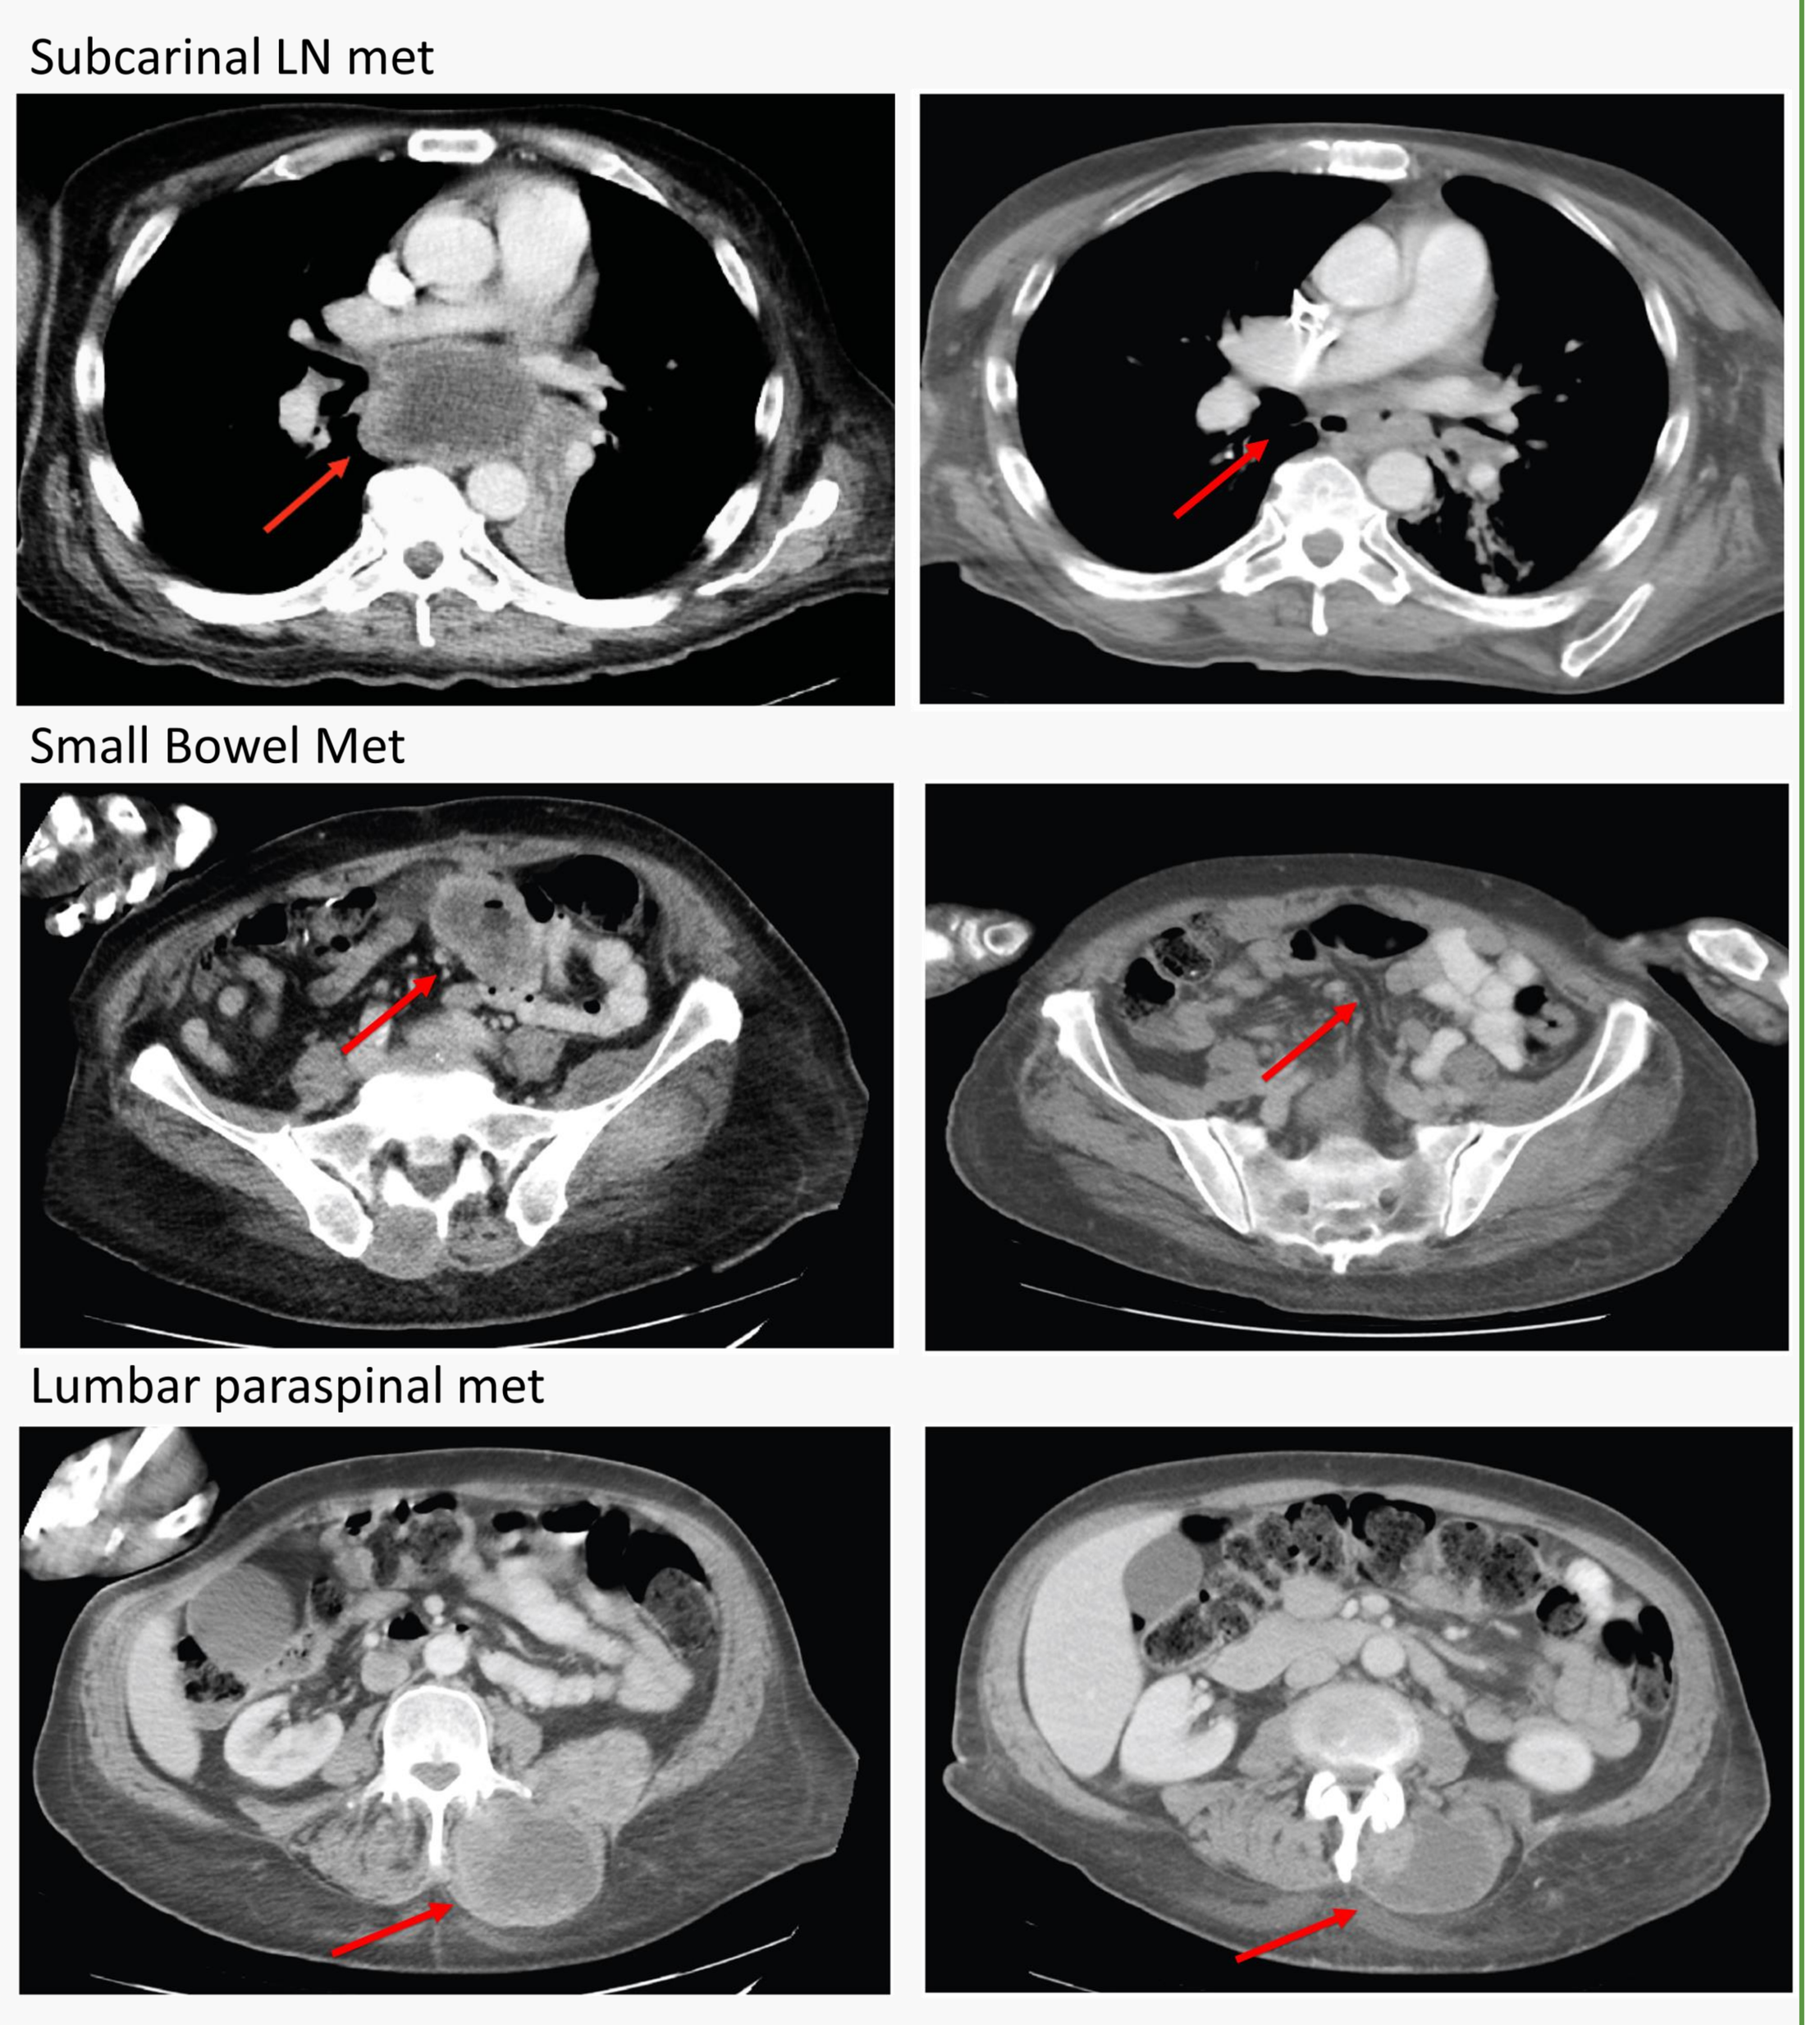

Subcarinal LN met
Small Bowel Met
Lumbar paraspinal met

Baseline **Cycle 6, 34% reduction**

Tipifarnib Response and Resolution of Disfiguring Skin Lesions s/p Immunotherapy Failure

- 69 yo man with recurrent oral cavity SCC (012-001)
- Prior therapies: TPEx (Docetaxel, CDDP, Cetuximab), nivolumab + lirilumab
- Molecular status: HRAS G12S, TP53 R248Q

Initial PR (40% tumor reduction) on Cycle 1 Day 15 (7 days tipifarnib + 7 days rest); 56% size reduction at Cycle 3

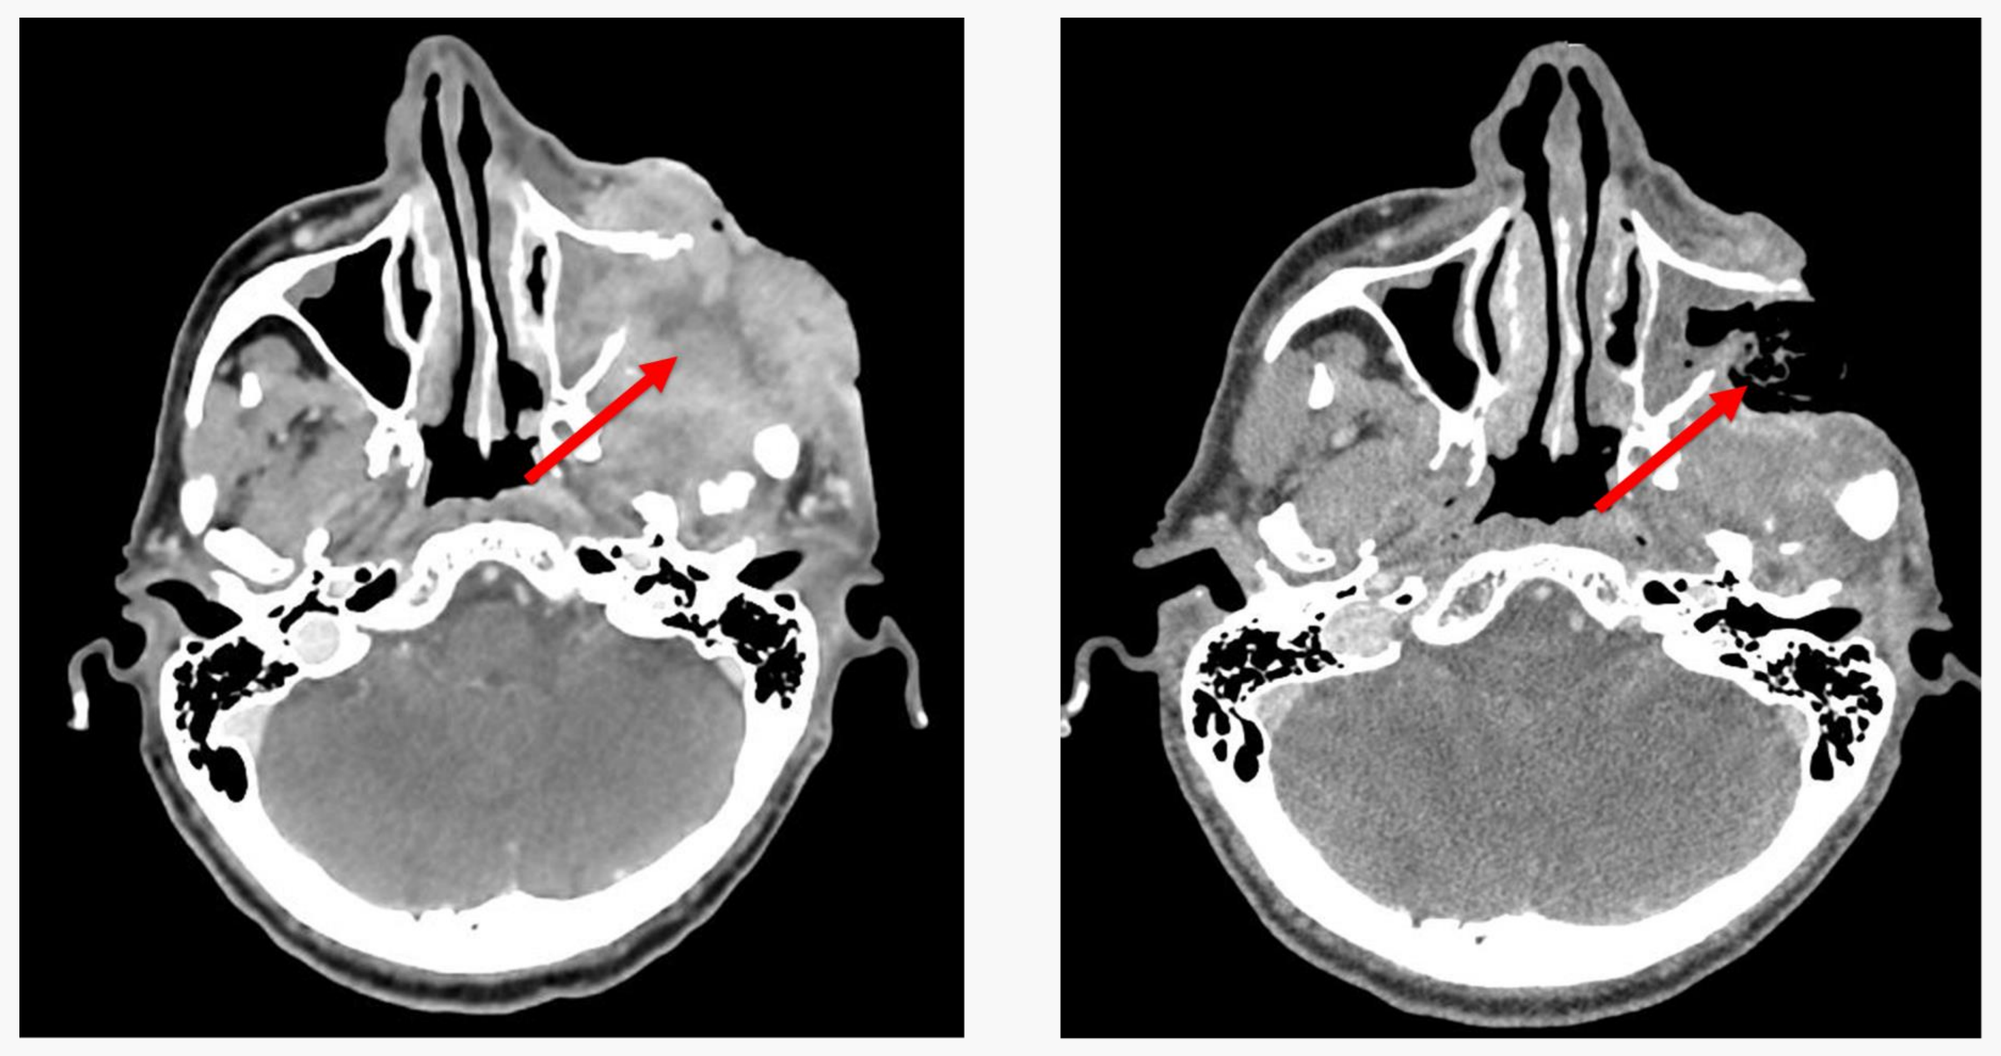

Baseline **Cycle 3, 56% reduction**

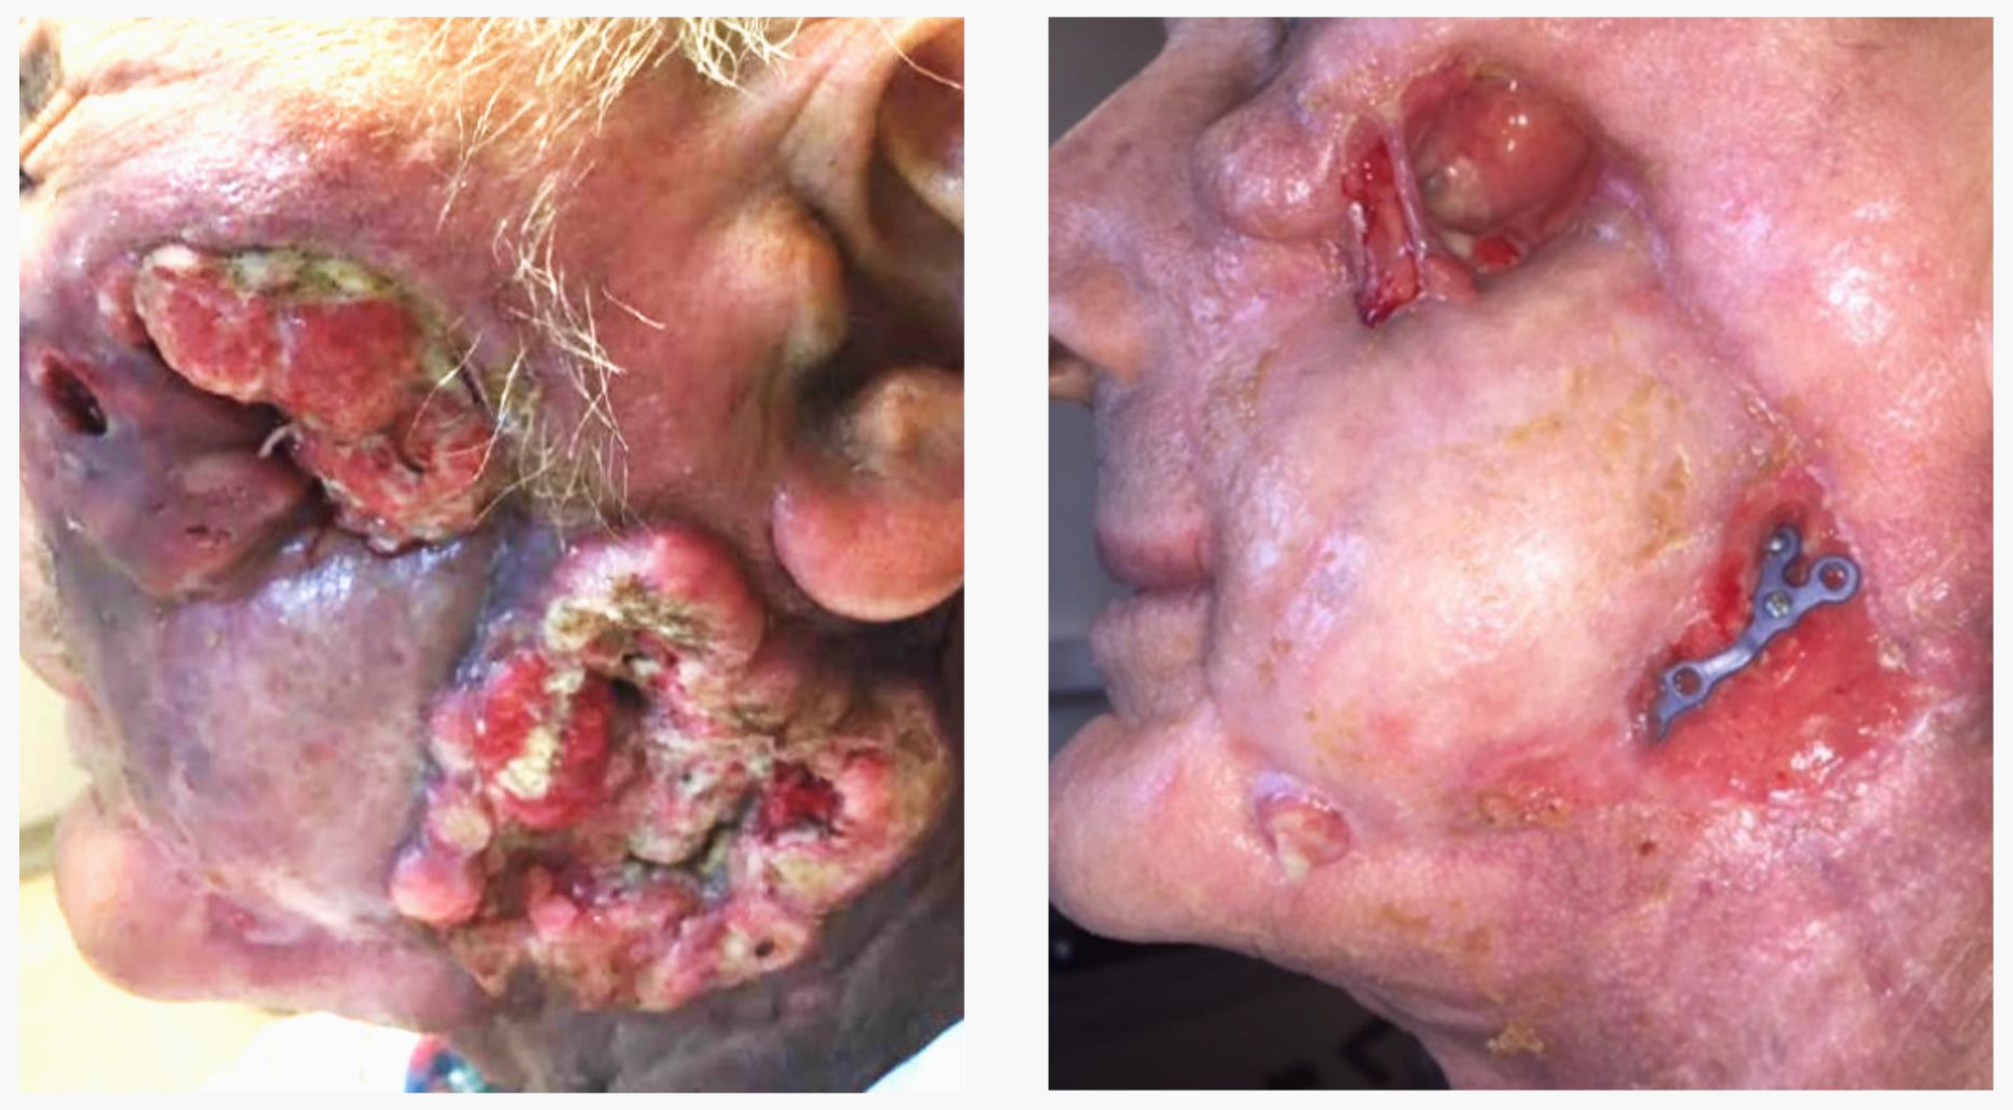

Cycle 1 Day 1 **Cycle 5 Day 1**

CONCLUSIONS

First clinical evidence that mutant HRAS is a targetable oncogene.

- Responses observed in patients with hotspot and non-hotspot mutations.

Phase 2 proof-of-concept of tipifarnib efficacy for recurrent / metastatic HNSCC carrying HRAS mutations.

- Confirmed PRs in 5 of 6 HNSCC patients (83%, 36-99.6% 95% CI)
- Rapid and durable responses (2 responses > 1.5 year)
- Activity in disease resistant to chemotherapy, cetuximab and immunotherapy
- Resolution of disfiguring lesions