C8

Farnesyl transferase inhibitor (FTI) darlifarnib (KO-2806) combined with cabozantinib (cabo) in clear cell renal cell carcinoma (ccRCC) patients after prior exposure to cabo: Preliminary phase 1 results from FIT-001

Yousef Zakharia[1], Adam E. Singer[2], Benjamin Garmezy[3], Jacob Thomas[4], Douglas E. Laux[5], Jason Henry[6], Liza Villaruz[7], Sanjay Goel[8], Paria Mahboub Johnson[9], Jenchun Kuan[10], Binaifer Balsara[9], Mollie Leoni[9], Adanma Ayanambakkam[11]

[1]Mayo Clinic Comprehensive Cancer Center, Phoenix, AZ, USA; [2]Division of Hematology-Oncology, University of California Los Angeles Medical Center, Los Angeles, CA, USA; [3]Sarah Cannon Research Institute, Nashville, TN, USA; [4]Division of Medical Oncology, Department of Medicine, University of Southern California Norris Comprehensive Cancer Center, Los Angeles, CA, USA; [5]Division of Hematology, Oncology and Blood & Marrow Transplantation, University of Iowa Hospitals and Clinics, Iowa City, IA, USA; [6]Sarah Cannon Research Institute at Health One, Denver, CO, USA; [7]UPMC Hillman Cancer Center, Pittsburgh, PA, USA; [8]Rutgers Cancer Institute of New Jersey, Brunswick, NJ, USA; [9]Clinical Development, Kura Oncology, Inc., San Diego, CA, USA; [10]Biometrics, Kura Oncology, Inc., San Diego, CA, USA; [11]Stephenson Cancer Center, University of Oklahoma Health Sciences Center, Oklahoma City, OK, USA

BACKGROUND

- Innate and adaptive resistance to VEGFR-targeted therapies in RCC, often mediated through activation of the mTOR pathway, highlights the need for alternative therapeutic options to improve patient outcomes
- Cabozantinib (cabo) is a standard RCC treatment that has demonstrated consistent clinical activity; however, adaptive resistance often leads to disease progression[1,2]
 - ORR with subsequent treatment after prior cabo or other TKI exposure ranges from ~17%–22%[3–5]
- **Darlifarnib (KO-2806)** – a potent, next-generation farnesyl transferase inhibitor – has been shown to enhance the therapeutic potential of cabo in preclinical models[6], and may address resistance mechanisms
 - Darlifarnib blocks farnesylation of RHEB, suppressing RHEB-mediated compensatory mTORC1 pathway activation
- In preclinical studies, darlifarnib in combination with cabo provides deep and durable responses in RCC xenograft models, and resensitizes tumors that have previously progressed on cabo and other TKIs by enhancing antiangiogenic activity[6,7]
- Preliminary clinical data in ccRCC demonstrate the potential of the darlifarnib + cabo combination[8]

AIM

- Here we report safety in RCC patients, and preliminary antitumor activity of the combination in prior cabo-exposed ccRCC patients, from the phase 1a portion of the FIT-001 study

METHODS

- FIT-001 (NCT06026410) is an ongoing first-in-human, multicenter, open-label, phase 1a/b dose-escalation/-expansion study of darlifarnib alone and in combination in patients with advanced solid tumors (**Figure 1**), including:
 - Darlifarnib + cabo combination in patients with locally advanced or metastatic ccRCC
- Darlifarnib 3, 5, or 8 mg was administered orally QD on Days 1–7 and 15–21 plus continuous cabo 40 mg or 60 mg orally QD in 28-day cycles
- Patients must have received ≥1 prior IO-based treatment
- Primary objective: safety
- Secondary objectives: antitumor activity, pharmacokinetics

Figure 1. FIT-001 study design

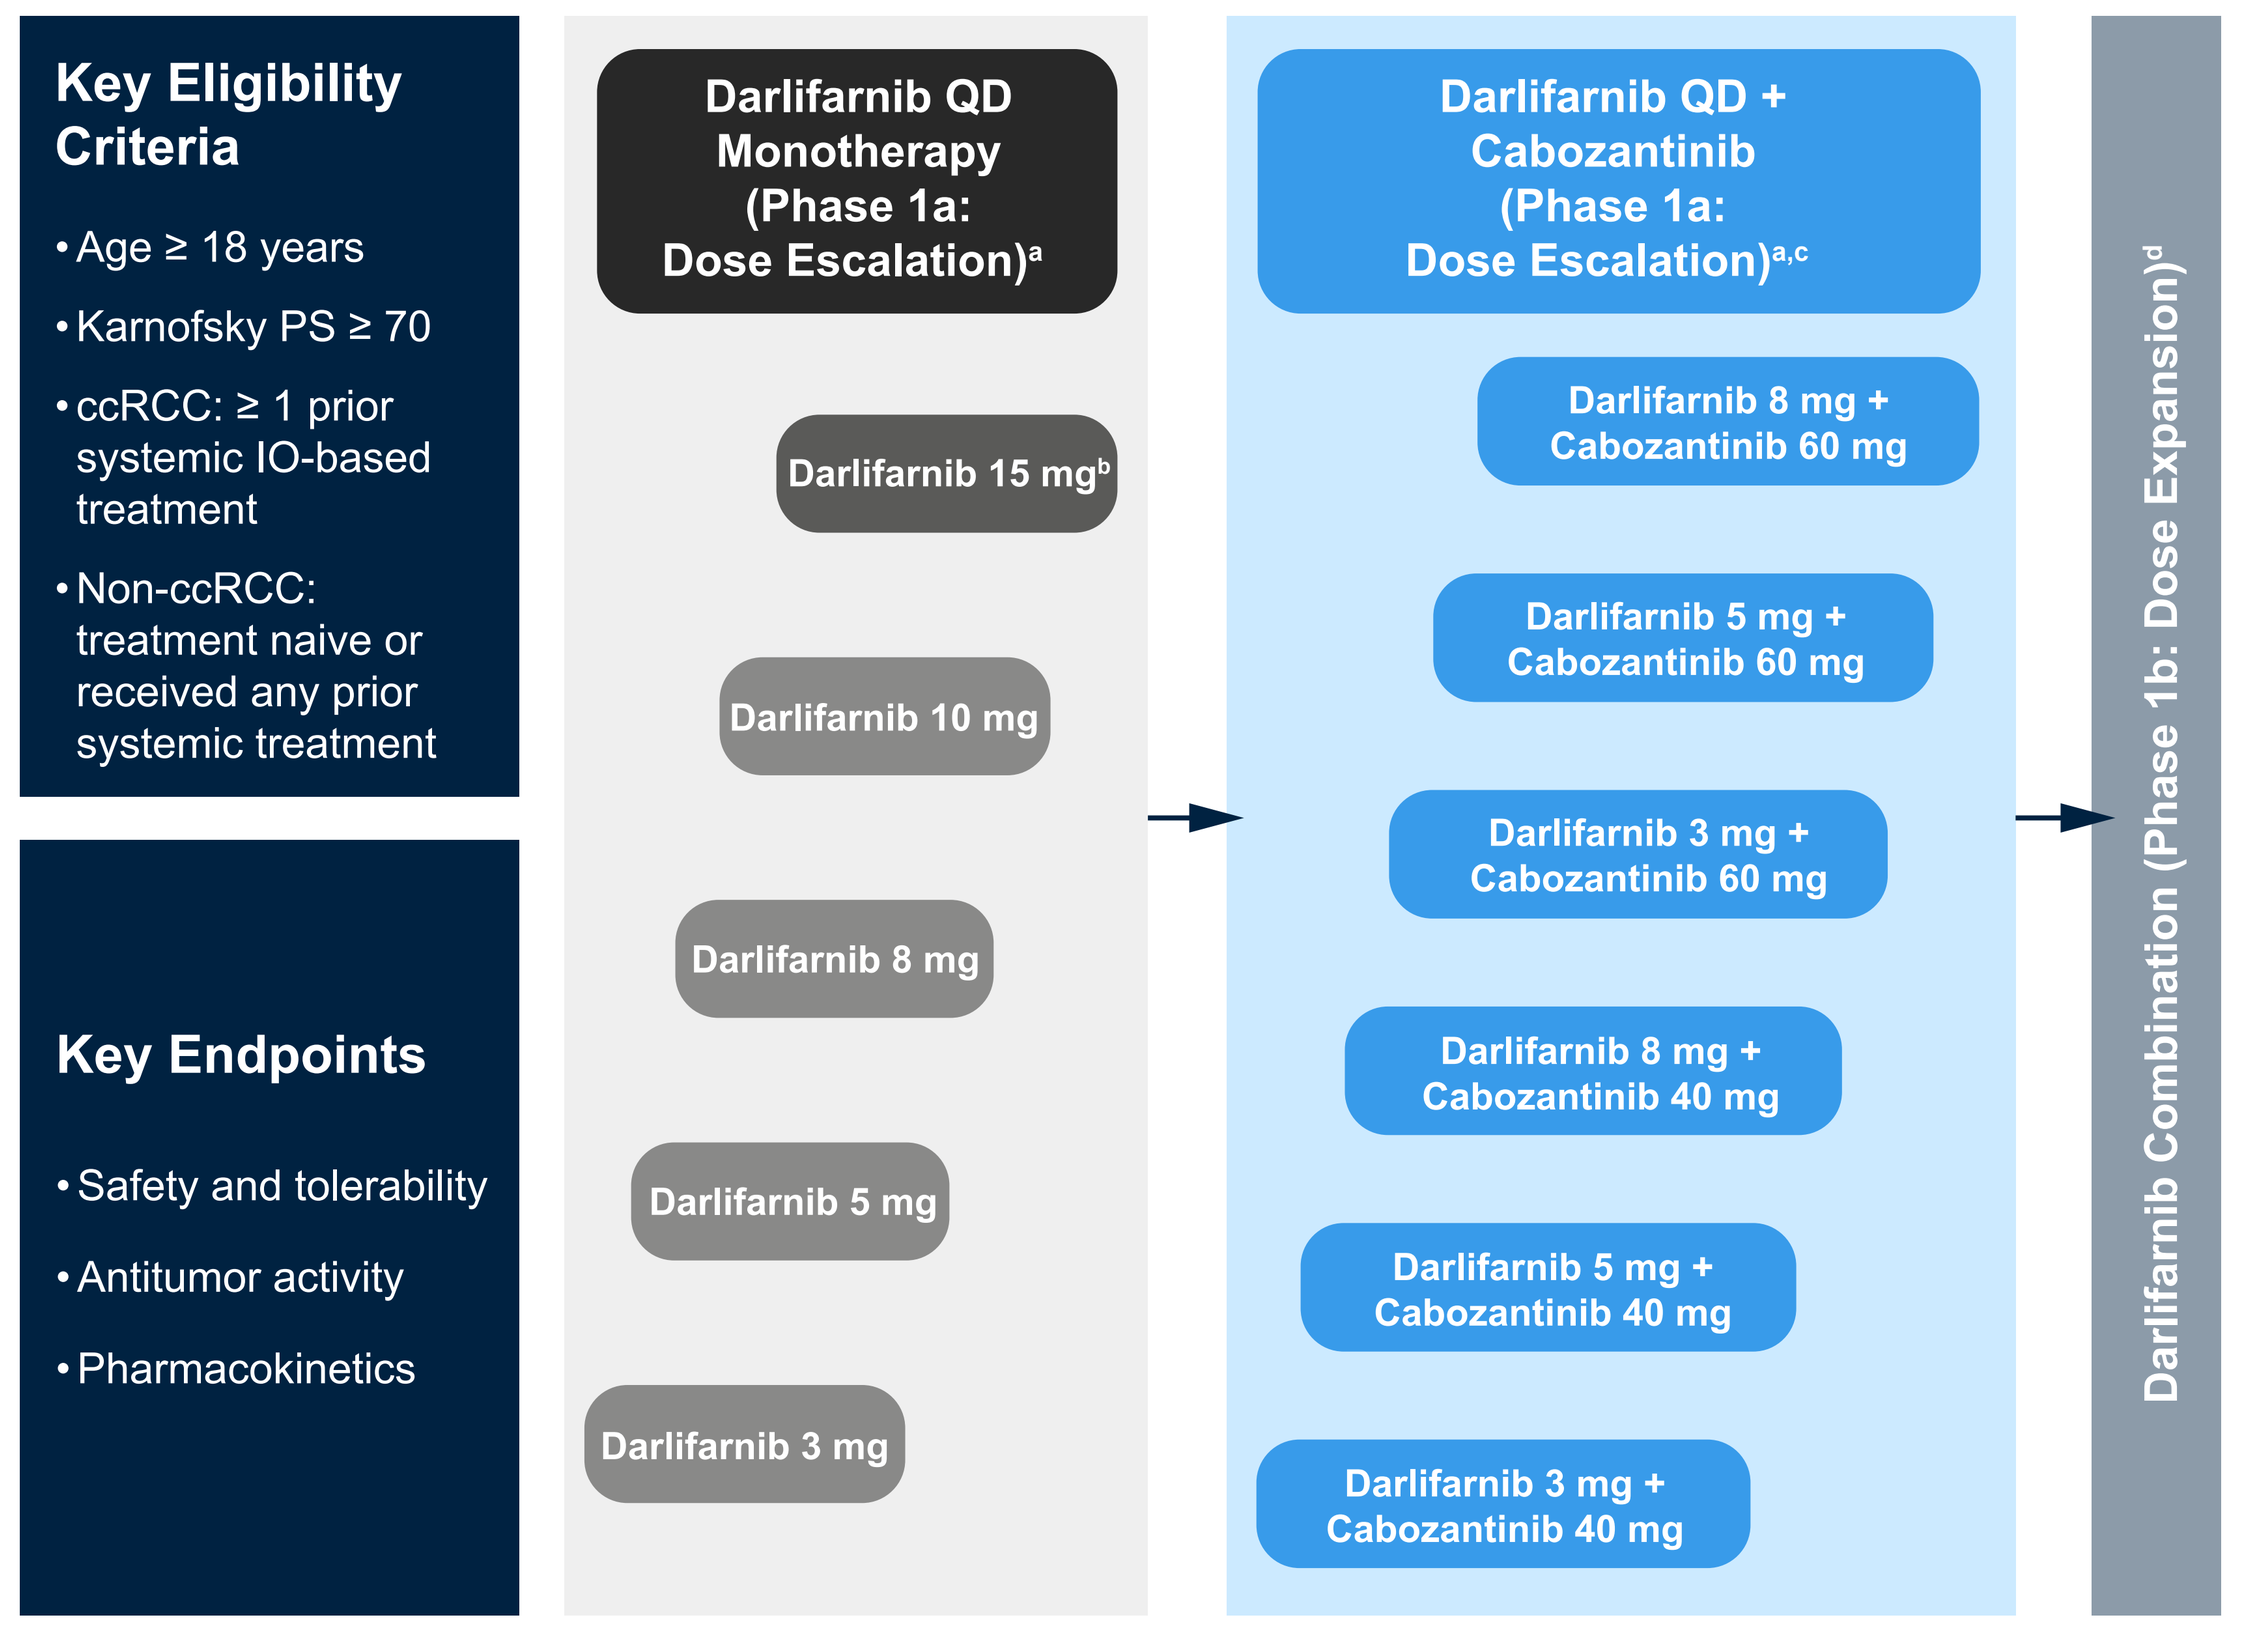

Key Eligibility Criteria
• Age ≥ 18 years
• Karnofsky PS ≥ 70
• ccRCC: ≥ 1 prior systemic IO-based treatment
• Non-ccRCC: treatment naive or received any prior systemic treatment
Key Endpoints
• Safety and tolerability
• Antitumor activity
• Pharmacokinetics
Darlifarnib QD Monotherapy (Phase 1a: Dose Escalation)a
Darlifarnib 15 mgb
Darlifarnib 10 mg
Darlifarnib 8 mg
Darlifarnib 5 mg
Darlifarnib 3 mg
Darlifarnib QD + Cabozantinib (Phase 1a: Dose Escalation)a,c
Darlifarnib 8 mg + Cabozantinib 60 mg
Darlifarnib 5 mg + Cabozantinib 60 mg
Darlifarnib 3 mg + Cabozantinib 60 mg
Darlifarnib 8 mg + Cabozantinib 40 mg
Darlifarnib 5 mg + Cabozantinib 40 mg
Darlifarnib 3 mg + Cabozantinib 40 mg
Darlifarnib Combination (Phase 1b: Dose Expansion)d

[a]Each individual patient will receive one of the planned DLs of darlifarnib. [b]Non-tolerated DL. [c]n=12 patients per dose level. [d]Up to 2 potential darlifarnib RP2Ds may be further evaluated in phase 1b dose expansion in combination with cabo.
cc, clear cell; DL, dose level; IO, immune checkpoint inhibition; PS, performance status; QD, once daily; RCC, renal cell carcinoma.

RESULTS

- As of Dec 8, 2025, 18 patients with cabo-exposed ccRCC were enrolled and treated with darlifarnib + cabo
- More than half of patients (56%; 10/18) received cabo as the immediate prior line (**Table 1**)
- Most patients (67%; 12/18) also had prior exposure to other TKIs
- More than half of patients (56%; 10/18) stopped prior cabo due to disease progression

Table 1. Baseline characteristics in cabo-exposed ccRCC patients

n (%)	Cabozantinib 40 mg: Darlifarnib 3 mg n=6	Cabozantinib 40 mg: Darlifarnib 5 mg n=6	Cabozantinib 40 mg: Darlifarnib 8 mg n=3	Cabozantinib 60 mg[c]: Darlifarnib 3 mg n=3	Total N=18
Median age, years (range)	63 (54–82)	66 (54–79)	56 (48–80)	70 (66–79)	67 (48–82)
Male	3 (50)	4 (67)	3 (100)	2 (67)	12 (67)
Race					
White	4 (67)	3 (50)	2 (67)	3 (100)	12 (67)
Non-White[a]	2 (33)	3 (50)	1 (33)	0	6 (33)
Karnofsky PS					
50–70	0	0	0	0	0
80–100	6 (100)	6 (100)	3 (100)	3 (100)	18 (100)
Distant metastasis[b]	6 (100)	6 (100)	3 (100)	2 (67)	17 (94)
Prior lines					
1	1 (17)	1 (17)	1 (33)	0	3 (17)
2	2 (33)	2 (33)	0	2 (67)	6 (33)
≥3	3 (50)	3 (50)	2 (67)	1 (33)	9 (50)
Prior therapy type(s)					
Cabo (immediate prior line)	3 (50)	3 (50)	2 (67)	2 (67)	10 (56)
Other TKI (any prior line)	3 (50)	4 (67)	2 (67)	3 (100)	12 (67)

[a]Includes Black or African American, Asian, American Indian or Alaska Native, Other, and Multiple. [b]Most common sites of metastases were lung, bone, liver, and lymph nodes. [c]Darlifarnib 5 and 8 mg + cabo 60 mg cohorts enrolled cabo-naive patients only.
Cabo, cabozantinib; ccRCC, clear cell renal cell carcinoma; PS, performance status; TKI, tyrosine kinase inhibitor.
Data cutoff: Dec 8, 2025.

Safety and tolerability in all RCC patients (N=70)

- Darlifarnib + cabo combination has exhibited a manageable safety profile across all dose levels in RCC patients (**Table 2**), including in cabo-exposed ccRCC patients
- An overall DLT rate of 6% was observed
- Neutropenia was the most commonly-reported darlifarnib-related grade ≥3 AE (n=21, 30%)
- While darlifarnib dose interruptions were more frequent with darlifarnib + cabo 60 mg versus 40 mg, very few patients required treatment discontinuation (6%; 4/70) or dose reduction (13%; 9/70), allowing patients to remain on therapy (**Table 3**)
 - Safety profile, including dose modifications, of cabo in this combination was generally consistent with that observed with cabo monotherapy[4,9]

Table 2. Treatment-related adverse events in all RCC patients[a] (N=70)

n (%)	Cabozantinib 40 mg: Darlifarnib 3 mg n=11	Cabozantinib 40 mg: Darlifarnib 5 mg n=12	Cabozantinib 40 mg: Darlifarnib 8 mg n=12	Cabozantinib 60 mg: Darlifarnib 3 mg n=11	Cabozantinib 60 mg: Darlifarnib 5 mg n=12	Cabozantinib 60 mg: Darlifarnib 8 mg n=12	Total N=70
Any-grade darlifarnib-related TEAEs	9 (82)	11 (92)	10 (83)	7 (64)	10 (83)	11 (92)	58 (83)
Neutropenia	1 (9)	5 (42)	8 (67)	2 (18)	6 (50)	7 (58)	29 (41)
Fatigue	4 (36)	2 (17)	6 (50)	3 (27)	2 (17)	4 (33)	21 (30)
Diarrhea	4 (36)	3 (25)	2 (17)	3 (27)	4 (33)	2 (17)	18 (26)
Nausea	4 (36)	4 (33)	1 (8)	1 (9)	4 (33)	4 (33)	18 (26)
Thrombocytopenia	1 (9)	3 (25)	3 (25)	2 (18)	3 (25)	3 (25)	15 (21)
Grade ≥3 darlifarnib-related TEAEs	4 (36)	7 (58)	8 (67)	2 (18)	7 (58)	7 (58)	35 (50)
Neutropenia	1 (9)	5 (42)	7 (58)	1 (9)	4 (33)	3 (25)	21 (30)
Anemia	1 (9)	1 (8)	2 (17)	0	0	1 (8)	5 (7)
Fatigue	0	1 (8)	1 (8)	0	0	2 (17)	4 (6)
Thrombocytopenia	1 (9)	1 (8)	1 (8)	0	0	0	3 (4)
Leukopenia	0	1 (8)	1 (8)	0	0	0	2 (3)
Diarrhea	0	0	0	0	2 (17)	0	2 (3)

[a]Includes all safety-evaluable RCC patients (cabo-exposed and cabo-naive patients).
RCC, renal cell carcinoma; TEAE, treatment-emergent adverse event.
Data cutoff: Dec 8, 2025.

Table 3. Overall safety summary in all RCC patients[a] (N=70)

n (%)	Cabozantinib 40 mg: Darlifarnib 3 mg n=11	Cabozantinib 40 mg: Darlifarnib 5 mg n=12	Cabozantinib 40 mg: Darlifarnib 8 mg n=12	Cabozantinib 60 mg: Darlifarnib 3 mg n=11	Cabozantinib 60 mg: Darlifarnib 5 mg n=12	Cabozantinib 60 mg: Darlifarnib 8 mg n=12	Total N=70
DLTs	0	1 (8)	1 (8)	0	1 (8)	1 (8)	4 (6)
SAEs related to darlifarnib	0	1 (8)	2 (17)	1 (9)	0	2 (17)	6 (9)
Any-grade cabo-related TEAEs	11 (100)	11 (92)	11 (92)	8 (73)	12 (100)	12 (100)	65 (93)
Grade ≥3 cabo-related TEAEs	3 (27)	8 (67)	6 (50)	2 (18)	7 (58)	6 (50)	32 (46)
SAEs related to cabo	0	3 (25)	1 (8)	1 (9)	2 (17)	2 (17)	9 (13)
Darlifarnib dose reduction	0	2 (17)	2 (17)	0	2 (17)	3 (25)	9 (13)
Darlifarnib interruption	5 (46)	7 (58)	7 (58)	3 (27)	9 (75)	11 (92)	42 (60)
Darlifarnib discontinuation	1 (9)	1 (8)	2 (17)	0	0	0	4 (6)
Cabo dose reduction	2 (18)	1 (8)	2 (17)	2 (18)	6 (50)	5 (42)	18 (26)
Cabo interruption	5 (46)	9 (75)	8 (67)	4 (36)	12 (100)	12 (100)	50 (71)

[a]Includes all safety-evaluable RCC patients (cabo-exposed and cabo-naive patients).
Cabo, cabozantinib; DLT, dose-limiting toxicity; RCC, renal cell carcinoma; TEAE, treatment-emergent adverse event; SAE, serious adverse event.
Data cutoff: Dec 8, 2025.

Antitumor activity

- Antitumor activity was observed across all dose levels (**Table 4**)
 - 44% (7/16) cabo-exposed patients had a response
 - DCR was 94%; 15/16 patients had a response of PR or SD

Table 4. Antitumor activity in response-evaluable[a] cabo-exposed ccRCC patients

	Cabozantinib 40 mg: Darlifarnib 3 mg n=5	Cabozantinib 40 mg: Darlifarnib 5 mg n=6	Cabozantinib 40 mg: Darlifarnib 8 mg n=2[b]	Cabozantinib 60 mg: Darlifarnib 3 mg n=3	Total N=16
ORR (uPR + PR), n (%)	2 (40)	2 (33)[c]	1 (50)	2 (67)	7 (44)
95% CI	5.3–85.3	4.3–77.7	1.3–98.7	9.4–99.2	19.8–70.1
DCR[d], n (%)	5 (100)	6 (100)	1 (50)	3 (100)	15 (94)
95% CI	47.8–100	54.1–100	1.3–98.7	29.2–100	69.8–99.8

[a]Response-evaluable patients had ≥1 post-baseline scan. [b]One patient had only one disease assessment (PD as best response). [c]One patient had uPR. [d]DCR includes patients with SD and PR of any duration.
Cabo, cabozantinib; ccRCC, clear cell renal cell carcinoma; CI, confidence interval; DCR, disease control rate; ORR, objective response rate; PD, progressive disease; PR, partial response; SD, stable disease; uPR, unconfirmed partial response.
Data cutoff: Dec 8, 2025

- Tumor shrinkage was observed in 12/16 (75%) patients (**Figure 2**)
- Tumor reduction ranged from 32% to 47% for patients who responded to treatment

Figure 2. Best overall response in response-evaluable[a] cabo-exposed ccRCC patients

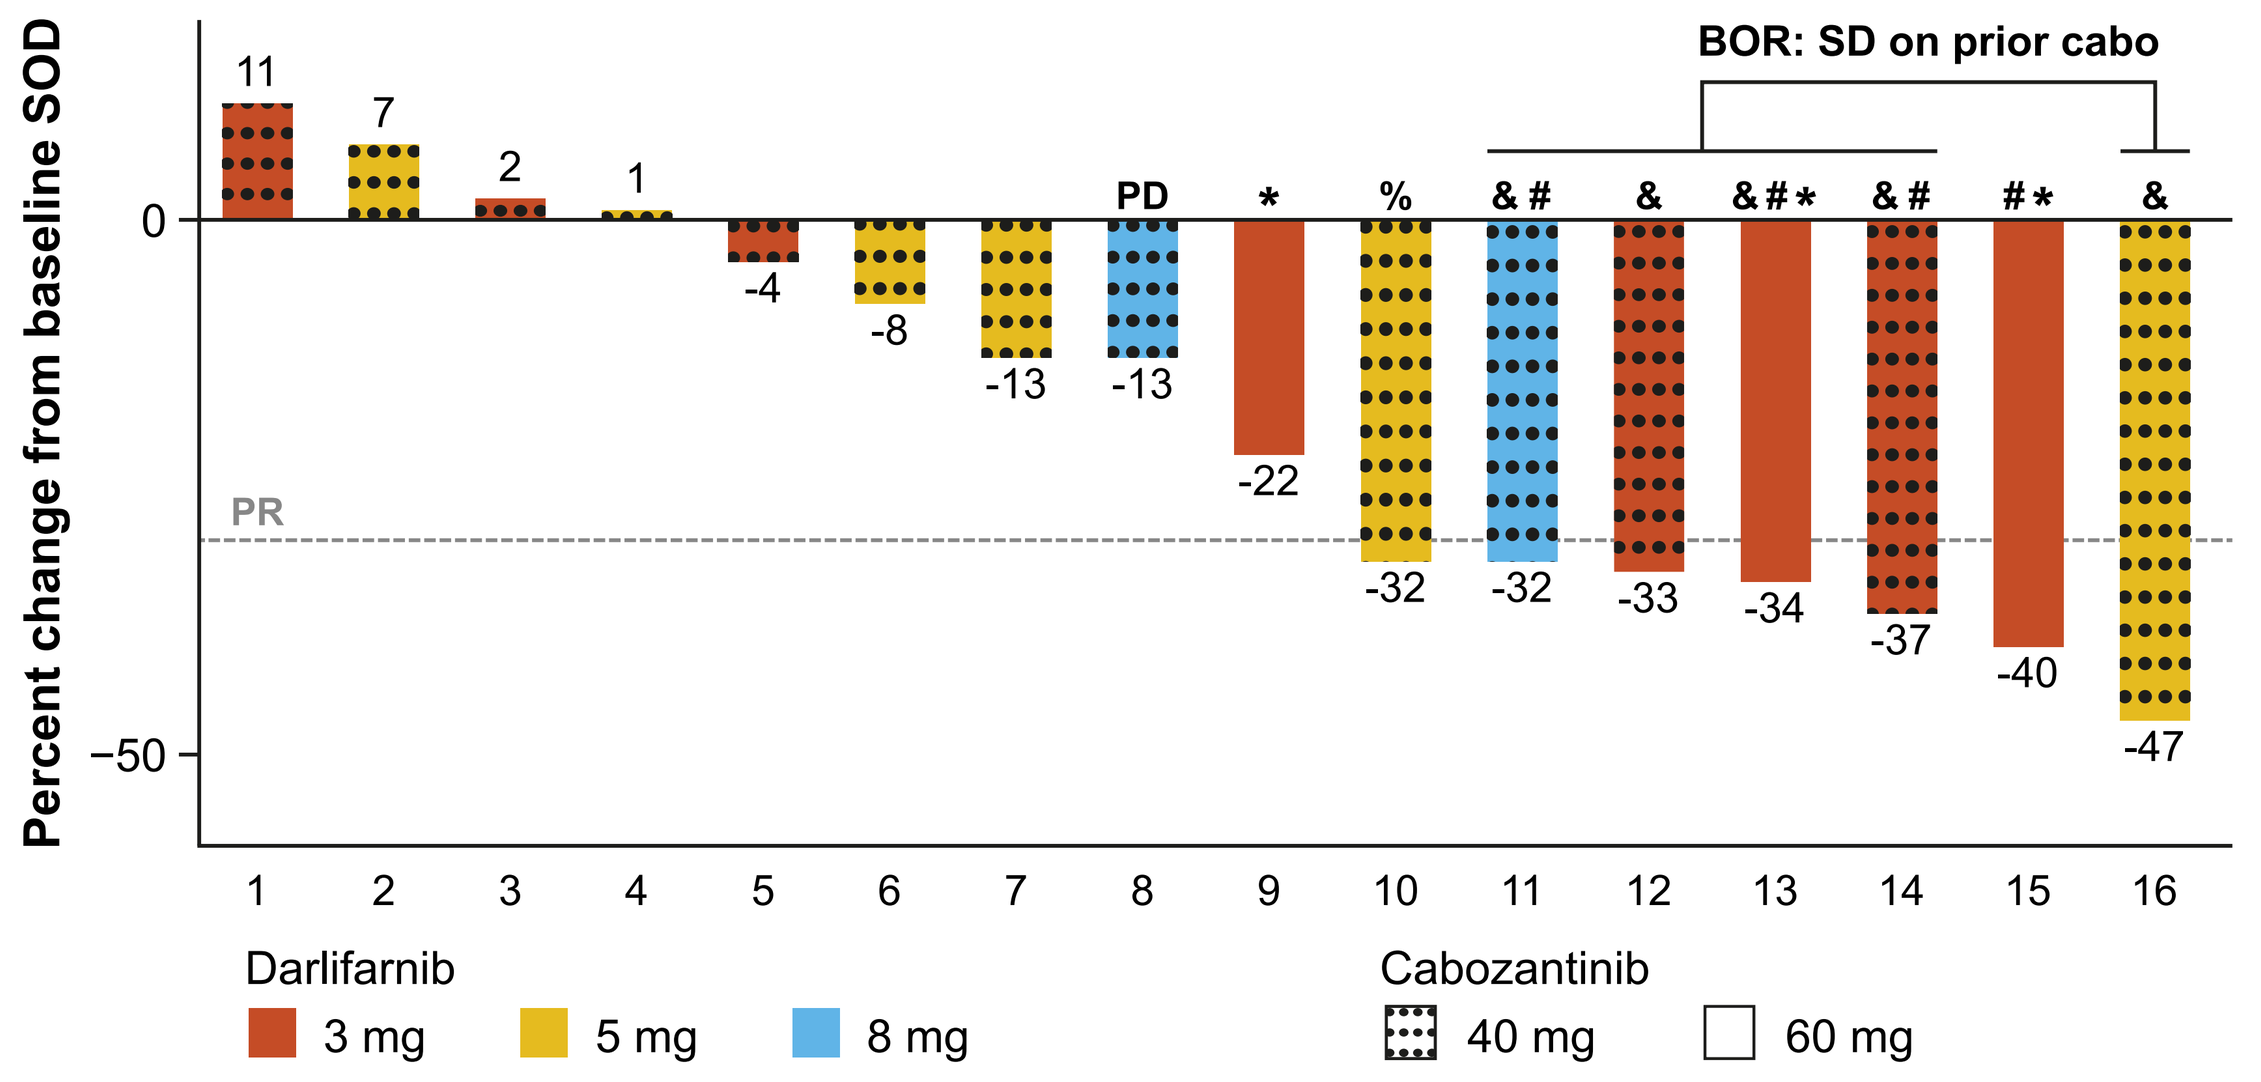

Percent change from baseline SOD
BOR: SD on prior cabo
0
−50
PR
11
7
2
1
-4
-8
-13
PD
-13
*
-22
%
-32
& #
-32
&
-33
& # *
-34
& #
-37
*
-40
&
-47
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16
Darlifarnib
3 mg
5 mg
8 mg
Cabozantinib
40 mg
60 mg

*Received darlifarnib 3 mg + cabo 60 mg; the remaining patients received cabo 40 mg in combination with darlifarnib 3, 5, or 8 mg.
#Immediate prior cabo exposure.
&BOR of SD on prior cabo.
%uPR.
[a]Response-evaluable patients had ≥1 post-baseline scan.
Cabo, cabozantinib; ccRCC, clear cell renal cell carcinoma; PD, progressive disease; PR, partial response; SOD, sum of diameters.
Data cutoff: Dec 8, 2025.

- Treatment duration ranged from 8–56 weeks and duration of response ranged from 8–32 weeks (**Figure 3**)
 - 6 patients remained on treatment at the time of data cutoff
- Darlifarnib + cabo provided clinical benefit for patients with prior cabo exposure, including in those with SD as best response to prior cabo and other TKIs (axitinib, lenvatinib, or sunitinib)
 - 57% (4/7) responders received cabo as immediate prior line
 - 71% (5/7) responders received other TKIs in addition to cabo
 - 71% (5/7) responders had BOR of SD on prior cabo

Figure 3. Duration of treatment and clinical outcomes in cabo-exposed ccRCC patients

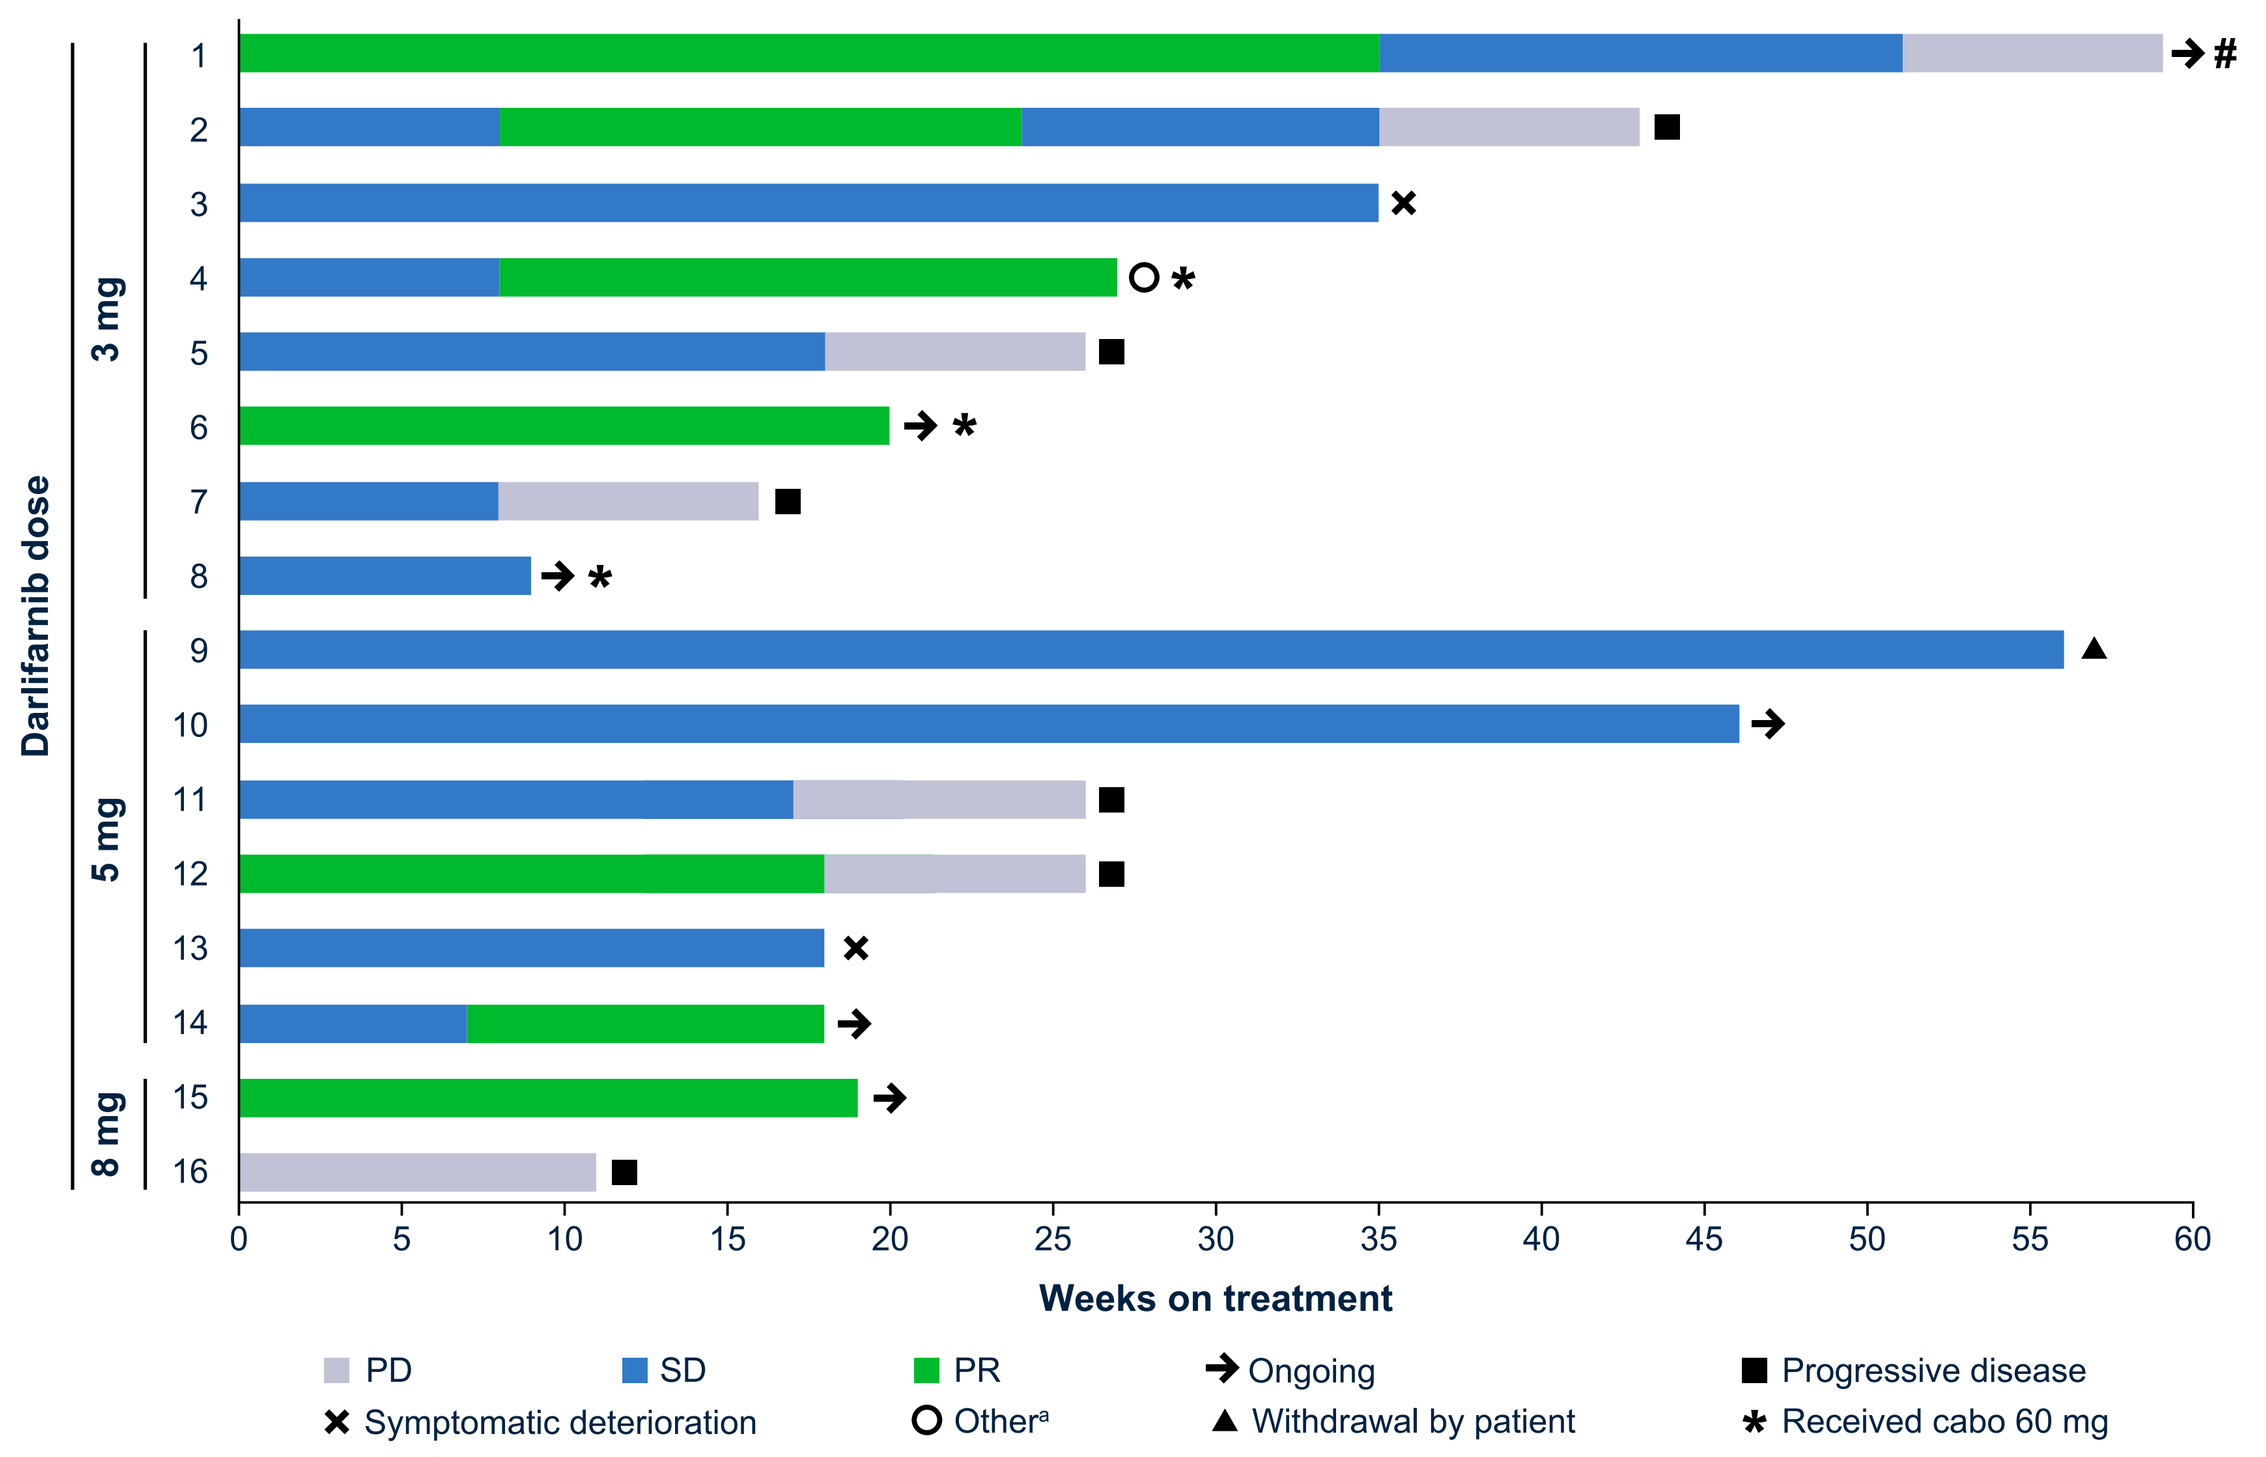

Darlifarnib dose
3 mg
5 mg
8 mg
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16
0 5 10 15 20 25 30 35 40 45 50 55 60
Weeks on treatment
PD
SD
PR
Ongoing
Progressive disease
Symptomatic deterioration
Othera
Withdrawal by patient
Received cabo 60 mg

#Patient was on treatment post-PD due to clinical benefit. [a]Adverse event.
Cabo, cabozantinib; ccRCC, clear cell renal cell carcinoma; PR, partial response; SD, stable disease; PD, progressive disease.
Data cutoff: Dec 8, 2025.

Figure 4. Scans[a] from a patient treated with darlifarnib 3 mg + cabo 40 mg with prior cabo exposure

- 59-year-old female with ccRCC diagnosed in May 2022 (**Figure 4**)

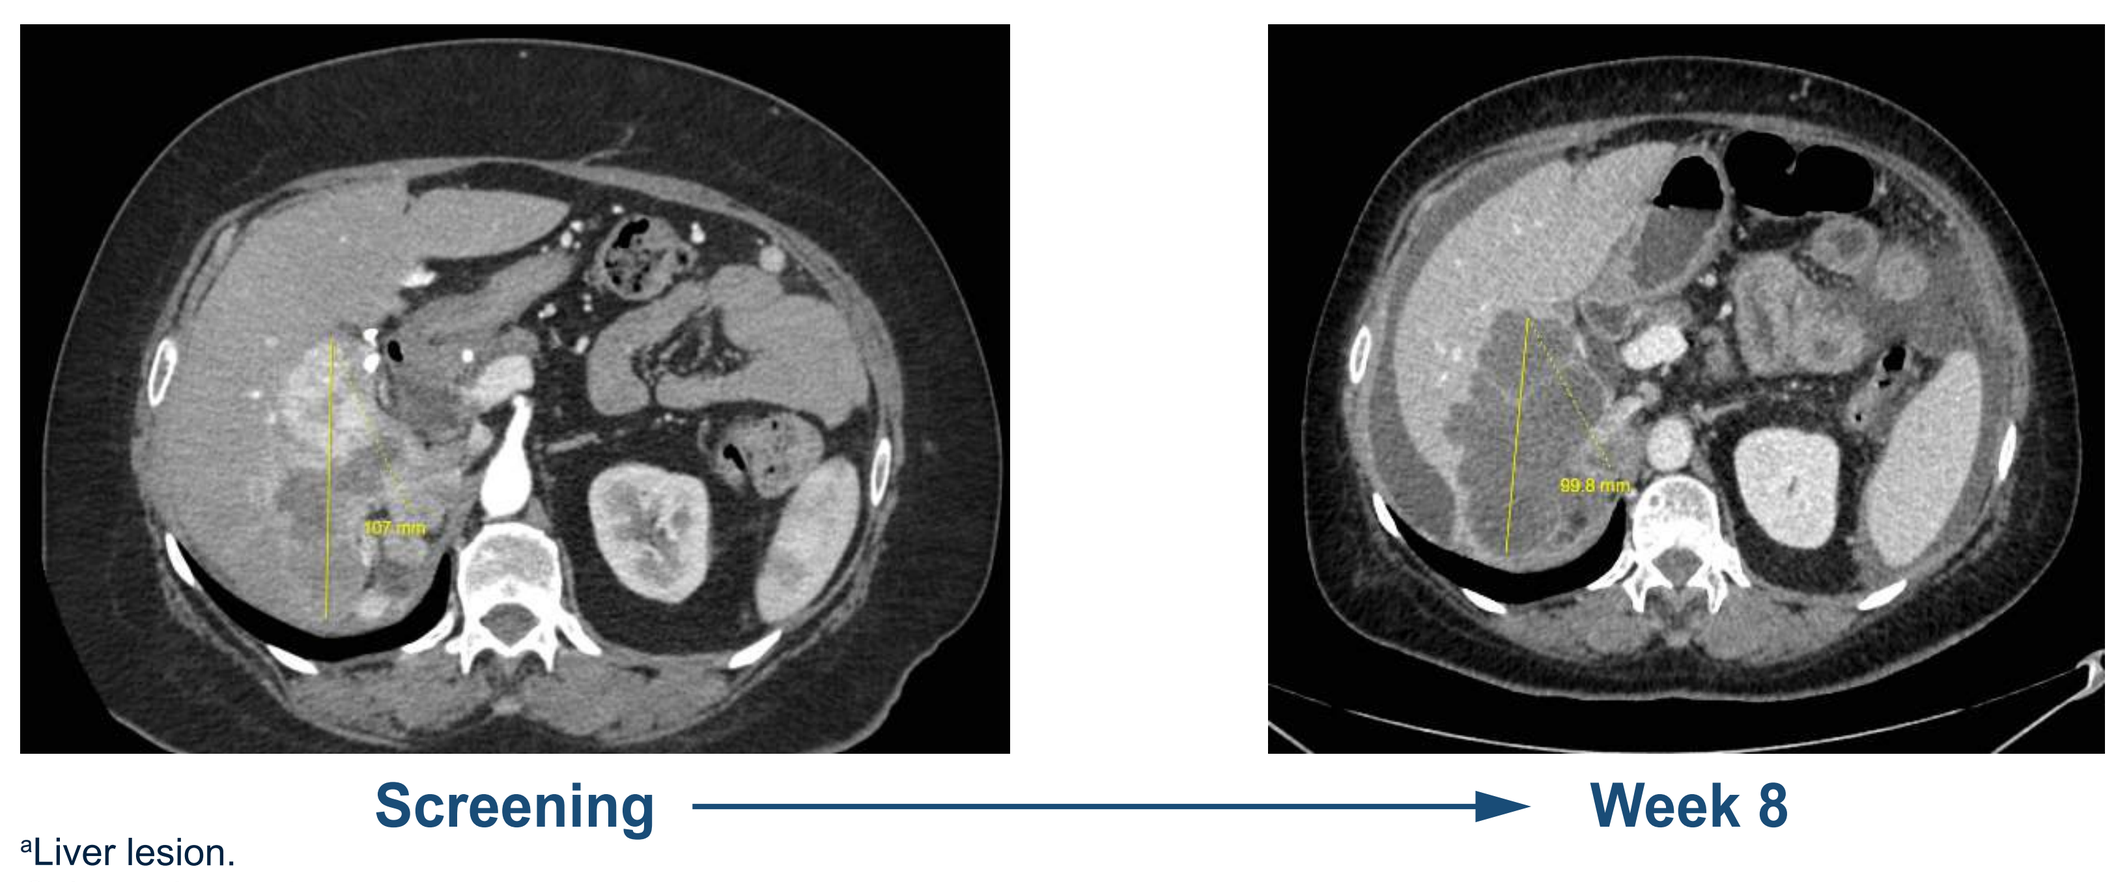

Screening
Week 8

[a]Liver lesion.
Cabo, cabozantinib.
Dec 8, 2025.

- Prior therapy:
 - 1L: Pembrolizumab + axitinib (BOR: PR)
 - 2L: **Cabo** (BOR: SD)
 - 3L: Belzutifan (BOR: PD)
- Patient progressed previously on pembrolizumab + axitinib, and belzutifan
- Initiated study treatment Sep 2025
- Patient continues on darlifarnib + cabo treatment with best response of SD since treatment initiation (6 months)

Key consideration:

- Patient had SD with darlifarnib + cabo, demonstrating encouraging responses in liver and peritoneal lesions, including extensive necrotic changes within these lesions

CONCLUSIONS

- **After progression on VEGFR-targeted therapies, subsequent treatment with single-agent therapies is associated with limited clinical benefit, possibly due to emergence of resistance mechanisms**
- **In the ongoing FIT-001 study, darlifarnib + cabo combination therapy demonstrated a manageable safety profile across all RCC patients, and encouraging antitumor activity in ccRCC patients with prior cabo exposure**
 - Activity of the combination in a post-cabo setting is suggestive of darlifarnib's contribution to antitumor activity in this patient population
 - This antitumor activity is notable given best responses were SD in patients on prior cabo or who had been treated with cabo and another TKI
 - These results suggest the potential impact of darlifarnib combination therapy as an option for patients who have progressed on prior cabo with or without other TKIs
- **These data support further investigation of combinations of darlifarnib and cabo in ccRCC patients, including in earlier lines of therapy**
- **The phase 1b portion of the FIT-001 study is currently enrolling cabo-naive patients (NCT06026410)**

References

1. Choueiri TK et al. *N Engl J Med* 2015;373:1814–23;
2. Sharma R et al. *J Exp Clin Cancer Res* 2021;40:186;
3. Choueiri TK et al. *Lancet Oncol* 2016 17:917–927;
4. Choueiri TK et al. *N Engl J Med* 2024;391:710–721;
5. Rini BI et al. *The Lancet* 2021;21:95–104;
6. Gasendo JG et al. Oral presentation (abstract #6370) presented at the AACR Annual Meeting, April 25–30, 2025, Chicago, IL, USA;
7. Gatchalian J et al. Poster (abstract #34634) presented at the 36th EORTC-NCI-AACR Symposium, October 11–15, 2023, Boston, MA, USA;
8. Ayanambakkam A et al. Poster (#2604P) presented at The European Society for Medical Oncology Annual Congress 2025, October 17–21, 2025, Berlin, Germany;
9. Lacy S et al. *Cancer Chemother Pharmacol* 2018;81:1061–1070.

Acknowledgments

The authors would like to thank the patients, their families, and their caregivers, as well as the FIT-001 study investigators and their study teams. The authors would also like to thank Jamie Kuipers for reviewing the data and poster draft. This study was sponsored by Kura Oncology, Inc. Editorial and artwork services were provided by Oxford PharmaGenesis, Inc of Wilmington, DE and funded by Kura Oncology, Inc.

Disclosures

YZ: Advisory Board - Advisory Board: Bristol Myers Squibb, Janssen, Merck, Astellas, Eisai, Exelixis, AstraZeneca, Pfizer, EMD Serono, Gilead, Kyowa Kirin, Xencor, AVEO. **AS**: Consulting or Advisory Role - Adaptyx Biosciences, AVEO. **JT**: Consulting or Advisory Role - Kura Oncology, Conjupro Biotherapeutics. **JH**: Employment - HCA Healthcare; Stock and Other Ownership Interests - HCA Healthcare; Research Funding - Abbisko (Inst); ABL Bio (Inst); accutar biotech (Inst); ADC Therapeutics (Inst); Agenus (Inst); Aileron Therapeutics (Inst); Amgen (Inst); Artios (Inst); AstraZeneca/Merck (Inst); Bicycle Therapeutics (Inst); BioAtla (Inst); Bioinvent (Inst); Biosplice (Inst); Black Diamond Therapeutics (Inst); Boehringer Ingelheim (Inst); cyteir (Inst); DaiichI Sankyo (Inst); Daiichi Sankyo/Lilly (Inst); Epizyme (Inst); Erasca, Inc (Inst); Exelixis (Inst); Fujifilm (Inst); GlaxoSmithKline (Inst); Hutchison MediPharma (Inst); Iconic Therapeutics (Inst); Iconic Therapeutics (Inst); Iconic Therapeutics (Inst); IgM Biosciences (Inst); Immunitas (Inst); Immunogen (Inst); Immunogen (Inst); Jacobio (Inst); Jazz Pharmaceuticals (Inst); Jounce Therapeutics (Inst); Jubilant Pharmaceuticals (Inst); Jubilant Pharmaceuticals (Inst); Lilly (Inst); Loxo (Inst); Merck (Inst); Metabomed (Inst); Mirati Therapeutics (Inst); Molecular Templates (Inst); Navire (Inst); NiKang Therapeutics (Inst); Oncorus (Inst); Poseida therapeutics (Inst); Prelude Therapeutics (Inst); PureTech (Inst); Pyramid Biosciences (Inst); Pyxis (Inst); RasCal (Inst); Regeneron (Inst); Relay Therapeutics (Inst); Rgenix (Inst); Ribon Therapeutics (Inst); Sapience Therapeutics (Inst); Sarah Cannon Research Institute (Inst); Sarah Cannon Research Institute (Inst); Seagan (Inst); Simcha Therapeutics (Inst); Sirnaomics (Inst); Stingthera (Inst); StingThera (Inst); Synthorx (Inst); Synthorx (Inst); Takeda (Inst); Takeda (Inst); Tallac Therapeutics (Inst); Tarveda Therapeutics (Inst); Teneothree (Inst); Tesaro (Inst); Turning Point Therapeutics (Inst); Xencor (Inst); Travel, Accommodations, Expenses - HCA/Sarah Cannon (Inst). **LV**: Consulting Fees - Caris, AstraZeneca, Johnson and Johnson, Amgen, Pfizer, Lilly, EMD Serono, Natera, Regeneron, Abbvie, Genentech, BMS. **BG**, **DL**, **SG**: Nothing to disclose. **PMJ**, **JK**: Employment, Stock and Other Ownership Interests - Kura Oncology, Inc. **BB**, **ML**: Employment; Stock and Other Ownership Interests; Patents, Royalties, Other Intellectual Property - Kura Oncology, Inc. **AA**: Consulting or Advisory Role – AVEO, Pfizer/Astellas, Johnson & Johnson, Kura Oncology; Travel, Accommodations, Expenses: Kura Oncology; Research Funding: Regeneron.

Abbreviations

1L, first-line; 2L, second-line; 3L, third-line; AE, adverse event; BOR, best overall response; Cabo, cabozantinib; ccRCC, clear cell renal cell carcinoma; DCR, disease control rate; DL, dose level; DLT, dose-limiting toxicity; FTI, farnesyl transferase inhibitor; IO, immuno-oncology; mTOR, mechanistic target of rapamycin kinase; mTORC1, mammalian target of rapamycin complex 1; ORR, objective response rate; PD, progressive disease; PR, partial response; PS, performance status; QD, once daily; RCC, renal cell carcinoma; RHEB, Ras homolog enriched in brain; SAE, serious adverse event; SD, stable disease; SOD, sum of diameters; TEAE, treatment-emergent adverse event; TKI, tyrosine kinase inhibitor; uPR, unconfirmed partial response; VEGFR, vascular endothelial growth factor receptor.

Poster PDF can be accessed through this Quick Response (QR) code. Copies obtained are for personal use only.

Additional information on the FIT-001 study can be accessed here.

Poster presented at: *The 2026 International Kidney Cancer Symposium: Europe, April 16–18, 2026, Paris, France*