Abstract #3038 Translating genotype to immunophenotype in HRAS mutated head and neck squamous cell carcinoma (HNSCC) to identify effective Tipifarnib partners for optimal patient outcomes

Theodoros Rampias[1], Miriam Anastasiou[2], Aris Spathis[3], Stergios Doumas[4], Panagiota Economopoulou[2], Ioannis Kotsantis[2], Anastasios Pantazopoulos[2], Anastasios Kyriazoglou[2], Georgios Kavourakis[2] , Periklis Foukas[1] and Amanda Psyrri[2]

[1]Biomedical Research Foundation Academy of Athens, Athens, Greece; [2]2Department of Internal Medicine, Section of Medical Oncology, Attikon University Hospital, National Kapodistrian University of Athens, Haidari, Athens, Greece; [3]2nd Department of Pathology, National and Kapodistrian University of Athens, Attikon University Hospital, Athens, Greece *[4] National and Kapodistrian University of Athens, 2nd Department of Otolaryngology, University General Hospital "Attikon", Athens, Greece.*

Background

tipifarnib is a highly selective farnesyltransferase inhibitor which has shown promising activity in HRAS mutant recurrent/metastatic HNSCC[1].

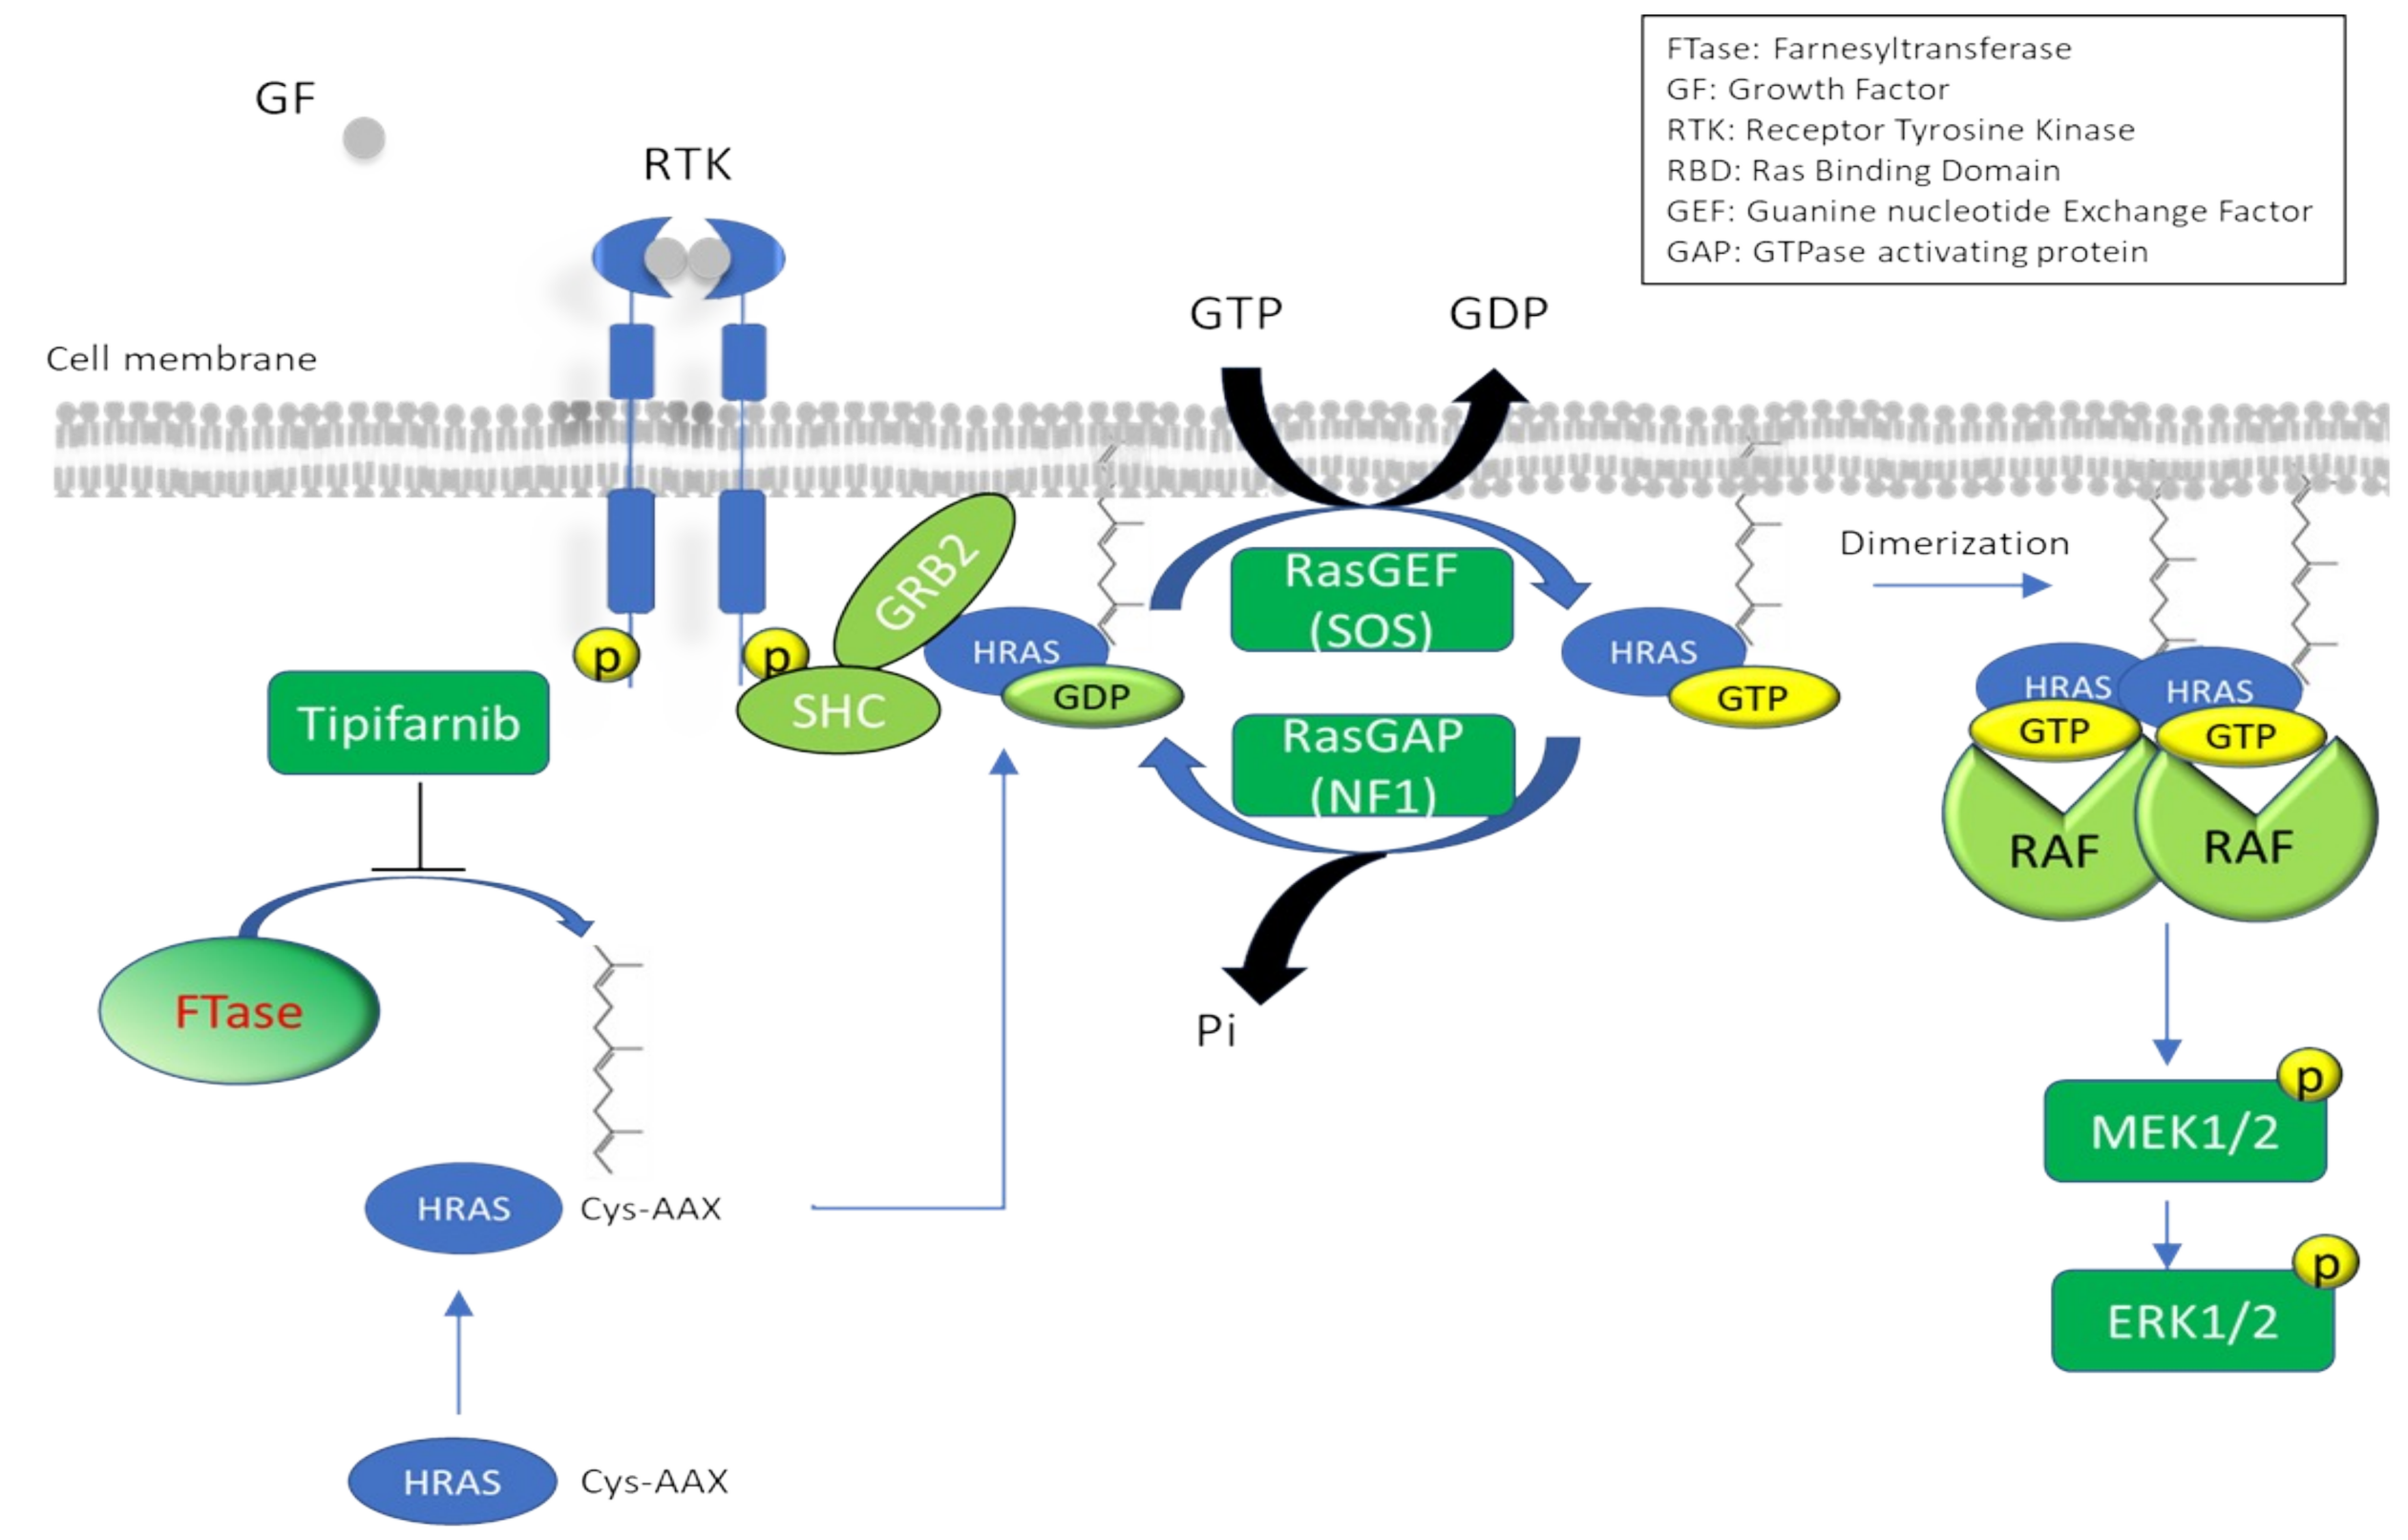
FTase: Farnesyltransferase
GF: Growth Factor
RTK: Receptor Tyrosine Kinase
RBD: Ras Binding Domain
GEF: Guanine nucleotide Exchange Factor
GAP: GTPase activating protein
GF
RTK
Cell membrane
GTP
GDP
RasGEF (SOS)
RasGAP (NF1)
GRB2
SHC
HRAS
Dimerization
RAF
MEK1/2
ERK1/2
Tipifarnib
FTase
Pi
Cys-AAX

Figure 1. Overview of HRAS activity and farnesyltransferase inhibition by tipifarnib.

Aims

- Since HRAS mutated HNSCC can be successfully targeted with Tipifarnib, we sought to analyze the immune and genomic landscape of these tumors to guide combinatorial strategies.

Materials and Methods

- To characterize the molecular and immune profile of HRAS mutant tumors in HNSCC, the TCGA mutational and transcriptome data were analyzed using the TIMER 2.0 platform for comprehensive analysis of tumor-infiltrating immune cells[2,3,4]

- Genomic DNA from baseline biopsies of tumors was purified and subjected to targeted sequencing. We identified thirteen patients with HRAS mutant tumors. To compare the genomic and immune profiles between HRAS mutant and wild type tumors, we included in the analysis 50 patients with HRAS wild-type tumors. PD-L1 expression in formalin-fixed tumour samples was assessed using the PD-L1 IHC 22C3 pharmDx assay (Agilent Technologies, Carpinteria, CA, USA) and characterized by the combined positive score (CPS). Assessment of PD-L1+ cells inside and at periphery of the tumor was performed. HRAS mutational status was correlated with PD-L1 status.

Results

1. TCGA data indicate that tumors with oncogenic HRAS display higher levels of CD8A+ infiltration in HNSCC.

Gene expression profiling using RNA-seq data has been widely used for immune infiltration estimation. In our study, the TIMER2.0 platform was used in order to retrieve the immune infiltration profile from the HNSCC TCGA samples and correlate them to HRAS mutation status. TIMER2.0 utilizes the Immunedeconv, an R package which integrates six state-of-the-art algorithms, including TIMER, xCell, MCP-counter, CIBERSORT, EPIC and quanTIseq. This analysis revealed that HPV- tumors with oncogenic HRAS display higher levels of CD8A+infiltration. The statistical significance of this correlation was tested against all different TIMER2.0 algorithms (Figure 2).

A.

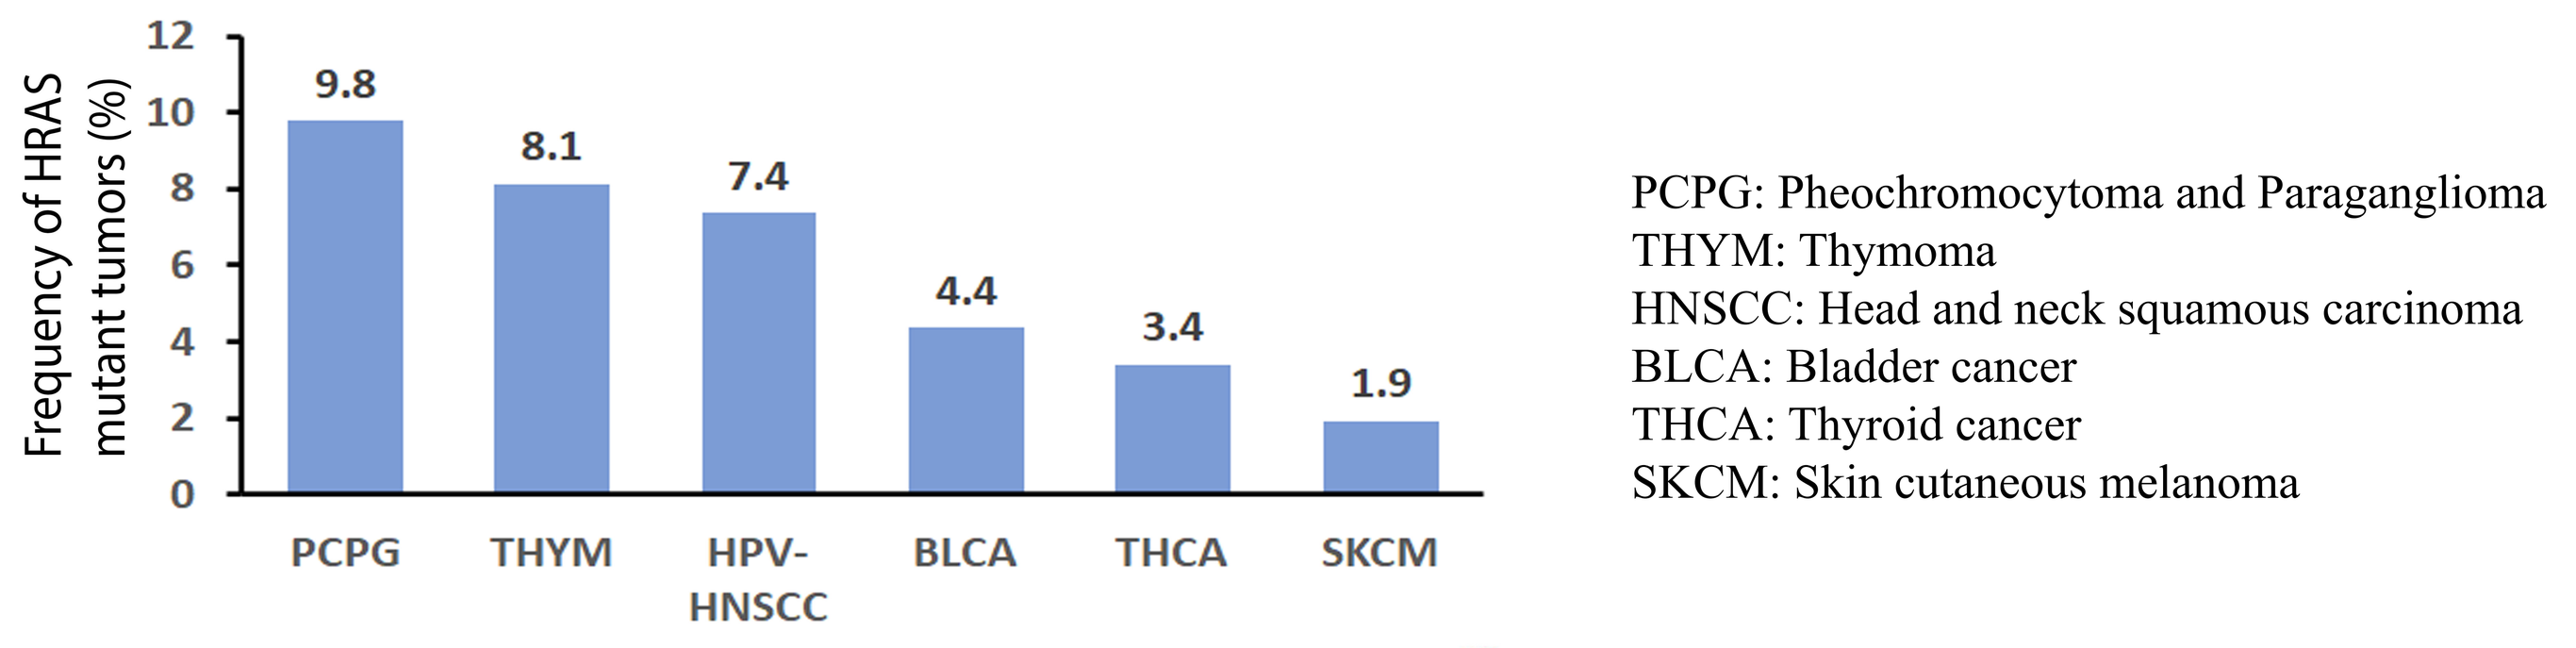
Frequency of HRAS mutant tumors (%)
9.8
8.1
7.4
4.4
3.4
1.9
PCPG
THYM
HPV- HNSCC
BLCA
THCA
SKCM
PCPG: Pheochromocytoma and Paraganglioma
THYM: Thymoma
HNSCC: Head and neck squamous carcinoma
BLCA: Bladder cancer
THCA: Thyroid cancer
SKCM: Skin cutaneous melanoma

B.

cancer	T cell CD8+ TIMER	T cell CD8+ EPIC	T cell CD8+ MCP-COUNTER	T cell CD8+ CIBERSORT	T cell CD8+ CIBERSORT-ABS	T cell CD8+ QUANTISEQ+	T cell CD8+ XCELL
HNSC-HPV- (n=412)	1.132	0.532	0.831	0.84	0.761	1.141	1.241

Log2(Fold Change) : higher level in mutants (p<0.05, log2FC>0)
Log2(Fold Change) : lower level in mutants (p<0.05, log2FC<0)
Log2(Fold Change) : not significant (p>0.05)

C.

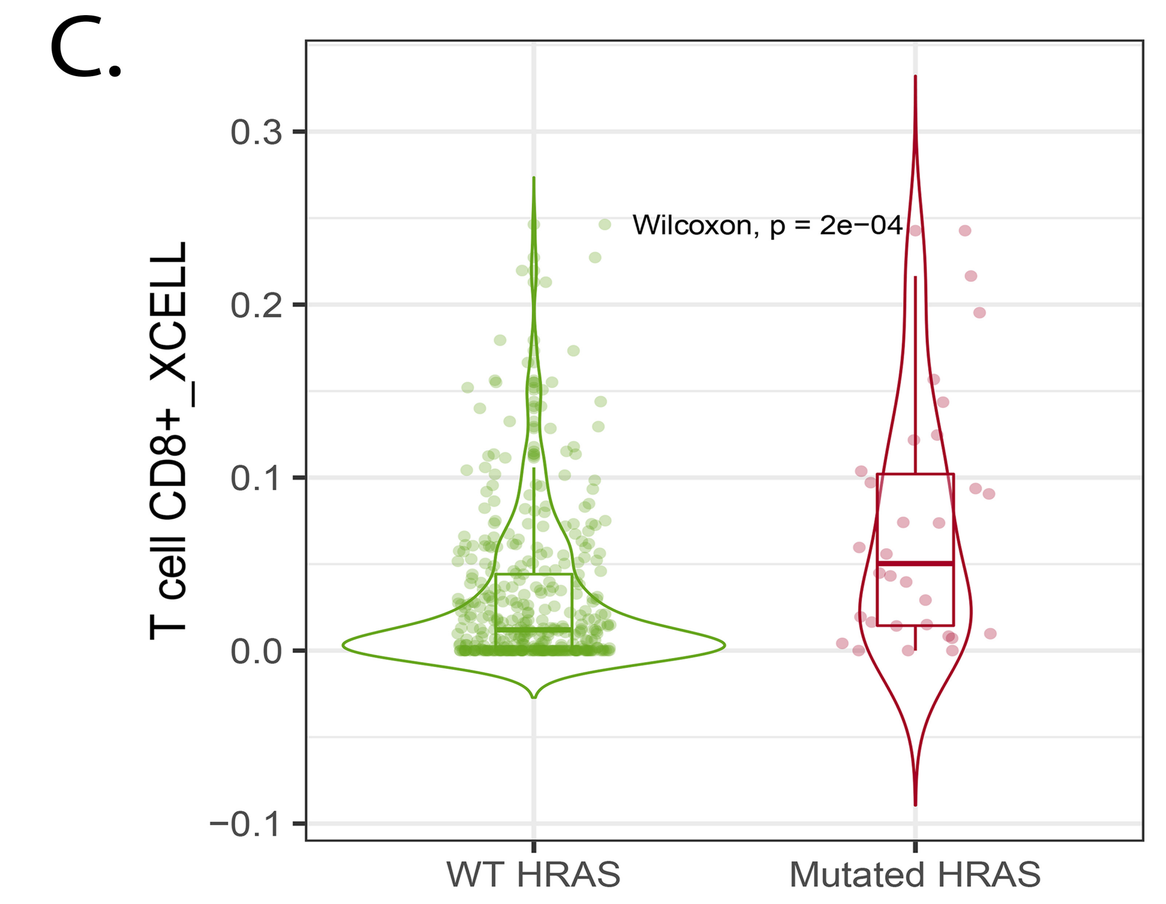
T cell CD8+_XCELL
Wilcoxon, p = 2e−04
WT HRAS
Mutated HRAS

Figure 2. Estimation of CD8A+ infiltration levels in HPV- HNSCCs. (A). Cancer types with high frequency of mutated HRAS based on TCGA mutation data. (B). Log2-fold changes of CD8A+ infiltration level between mutated HRAS and wild type HRAS tumors in HPV- HNSCC. (C). Violin plot of CD8A+ immune infiltration distribution based on XCELL algorithm.

2. Pan-Cancer TCGA data indicate a cancer type specific correlation between HRAS mutation status and CD8A+ infiltration.

The pan–cancer correlation between the HRAS mutation status and the transcriptomic data related to signatures of CD8A+ immune cells by TIMER 2.0, revealed that a statistically significant upregulation of CD8A+ levels is exclusively assigned to the cluster of HPV- HNSCCs among a wide variety of cancer types. This association was indicated by all different TIMER2.0 algorithms (Figure 3).

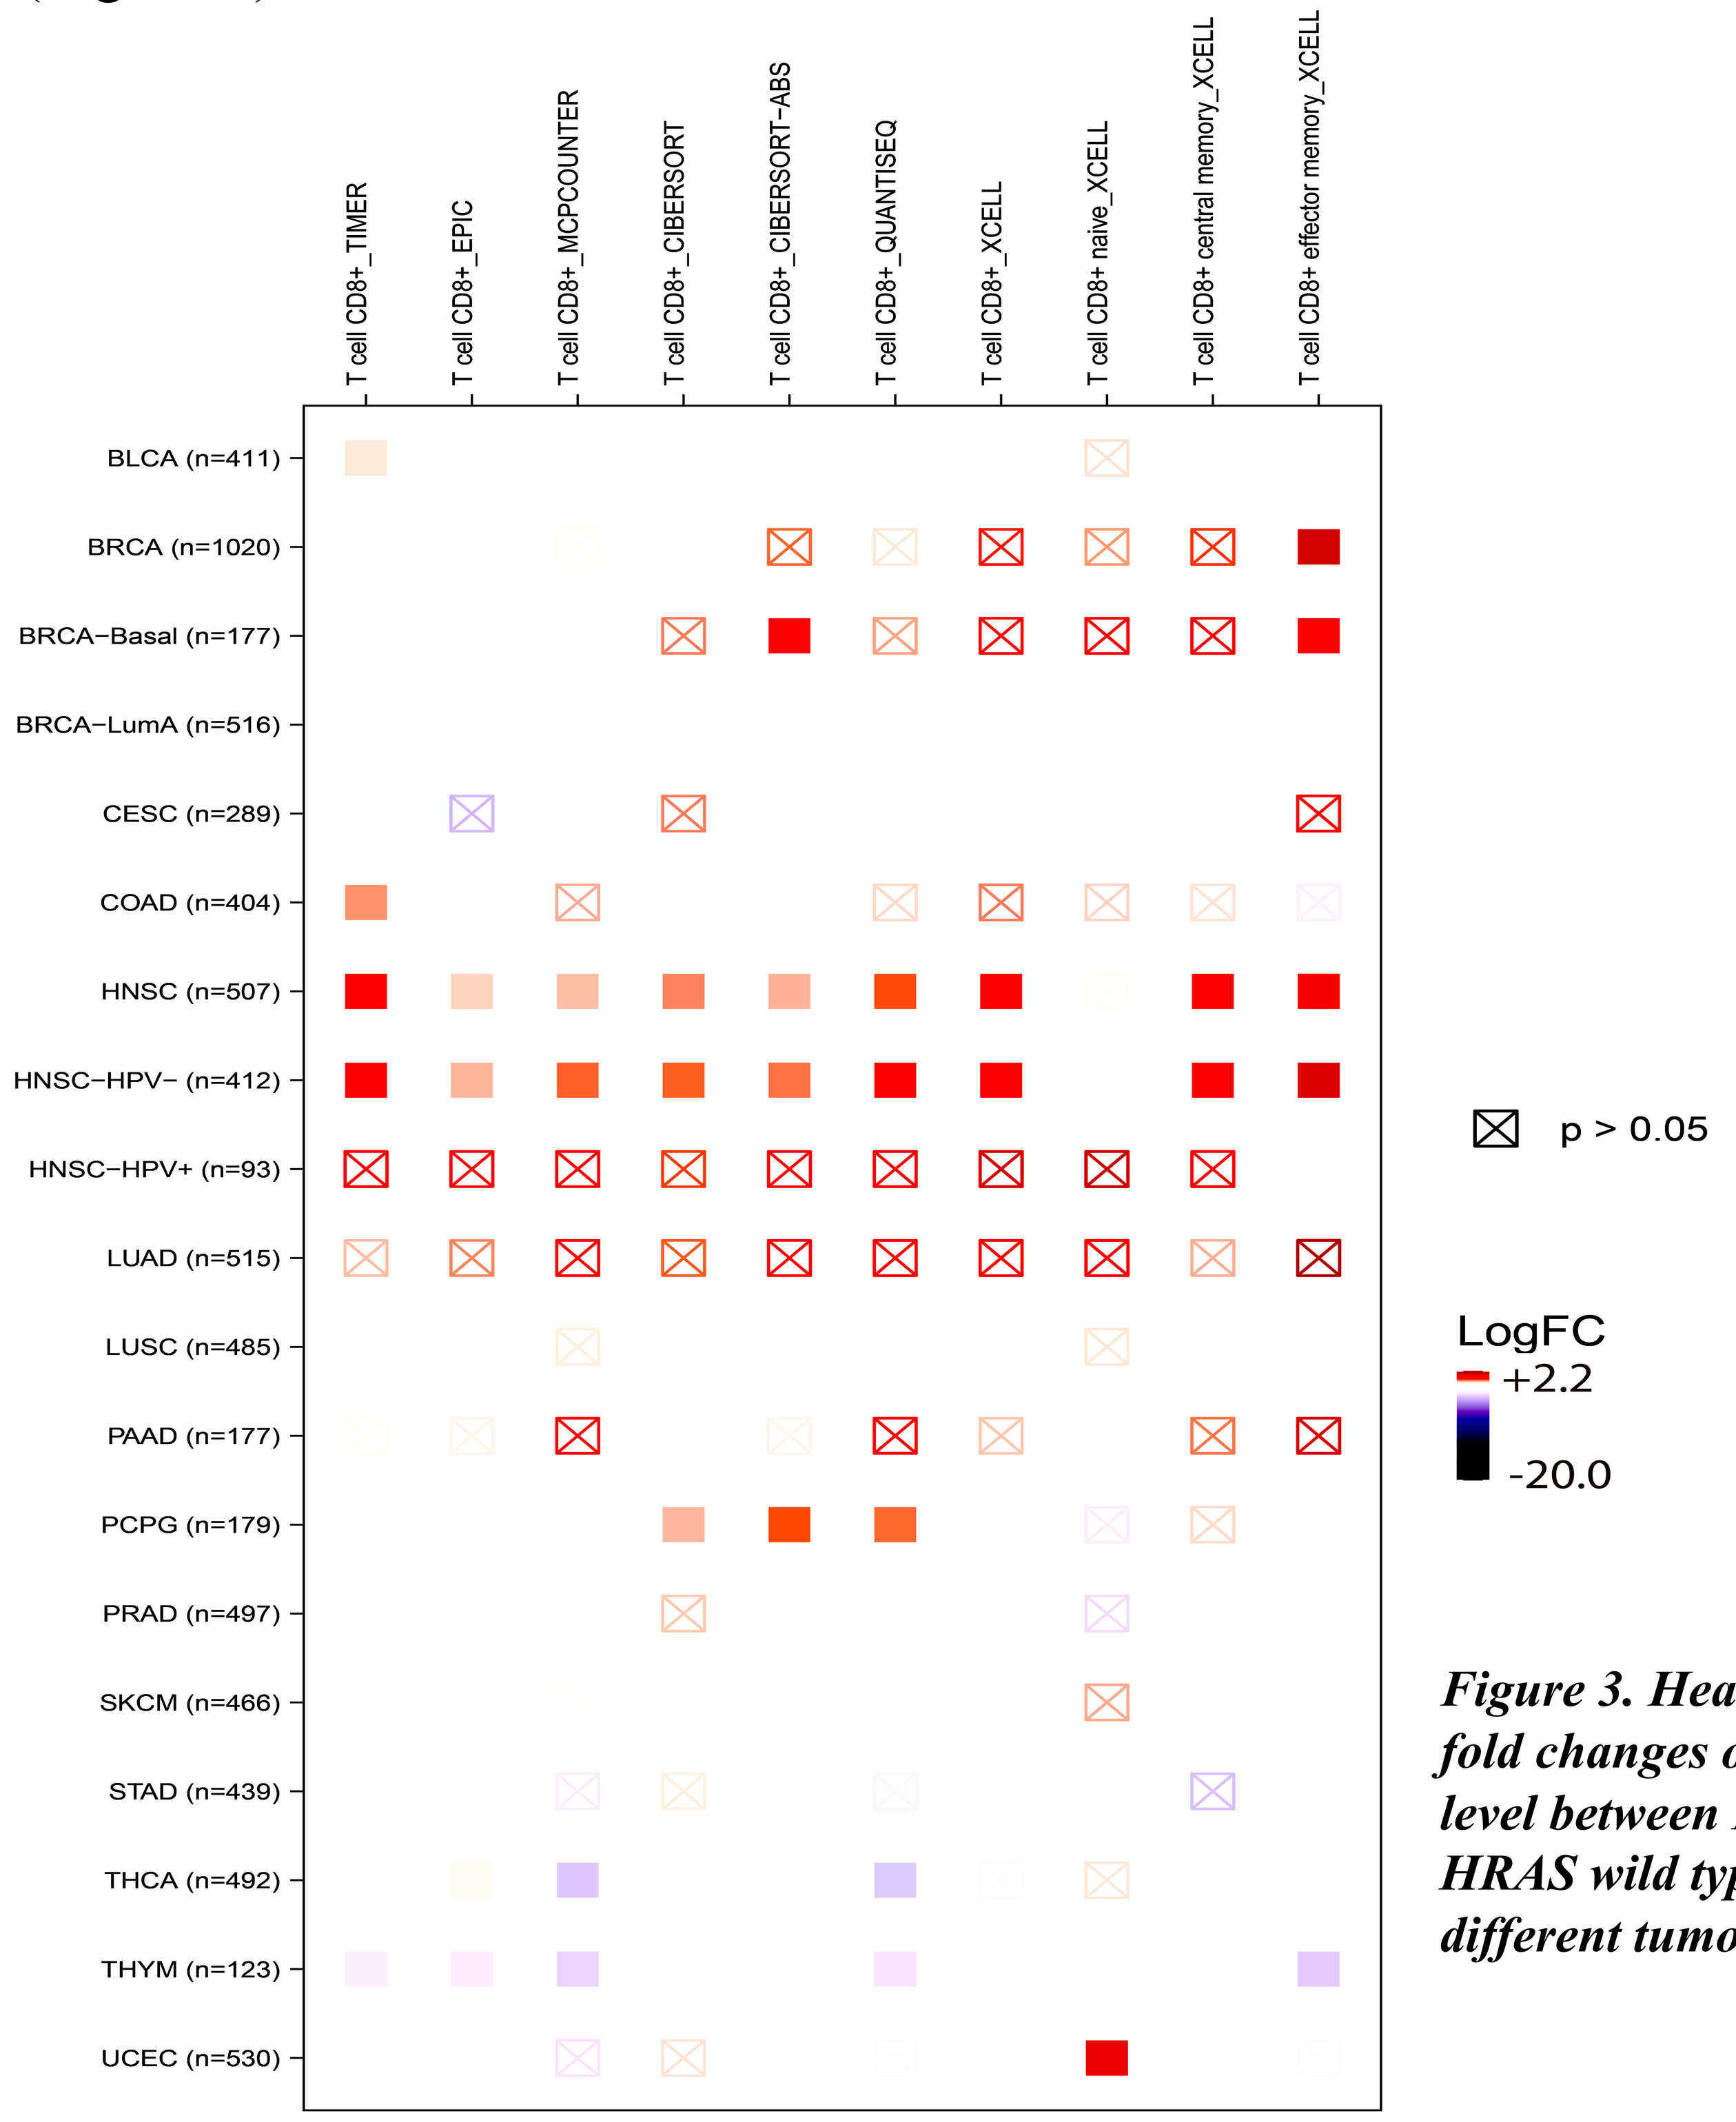
T cell CD8+_TIMER
T cell CD8+_EPIC
T cell CD8+_MCPCOUNTER
T cell CD8+_CIBERSORT
T cell CD8+_CIBERSORT-ABS
T cell CD8+_QUANTISEQ
T cell CD8+_XCELL
T cell CD8+ naive_XCELL
T cell CD8+ central memory_XCELL
T cell CD8+ effector memory_XCELL
BLCA (n=411)
BRCA (n=1020)
BRCA−Basal (n=177)
BRCA−LumA (n=516)
CESC (n=289)
COAD (n=404)
HNSC (n=507)
HNSC−HPV− (n=412)
HNSC−HPV+ (n=93)
LUAD (n=515)
LUSC (n=485)
PAAD (n=177)
PCPG (n=179)
PRAD (n=497)
SKCM (n=466)
STAD (n=439)
THCA (n=492)
THYM (n=123)
UCEC (n=530)
p > 0.05
LogFC
+2.2
-20.0

Figure 3. Heatmap table of the log-fold changes of CD8A+ infiltration level between HRAS mutated and HRAS wild type tumors among different tumor types.

3. The presence of HRAS mutations is associated with higher PD-L1 expression.

-PD-L1 expression levels were assessed by IHC in a group of 13 HRAS mutated HNSCCs and in a group of 50 HNSCCs harboring wild type HRAS.

-Assessment of PD-L1+ cells inside and at periphery of the tumor was performed for all cases independently. Combined positive score (CPS) and tumor proportion score (TPS) were both evaluated.

-The presence of HRAS mutations was associated with higher TPS PD-L1 scores (26% vs 40% vs 100% in TPS <1%, 1-49% and >50% respectively, p=0.043). Similar results were obtained for CPS (28% vs 80% in CPS <10% and =>10%, p=0.047).

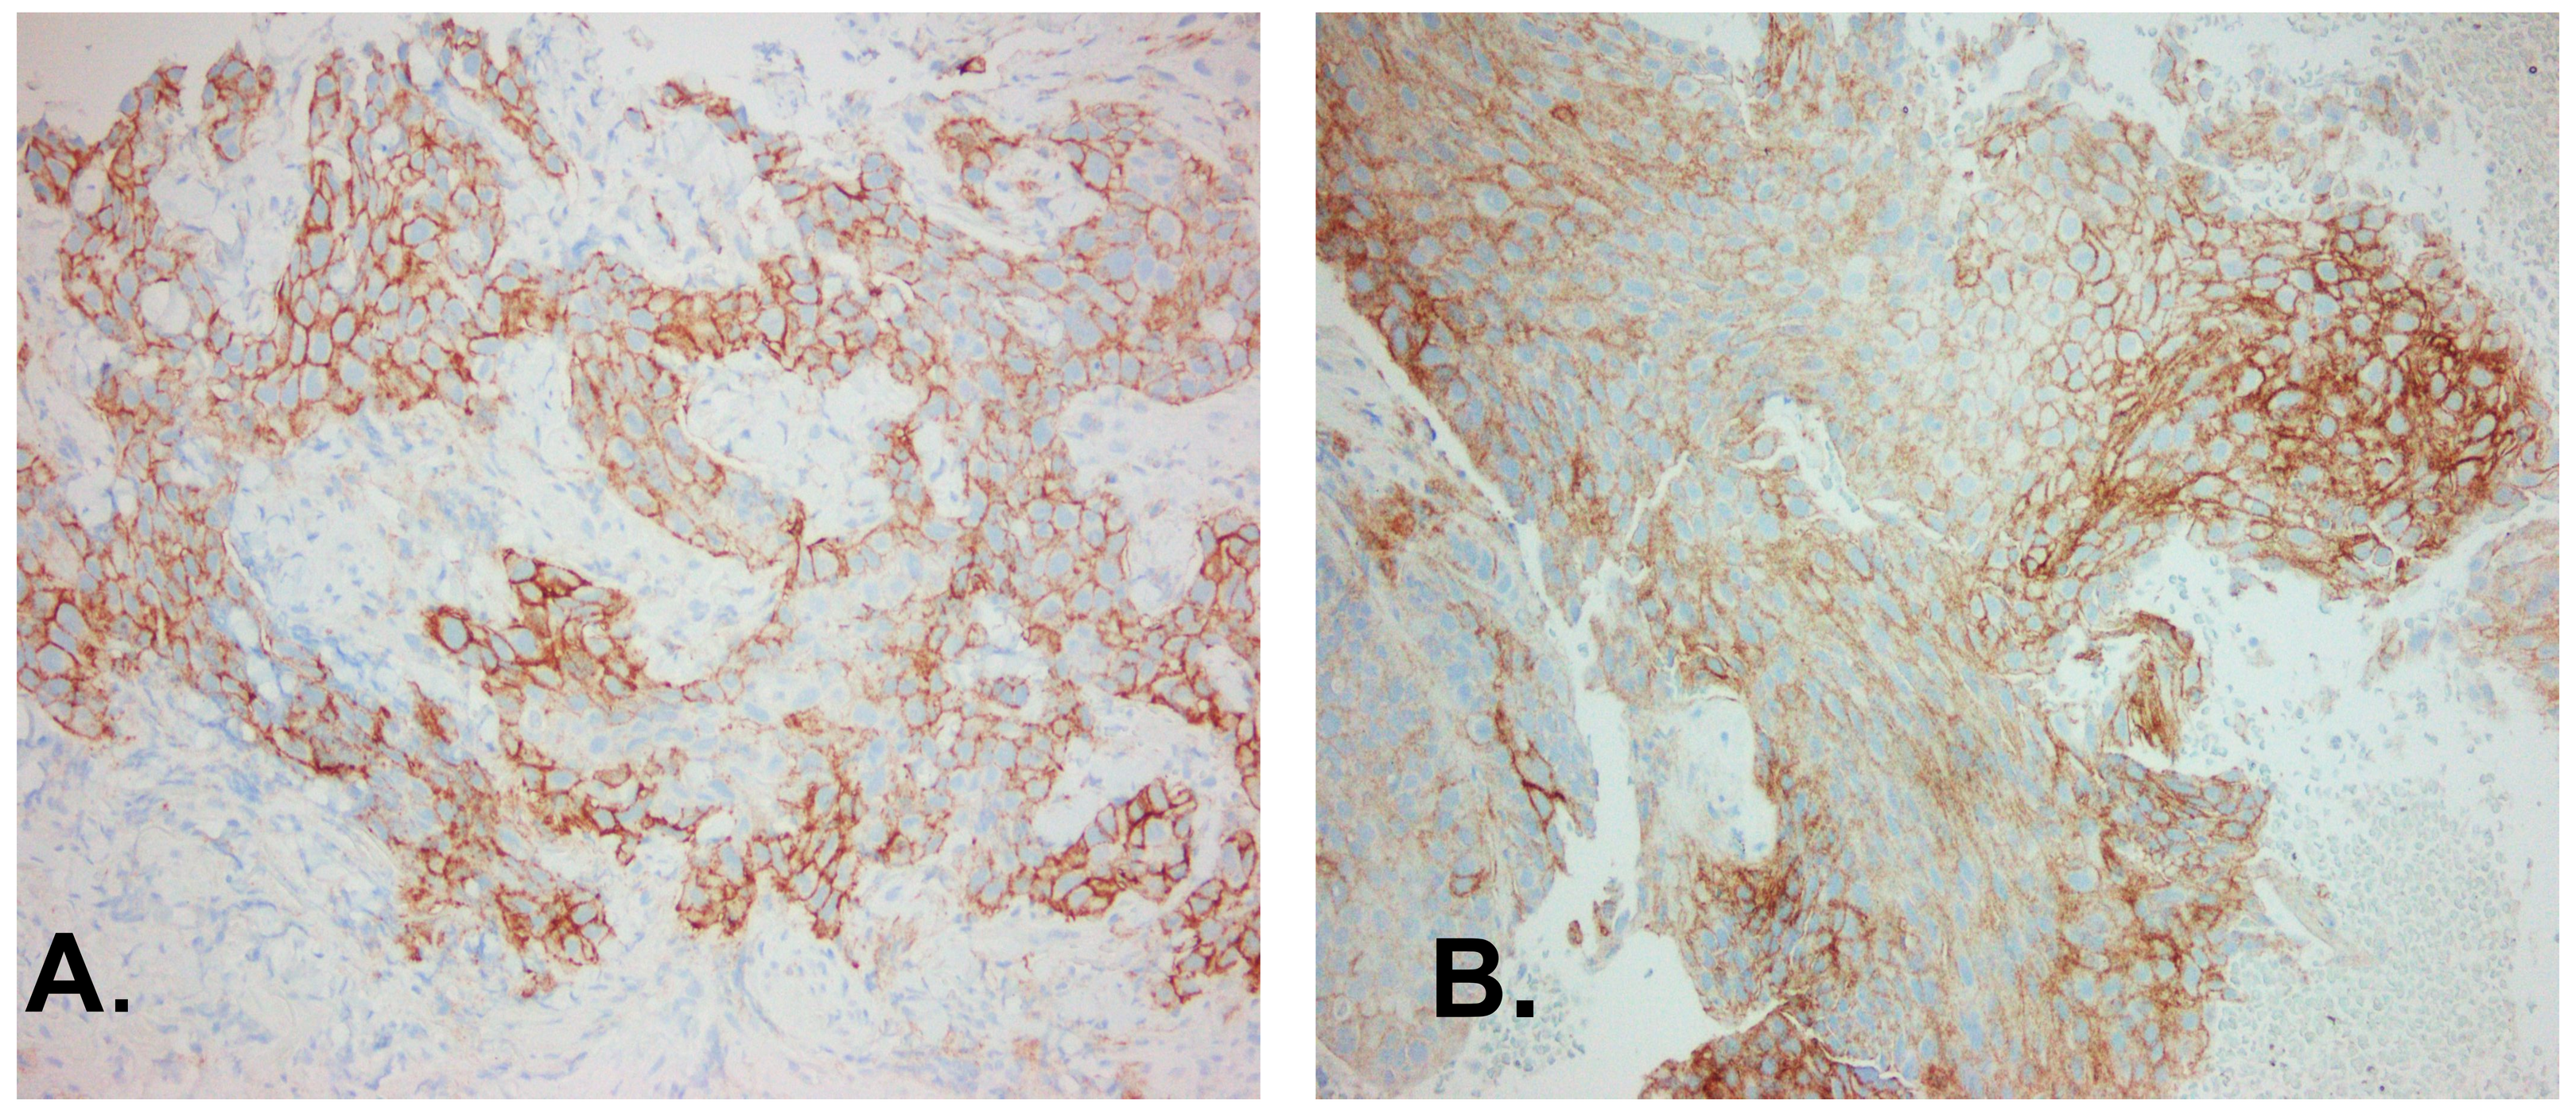
A.
B.

Figure 4. Representative PD-L1 staining images (A,B) from two HRAS mutant tumors with concomitant high PD-L1 expression..

Conclusions

-HNSCC TCGA transcriptomic data, indicate that HPV- tumors with mutant HRAS display higher levels of CD8A+ infiltration compared to wild type HRAS tumors.

-The pan-cancer analysis indicate that the association between HRAS oncogenic mutations and higher CD8A+ infiltration is statistically significant only in HPV- HNSCC among different cancer types.

-Our experimental data indicate that HRAS mutant HNSCCs are associated with high PD-L1 expression. Combinations of tipifarnib with an anti-PD1 antibody, an anti-PDL1 antibody, or an anti-CTLA-4 antibody warrants clinical investigation in this rare subset of HNSCC.

References

1. Cox AD and Der CJ. 2010. "Ras history: The saga continues". Small GTPases 1(1):2-27.
2. Li T et al., TIMER2.0 for analysis of tumor-infiltrating immune cells. Nucleic Acids Res. 2020
3. Liu CC et al., Computational approaches for characterizing the tumor immune microenvironment. Immunology. 2019
4. Li B et al., Comprehensive analyses of tumor immunity: implications for cancer immunotherapy. Genome Biol. 2016

Acknowledgements

- We thank KURA ONCOLOGY Inc for their support on this study.