THE CLINICAL MENIN INHIBITOR ZIFTOMENIB AND THE NUCLEAR EXPORT INHIBITOR SELINEXOR SYNERGISTICALLY INHIBIT THE GROWTH OF MLL-R AML

M. UDDIN[1], A. ABOUKAMEEL[1], S. DHIMAN [1], H. KHAN[1], S. SIDDIQUI [1], S. BANNOURA[1], A. DEOL[1], J. YANG[1], G. DYSON[1], A. AZMI[1], L. POLIN[1], J. MACIEJEWSKI[2] and S. BALASUBRAMANIAN[1,2]

1. Department of Oncology, Karmanos Cancer Institute, Wayne State University, Detroit, MI.
2. Translational Hematology and Oncology Research, Cleveland Clinic Foundation/ Taussig Cancer Institute, Cleveland, OH.

INTRODUCTION

- Menin is a scaffold protein that interacts with oncogenic histone-lysine-N-methyltransferase MLL1 (KMT2A)-fusion protein (FP) complex in MLL-rearranged (MLL-r) AML.
- Menin inhibitors that evict menin from this interaction have been shown to be active against MLL-r and NPM1MT leukemia by modulating the expression of the leukemogenic homeobox A9 (HOXA9) gene and its co-factor, MEIS1 (top and bottom panel).
- Recent evidence showed that menin inhibitors can activate a tumor-suppressive network via a non-canonical transcriptional program through UTX (bottom panel).

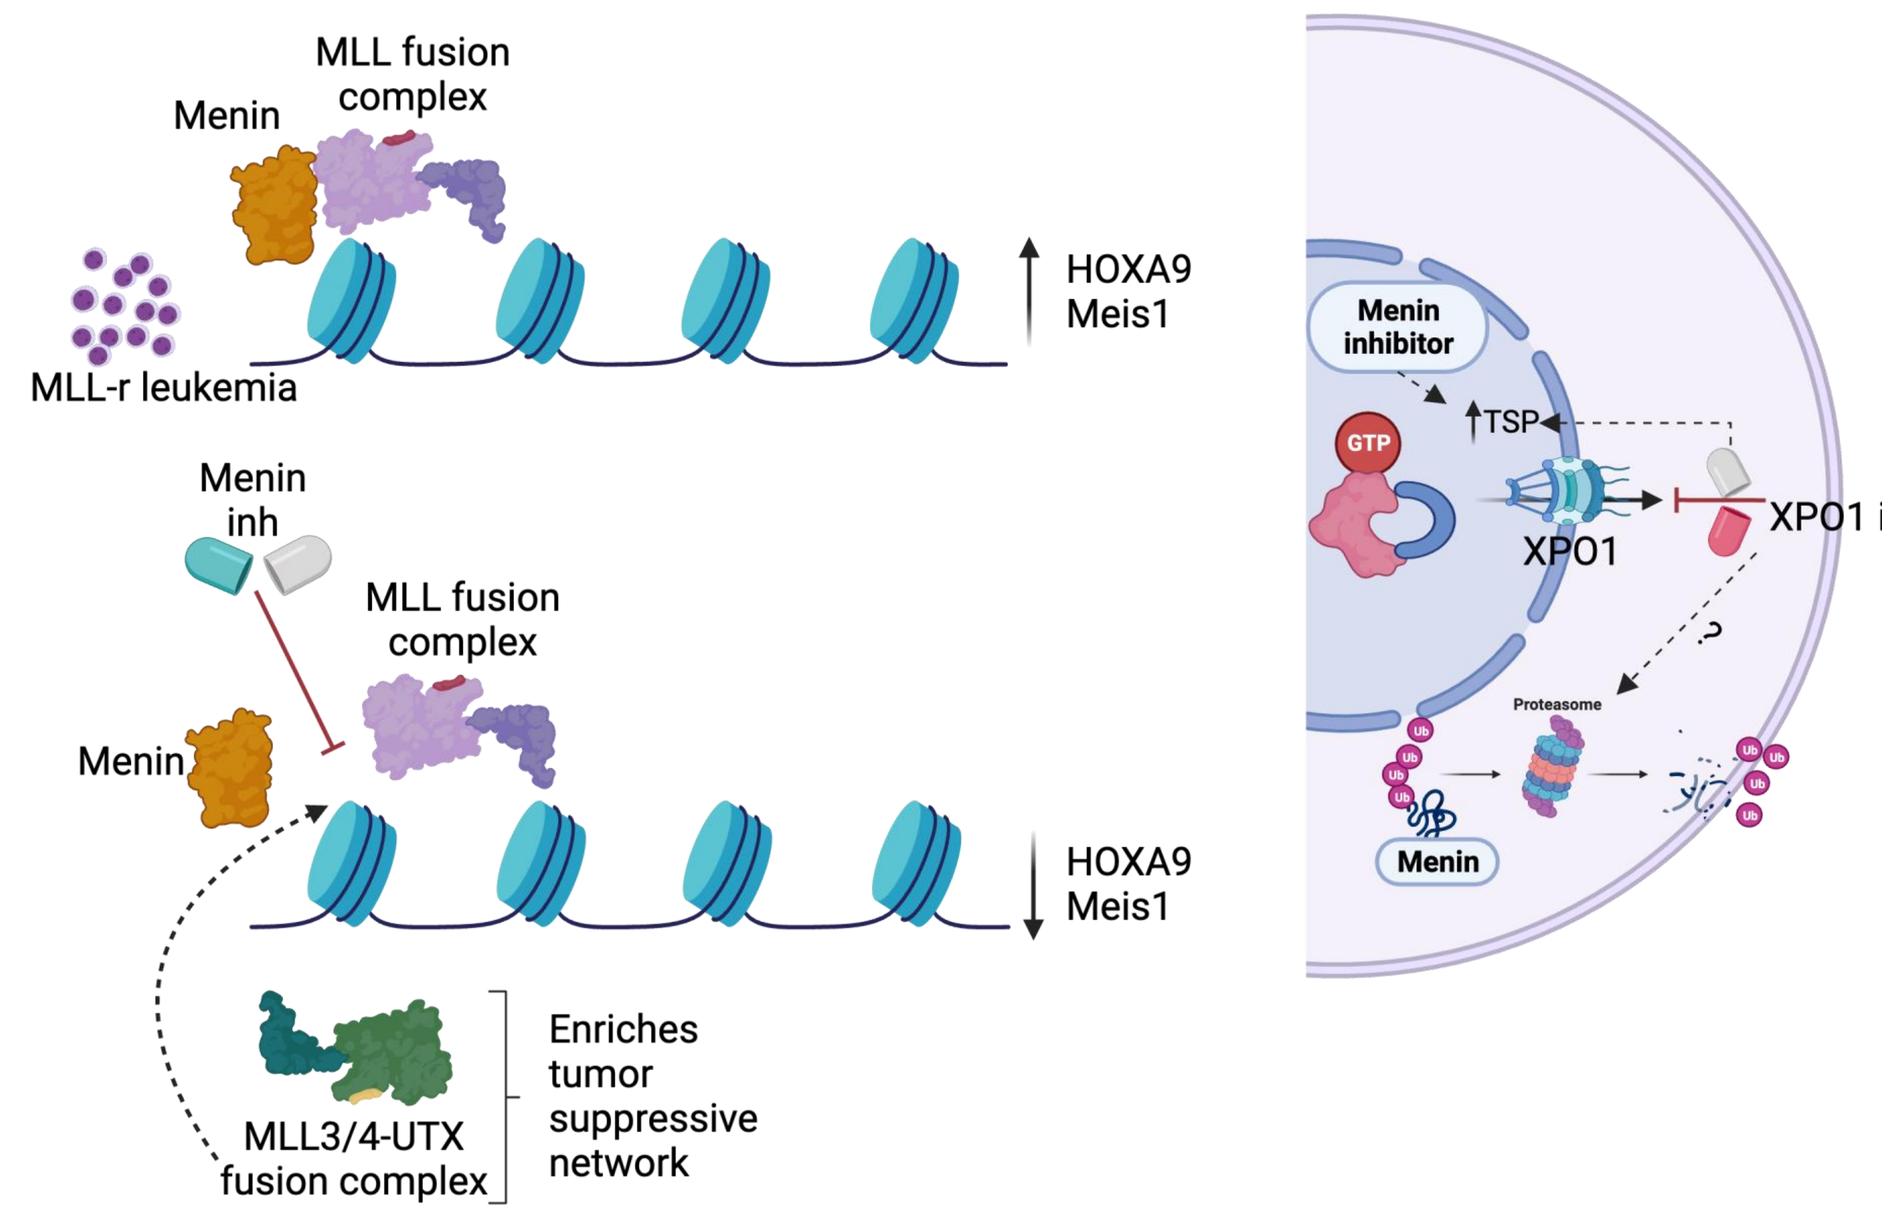
MLL fusion complex
Menin
MLL-r leukemia
HOXA9
Meis1
Menin inh
MLL fusion complex
Menin
HOXA9
Meis1
MLL3/4-UTX fusion complex
Enriches tumor suppressive network
Menin inhibitor
GTP
TSP
XPO1
XPO1 inh
Proteasome
Menin

- On the other hand, selective inhibitor of nuclear export (SINE) against XPO1/CRM1 has been shown to have robust anti-leukemia activity via enrichment of tumor suppressor proteins in the nucleus (right panel).
- In the present study, we hypothesized that menin and nuclear export inhibition would synergistically suppress AML cell proliferation and possibly sensitize menin inhibitor-resistant cells.
- We have used the clinical-stage menin inhibitor ziftomenib and SINE compound selinexor to simultaneously target menin-KMT2A protein-protein interactions and nuclear export.

RESULTS

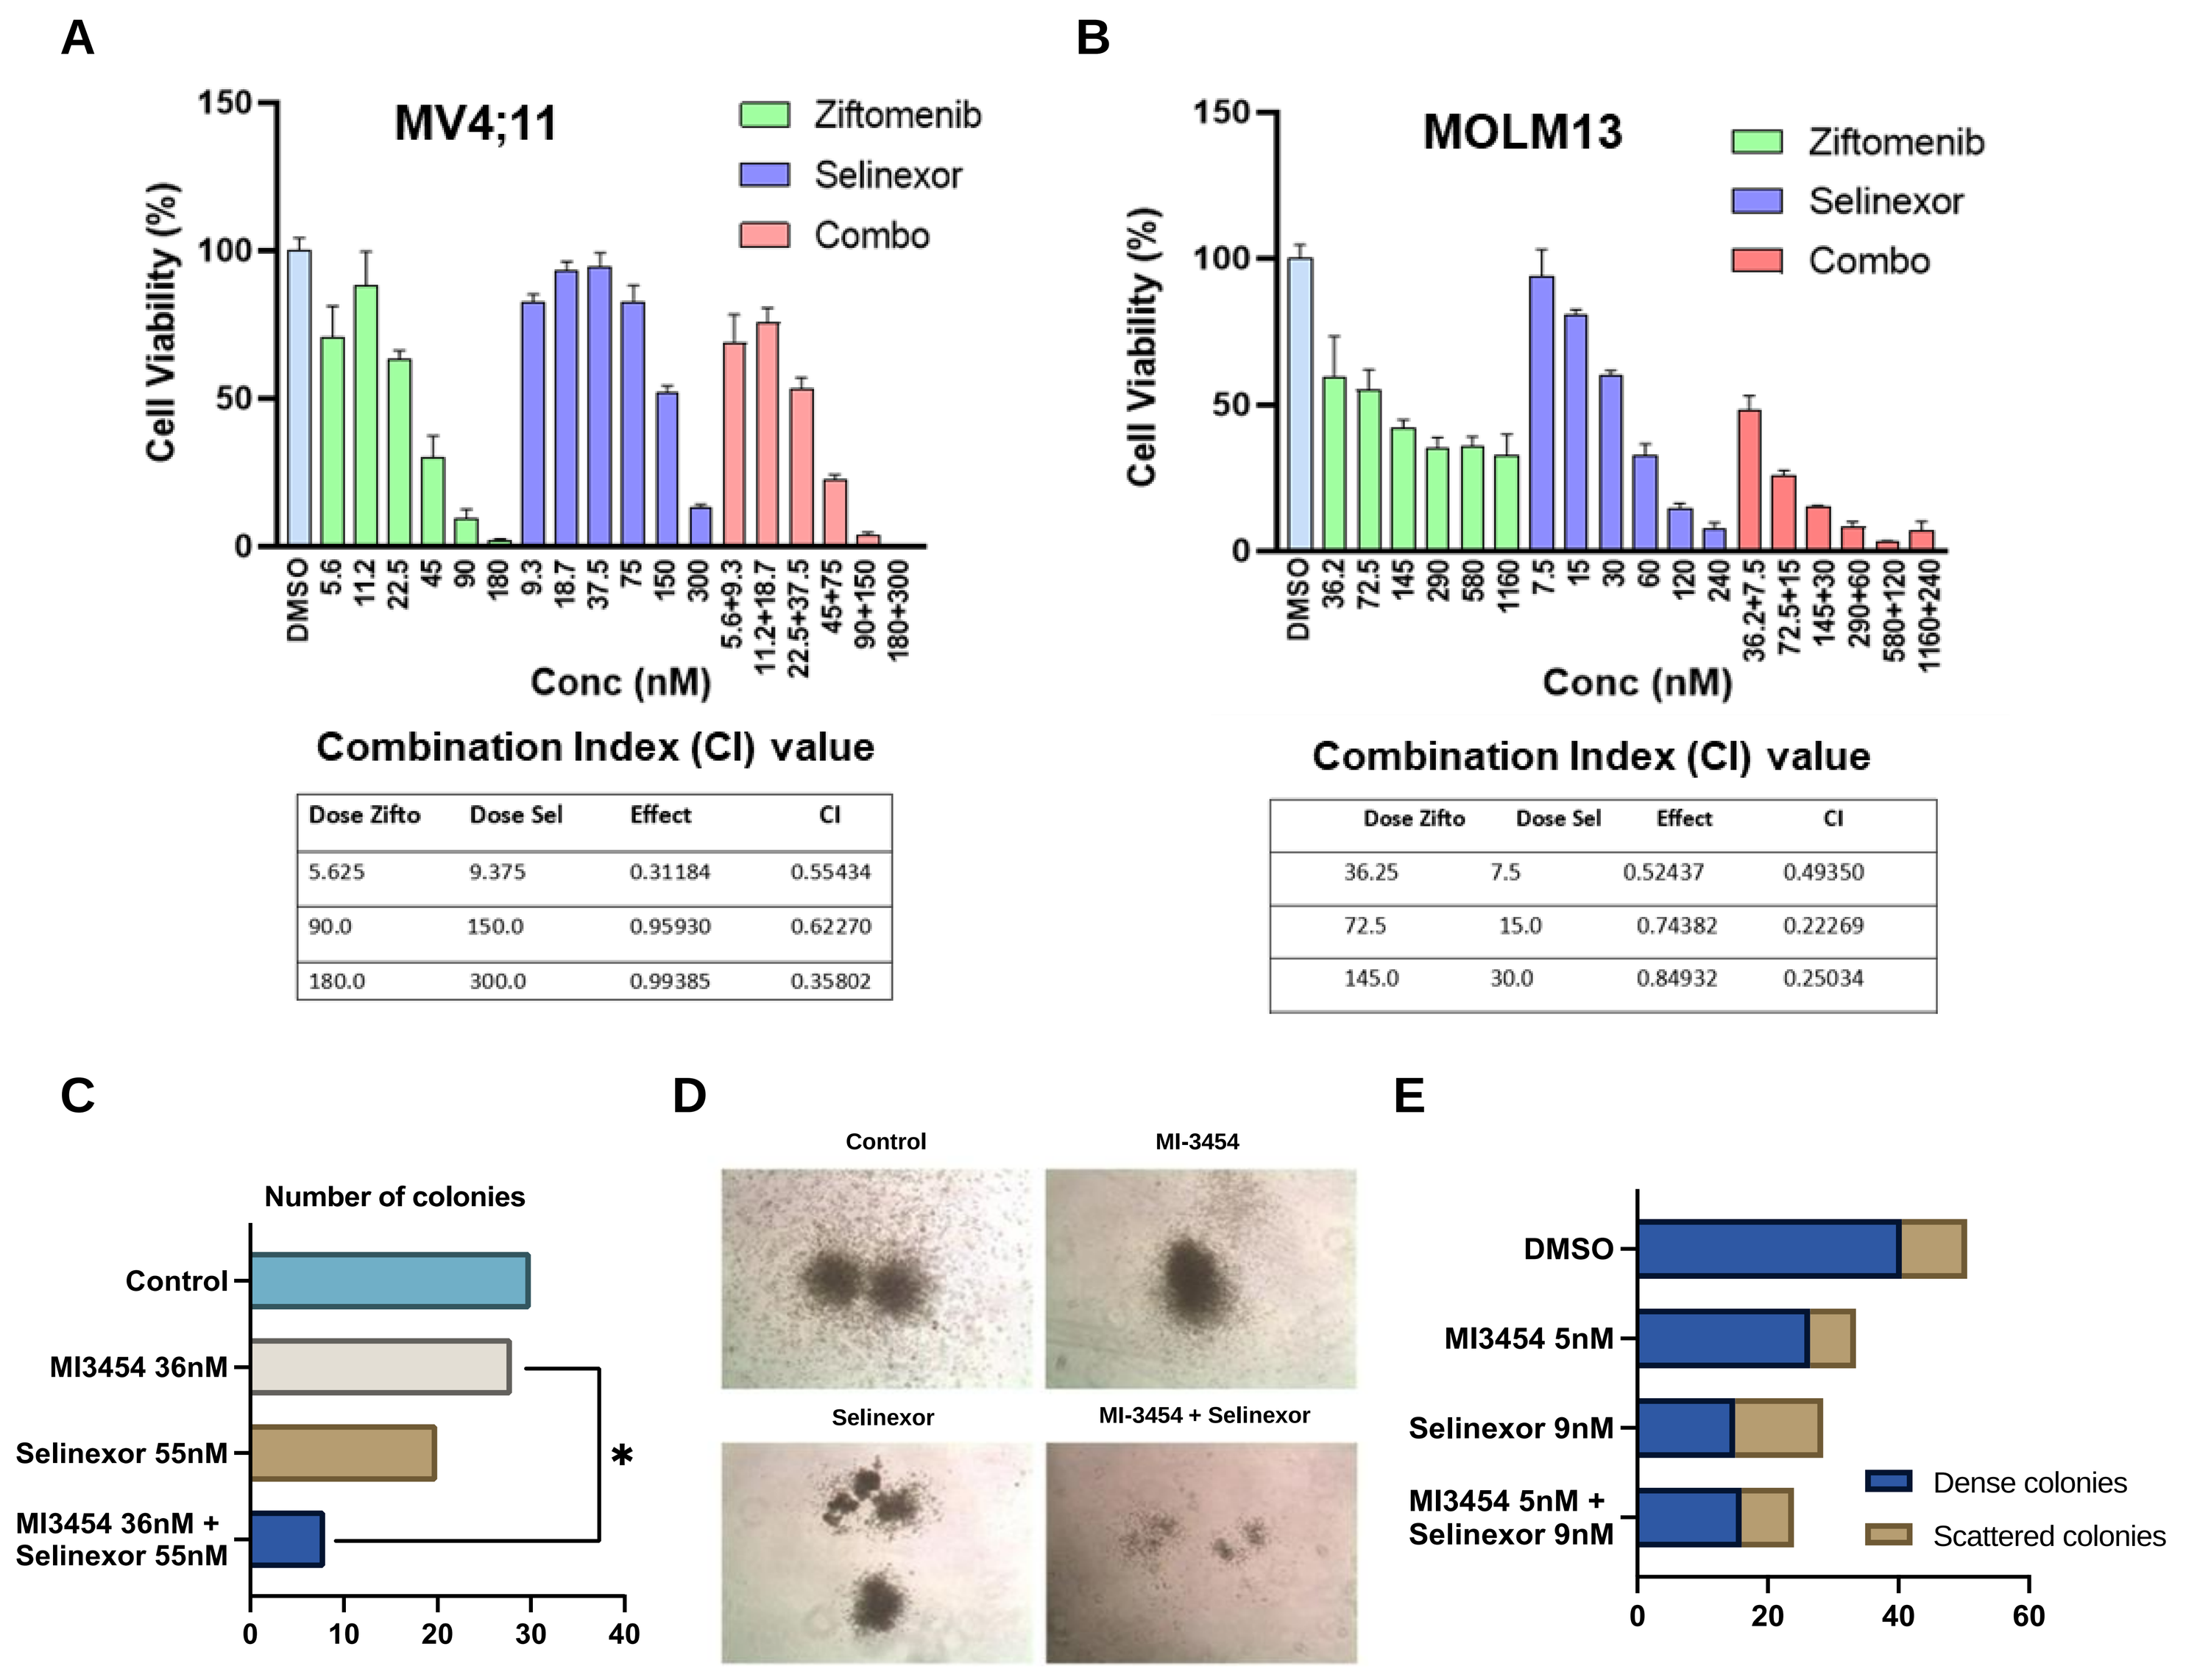
A
MV4;11
Ziftomenib
Selinexor
Combo
Cell Viability (%)
Conc (nM)
B
MOLM13
C
Number of colonies
Control
MI3454 36nM
Selinexor 55nM
MI3454 36nM + Selinexor 55nM
D
Control
MI-3454
Selinexor
MI-3454 + Selinexor
E
DMSO
MI3454 5nM
Selinexor 9nM
MI3454 5nM + Selinexor 9nM
Dense colonies
Scattered colonies

Combination Index (CI) value (MV4;11)

Dose Zifto	Dose Sel	Effect	CI
5.625	9.375	0.31184	0.55434
90.0	150.0	0.95930	0.62270
180.0	300.0	0.99385	0.35802

Combination Index (CI) value (MOLM13)

Dose Zifto	Dose Sel	Effect	CI
36.25	7.5	0.52437	0.49350
72.5	15.0	0.74382	0.22269
145.0	30.0	0.84932	0.25034

Figure 1: Menin and nuclear export (XPO1) inhibitors synergistically inhibit the growth of MLL-r AML. [A-B] MV4;11 and MOLM13 cells were treated with varying concentrations of ziftomenib or selinexor for 72 hrs (upper panels). Cell titer glo assay was performed to determine the growth inhibition. The lower panel shows the combination index (CI) values generated from Calcusyn 2.1 software. [C] The combination of menin and XPO1 inhibitors in the suppression of colony formation of CD34+ MLL-r progenitor cells derived from primary patient samples. [D] Representative colonies from all four groups. [E] The combination of menin and XPO1 inhibitors in the suppression of both dense and scattered colony formation of CD34+ MLL-r progenitor cells derived from primary patient samples.

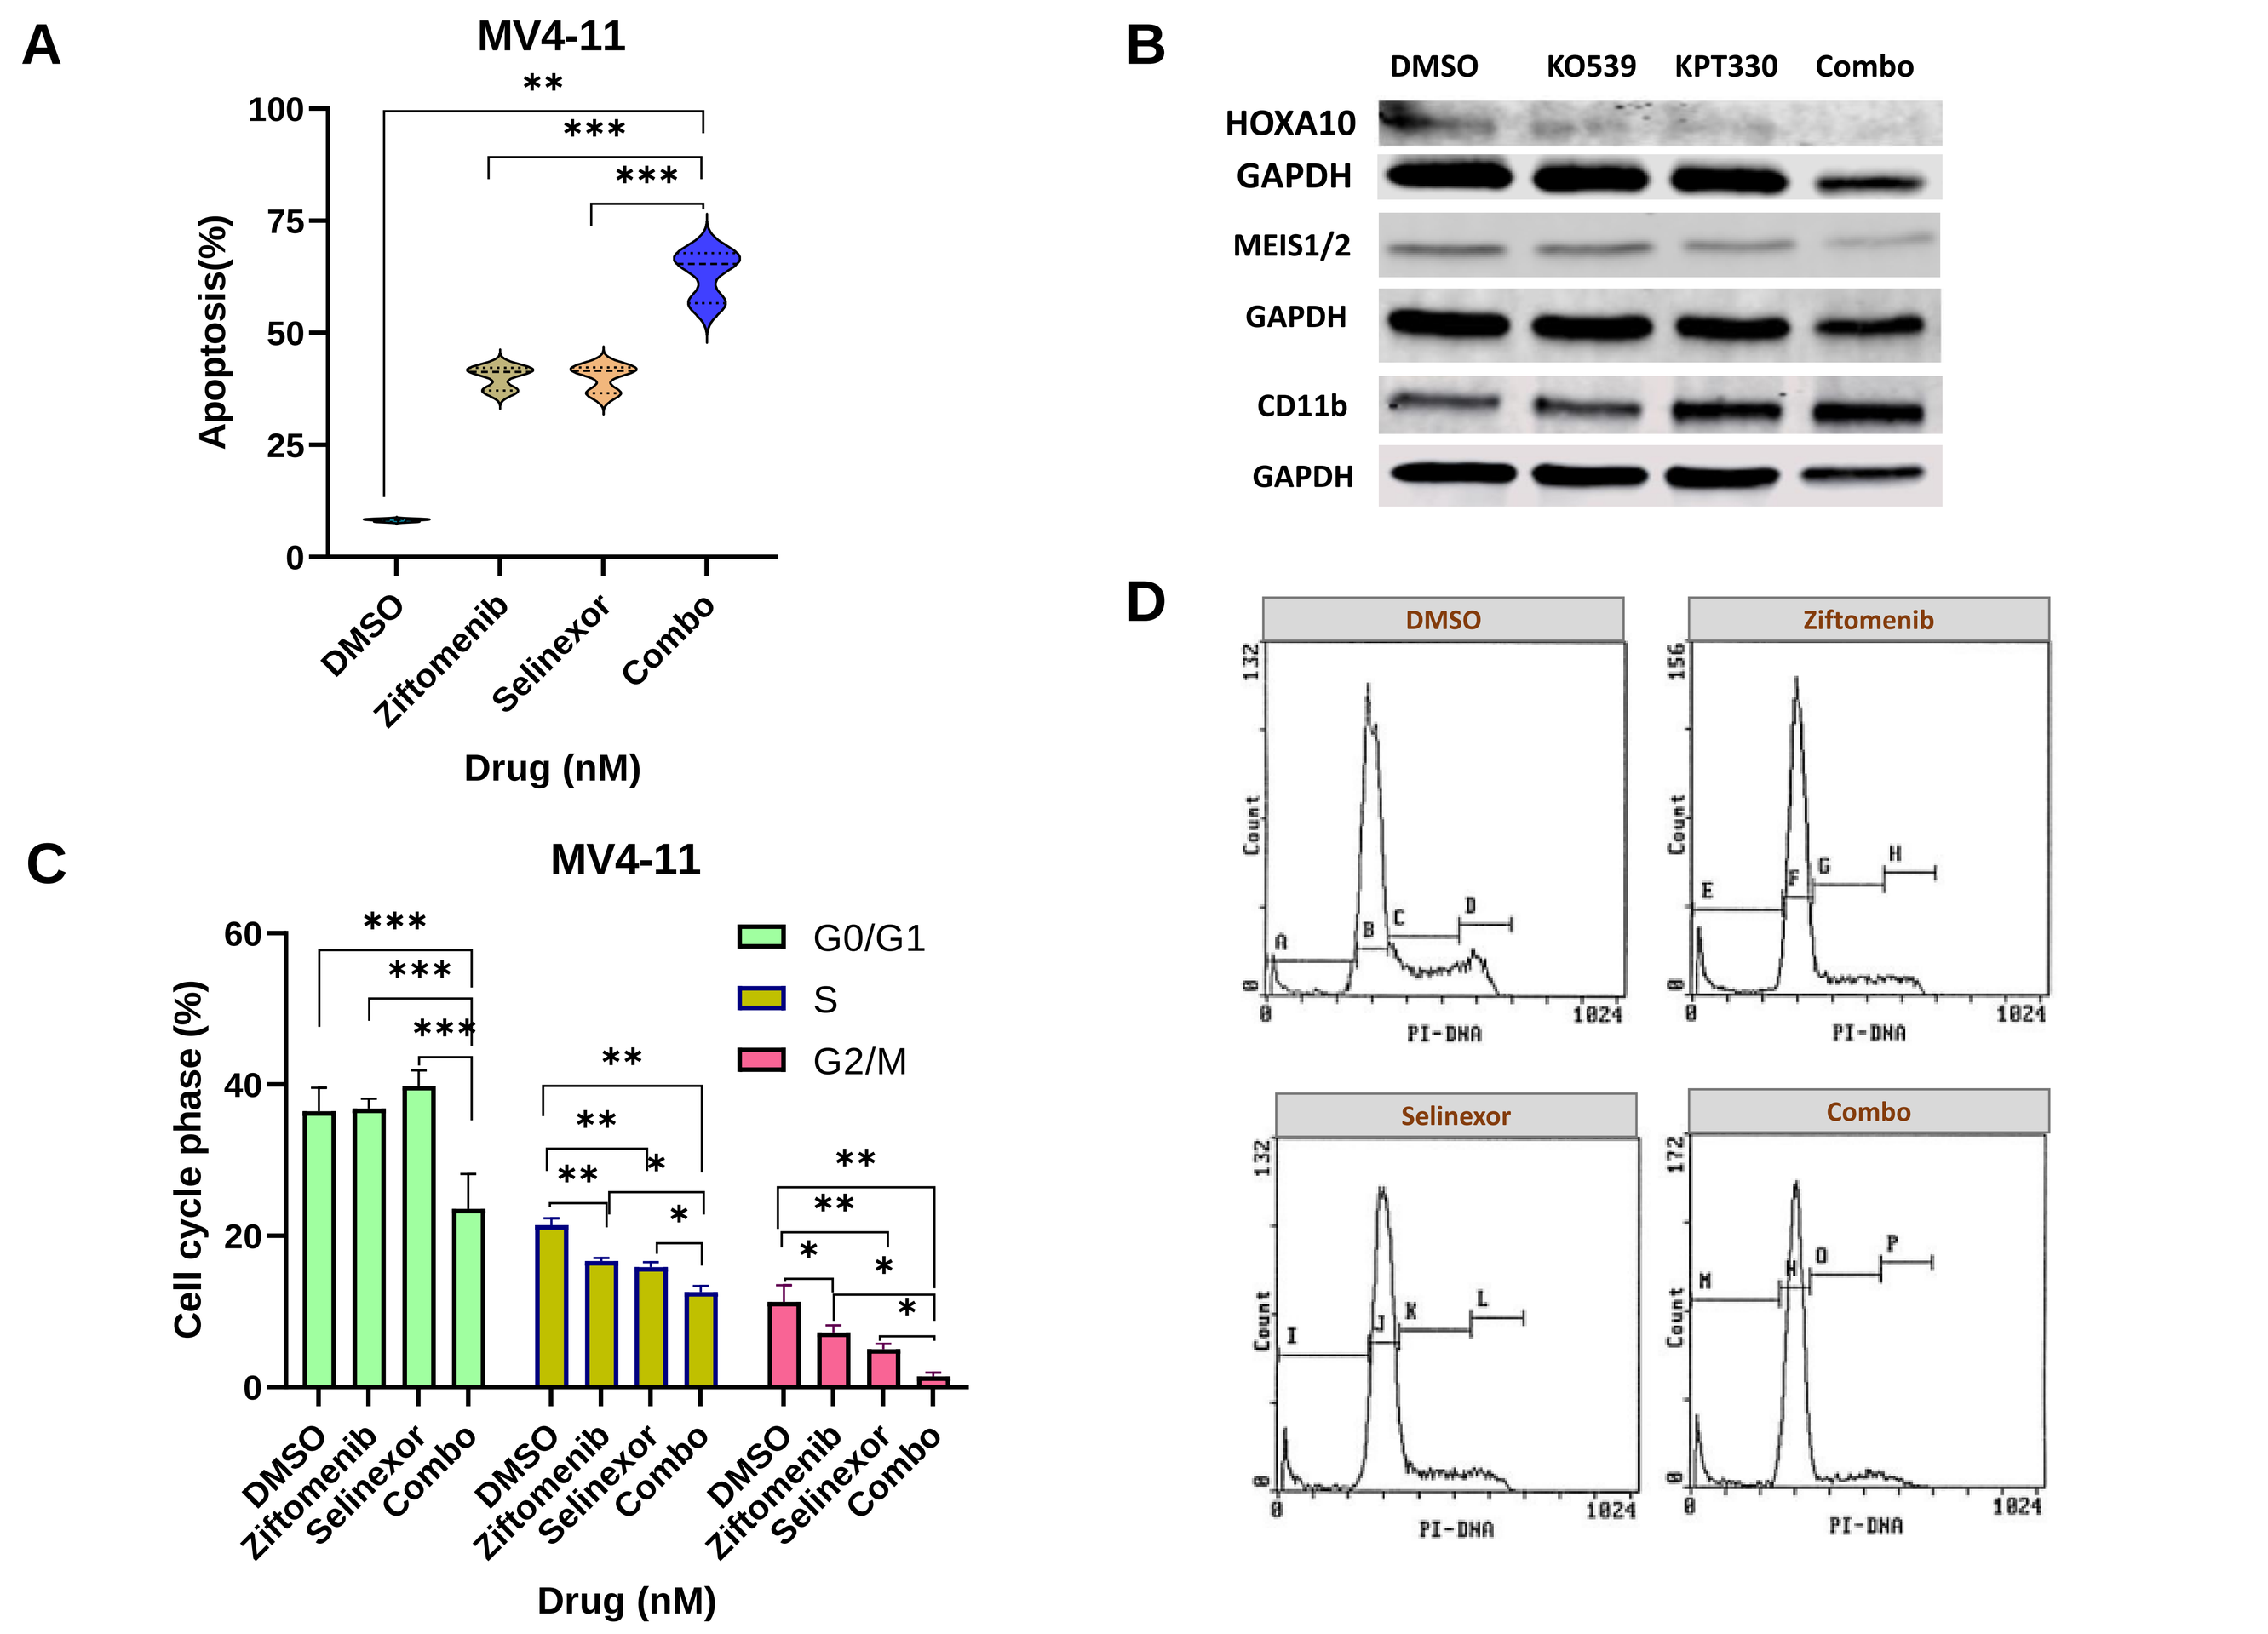
A
MV4-11
Apoptosis(%)
DMSO
Ziftomenib
Selinexor
Combo
Drug (nM)
B
DMSO
KO539
KPT330
Combo
HOXA10
GAPDH
MEIS1/2
GAPDH
CD11b
GAPDH
C
MV4-11
Cell cycle phase (%)
G0/G1
S
G2/M
Drug (nM)
D
DMSO
Ziftomenib
Selinexor
Combo
PI-DNA

Figure 2: [A] Apoptotic cell death with ziftomenib and selinexor in MV4;11 AML cells. Apoptosis was determined by the flow cytometric detection of annexin-V and propidium iodide (PI). [B] Western blot analysis. [C] Cell cycle analysis from ziftomenib and selinexor treatment alone or in combination. [D] Representative flow cytometric image of cell cycle. *, *p* < .05 ; **, *p* < .01; *, *p* < .001.**

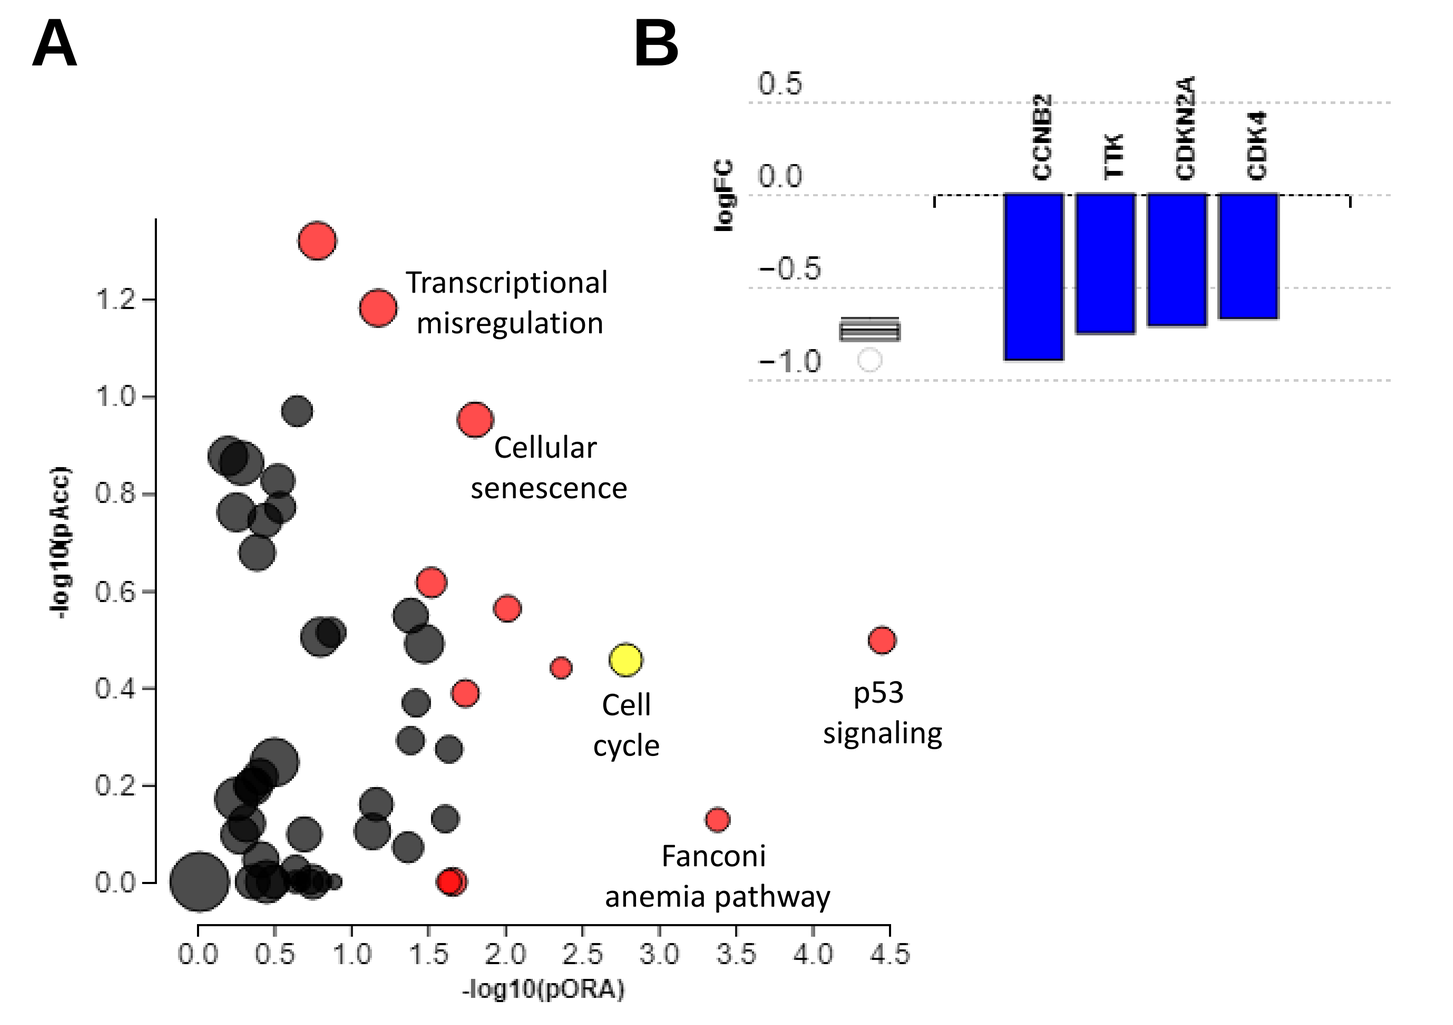
A
B
Transcriptional misregulation
Cellular senescence
Cell cycle
p53 signaling
Fanconi anemia pathway
-log10(pAcc)
-log10(pORA)
logFC
CCNB2
TTK
CDKN2A
CDK4

Figure 3: Affected pathways in the combination compared to control from proteomic data analysis. [A] Pathways that are significantly perturbed shown in red or yellow circle. Diameter of the circle is the representation of differentially expressed protein number. [B] Differentially expressed proteins in cell cycle regulatory pathways.

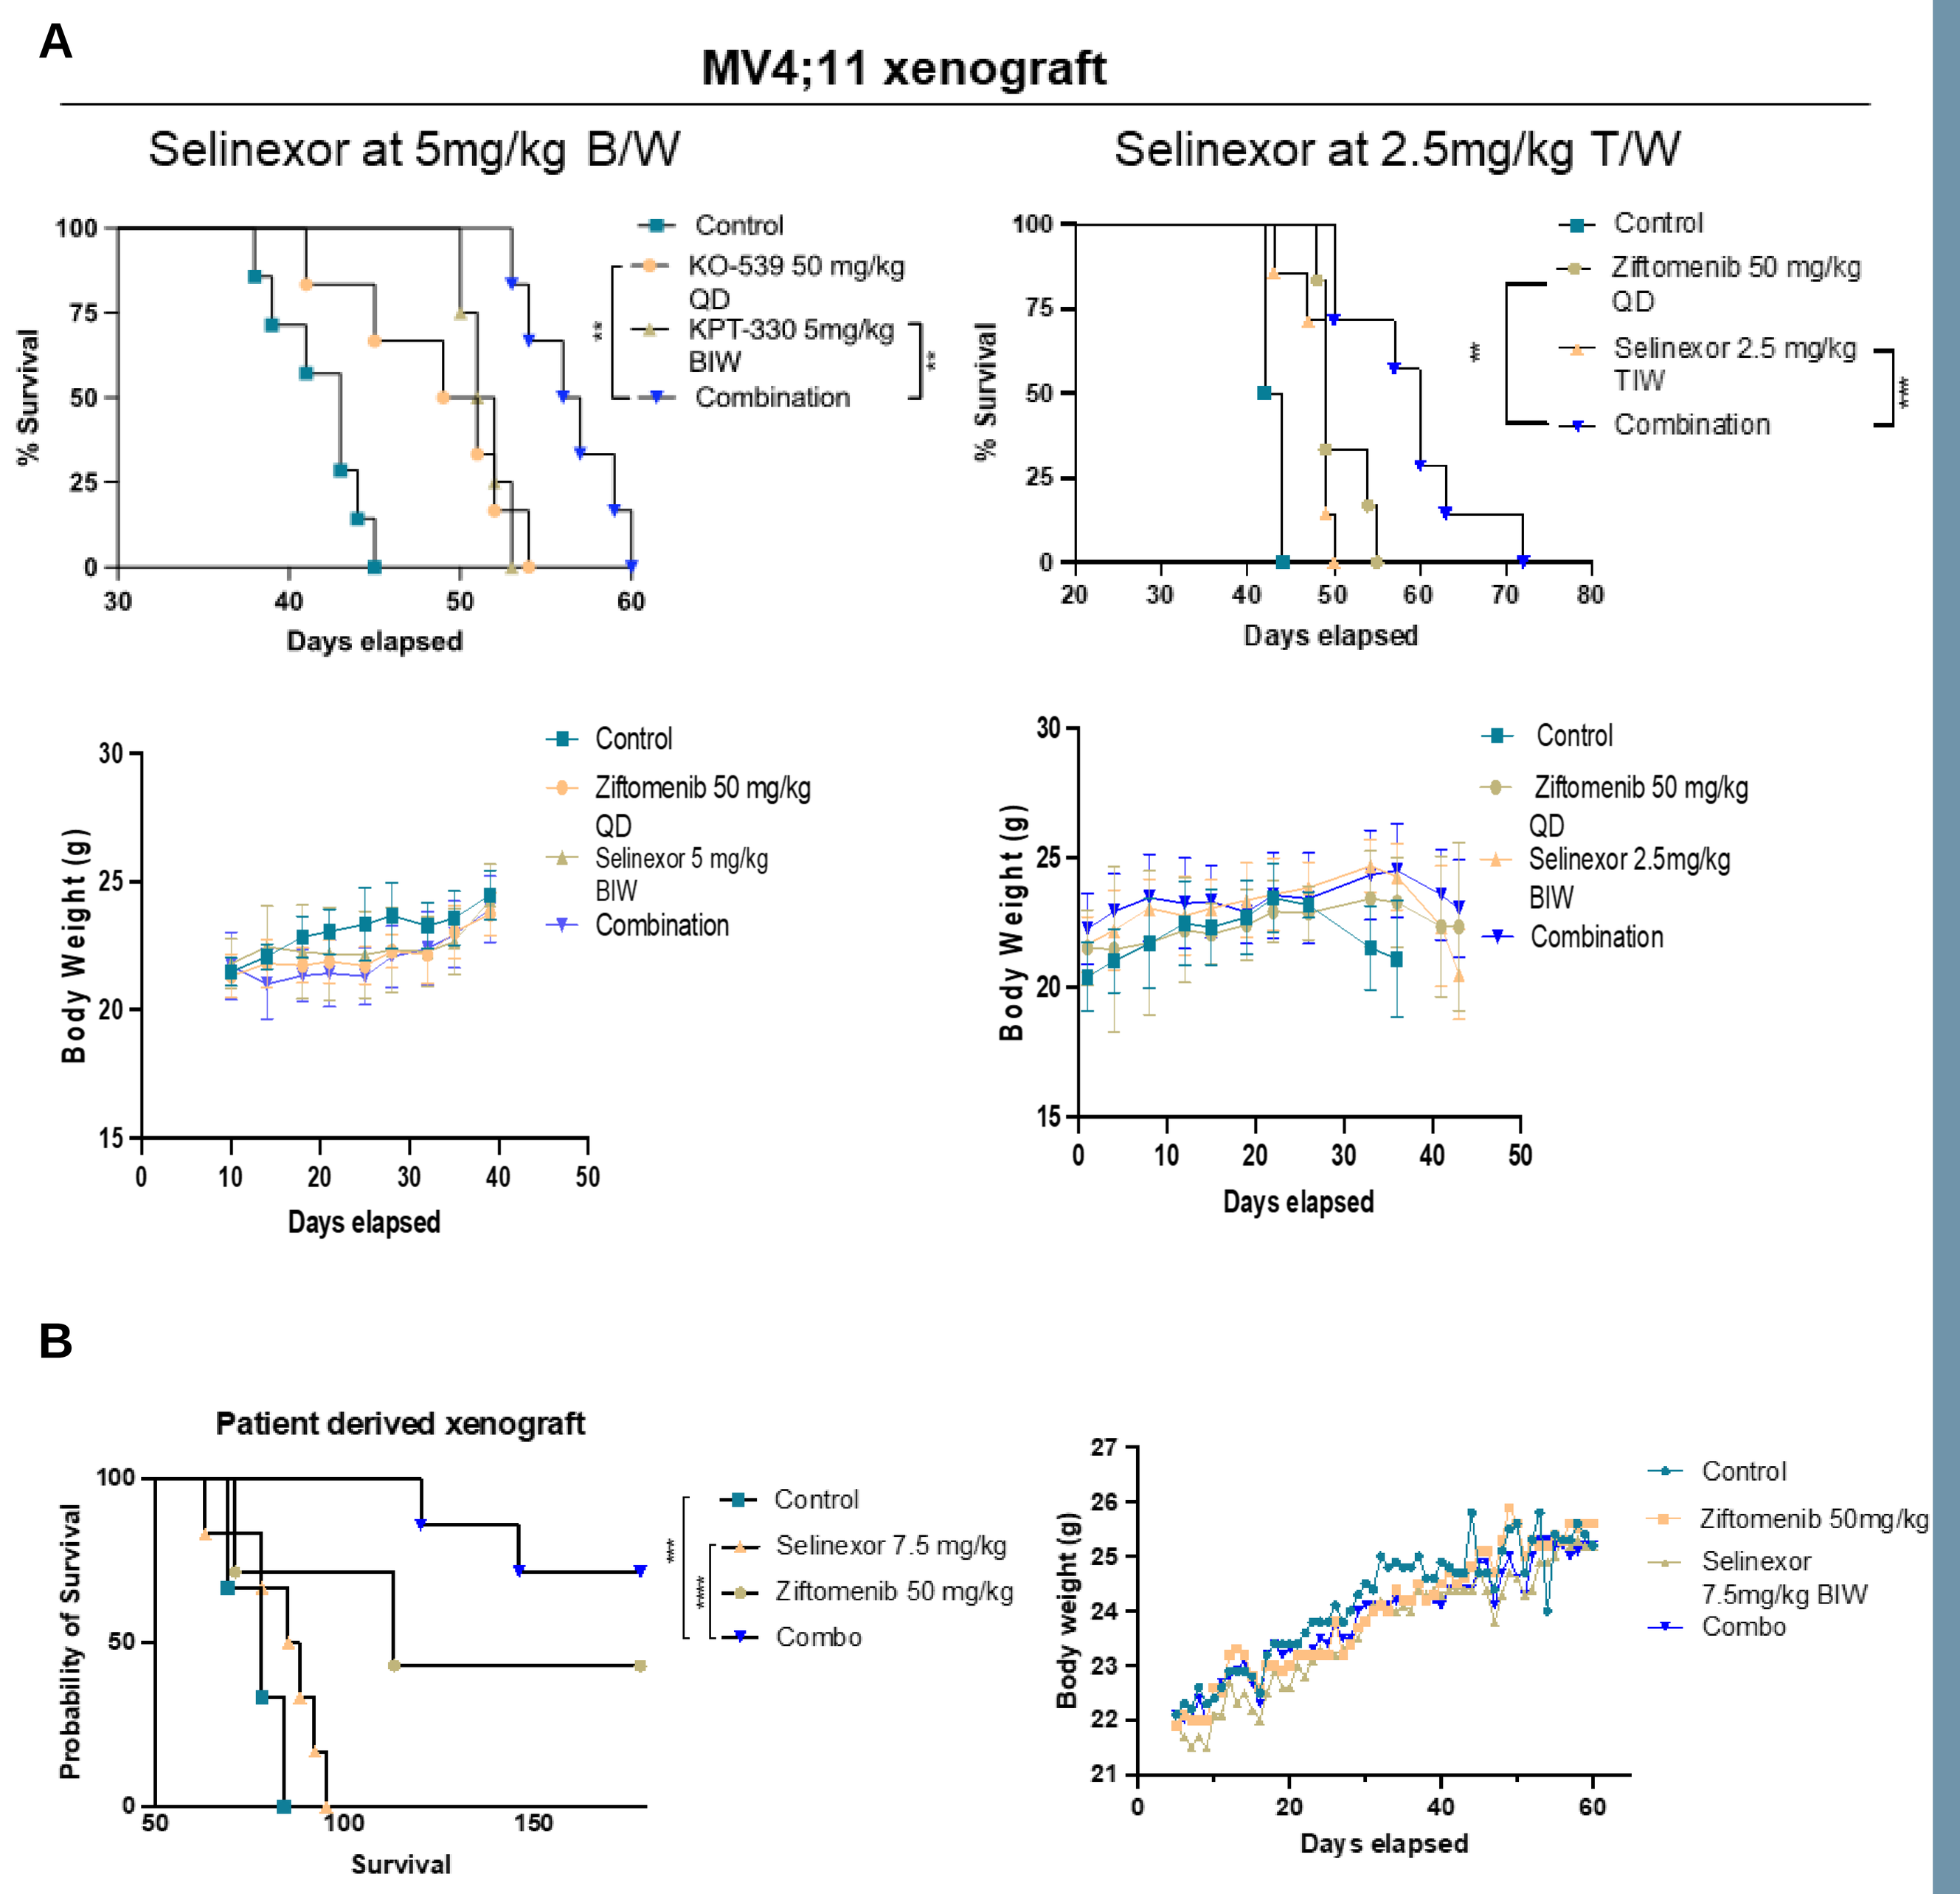
A
MV4;11 xenograft
Selinexor at 5mg/kg B/W
Selinexor at 2.5mg/kg T/W
% Survival
Days elapsed
Control
KO-539 50 mg/kg QD
KPT-330 5mg/kg BIW
Combination
Ziftomenib 50 mg/kg QD
Selinexor 2.5 mg/kg TIW
Body Weight (g)
Selinexor 5 mg/kg BIW
Selinexor 2.5mg/kg BIW
B
Patient derived xenograft
Probability of Survival
Survival
Selinexor 7.5 mg/kg
Ziftomenib 50 mg/kg
Combo
Body weight (g)
Ziftomenib 50mg/kg
Selinexor 7.5mg/kg BIW

Figure 4: [A] Cell line derived xenografts (CDX) in NSG mice using GFP/Luciferase expressing MV4;11 cells. About 2 million cells were injected through the tail vein. Mice were randomized based on the luciferase intensity on bioluminescent imaging on Day 8 and received a fixed dose of KO-539/Ziftomenib and different doses of selinexor. Survival of vehicle or inhibitors treated mice (upper panel). Body weight (lower panel) [B] Patient derived xenografts (PDX) in NSG mice. 0.75 Million Primary MLL^r cells were injected through the tail vein. Mice received 50 mg/kg dose of KO-539/Ziftomenib and 7.5 mg/kg dose of selinexor. Survival of vehicle or inhibitors treated mice (left panel). Body weight (right panel).

METHODS

- An ATP-based cell proliferation assay was used to assess growth inhibition.
- Combination synergy was determined using CalcuSyn Version 2.0 synergy software.
- Stem-like progenitor cells were isolated using the StemSpan CD34+ expansion kit (StemCell Tech).
- Colony formation efficiency was determined using a Methocult assay (StemCell Tech).
- Gene and protein expression and cell death were detected using quantitative real-time PCR, western blotting.
- Flow cytometric analysis was performed to detect cell death and cell cycle status.
- We performed whole transcriptomic analysis via RNA sequencing approach and global proteomic analysis.

CONCLUSIONS

- The clinical candidate ziftomenib, in combination with selinexor, synergistically inhibited the growth of MLL-r AML cells (MV4;11, MOLM13, and SEM; CI < 1).
- The combination affected cell cycle pathway via the downregulation of multiple proteins, including CDK4.
- These preclinical findings demonstrate that simultaneous inhibition of the menin-KMT2A interaction and nuclear export is a viable strategy for treating MLL-r AML.
- Further studies on menin inhibitor resistance cells and other xenograft studies are ongoing.

REFERENCES

Klossowski S, et al. Menin inhibitor MI-3454 induces remission in MLL1-rearranged and NPM1-mutated models of leukemia. J Clin Invest. 2020;130(2):981-997. https://doi.org/10.1172/JCI129126.

Azmi A, et al. The nuclear export protein XPO1 - from biology to targeted therapy. Nat Rev Clin Oncol. (2021) 18(3):152-169. doi: 10.1038/s41571-020-00442-4.

Fiskus W, et al. Effective Menin inhibitor-based combinations against AML with MLL rearrangement or NPM1 mutation (NPM1c). Blood Can. J. (2022) 12:5;https://doi.org/10.1038/s41408-021-00603-3.

Barajas J, et al. Acute myeloid leukemias with UBTF tandem duplications are sensitive to Menin inhibitors. Blood (2023). Tracking no: BLD-2023-021359R1. https://doi.org/10.1182/blood.2023021359.

Kollmann S, et al. A STAT5B–CD9 axis determines self-renewal in hematopoietic and leukemic stem cells. Blood (2021) 138:23.

Aubrey B, et al. IKAROS and MENIN coordinate therapeutically actionable leukemogenic gene expression in MLL-r acute myeloid leukemia. Nature Cancer (2022) 3: 595–613.

ACKNOWLEDGEMENTS

Funding source: 1. Kura Oncology

2. 5P30CA022453-41 [CCSG KCI]

Figure (I),(II)&(III) were created with BioRender.com

@malignantheme

KARMANOS CANCER INSTITUTE