A phase 1 clinical trial of the novel farnesyl transferase inhibitor KO-2806 (FIT-001) alone or as part of combination therapy for advanced solid tumors

Abstract presentation CT163

Glenn J Hanna[1], Douglas R Adkins[2], Jacob S Thomas[3], Justine Y Bruce[4], Manish R Patel[5], Guru Sonpavde[6], Jason Henry[7], Nawal Bendris[8], Zijing Zhang[8], Amitava Mitra[8], Andrew Saunders[8], Stephen Dale[8]

[1]Dana-Farber Cancer Institute, Harvard Medical School, Boston, MA, USA; [2]Washington University School of Medicine, St Louis, MO, USA; [3]University of Southern California, Norris Comprehensive Cancer Center, Los Angeles, CA, USA; [4]University of Wisconsin Carbone Cancer Center, Madison, WI, USA; [5]Florida Cancer Specialists, Sarah Cannon Research Institute, Sarasota, FL, USA; [6]AdventHealth Cancer Institute and the University of Central Florida, Orlando, FL, USA; [7]Sarah Cannon Research Institute at HealthONE, Denver, CO, USA; [8]Kura Oncology, Inc., Boston, MA, USA

BACKGROUND

- RAS mutations are found in a wide range of human cancers, including lung, colon and pancreatic cancers; approximately 30% of human tumors express a mutation in a RAS gene.[1,2]
- HRAS, unlike NRAS and KRAS, is completely dependent on the post-translational modification farnesylation for downstream activity, making it particularly sensitive to farnesyl transferase inhibitors (FTIs).[2,3]
- Clinical trials (NCT02383927, NCT03719690) of the FTI tipifarnib in patients with *HRAS*-mutant (-m) head and neck squamous cell carcinoma (HNSCC) with high-variant allele frequency mutations (20% or higher) had an objective response rate (ORR) of 52.6% or 30%, and favorable long-term outcomes.[3–5]
- KO-2806 is a next-generation FTI with increased potency and improved pharmacokinetic (PK) properties compared with current FTIs; in preclinical studies, KO-2806 enhanced the antitumor activity of:
 - tyrosine kinase inhibitors, including cabozantinib, in clear cell renal cell carcinoma (ccRCC) cell line-derived xenograft (CDX) and patient-derived xenograft (PDX) models[6]
 - KRAS inhibitors in *KRAS*-m CDX and PDX models, including adagrasib in non-small cell lung cancer (NSCLC), and MRTX1133 in colorectal cancer (CRC) and pancreatic ductal adenocarcinoma (PDAC).[7,8]

OBJECTIVE

- To assess the safety, tolerability, PK, pharmacodynamics (PD) and preliminary antitumor activity of KO-2806, as a monotherapy and in combination, in adult patients with advanced solid tumors.

FIGURE 1. OVERALL STUDY DESIGN

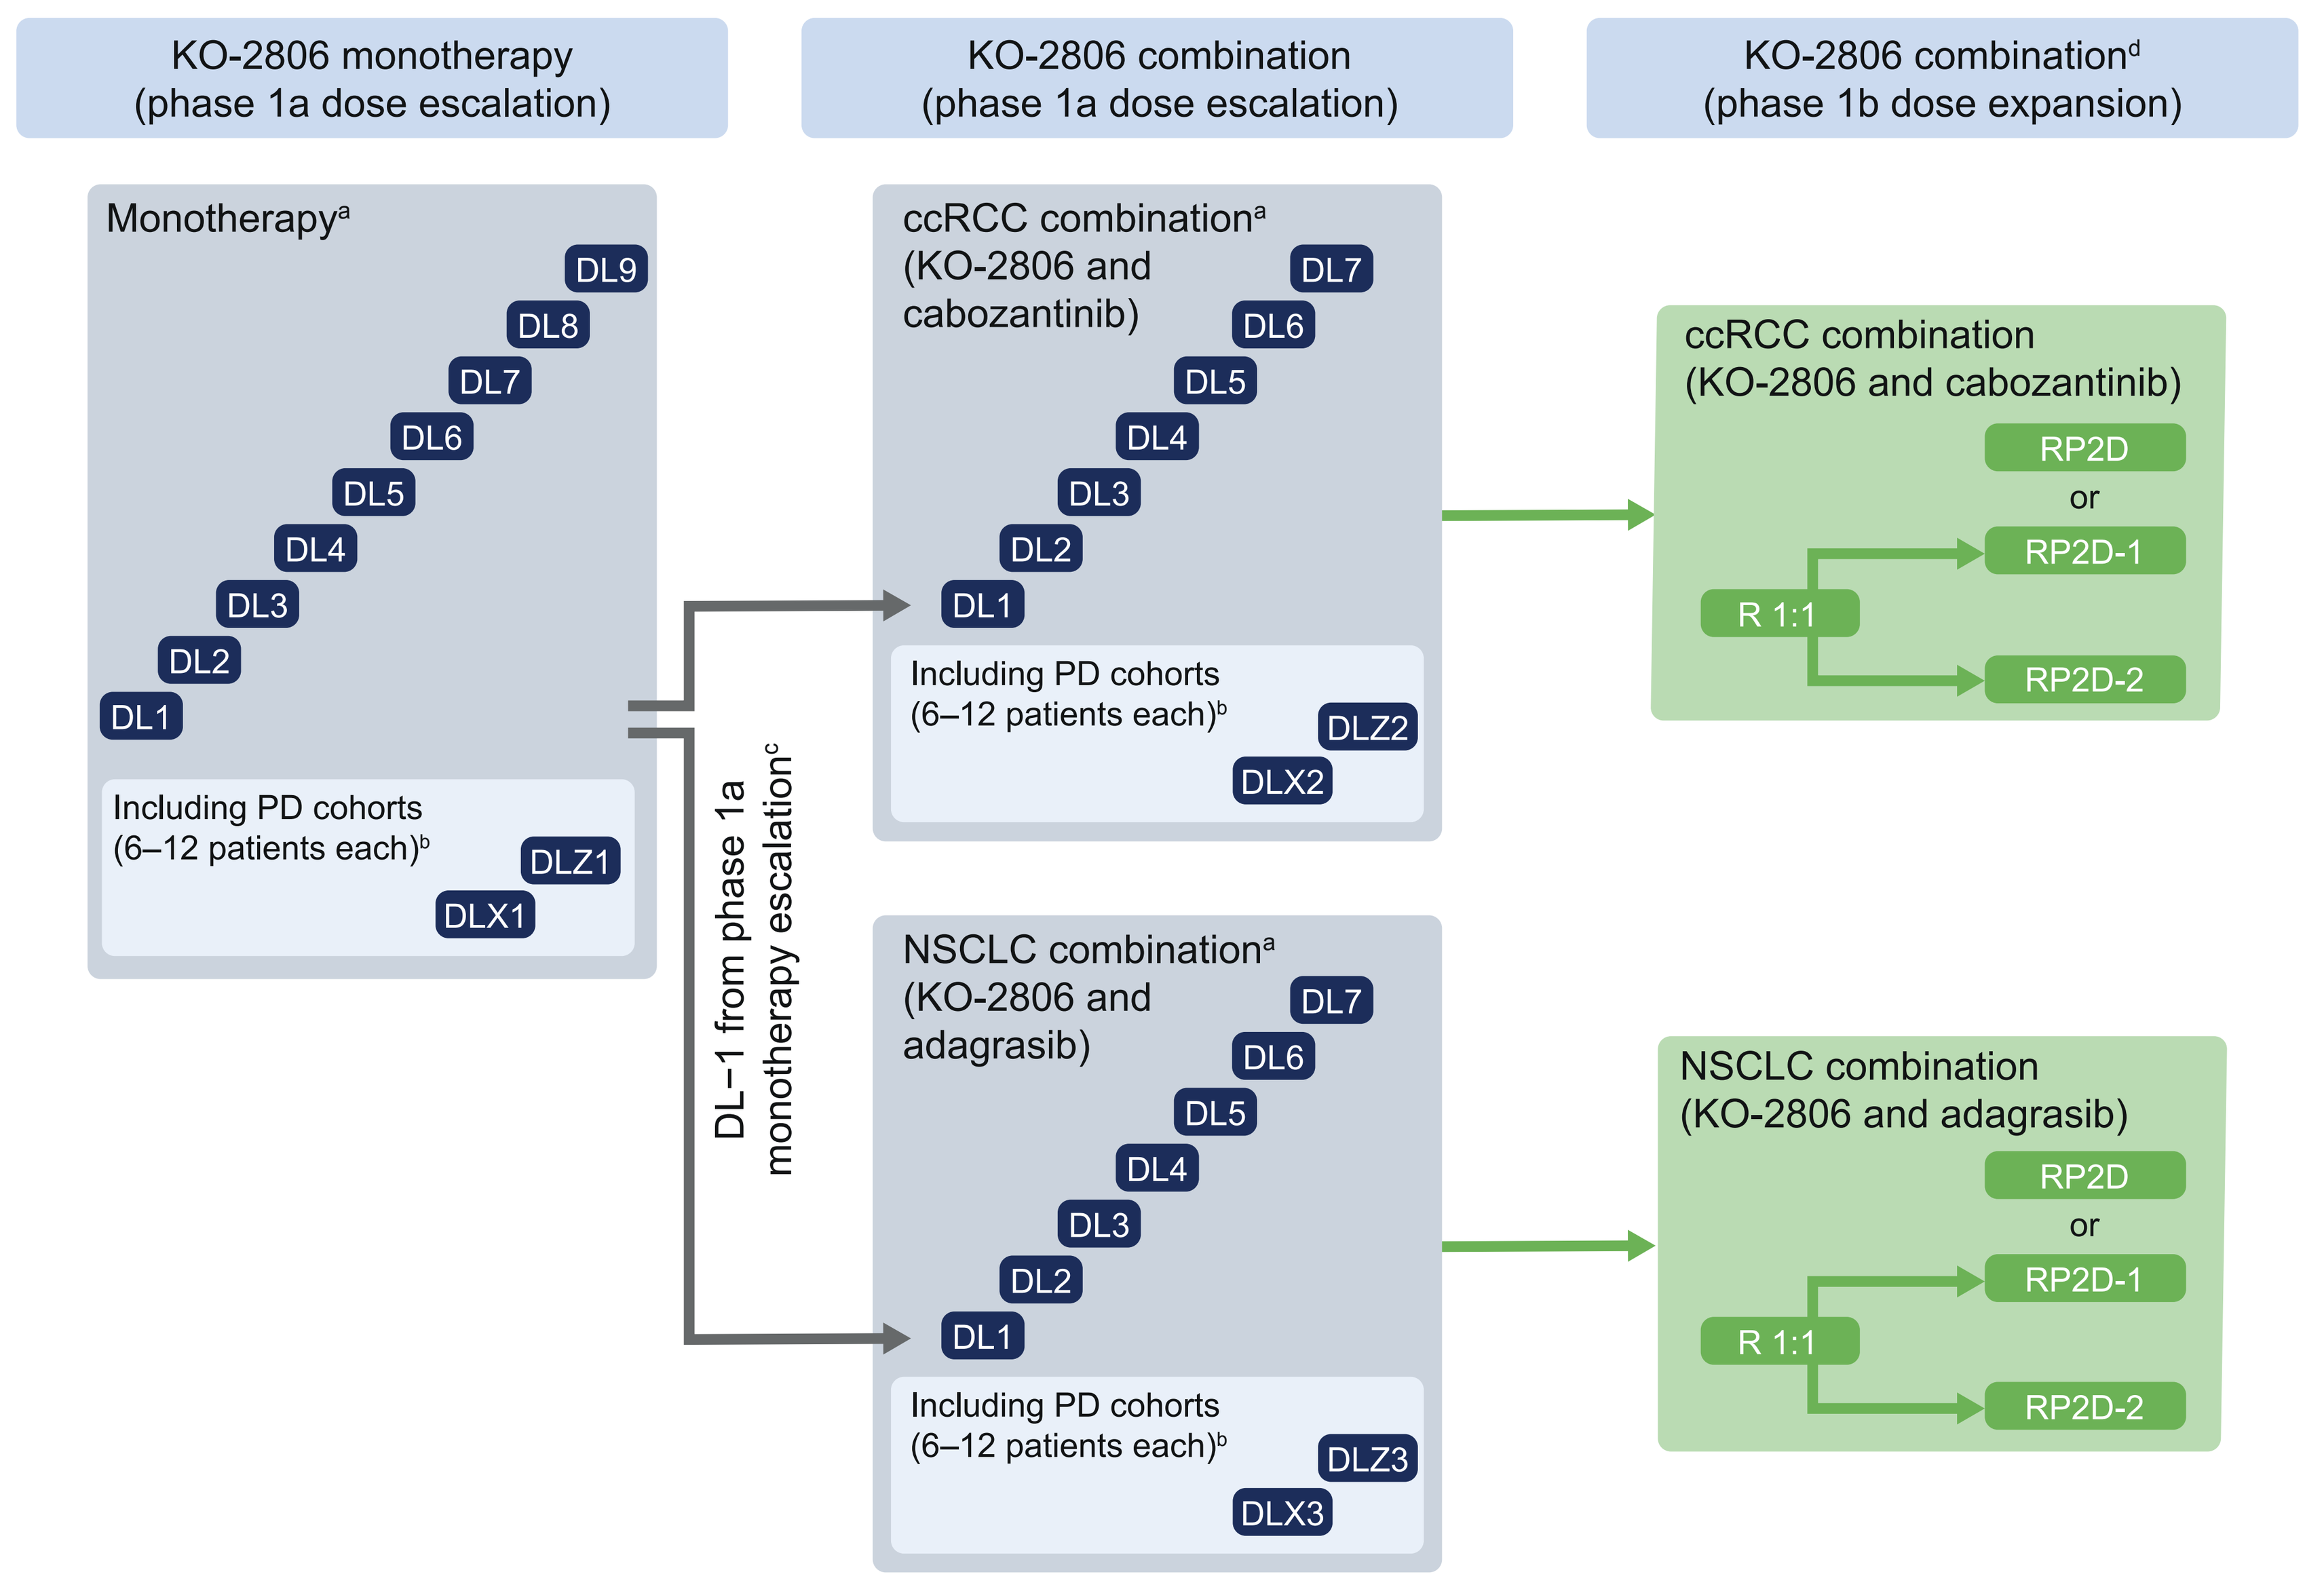
KO-2806 monotherapy (phase 1a dose escalation)
KO-2806 combination (phase 1a dose escalation)
KO-2806 combination[d] (phase 1b dose expansion)
Monotherapy[a]
DL9
DL8
DL7
DL6
DL5
DL4
DL3
DL2
DL1
Including PD cohorts (6–12 patients each)[b]
DLZ1
DLX1
DL−1 from phase 1a monotherapy escalation[c]
ccRCC combination[a] (KO-2806 and cabozantinib)
DL7
DL6
DL5
DL4
DL3
DL2
DL1
Including PD cohorts (6–12 patients each)[b]
DLZ2
DLX2
NSCLC combination[a] (KO-2806 and adagrasib)
DL7
DL6
DL5
DL4
DL3
DL2
DL1
Including PD cohorts (6–12 patients each)[b]
DLZ3
DLX3
ccRCC combination (KO-2806 and cabozantinib)
RP2D
or
R 1:1
RP2D-1
RP2D-2
NSCLC combination (KO-2806 and adagrasib)
RP2D
or
R 1:1
RP2D-1
RP2D-2

[a]Each individual patient will receive one of the planned dose levels of KO-2806.
[b]Mandatory pretreatment and on-treatment fresh tumor biopsies.
[c]KO-2806 combination dose level will start one level lower than the recommended monotherapy dose.
[d]Patients will receive the RP2D of KO-2806 or will be randomized if two potential RP2Ds are identified.

STUDY POPULATION

- FIT-001 is a first-in-human, multicenter, open-label, phase 1a/b dose escalation/expansion clinical trial (KO-2806-001; NCT06026410) (**Figure 1**).
- Approximately 270 patients will be enrolled in phases 1a and 1b across 50 sites globally.
- Phase 1a (dose escalation) consists of three arms with specific eligibility criteria shown in the table below.
- Based on emerging data from phase 1a, up to two PD cohorts may be explored for each of the monotherapy, ccRCC combination and NSCLC combination.
 - In each cohort, 6 to 12 patients may be enrolled; taking paired fresh tumor biopsies at screening and on day 21 will be mandatory.
- As of March 9, 2024, arms 1 and 2 are recruiting; the first patient dose for arm 3 is expected by mid-2024.
- In phase 1b (dose expansion), patients will receive the recommended phase 2 dose (RP2D) of KO-2806 with cabozantinib (in ccRCC) or adagrasib (in NSCLC), or will be randomized if two potential RP2Ds are identified for combination therapy.
- Other combination arms may also be considered.

KEY ELIGIBILITY CRITERIA

	Arm 1 monotherapy	Arm 2 ccRCC combination (KO-2806 and cabozantinib)	Arm 3 NSCLC combination (KO-2806 and adagrasib)
Key inclusion criteria			
Age	≥ 18 years		
Diagnosis of advanced solid tumor	Histologically or cytologically confirmed advanced solid tumor		
Disease severity	• Measurable disease per RECIST (version 1.1) • Karnofsky Performance Status Scale score of ≥ 70 with no clinically significant deterioration in the previous 2 weeks • Acceptable endocrine, hematologic, liver and renal functions		
Arm-specific criteria	*HRAS* mutation and/or amplification (any solid tumor type); *HRAS* overexpression (HNSCC only); *KRAS*, *NRAS* and/or *HRAS* mutation and/or amplification (NSCLC or CRC); *KRAS* mutation and/or amplification (PDAC)	Must have received at least one prior systemic therapy with an immunotherapy for locally advanced or metastatic ccRCC	Must have received at least one prior systemic therapy for *KRAS-G12C*-m locally advanced or metastatic NSCLC
Key exclusion criteria			
Prior or ongoing treatment/conditions	• Prior treatment with an FTI or HRAS inhibitor • Major surgery in the previous 28 days or still recovering from prior surgery • Spinal cord compression, leptomeningeal disease or clinically active CNS metastases • Active or prior documented autoimmune or inflammatory disorders in the previous 5 years (with exceptions) • Inability to swallow, impairment of GI function or GI disease that may significantly alter the absorption of the trial drugs • Inadequate cardiac and/or vascular function, including mean QTcF ≥ 470 ms, presence of acute coronary syndrome in the past 6 months or class II or greater congestive heart failure • Other invasive malignancy in the previous 2 years		

OUTCOME MEASURES

Objectives	Endpoints
Primary	
Safety	
• Evaluate safety and tolerability of KO-2806 as monotherapy and in combination in patients with advanced solid tumors (phase 1a) • Determine MTD/HPDD and/or OBAD of KO-2806 as monotherapy and in combination (phase 1a) • Define RP2D of KO-2806 in combination (phase 1b)	• Incidence and severity of treatment-emergent AEs, incidence of SAEs, dose-limiting toxicities, abnormal laboratory parameters, abnormal vital signs and abnormal ECG results • Incidence of dose interruptions, reductions and discontinuations due to AEs
Efficacy	
• Evaluate antitumor activity of KO-2806 in combination in patients with advanced solid tumors (phase 1b)	• ORR per RECIST (version 1.1)
Secondary[a]	
Safety	
• Evaluate safety and tolerability of KO-2806 in combination in patients with advanced solid tumors (phase 1b)	• Incidence and severity of treatment-emergent AEs, incidence of SAEs, dose-limiting toxicities, abnormal laboratory parameters, abnormal vital signs and abnormal ECG results • Incidence of dose interruptions, reductions and discontinuations due to AEs
Efficacy	
• Evaluate preliminary antitumor activity of KO-2806 as monotherapy or in combination in patients with advanced solid tumors (phase 1a)	• ORR, DCR, DoR, PFS per RECIST (version 1.1) per Investigator assessment and OS
• Further evaluate the antitumor activity of KO-2806 in combination in patients with advanced solid tumors (phase 1b)	• DCR, DoR, PFS per RECIST (version 1.1) per Investigator assessment and OS
PK	
• Characterize PK of KO-2806 as monotherapy and in combination (phase 1a and 1b)	• Systemic plasma concentration of KO-2806 and the combination agent • PK parameters, including AUC_{last}, C_{max}, T_{max}, $t_{½}$ and CL/F
Other objectives	
• Evaluate effect of food on the PK of KO-2806 as monotherapy • Evaluate relationship between plasma concentrations and QTc intervals for KO-2806 and adagrasib as monotherapy and KO-2806 and adagrasib in combination • Characterize urine PK for KO-2806 monotherapy	

[a]Not a comprehensive list.

CONCLUSIONS

- **This study is exploring the safety and preliminary antitumor activity of KO-2806 for patients with advanced solid tumors; the results will inform the design of future trials.**
- **The data will also allow evaluation of the safety and preliminary antitumor activity of KO-2806 administered in combination with cabozantinib in ccRCC or adagrasib in NSCLC, and other potential combinations.**

Disclosures

Glenn J Hanna receives grant/institutional research support and serves in a consulting/advisory role for Kura Oncology. **Douglas R Adkins** has received research support from AstraZeneca, Blueprint, Bristol Myers Squibb/Celgene, Celldex, Cue Biopharma, Enzychem, Exelixis, Innate Pharma, Kura Oncology, Lilly, Merck, Novartis, Oncolys, Pfizer and Sensei Biotherapeutics; and consulting fees from Blueprint, Coherus BioSciences, Cue Biopharma, Kura Oncology, Lilly, Merck, Oncolys, twoXAR, Vaccinex and Xilio. **Jacob S Thomas** reports no potential conflicts of interest. **Justine Y Bruce** is on the safety monitoring committee for the FIT-001 trial. **Manish R Patel** has leadership involvement in ION Pharma; has received honoraria from Janssen Oncology; has served in a consulting/advisory role for Accutar Biotech, Daiichi Sankyo/UCB Japan, Kura Oncology, Nurix and Olema Pharmaceuticals; and has received research support from Accutar Biotech, Acerta Pharma, Adagene, ADC Therapeutics, Agenus, Allorion Therapeutics, Artios, Astellas, AstraZeneca, BioNTech AG, BioTHeryX, Blueprint Pharmaceuticals, Boehringer Ingelheim, Bristol Myers Squibb/Celgene, Celgene, Compugen, Conjupro Biotherapeutics, Cullinan Oncology, Cyteir Therapeutics, Daiichi Sankyo, Erasca Inc., Genentech/Roche, Georgiamune, Gilead Sciences, GlaxoSmithKline, H3 Biomedicine, Hengrui Therapeutics, Hotspot Therapeutics, Hutchison MediPharma, Immune-Onc Therapeutics, Immunitas, Immunogen, Incyte, Janssen, Kineta, Klus Pharma, Kura Oncology, Kymab, Lilly, Loxo, LSK Biopartners, MabSpace Biosciences, Macrogenics, Merck, Millennium, Moderna Therapeutics, Novartis, Nurix, Olema, ORIC, Pfizer, Pionyr, Prelude Therapeutics, Puretech, Ribon Therapeutics, Seven and Eight Biopharmaceuticals, Step Pharma, Syndax, Taiho Pharmaceutical, TeneoBio, Tesaro, Treadwell Therapeutics, Vividion Therapeutics and Zymeworks. **Guru Sonpavde** receives grant/institutional research support from Bristol Myers Squibb, EMD Serono and Jazz Pharmaceuticals; has served in a consulting/advisory role for Atkis, Bicycle Therapeutics, Bristol Myers Squibb, Eli Lilly/Loxo Oncology, EMD Serono, Gilead, Janssen, Kura Oncology, Merck, Pfizer, PrecisCa, Scholar Rock, Seattle Genetics/Astellas, Servier, Syapse, Syncorp and Vial; has received speaker/travel fees from Astellas, Aveo, Bayer, Bristol Myers Squibb, Exelixis, Gilead, Janssen, Merck, Natera, Pfizer and Seagen; has received honoraria from Mereo; and their spouse is employed by Myriad. **Jason Henry** is an employee of the Sarah Cannon Research Institute at HealthONE and has stocks/shares in HCA. **Nawal Bendris** and **Zijing Zhang** are employees and stockholders of Kura Oncology. **Amitava Mitra** and **Stephen Dale** are employees and stockholders of Kura Oncology and are named inventors on planned, issued or pending patents that are owned by Kura Oncology. **Andrew Saunders** has received consulting/advisory fees from Kura Oncology and is a stockholder of Kura Oncology.

References

1. Cox AD, Der CJ. *Small GTPases* 2010;1:2–27.
2. Kessler L *et al. Cancers (Basel)* 2021;13:5310.
3. Ho AL *et al. J Clin Oncol* 2021;39:1856–64.
4. ClinicalTrials.gov. Safety and Efficacy of Tipifarnib in Head and Neck Cancer With HRAS Mutations and Impact of HRAS on Response to Therapy (AIM-HN/SEQ-HN). Available from: https://www.clinicaltrials.gov/study/NCT03719690 (Accessed February 27, 2024).
5. Ho AL *et al.* Poster (abstract #LBA47) presented at the European Society for Medical Oncology Congress, October 20–24, 2023, Madrid, Spain.
6. Gatchalian J *et al.* Poster (abstract #34634) presented at the 36th EORTC-NCI-AACR Symposium, October 11–15, 2023, Boston, MA, USA.
7. Patel HV *et al.* Poster (abstract #34968) presented at the 36th EORTC-NCI-AACR Symposium, October 11–15, 2023, Boston MA, USA.
8. Smith A *et al.* Poster (abstract #34971) presented at the 36th EORTC-NCI-AACR Symposium, October 11–15, 2023, Boston MA, USA.

Abbreviations

-m, mutant; AE, adverse event; AUC_{last}, AUC from time 0 to last measurable concentration; ccRCC, clear cell renal cell carcinoma; CDX, cell line-derived xenograft; CL/F, total apparent clearance; C_{max}, maximum plasma concentration; CNS, central nervous system; CRC, colorectal cancer; DCR, disease control rate; DL, dose level; DLX, dose for the first PD cohort; DLZ, dose for the second PD cohort; DoR, duration of response; ECG, electrocardiogram; FTI, farnesyl transferase inhibitor; GI, gastrointestinal; HNSCC, head and neck squamous cell carcinoma; HPDD, highest protocol-defined dose; MTD, maximum tolerated dose; NSCLC, non-small cell lung cancer; OBAD, optimal biologically active dose; ORR, objective response rate; OS, overall survival; PD, pharmacodynamic(s); PDAC, pancreatic ductal adenocarcinoma; PDX, patient-derived xenograft; PFS, progression-free survival; PK, pharmacokinetic(s); QTc, corrected QT interval; QTcF, QT interval corrected for heart rate by Fridericia's cube-root formula; R, randomization; RECIST, Response Evaluation Criteria in Solid Tumors; RP2D, recommended phase 2 dose; SAE, serious adverse event; $t_{½}$, half-life; T_{max}, time to maximum observed plasma concentration.

Acknowledgments

The authors would like to thank the patients and their families for their involvement in the study.

Author contributions

All authors contributed to study conception/design or data collection/analysis/interpretation, development of this poster or critical review of the content; and gave their approval for the final poster.

Medical writing support

The authors thank Rebecca Burge PhD of Oxford PharmaGenesis, Oxford, UK for providing medical writing support and editorial assistance, which was funded by Kura Oncology, Inc., in accordance with Good Publications Practice guidelines (GPP 2022).

Presented at the American Association for Cancer Research (AACR) Annual Meeting, April 5–10, 2024, San Diego, CA, USA.

For further information, please contact:
medicalaffairs@kuraoncology.com

This study was funded by Kura Oncology, Inc.