Tipifarnib is Highly Active in HRAS-mutant Head and Neck Squamous Cell Carcinoma (HNSCC) Tumor Models

Mara Gilardi[2], Zhiyong Wang[2], Linda Kessler[1], Antonio Gualberto[1], Yi Liu[1], J. Silvio Gutkind[2], Francis Burrows[1]
[1]*Kura Oncology, Inc., San Diego, CA, USA*, [2]*University of California, San Diego*

BACKGROUND AND RATIONALE

Tipifarnib is a potent and selective inhibitor of farnesyltransferase (FT). FT catalyzes the post-translational attachment of farnesyl groups to signaling proteins that are requisite for localization to the inner cell membrane. Although all RAS isoforms are FT substrates, only HRAS is exclusively dependent upon farnesylation for membrane localization and signaling activation, making HRAS mutant tumors uniquely susceptible to tipifarnib mediated inhibition of FT. Based upon this rationale, the safety and efficacy of tipifarnib is currently being evaluated in patients with HRAS mutant head and neck squamous cell carcinoma (HNSCC) in an international, multicenter, open-label 2 cohort, non-comparative, pivotal Phase 2 trial (NCT03719690).

Here we report the characterization of tipifarnib in CDX and PDX models of HRAS-mutant HNSCC *in vitro* and *in vivo*

HRAS MUTATIONS DEFINE A UNIQUE MOLECULAR SUBSET OF HNSCC

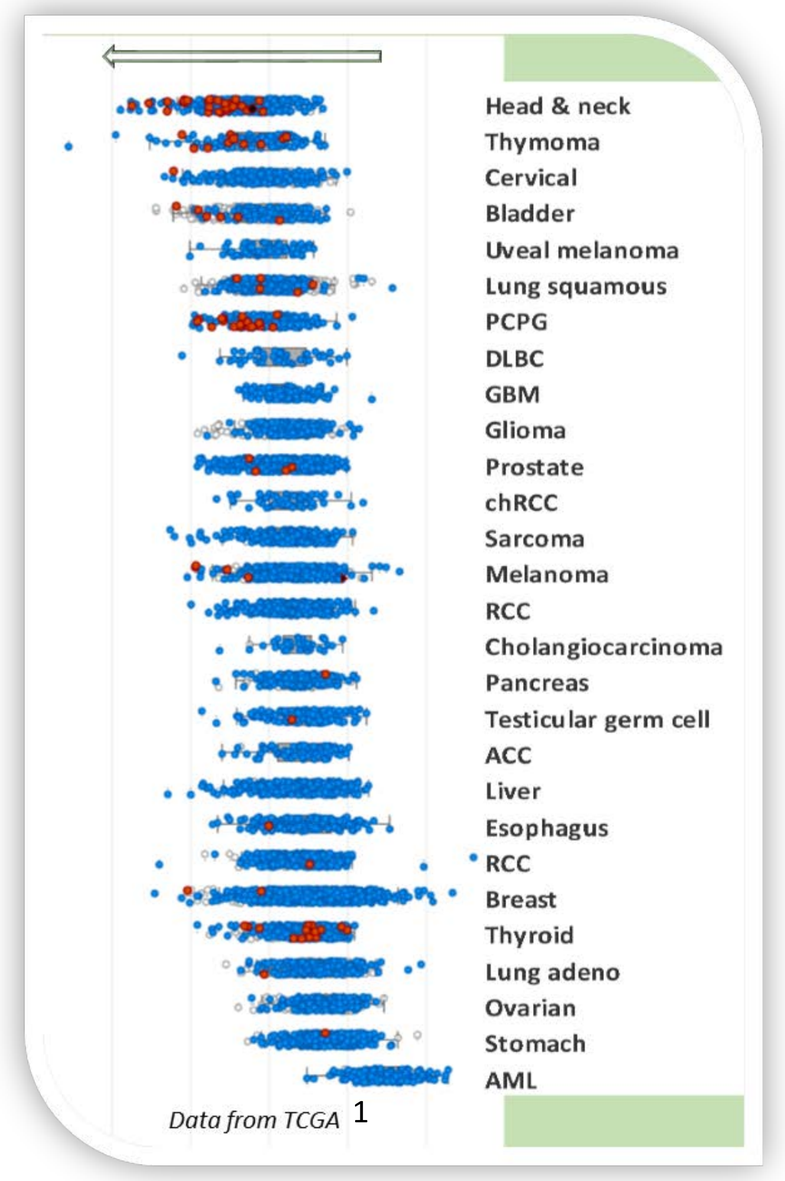
Head & neck
Thymoma
Cervical
Bladder
Uveal melanoma
Lung squamous
PCPG
DLBC
GBM
Glioma
Prostate
chRCC
Sarcoma
Melanoma
RCC
Cholangiocarcinoma
Pancreas
Testicular germ cell
ACC
Liver
Esophagus
RCC
Breast
Thyroid
Lung adeno
Ovarian
Stomach
AML
Data from TCGA 1

HNSCC TCGA (N=30)

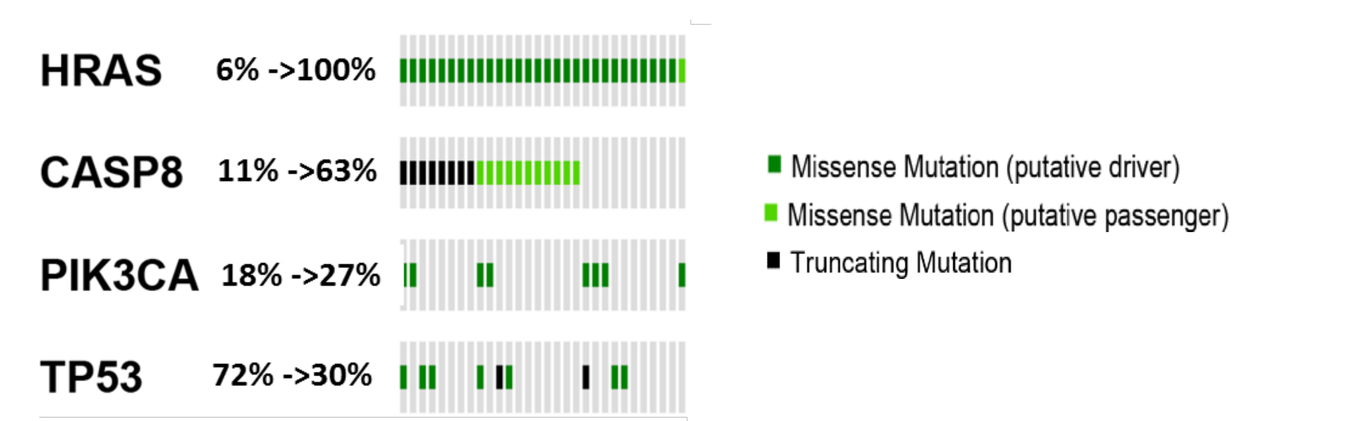
HRAS 6% ->100%
CASP8 11% ->63%
PIK3CA 18% ->27%
TP53 72% ->30%
Missense Mutation (putative driver)
Missense Mutation (putative passenger)
Truncating Mutation

- HNSCC express high levels of HRAS[2]
- HRAS mutant SCCHN (~6% at initial diagnosis) characterized by frequent CASP8 mutations and low rate of TP53 mutation[3]
- HRAS mutation may result from toxin (*e.g.* tobacco) exposure[4]
- HRAS mutation is associated with innate and acquired resistance to cetuximab[5]
 - 3/13 (23%) of patients with on-treatment disease progression during cetuximab/platinum/5-FU therapy acquired HRAS mutations

1. TCGA data. Gao *et al.* 2013. *Sci Signal* 6:pl1; Cerami *et al* 2012. *Cancer Discov.* 2:401-4
2. Cancer Genome Atlas Network 2015. *Nature* 517:576-82.
3. Cancer Genome Atlas Research Network 2012. *Nature* 489:519-25
4. Sathyan *et al.* 2007. *Modern Path* 20, 1141-8
5. Braig *et al. Oncotarget*. 2016. 7, 42988-42995

Molecular basis of tipifarnib activity in HRAS-mutant HNSCC

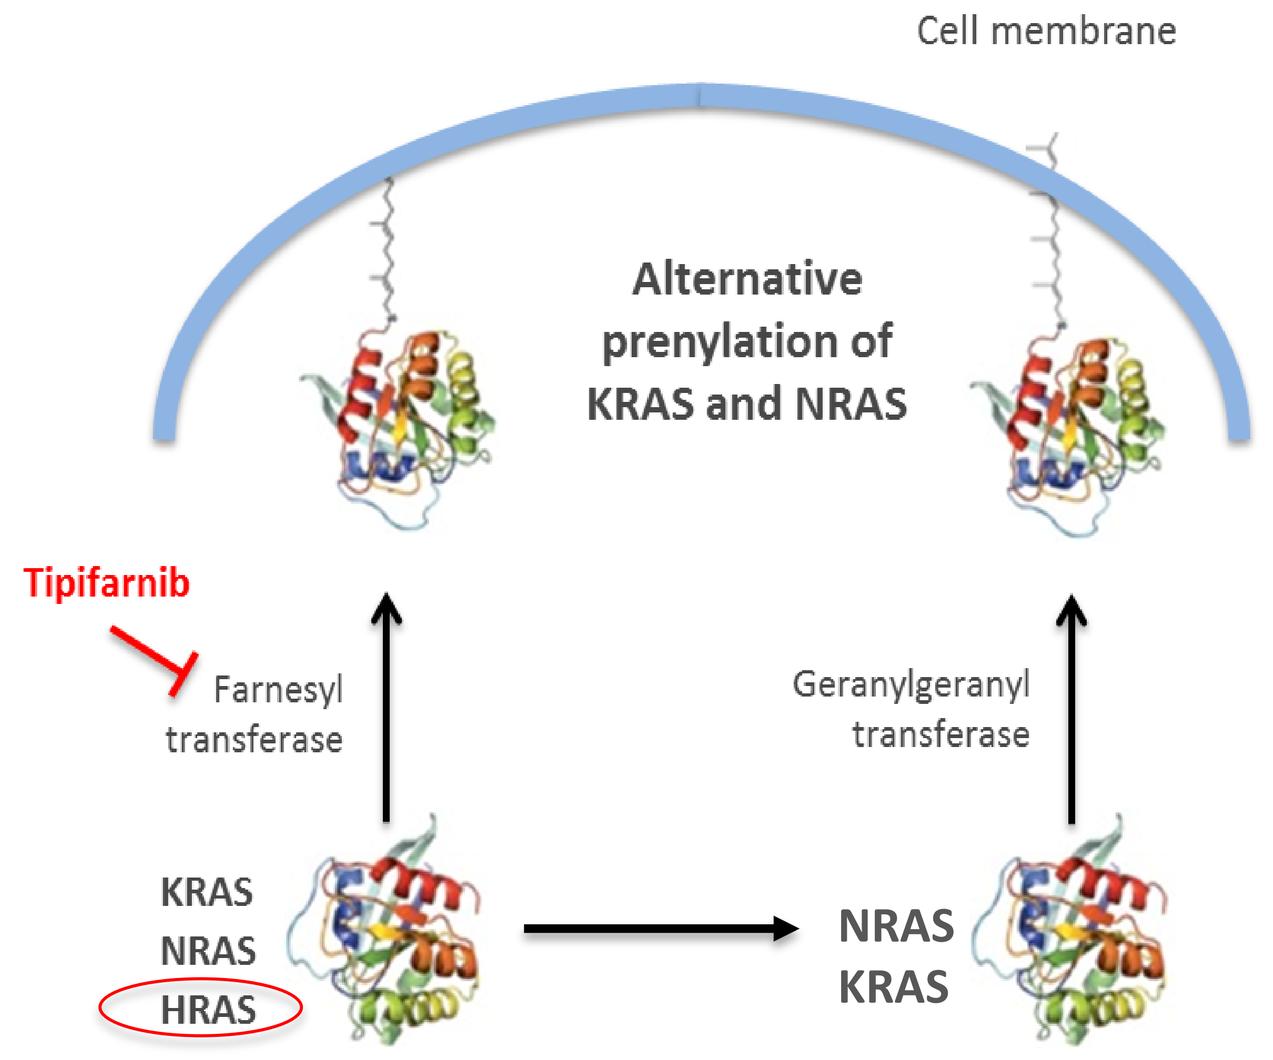
Cell membrane
Alternative prenylation of KRAS and NRAS
Tipifarnib
Farnesyl transferase
Geranylgeranyl transferase
KRAS
NRAS
HRAS
NRAS
KRAS

TIPIFARNIB DEPRENYLATES AND DISPLACES HRAS FROM MEMBRANES

Tipifarnib deprenylates HRAS *in vivo*

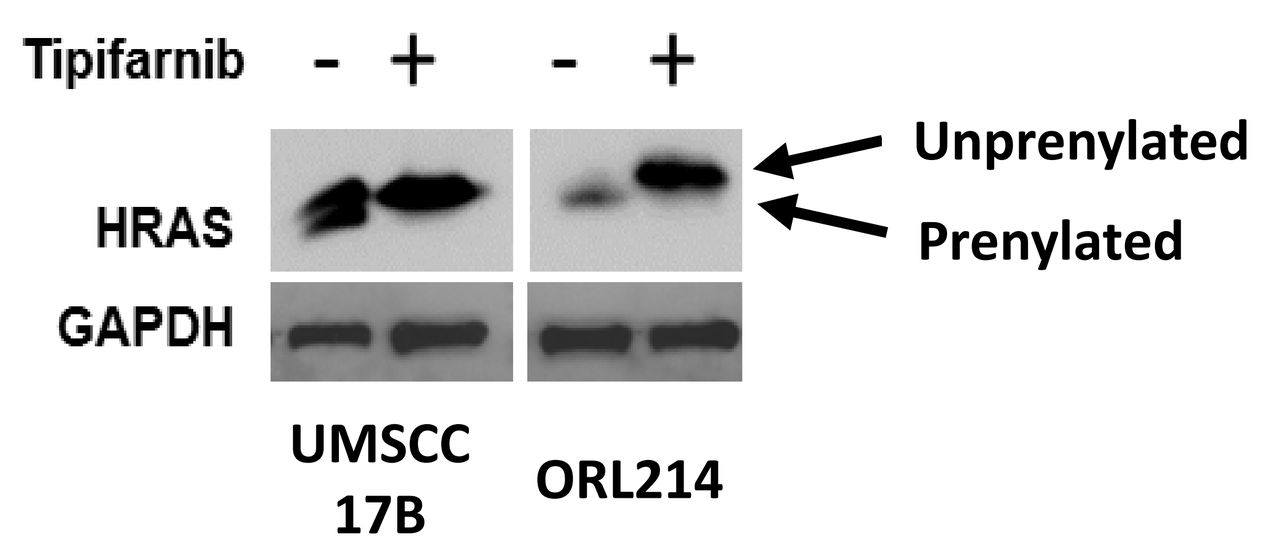
Tipifarnib - + - +
HRAS
GAPDH
Unprenylated
Prenylated
UMSCC 17B
ORL214

HRAS-mutant UMSCC17B and ORL214 cells were treated with tipifarnib (100nM, 48h) and stained with anti-HRAS antibodies by western blotting (left) or immunofluorescence (right). The fluorescence profile relative to the distribution of HRAS (green) was compared to that of DAPI (blue) and the membrane marker WGA lectins (red) by confocal microscopy

HRAS colocalization with cellular membranes is lost in tipifarnib-treated HNSCC cells

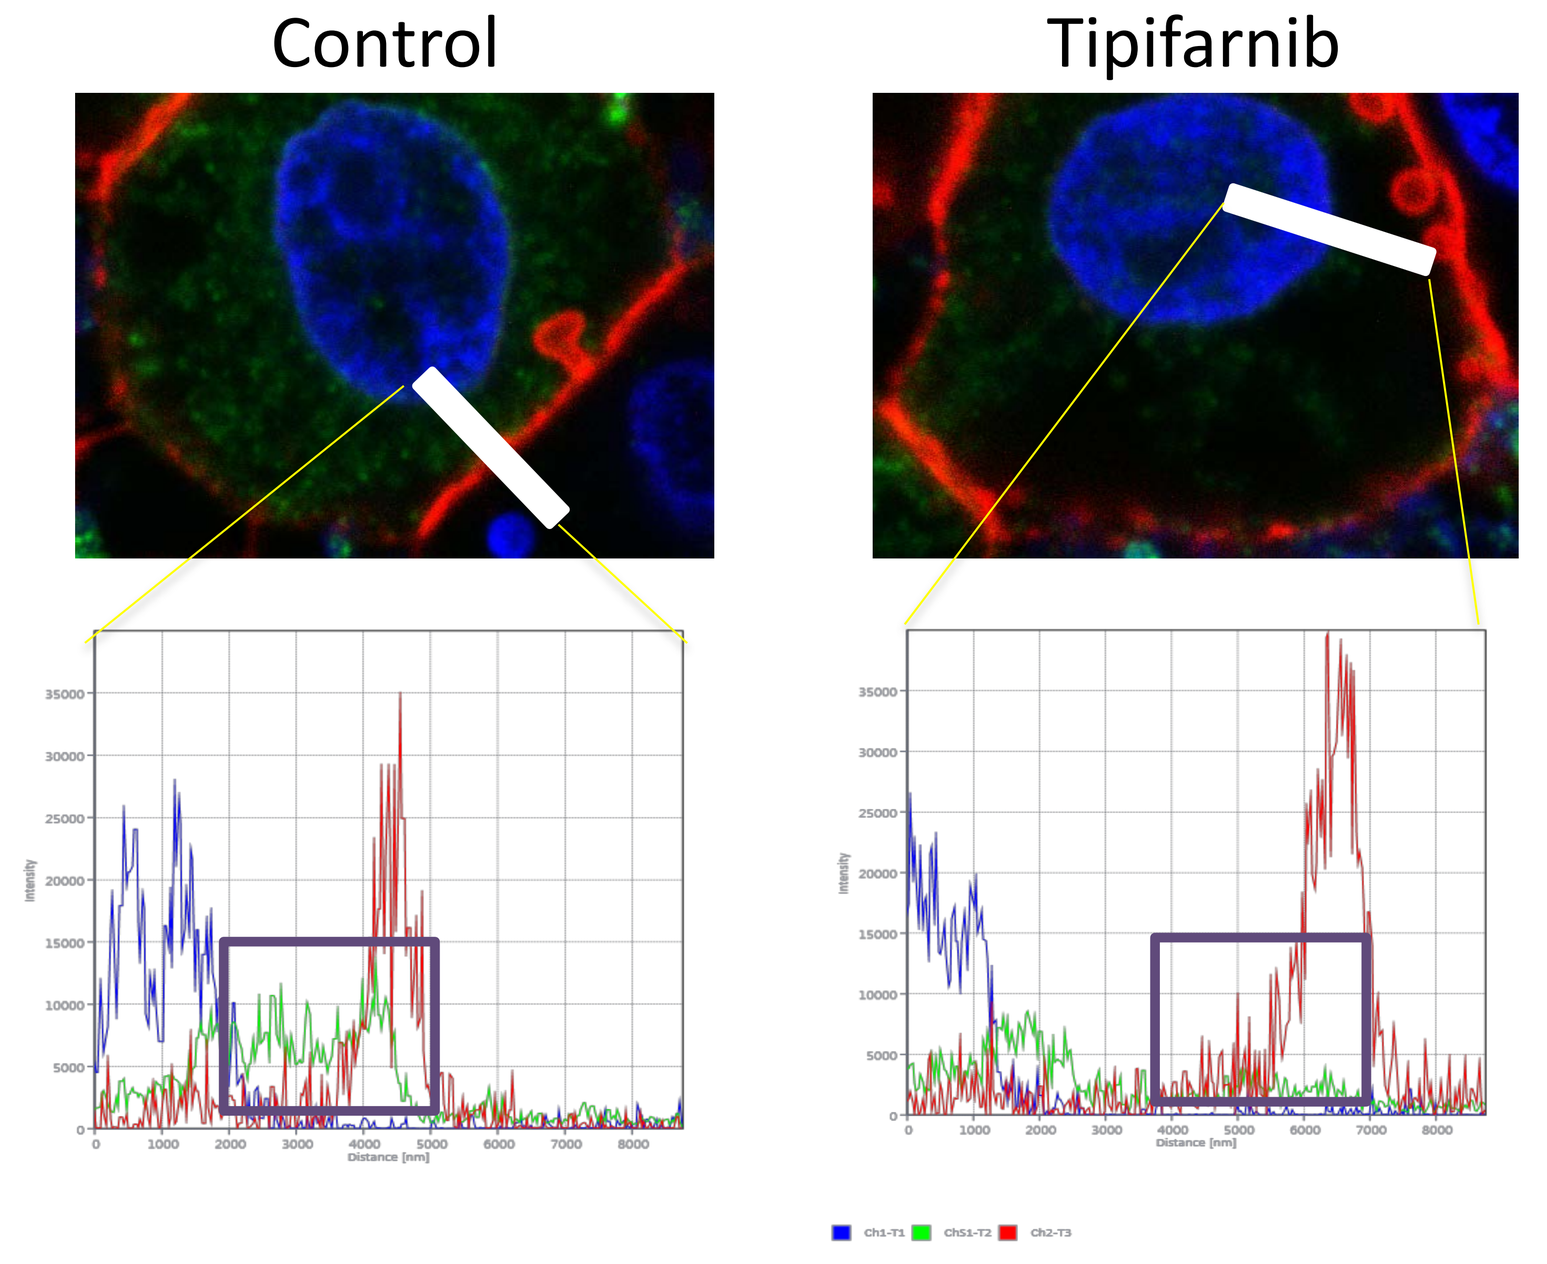
Control
Tipifarnib

TIPIFARNIB IS SELECTIVELY CYTOTOXIC TO HRAS-MUTANT HNSCC CELLS IN VITRO

2D monolayer assay

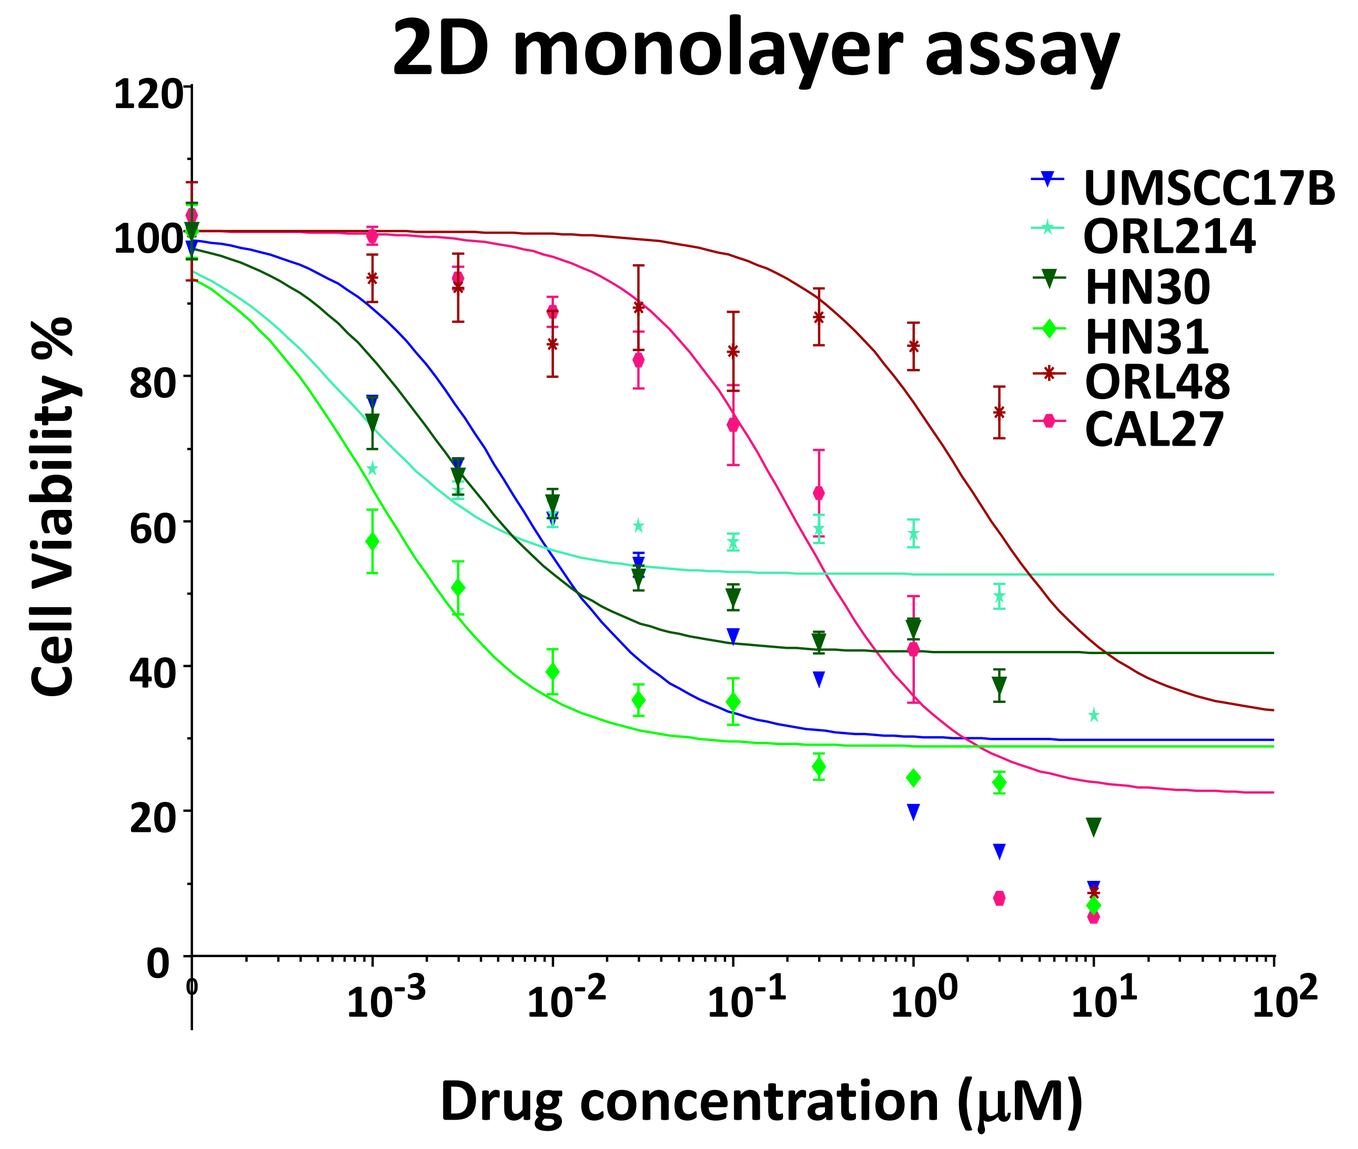
UMSCC17B
ORL214
HN30
HN31
ORL48
CAL27
Cell Viability %
Drug concentration (μM)

HRAS	
MUTANT	WILD TYPE
UMSCC17B	ORL48
ORL214	CAL27
HN30	
HN31	

*** p < 0.0001
** p < 0.011
* p < 0.01

3D sphere formation assay

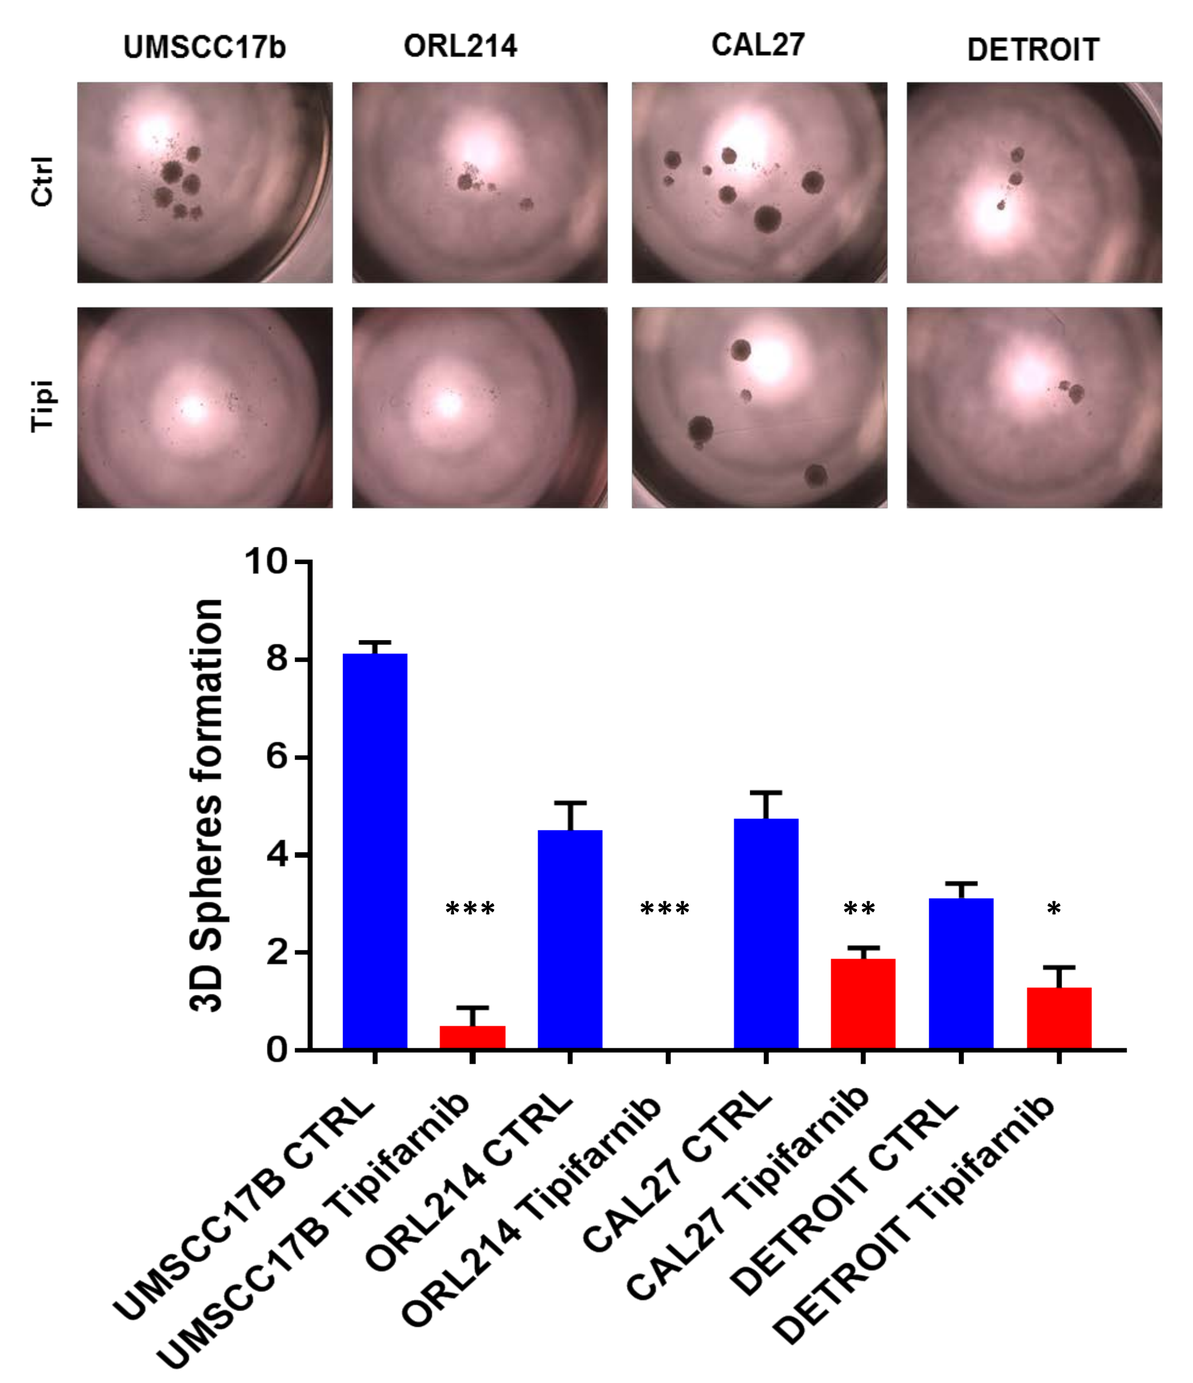
UMSCC17b
ORL214
CAL27
DETROIT
Ctrl
Tipi
3D Spheres formation
UMSCC17B CTRL
UMSCC17B Tipifarnib
ORL214 CTRL
ORL214 Tipifarnib
CAL27 CTRL
CAL27 Tipifarnib
DETROIT CTRL
DETROIT Tipifarnib

HNSCC cells were treated with a titration of tipifarnib for 72 h (left panel) or with vehicle or 300 nM tipifarnib under anchorage-independent conditions for 14 days (right panels)

TIPIFARNIB IS HIGHLY ACTIVE IN HRAS-MUTANT HNSCC XENOGRAFTS

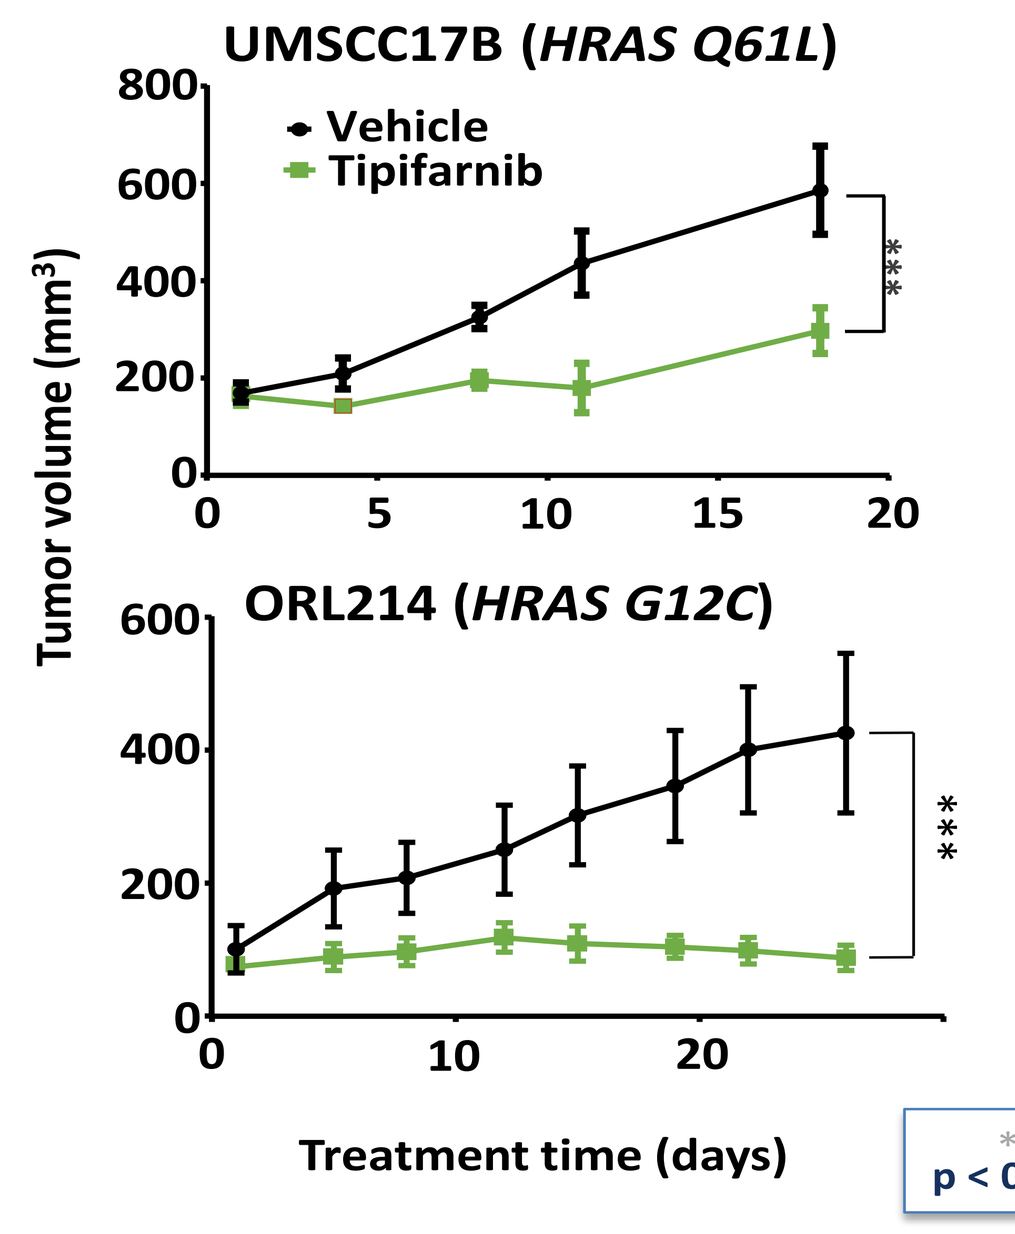
UMSCC17B (HRAS Q61L)
Vehicle
Tipifarnib
ORL214 (HRAS G12C)
Tumor volume (mm3)
Treatment time (days)

*** p < 0.01

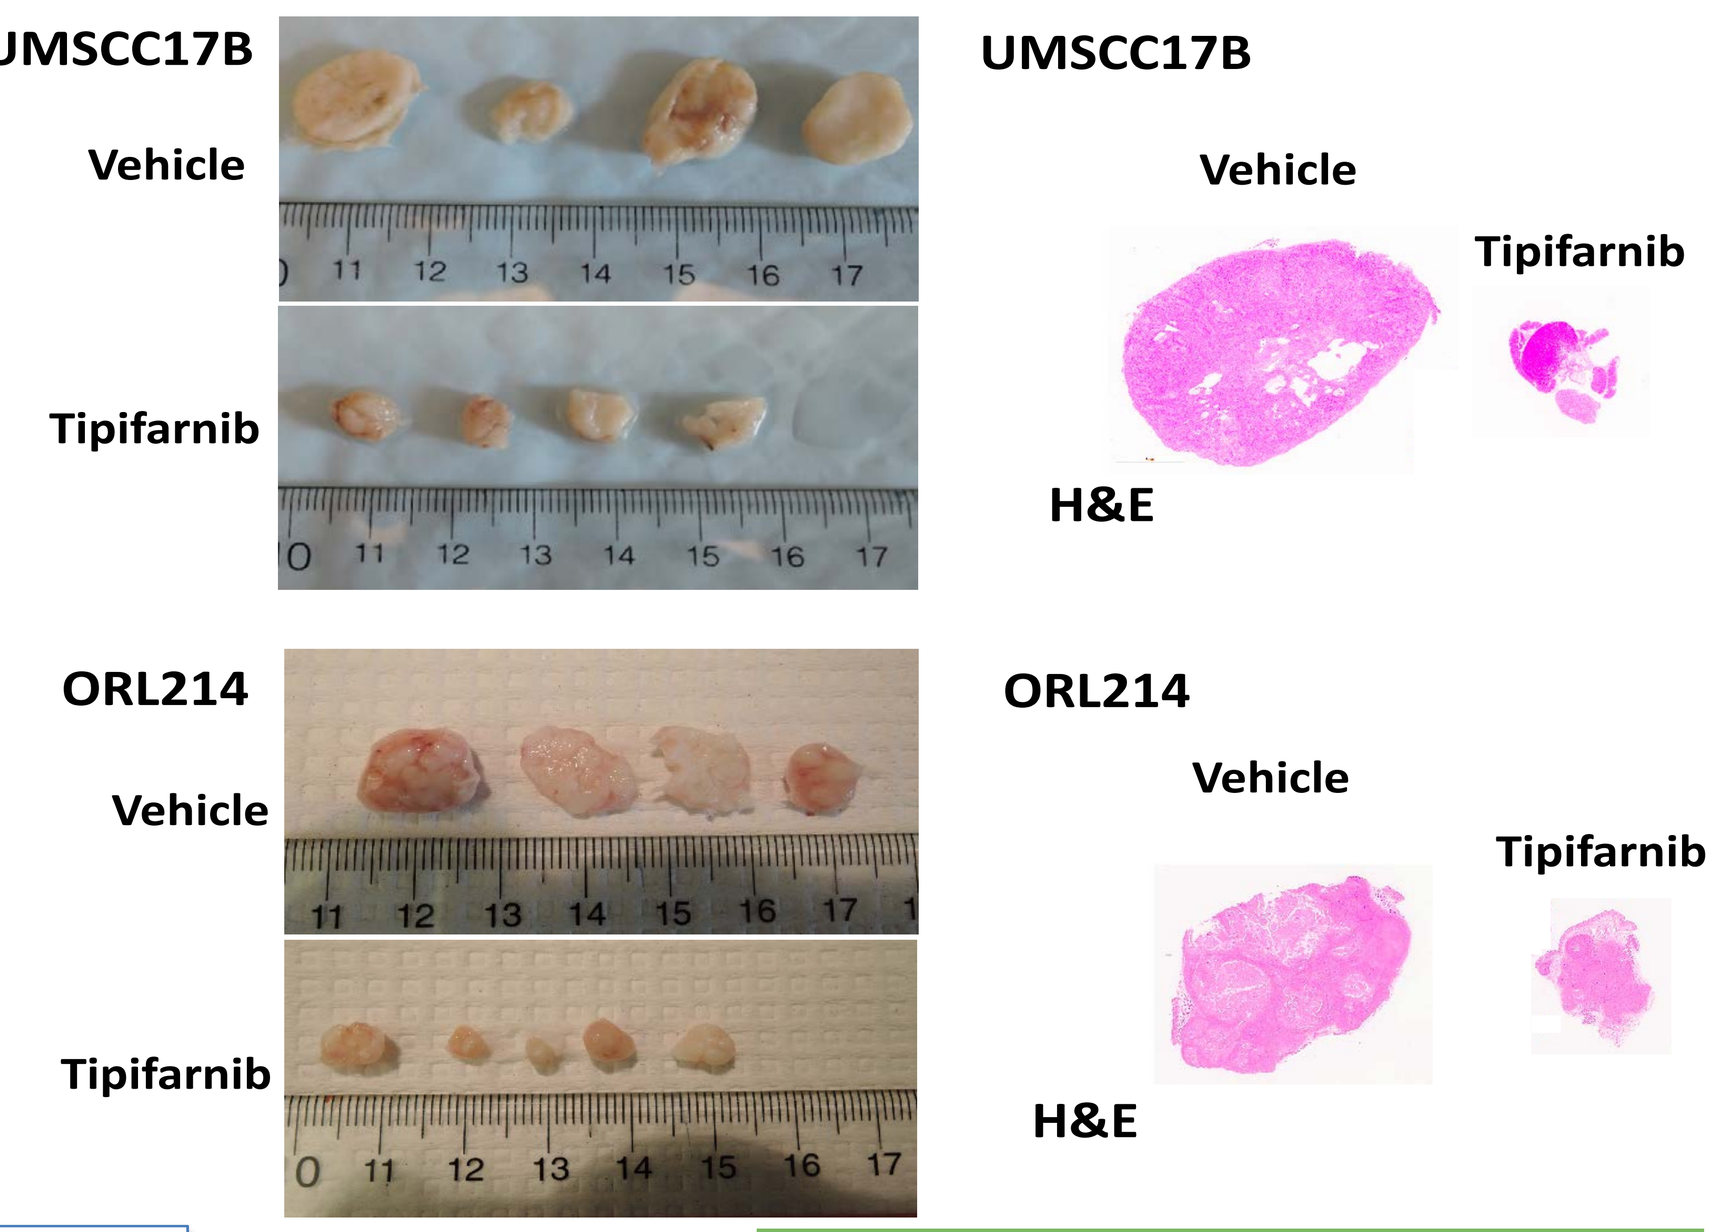
UMSCC17B
Vehicle
Tipifarnib
H&E
ORL214
Vehicle
Tipifarnib
H&E

All animals dosed orally at 60mg/kg BID

ACTIVITY OF TIPIFARNIB IN HRAS-MUTANT SCC IS INDEPENDENT OF GENOTYPE

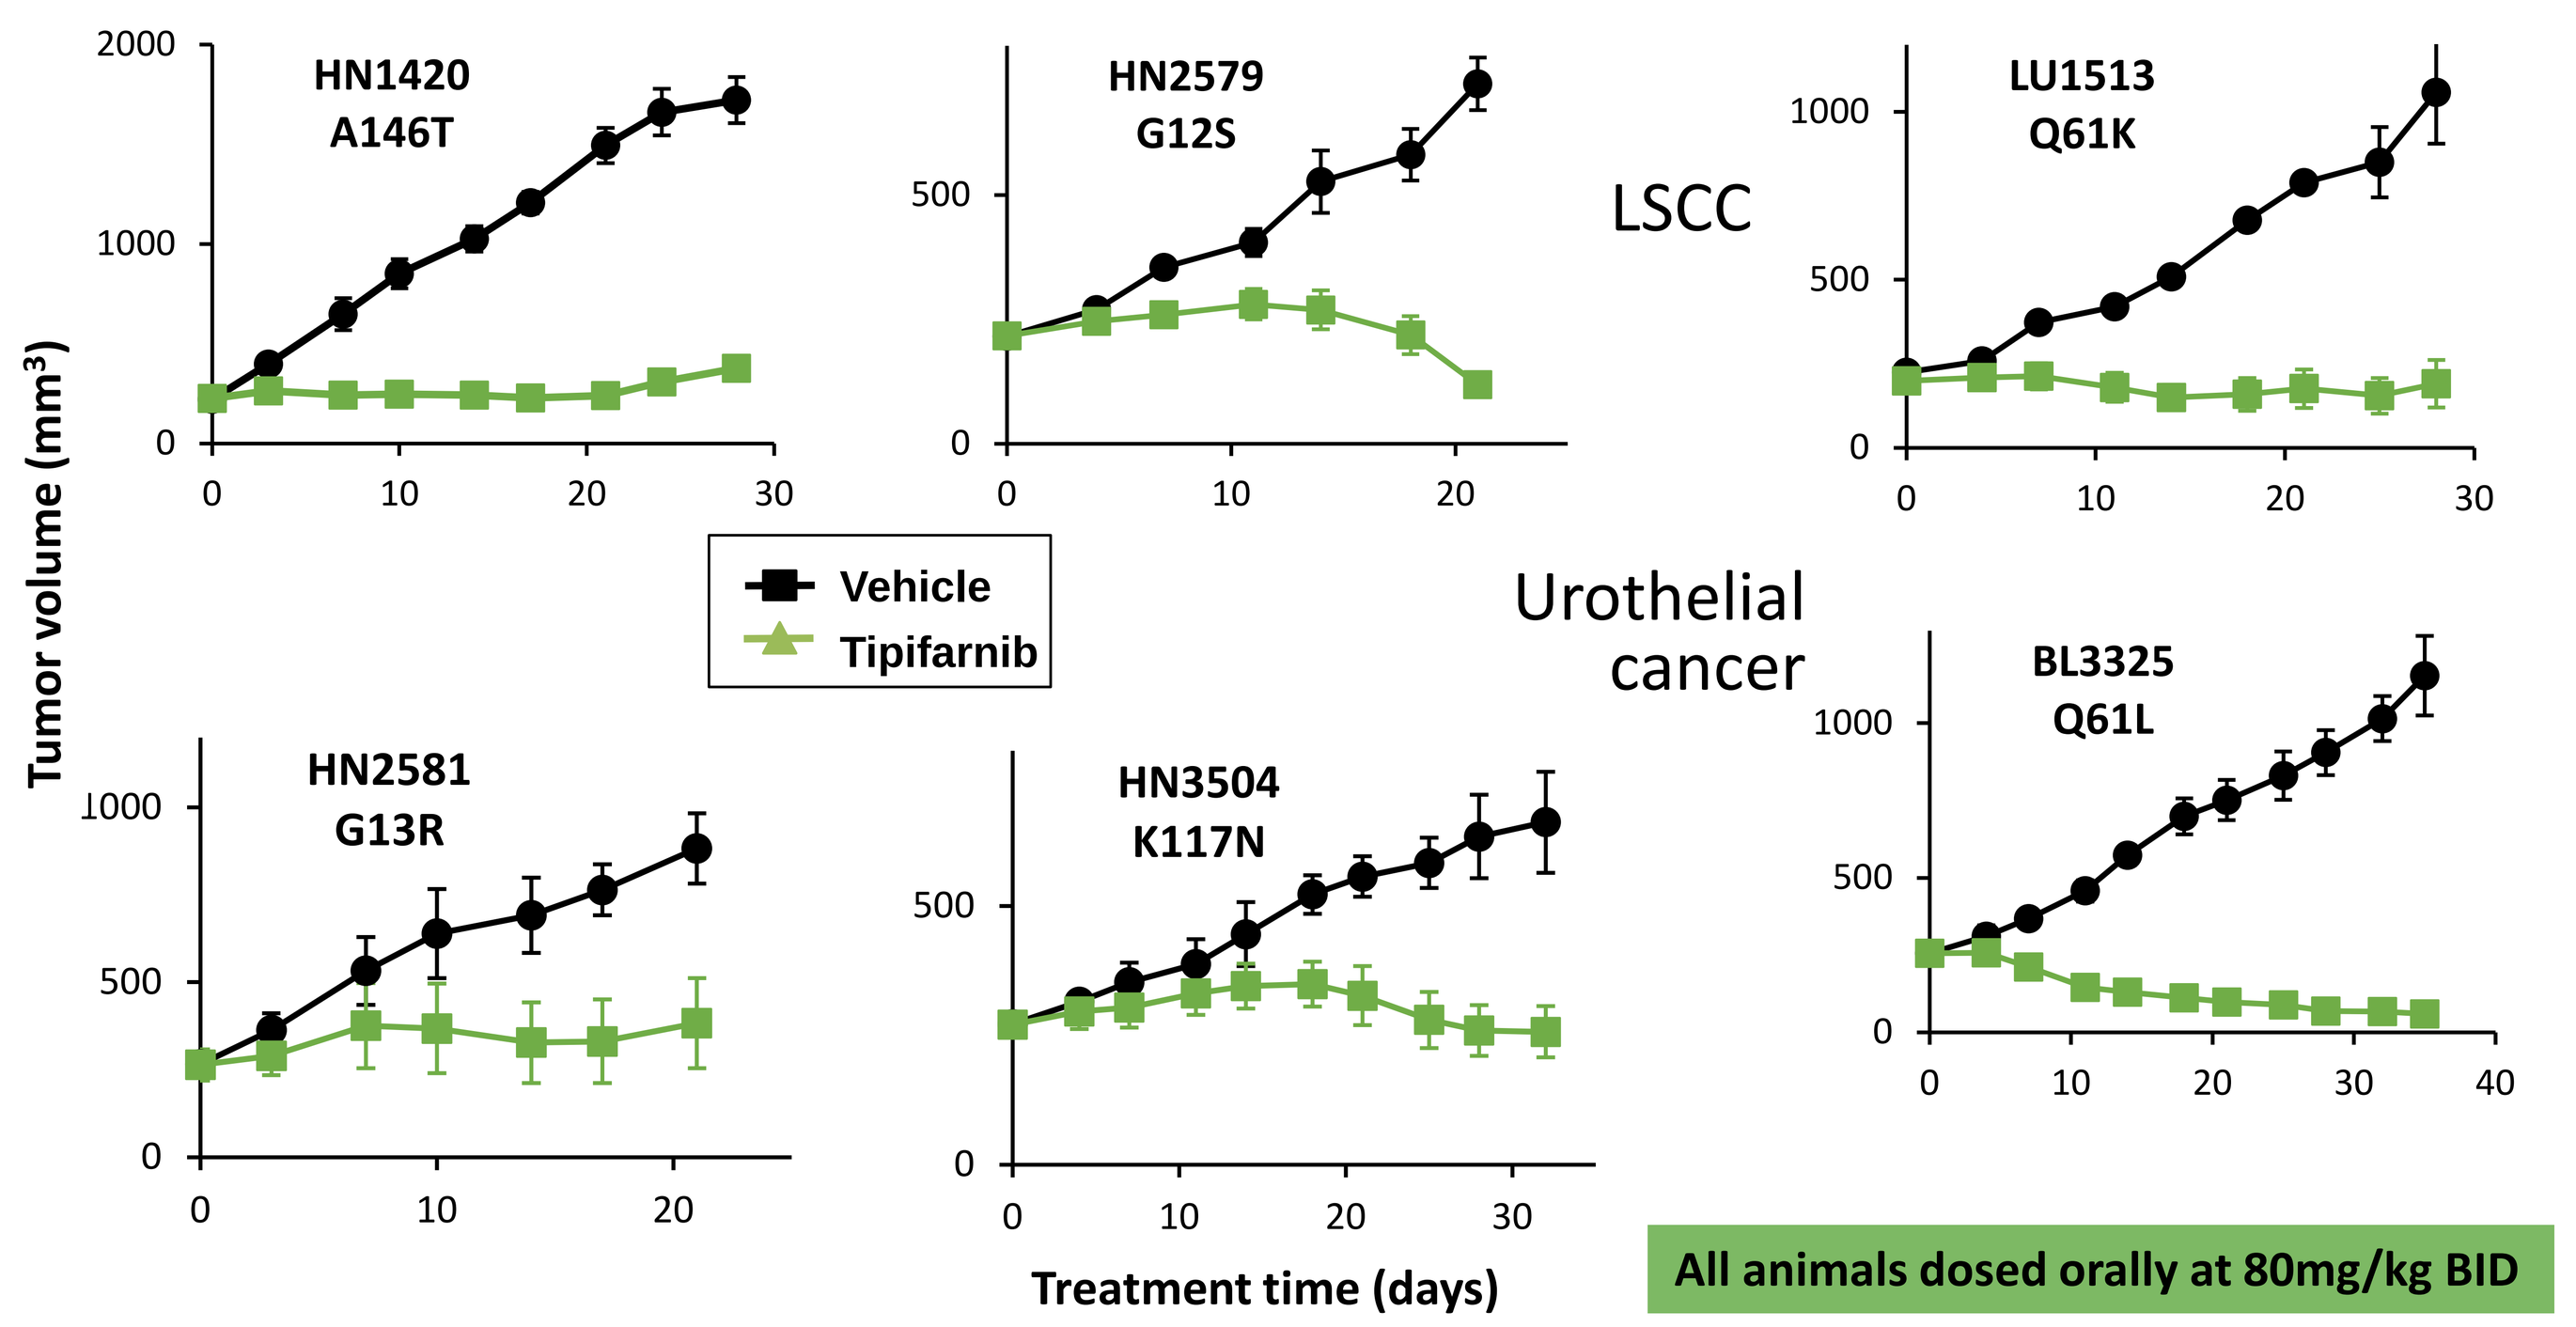
HNSCC
HN1420 A146T
HN2579 G12S
LU1513 Q61K
LSCC
HN2581 G13R
HN3504 K117N
BL3325 Q61L
Urothelial cancer
Vehicle
Tipifarnib
Tumor volume (mm3)
Treatment time (days)

All animals dosed orally at 80mg/kg BID

TIPIFARNIB INHIBITS ONCOGENIC SIGNALING AND PROLIFERATION AND INDUCES APOPTOSIS IN HRAS-MUTANT HNSCC XENOGRAFTS IN VIVO

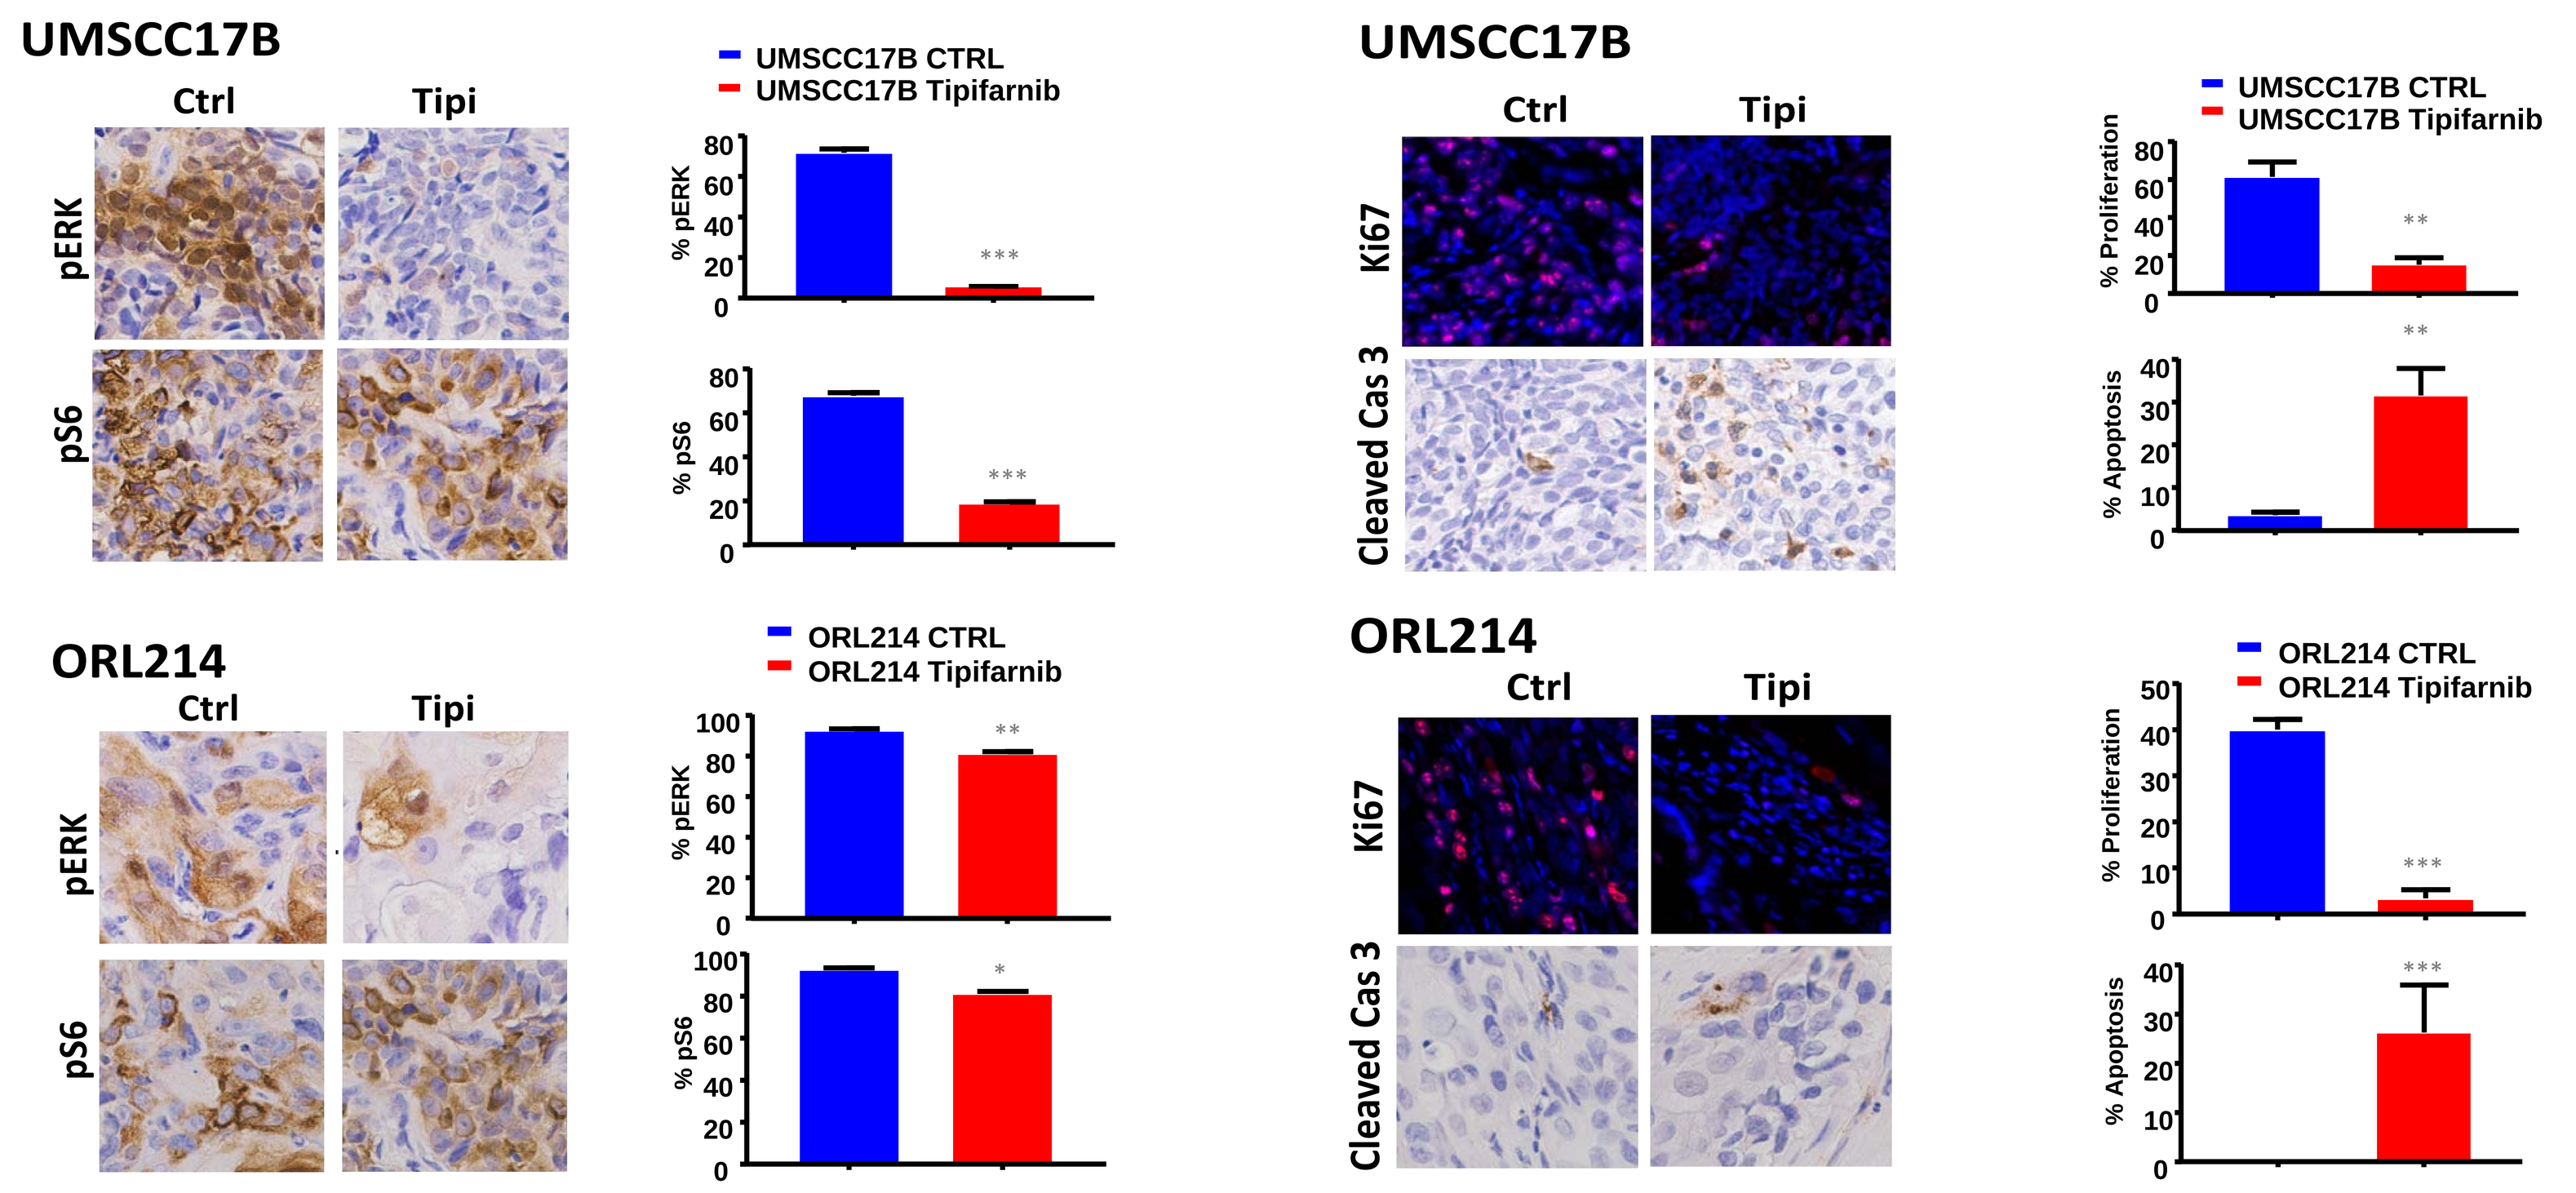
UMSCC17B
Ctrl
Tipi
pERK
pS6
UMSCC17B CTRL
UMSCC17B Tipifarnib
% pERK
% pS6
Ki67
Cleaved Cas 3
% Proliferation
% Apoptosis
ORL214
ORL214 CTRL
ORL214 Tipifarnib

*** p < 0.001
** p < 0.01
* NS

TIPIFARNIB INHIBITS ANGIOGENESIS IN VITRO AND IN VIVO

***In vitro* tube formation**

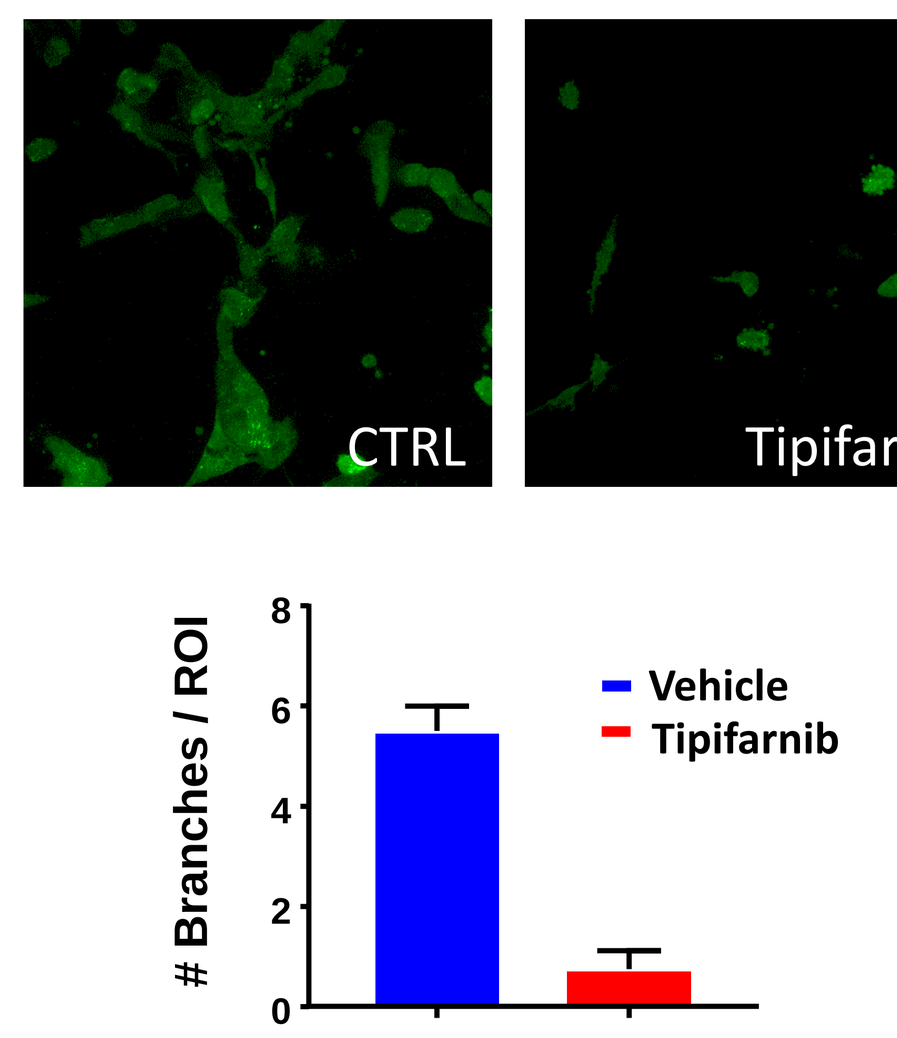
CTRL
Tipifarnib
Branches / ROI
Vehicle
Tipifarnib

***In vivo* vessel density**

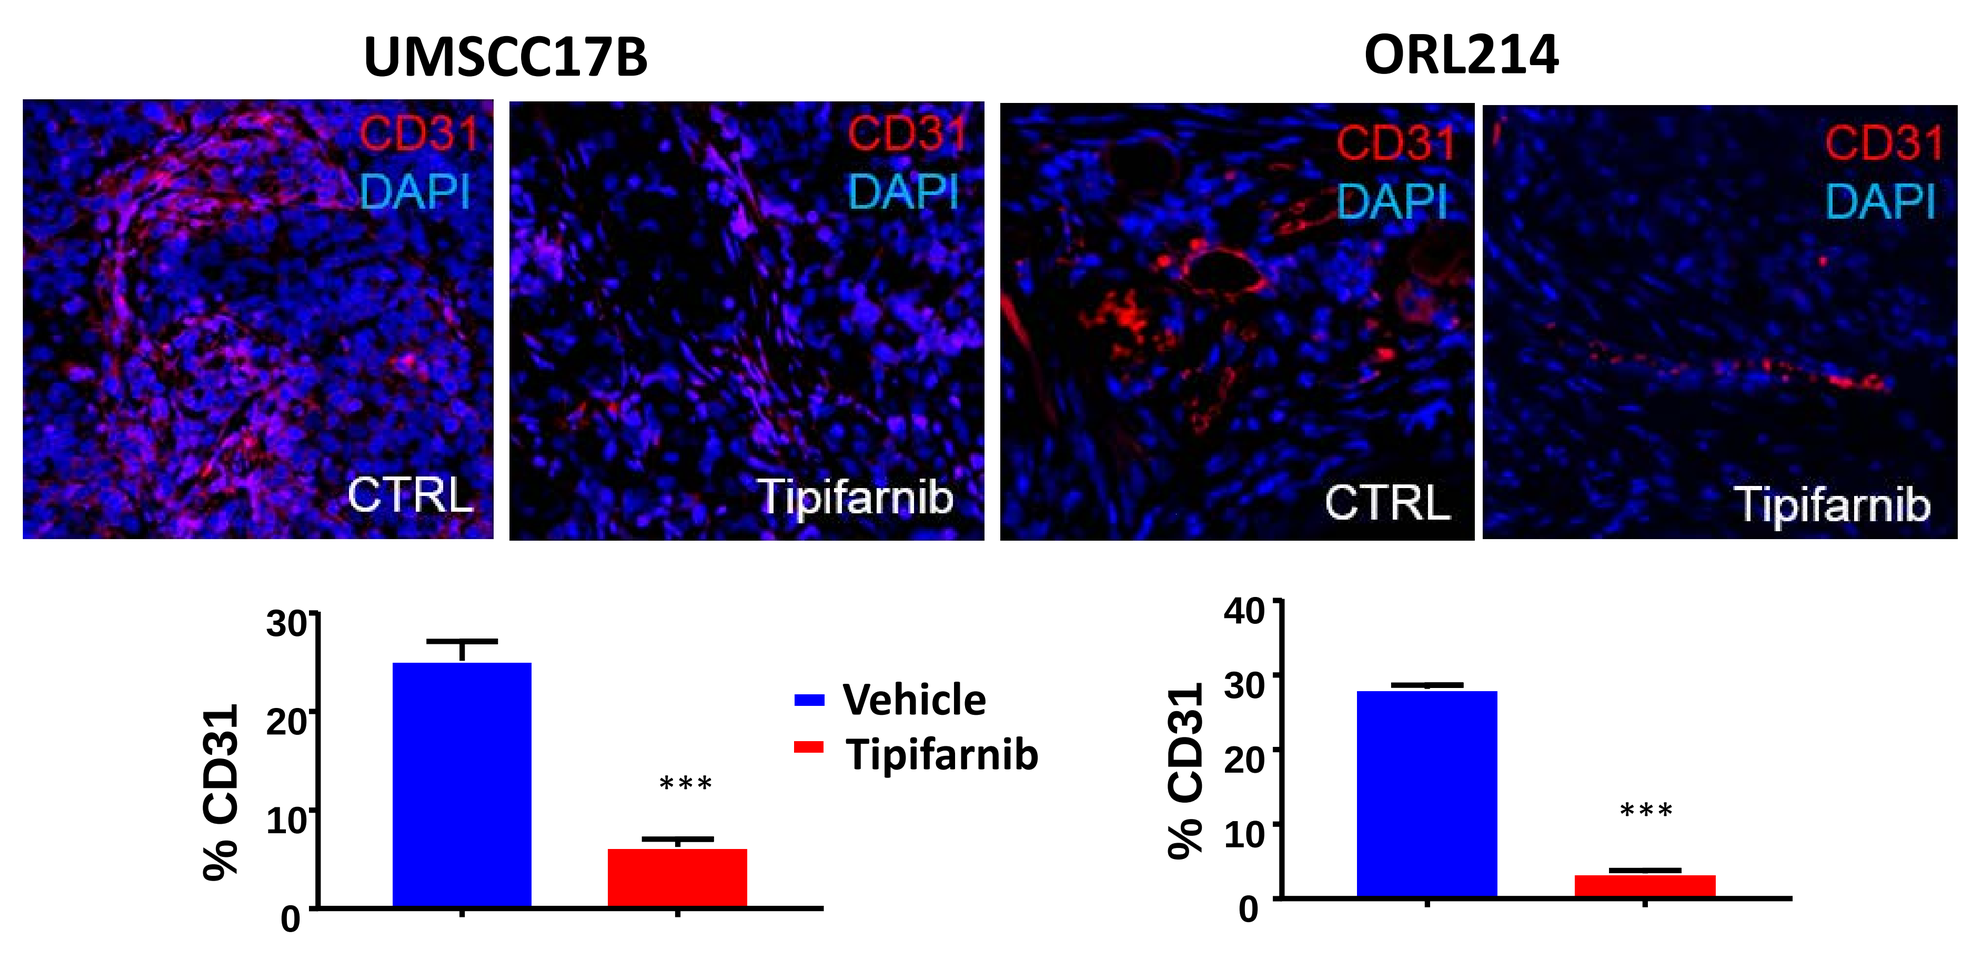
UMSCC17B
ORL214
CD31
DAPI
CTRL
Tipifarnib
% CD31
Vehicle
Tipifarnib

CONCLUSIONS

- *HRAS* is a targetable oncogene in squamous cell carcinomas using tipifarnib
- Tipifarnib prevents *HRAS* prenylation and membrane insertion
- *HRAS*-mutant cells are highly sensitive *in vitro* under both adherent and anchorage-independent conditions
- Tipifarnib induced stasis or regression in HRAS-mutant HNSCC xenograft models, independently of *HRAS* genotype
- Antitumor activity *in vivo* is associated with inhibition of MAPK and PI3K-mTOR signaling, cell cycle arrest and robust activation of apoptosis
- Direct antitumor effects may be enhanced by antiangiogenic activity of tipifarnib *in vivo*
- Tipifarnib is under active clinical evaluation in HRAS-mutant SCC including HNSCC

Poster PB-083, EORTC-NCI-AACR Molecular Targets and Cancer Therapeutics Symposium November13-16th, 2018