Abstract #1071

Tipifarnib synergizes with a TKI in clear cell renal cell carcinoma models

Jovylyn Gatchalian[1], Linda Kessler[1], Hetika Vora Patel[1], Stacia Chan[1], Francis Burrows[1], and Shivani Malik[1]

[1] Kura Oncology, Inc, San Diego, CA

BACKGROUND

- Clear cell renal cell carcinoma (ccRCC) is a highly vascularized tumor type, primarily due to loss of the Von Hippel-Lindau (VHL) gene, which is observed in ~88% of ccRCC cases[1]
- Deletion of VHL stabilizes hypoxia-inducible factor alpha proteins, HIF1α and HIF2α, primary drivers of the angiogenic response in this cell type[1]
- Anti-angiogenic tyrosine kinase inhibitor (TKIs) such as the VEGFR1/2/3 inhibitor axitinib have demonstrated therapeutic benefit in ccRCC patients by exploiting the critical dependency of the tumors on the vasculature.[2]
- However, resistance to TKIs is commonly observed, necessitating development of rational combination strategies
- Tipifarnib, a farnesyltransferase inhibitor, potentially blocks hyperactivated growth factor signaling at multiple nodes[3,4]
- **Here, we utilize cell line and PDX models to establish the scope and mechanistic underpinnings of tipifarnib-axitinib effects and strengthen the scientific rationale for combining TKIs with tipifarnib in the treatment of patients with ccRCC**

RESULTS

Tipifarnib-axitinib combination causes tumor regression or stasis in VHL-mutant and VHL-wildtype ccRCC models

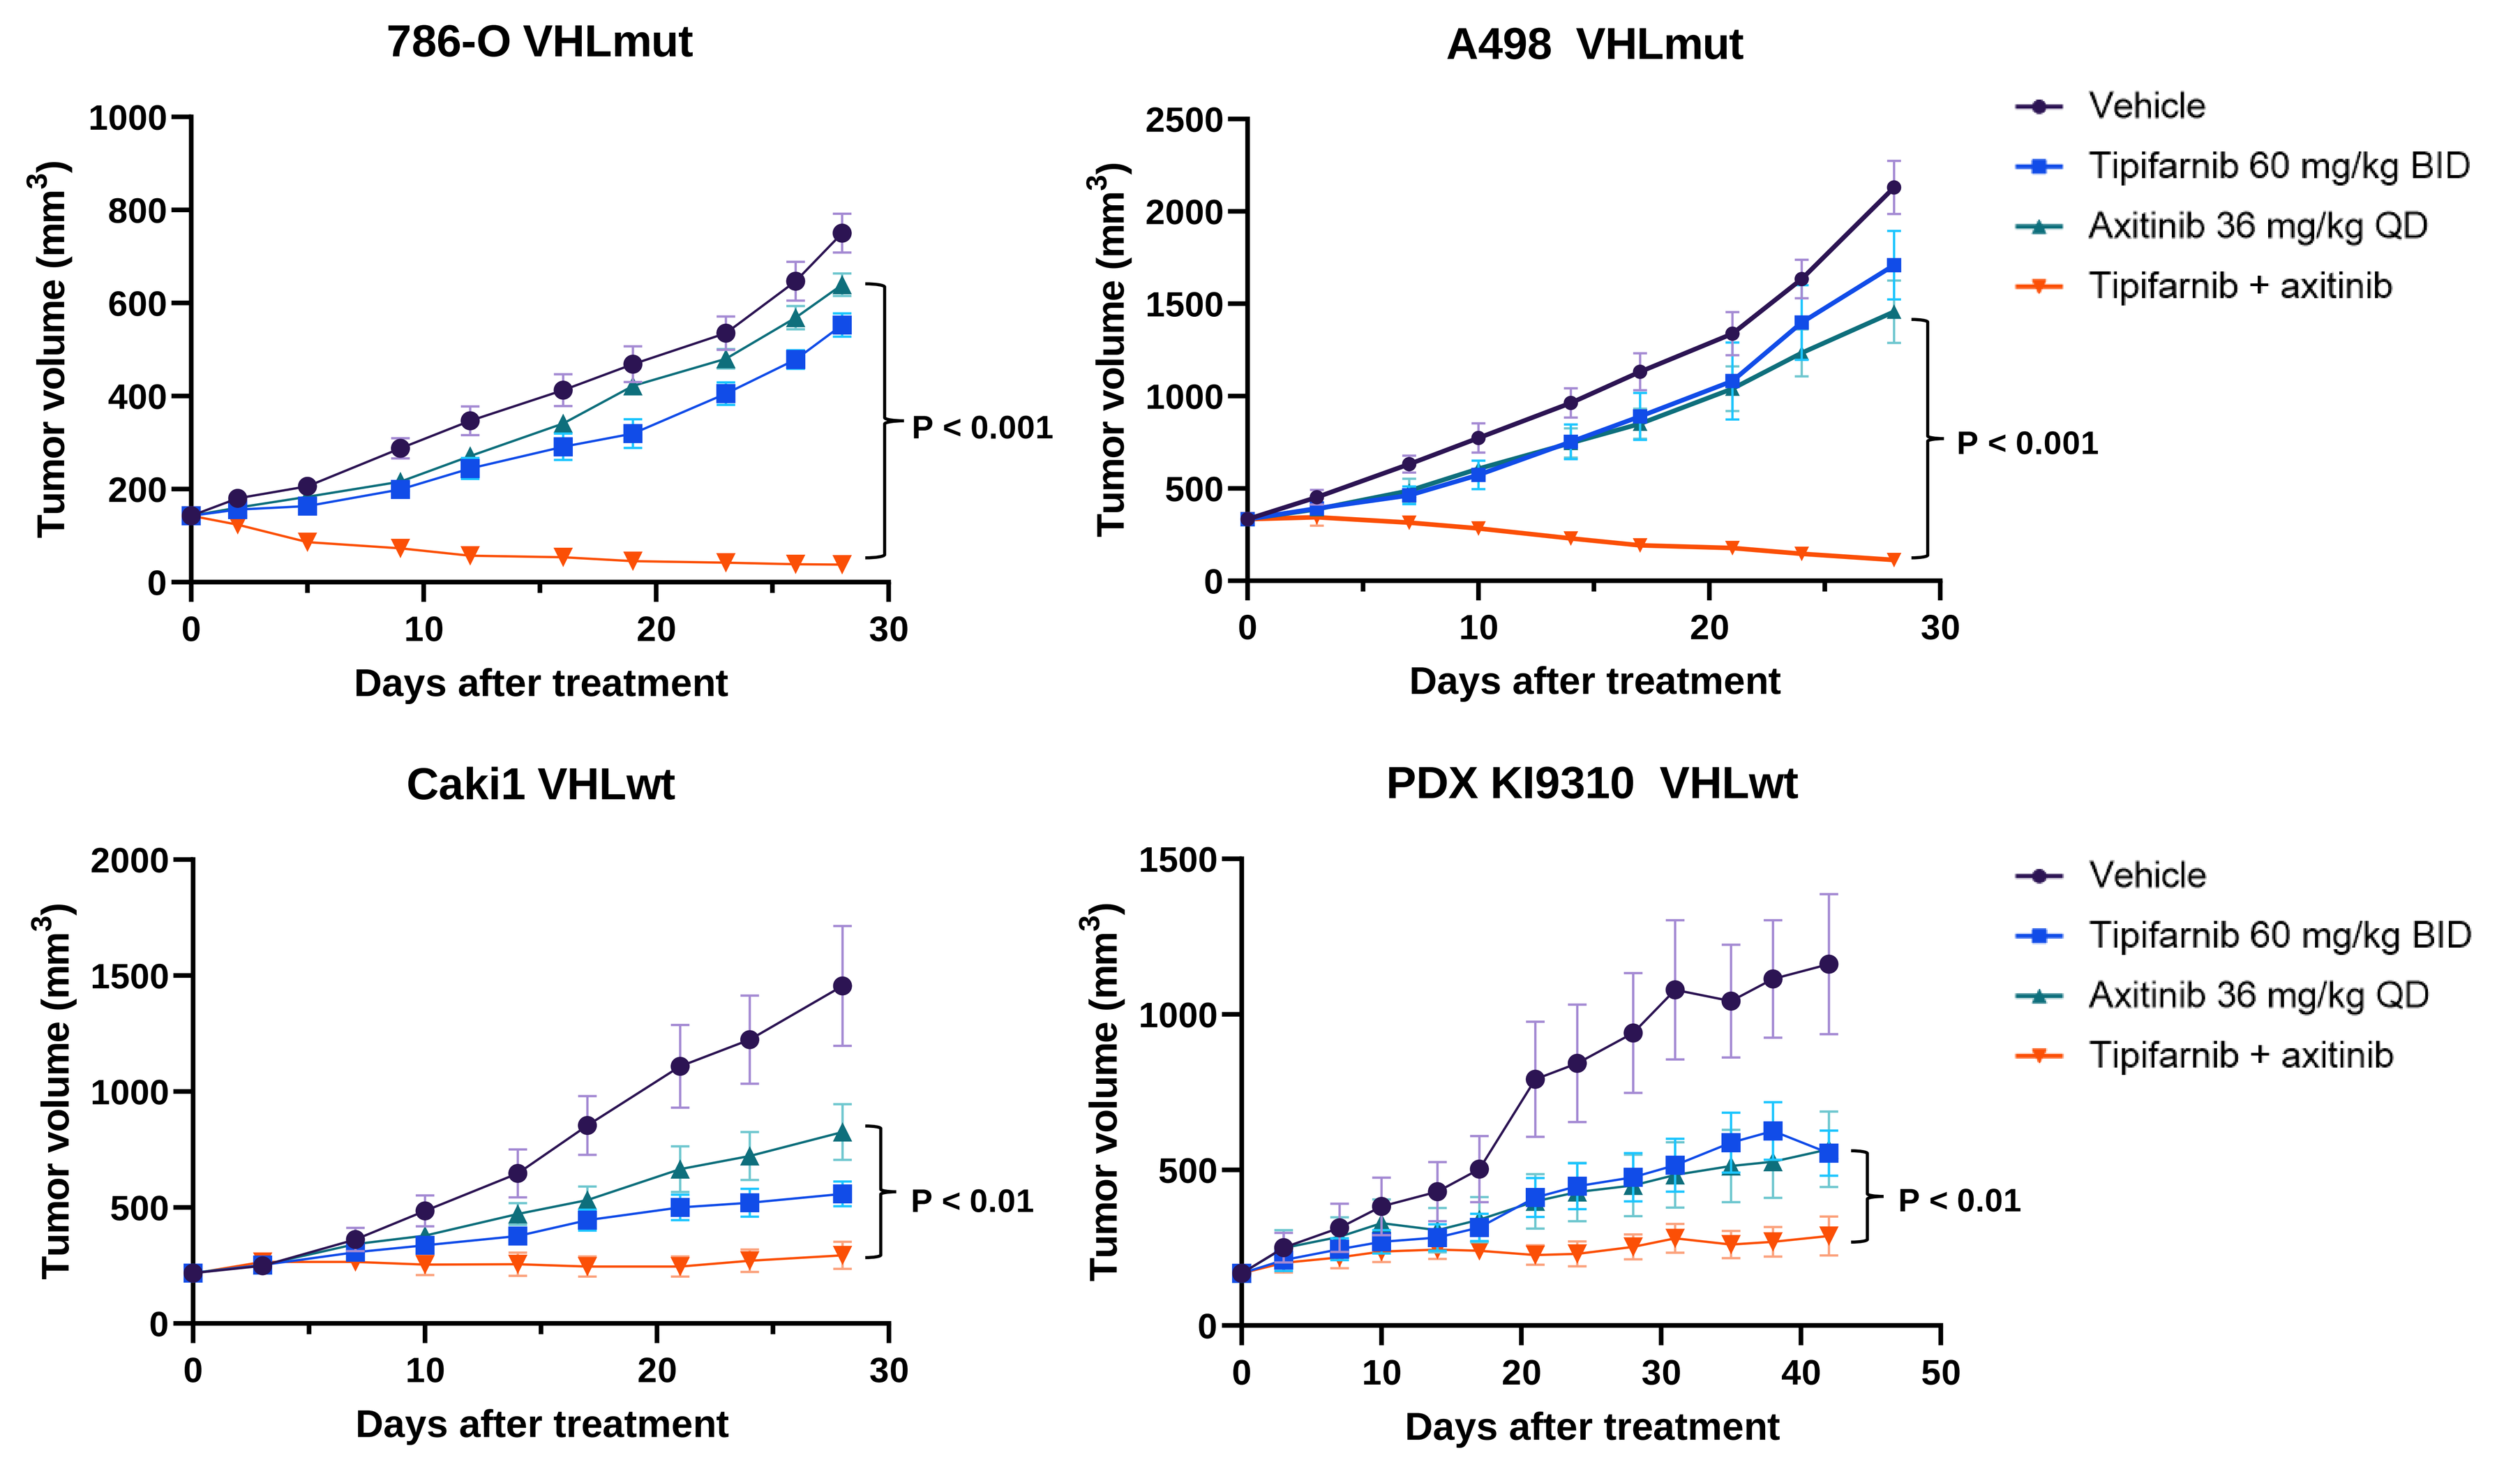
786-O VHLmut
A498 VHLmut
Caki1 VHLwt
PDX KI9310 VHLwt
Tumor volume (mm3)
Days after treatment
Vehicle
Tipifarnib 60 mg/kg BID
Axitinib 36 mg/kg QD
Tipifarnib + axitinib
P < 0.001
P < 0.01

Figure 1. Continuous treatment of tipifarnib plus the anti-angiogenic TKI axitinib robustly inhibits the growth of ccRCC cell line and patient derived xenograft models. Tumor growth curves of ccRCC CDX and PDX models harboring either VHL mutant (VHLmut) or VHL wildtype (VHLwt), treated with vehicle, tipifarnib (60 mg/kg BID), axitinib (36 mg/kg QD), or the combination.

Tipifarnib enhances the anti-angiogenic activity of axitinib *in vivo*

A)

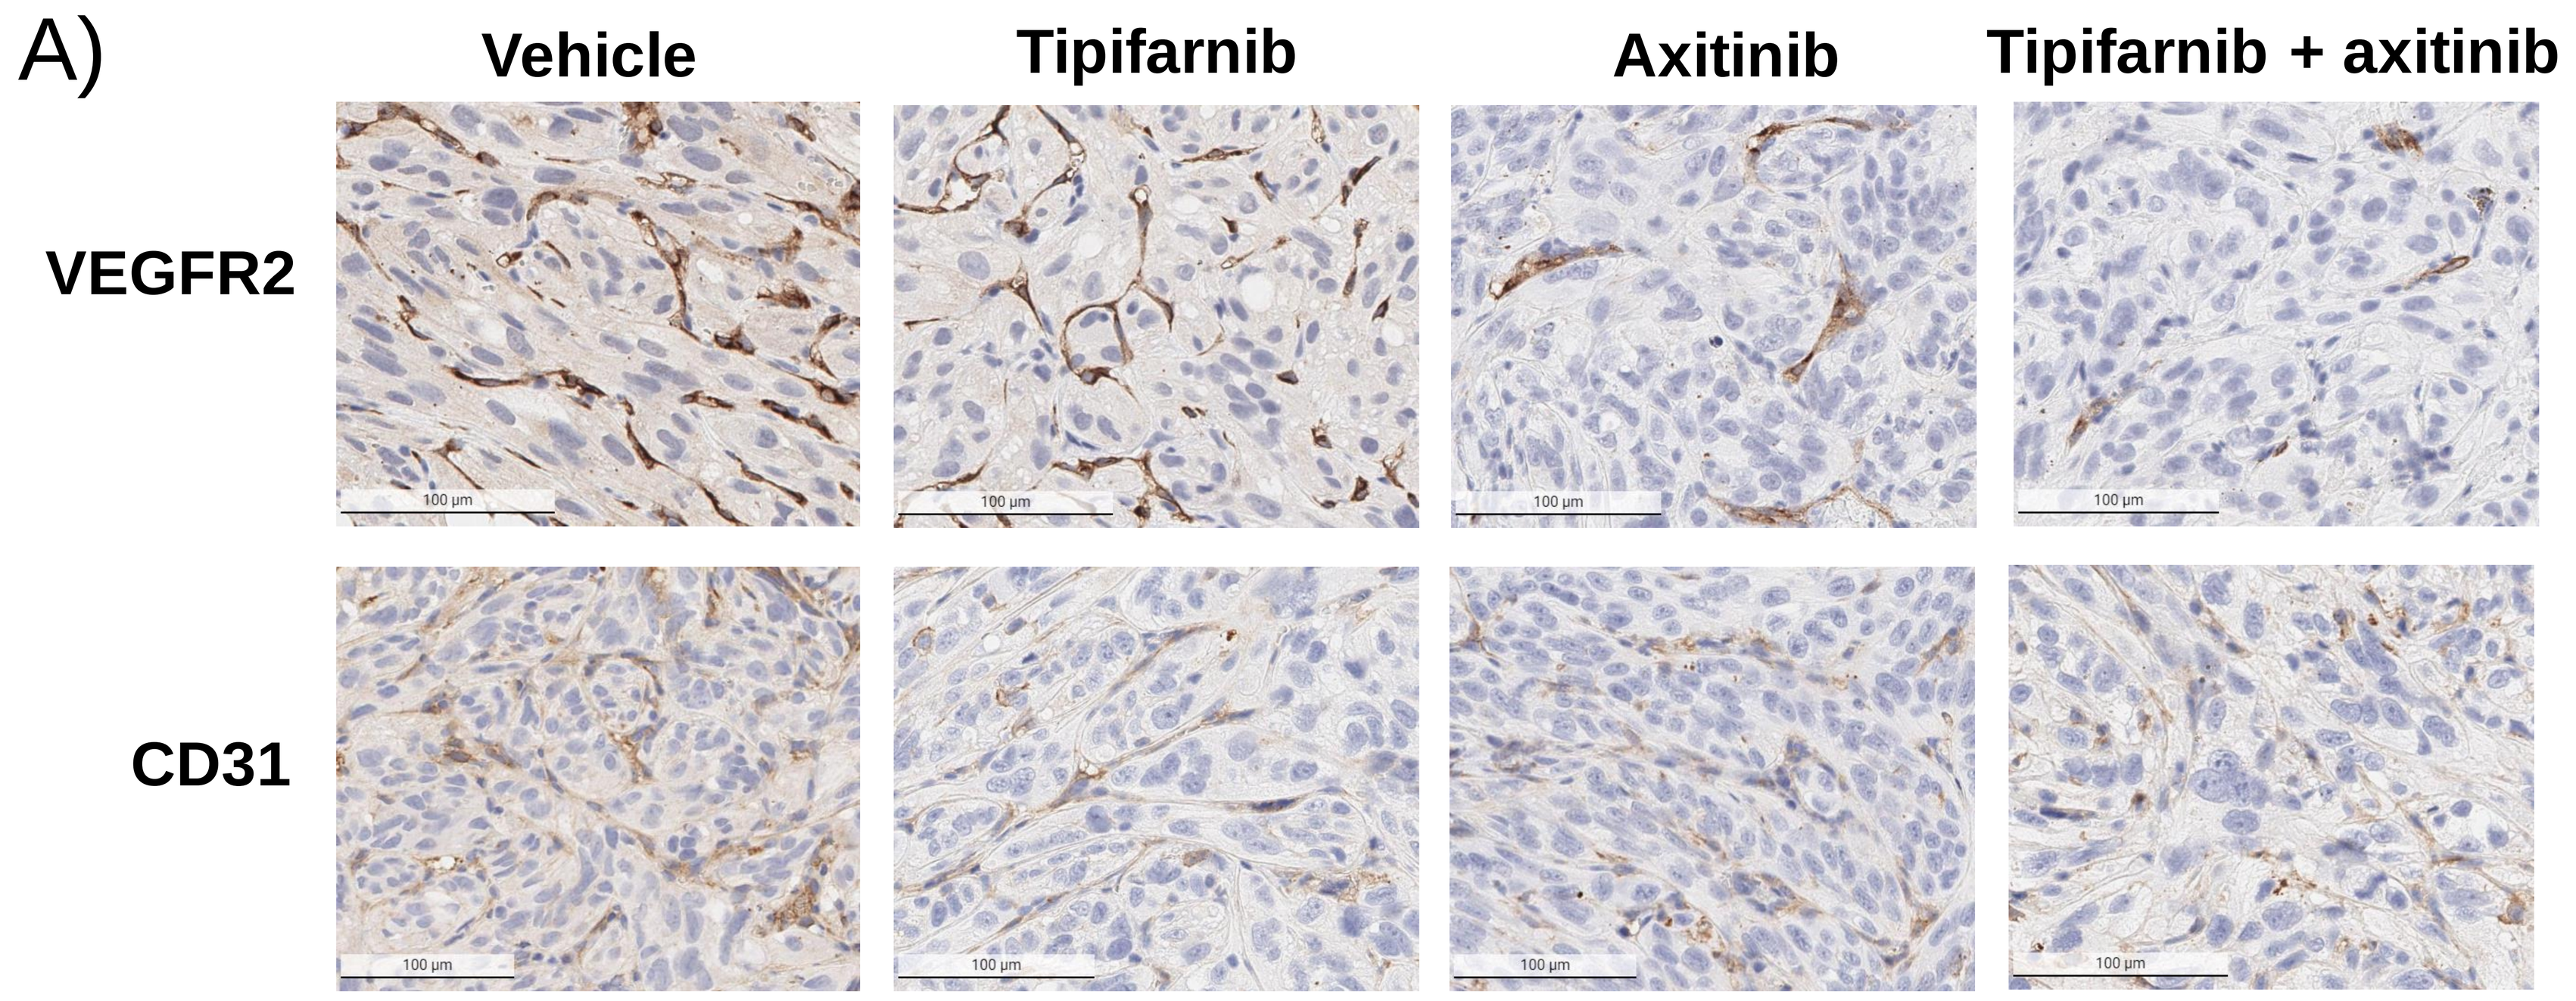
Vehicle
Tipifarnib
Axitinib
Tipifarnib + axitinib
VEGFR2
CD31

B)

% of area	Vehicle	Tipifarnib	Axitinib	Tipifarnib + axitinib
VEGFR2	29.1	13.5	3	0.01
CD31	39.9	11.5	10.9	1.7

Figure 2. Tipifarnib plus axitinib combination leads to decreased expression of endothelial cell markers *in vivo*. A) Representative images of immunohistochemistry analysis for vascular markers in 786-O tumors collected after thirteen days of treatment with vehicle, tipifarnib (60 mg/kg BID), axitinib (36 mg/kg QD), or the combination. VEGFR2 and CD31 are markers of endothelial cells. B) Quantitation of indicated target expression is expressed by percent of area with positive stain over the tumor area, for each of the tumors shown in A. Measurements were performed by a pathologist, who was blinded to the identity of the treatment groups.

Tipifarnib and axitinib inhibit endothelial cell tube formation *in vitro*

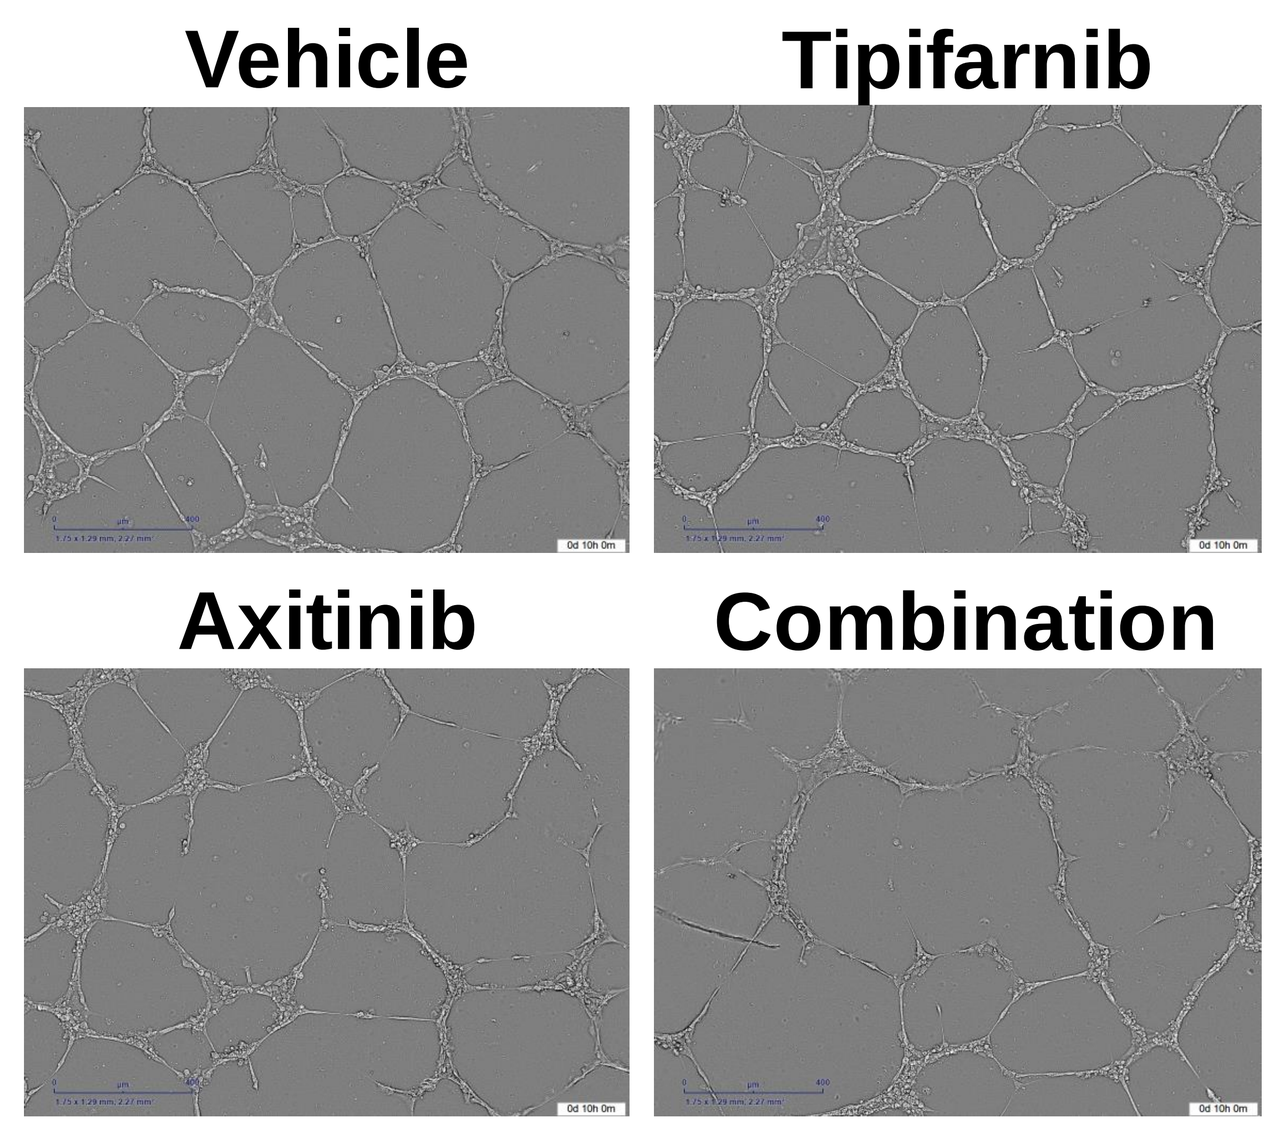
Vehicle
Tipifarnib
Axitinib
Combination

	# of nodes	# of segments	% mesh area
Vehicle	228	86	37.8
Tipifarnib	159	64	25.2
Axitinib	120	40	23.9
Combination	120	41	21.2

Figure 3. Tipifarnib and axitinib inhibit tube formation of primary human umbilical vein endothelial cells (HUVEC) *in vitro*. Left, representative images of tubes formed by HUVECs on matrix proteins after 7 hours of treatment with vehicle, 1 μM tipifarnib, 100 nM axitinib, or the combination. Right, quantitation of tube parameters using ImageJ plugin Angiogenesis Analyzer.

Potential mechanisms of synergy for tipifarnib plus axitinib combination in ccRCC

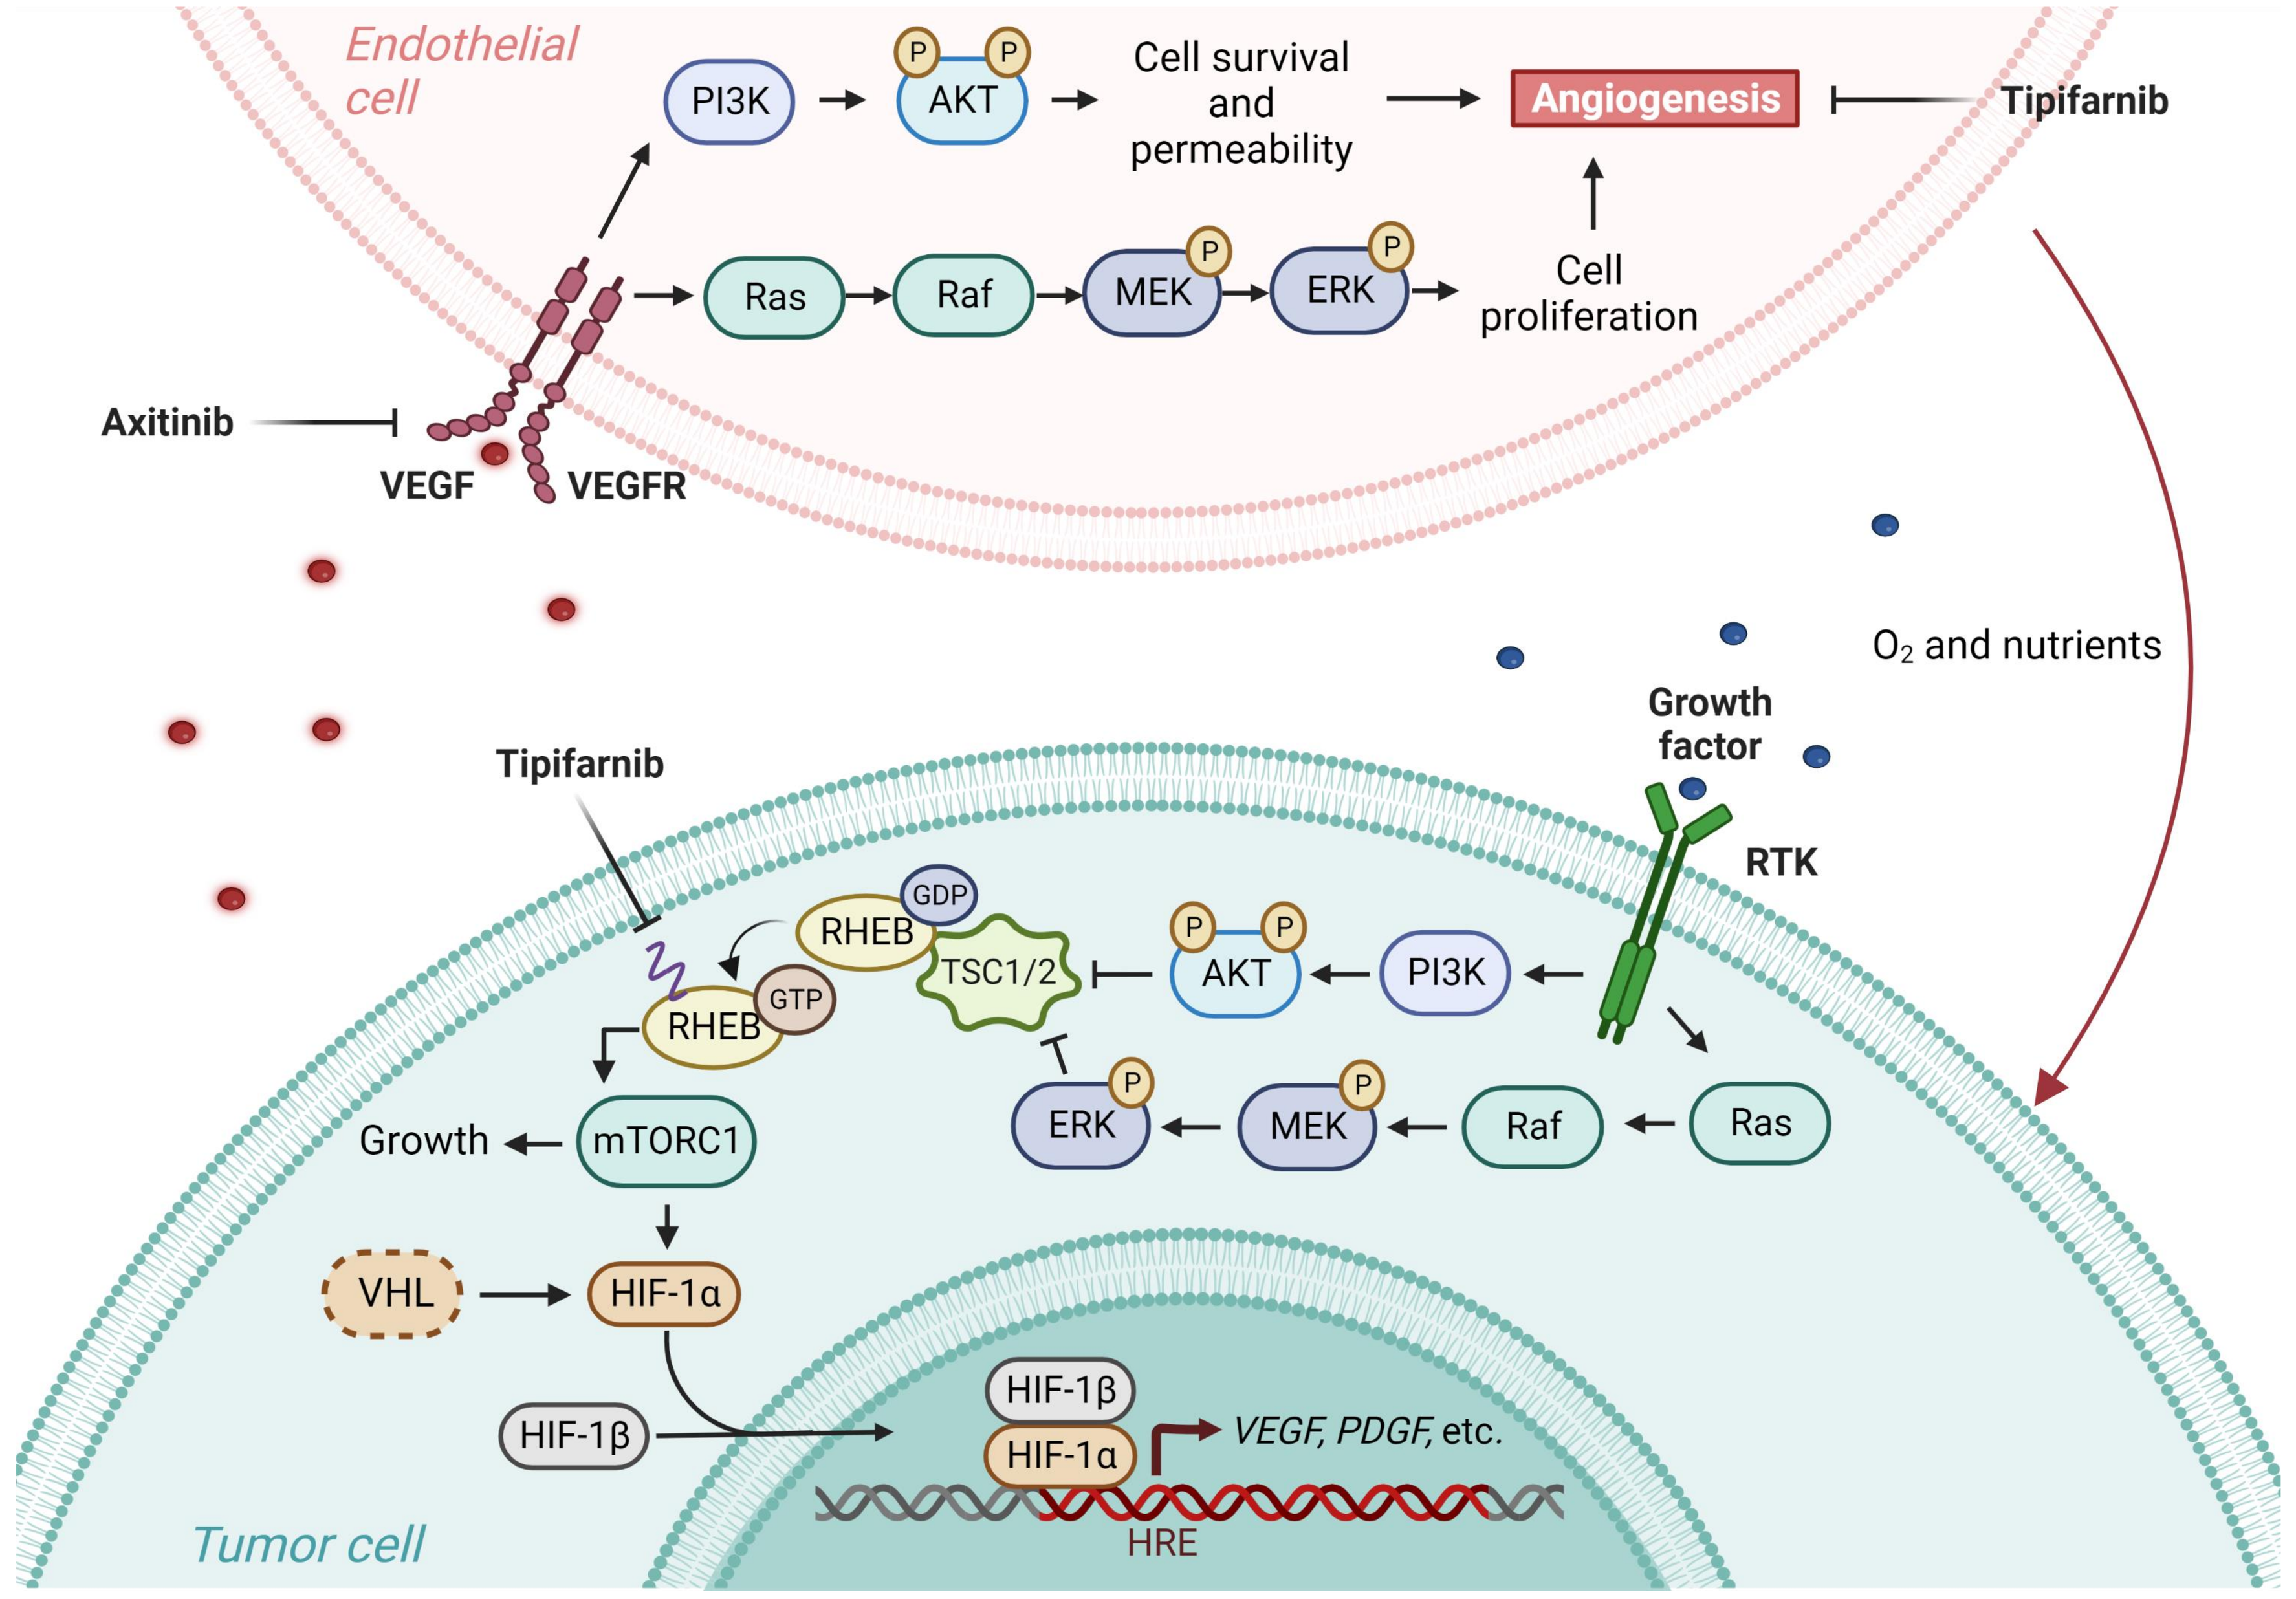
Endothelial cell
PI3K
AKT
P
Cell survival and permeability
Angiogenesis
Tipifarnib
Ras
Raf
MEK
ERK
Cell proliferation
Axitinib
VEGF
VEGFR
O2 and nutrients
Growth factor
RTK
Tipifarnib
RHEB
GDP
GTP
TSC1/2
Growth
mTORC1
VHL
HIF-1α
HIF-1β
VEGF, PDGF, etc.
HRE
Tumor cell

Figure 4. Tipifarnib can affect signaling pathways in both endothelial and tumor cells, providing a potential explanation for the synergistic effect with axitinib. Tipifarnib potentially blocks VEGFA-induced angiogenesis pathways in endothelial cells and/or mTOR-dependent signaling in tumor cells. For the latter, we hypothesize that in response to hypoxia induced by decreased endothelial function and support, tumor cells become more dependent on mTOR signaling, which is sensitive to tipifarnib treatment.[4]

CONCLUSIONS

- **Tipifarnib and axitinib synergize to induce tumor regression or stasis in ccRCC CDX and PDX models.**
- **Tipifarnib enhances the anti-angiogenic activity of axitinib *in vivo,* as observed by decreased expression of endothelial cell markers in 786-O tumors.**
- **The combination of tipifarnib and axitinib holds potential for the treatment of ccRCC. Studies are ongoing to define the basis of the synergy of the combination.**

References
1) Clark P. "The role of VHL in clear cell renal cell carcinoma and its relation to targeted therapy." Kidney International (2009) 76:939-45.
2) Rini, BI. "Comparative effectiveness of axitinib versus sorafenib in advanced renal cell carcinoma (AXIS): a randomized phase 3 trial." The Lancet (2011) 378(9807):1931-39.
3) Gilardi M, et al. "Tipifarnib as a precision therapy for HRAS-mutant head and neck squamous cell carcinomas." Mol Cancer Ther (2020) 19(9):1784-96.
4) Smith AE, et al. "Tipifarnib potentiates the antitumor effects of PI3Kα inhibition in *PIK3CA*- and *HRAS*-dysregulated HNSCC via convergent inhibition of mTOR activity." BioRxiv 2023.01.17.523964.