Combination of tipifarnib with KRAS^{G12C} inhibitors to prevent adaptive resistance

Hetika Vora Patel[1], Alison Smith[1], Stacia Chan[1], Linda Kessler[1], Francis Burrows[1], and Shivani Malik[1]

[1] Kura Oncology, Inc, San Diego, CA

Abstract #1079

BACKGROUND

Selective, irreversible KRAS^{G12C} inhibitors, including MRTX849 (adagrasib) and AMG510 (sotorasib), exhibit clinical activity with response rates of 45% and 37%, respectively, in patients with *KRAS^{G12C}* NSCLC[1-3]. However, feedback reactivation of the MAPK and/or mTOR signaling pathways seems to limit therapeutic efficacy of single agent KRAS^{G12C} inhibitors, warranting rational combination strategies[1,4,5]. We have shown tipifarnib, a farnesyltransferase inhibitor, blocks compensatory feedback reactivation through inhibition of mTOR signaling[6]. Based on our previous work, we hypothesize that tipifarnib in combination with KRAS^{G12C} inhibitors could potentially inhibit mTOR pathway reactivation, thus enhancing single agent efficacy and durability in *KRAS^{G12C}* NSCLC. **In this study, we utilize xenograft models to evaluate the therapeutic effects of the combination of tipifarnib with a KRAS^{G12C} inhibitor, including adagrasib and sotorasib, in *KRAS^{G12C}* NSCLC.**

RESULTS

Combination of tipifarnib and KRAS^{G12C} inhibitors decrease spheroid growth of *KRAS^{G12C}* NSCLC cell lines in a dose-dependent manner

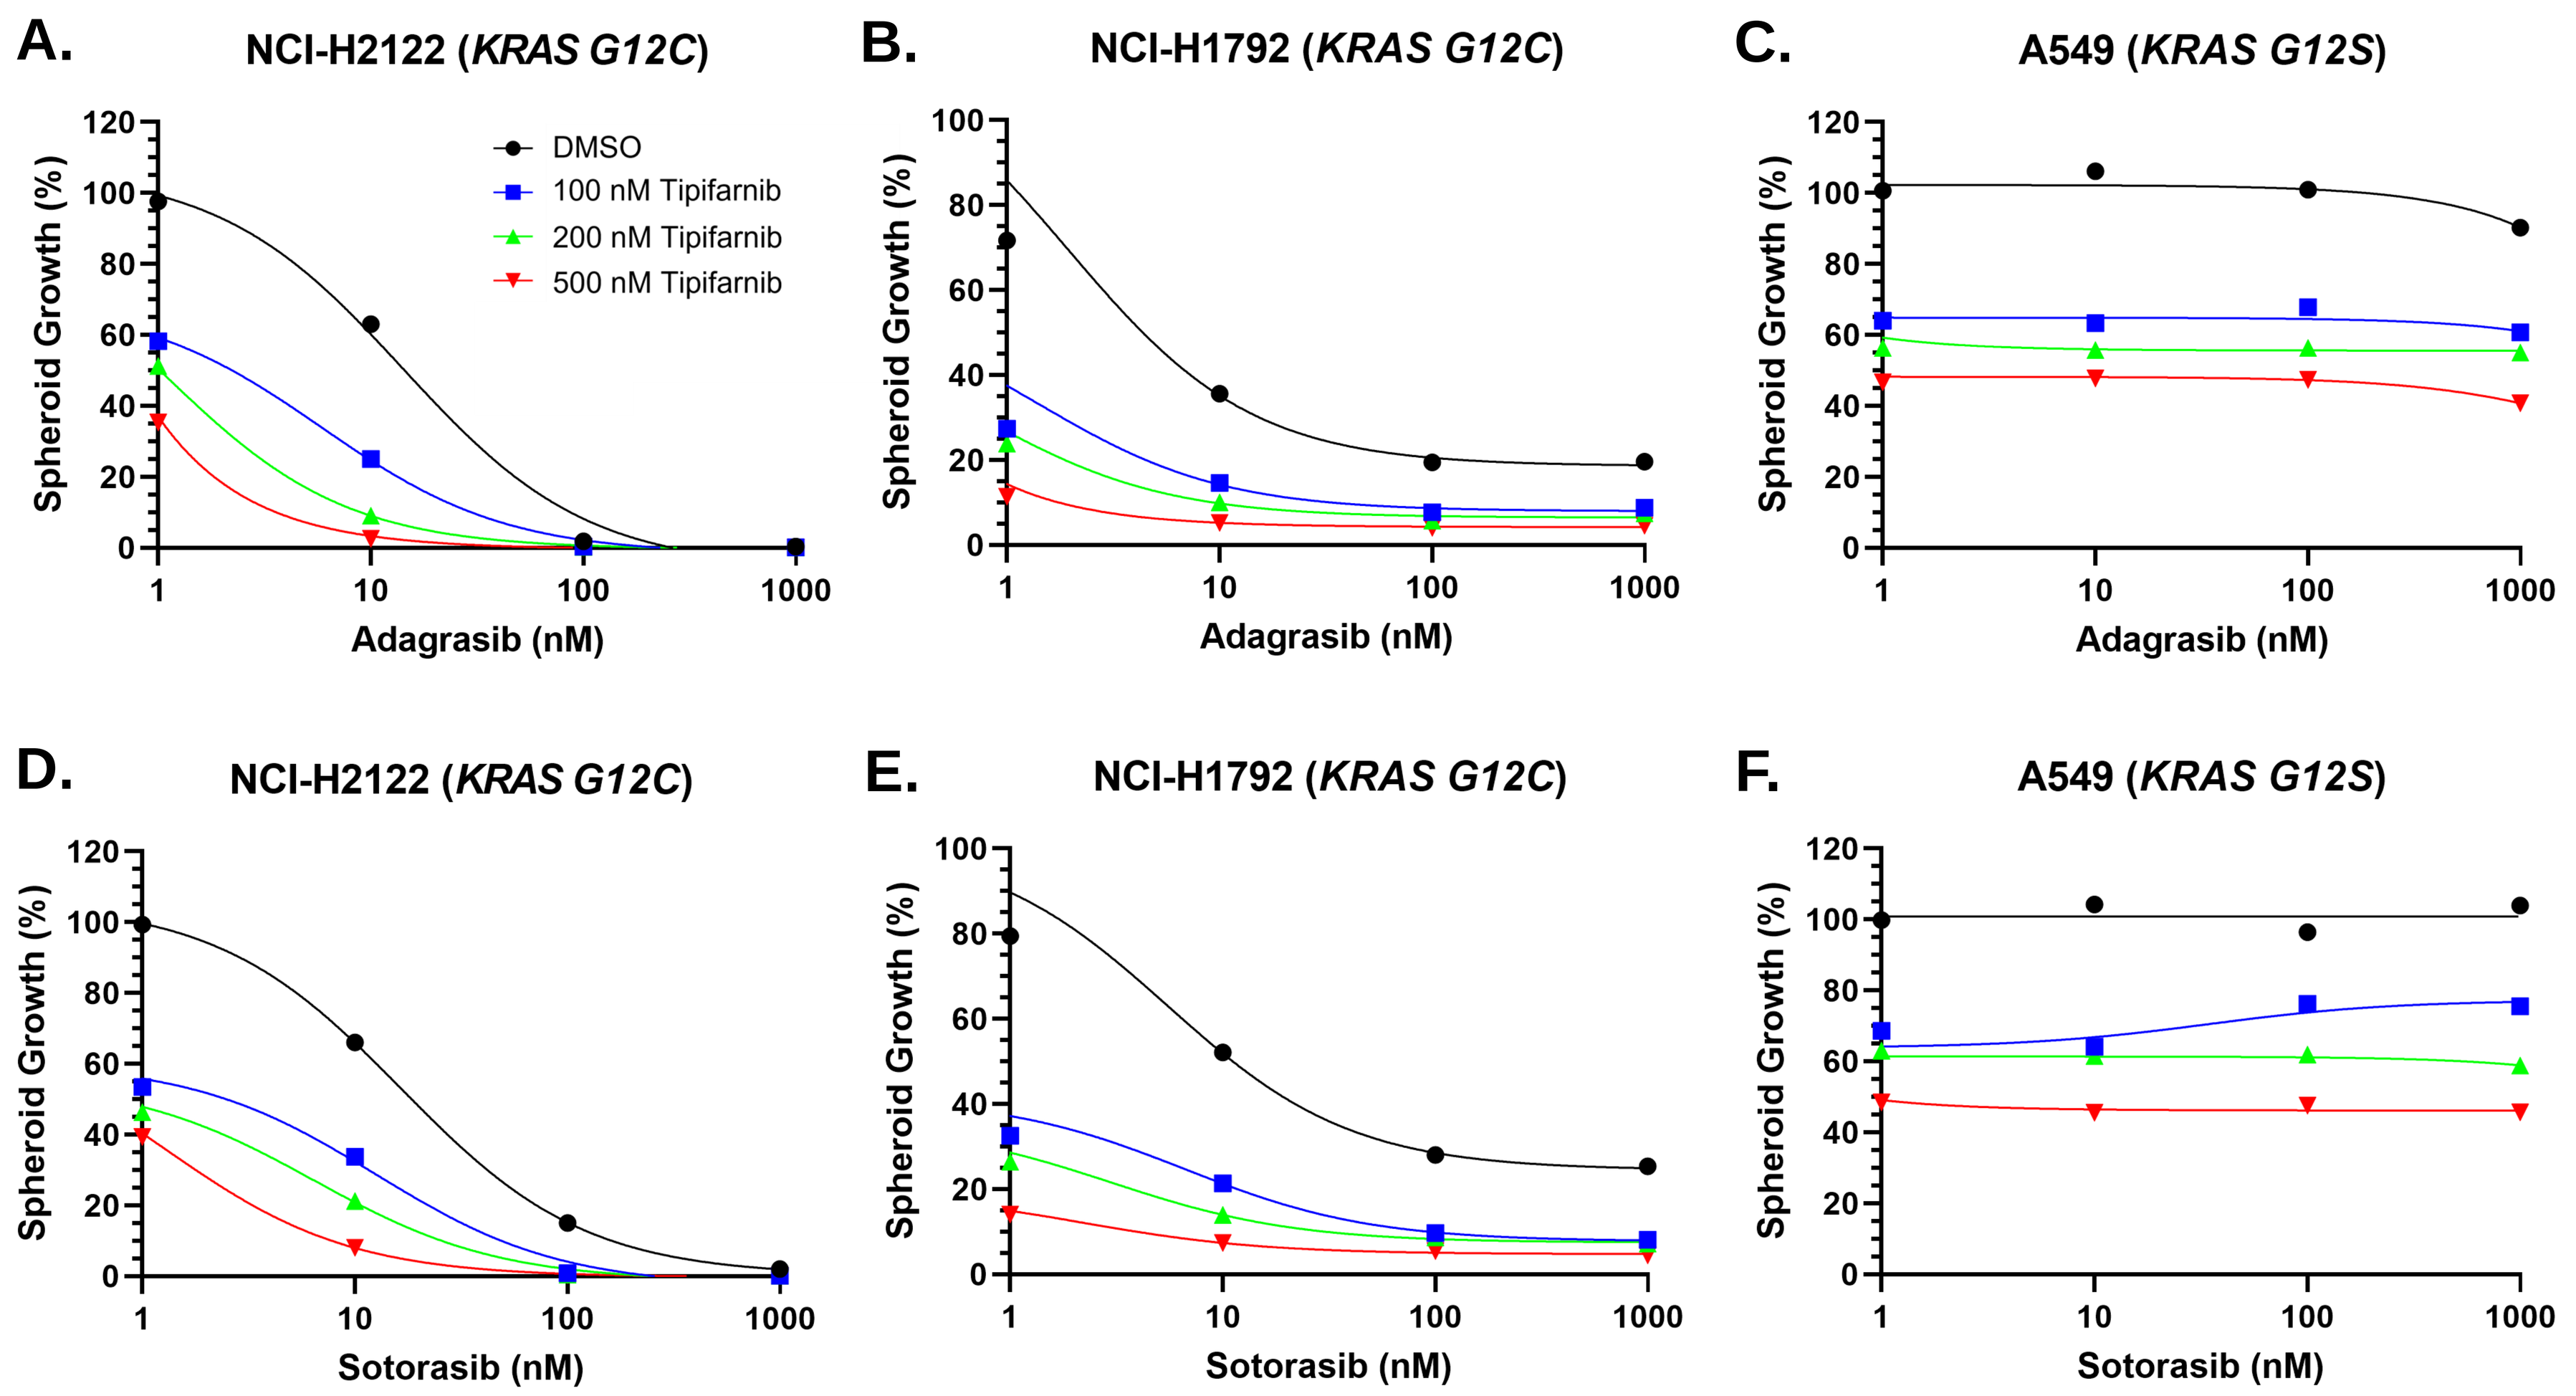
A. NCI-H2122 (KRAS G12C)
B. NCI-H1792 (KRAS G12C)
C. A549 (KRAS G12S)
D. NCI-H2122 (KRAS G12C)
E. NCI-H1792 (KRAS G12C)
F. A549 (KRAS G12S)
DMSO
100 nM Tipifarnib
200 nM Tipifarnib
500 nM Tipifarnib
Spheroid Growth (%)
Adagrasib (nM)
Sotorasib (nM)

Figure 1. Tipifarnib and KRAS^{G12C} inhibitors, adagrasib and sotorasib, inhibit the spheroid growth of *KRAS^{G12C}* NSCLC cell line models. *KRAS^{G12C}* NSCLC cell lines were cultured as 3D spheroids and treated for 7 days. Spheroid growth was inhibited in a dose-dependent manner in various *KRAS^{G12C}* NSCLC cell lines. NCI-H2122 *KRAS^{G12C}* NSCLC cell line was treated with tipifarnib and (A) adagrasib or (D) sotorasib, and NCI-H1792 *KRAS^{G12C}* NSCLC cell line was treated with tipifarnib and (B) adagrasib or (E) sotorasib. A549 *KRAS^{G12S}* NSCLC cell line was used as a control for KRAS^{G12C} inhibitor treatment, and this control cell line was treated with tipifarnib and (C) adagrasib or (F) sotorasib.

Combination of tipifarnib with KRAS^{G12C} inhibitors causes tumor regression in patient-derived and cell-derived NSCLC xenografts

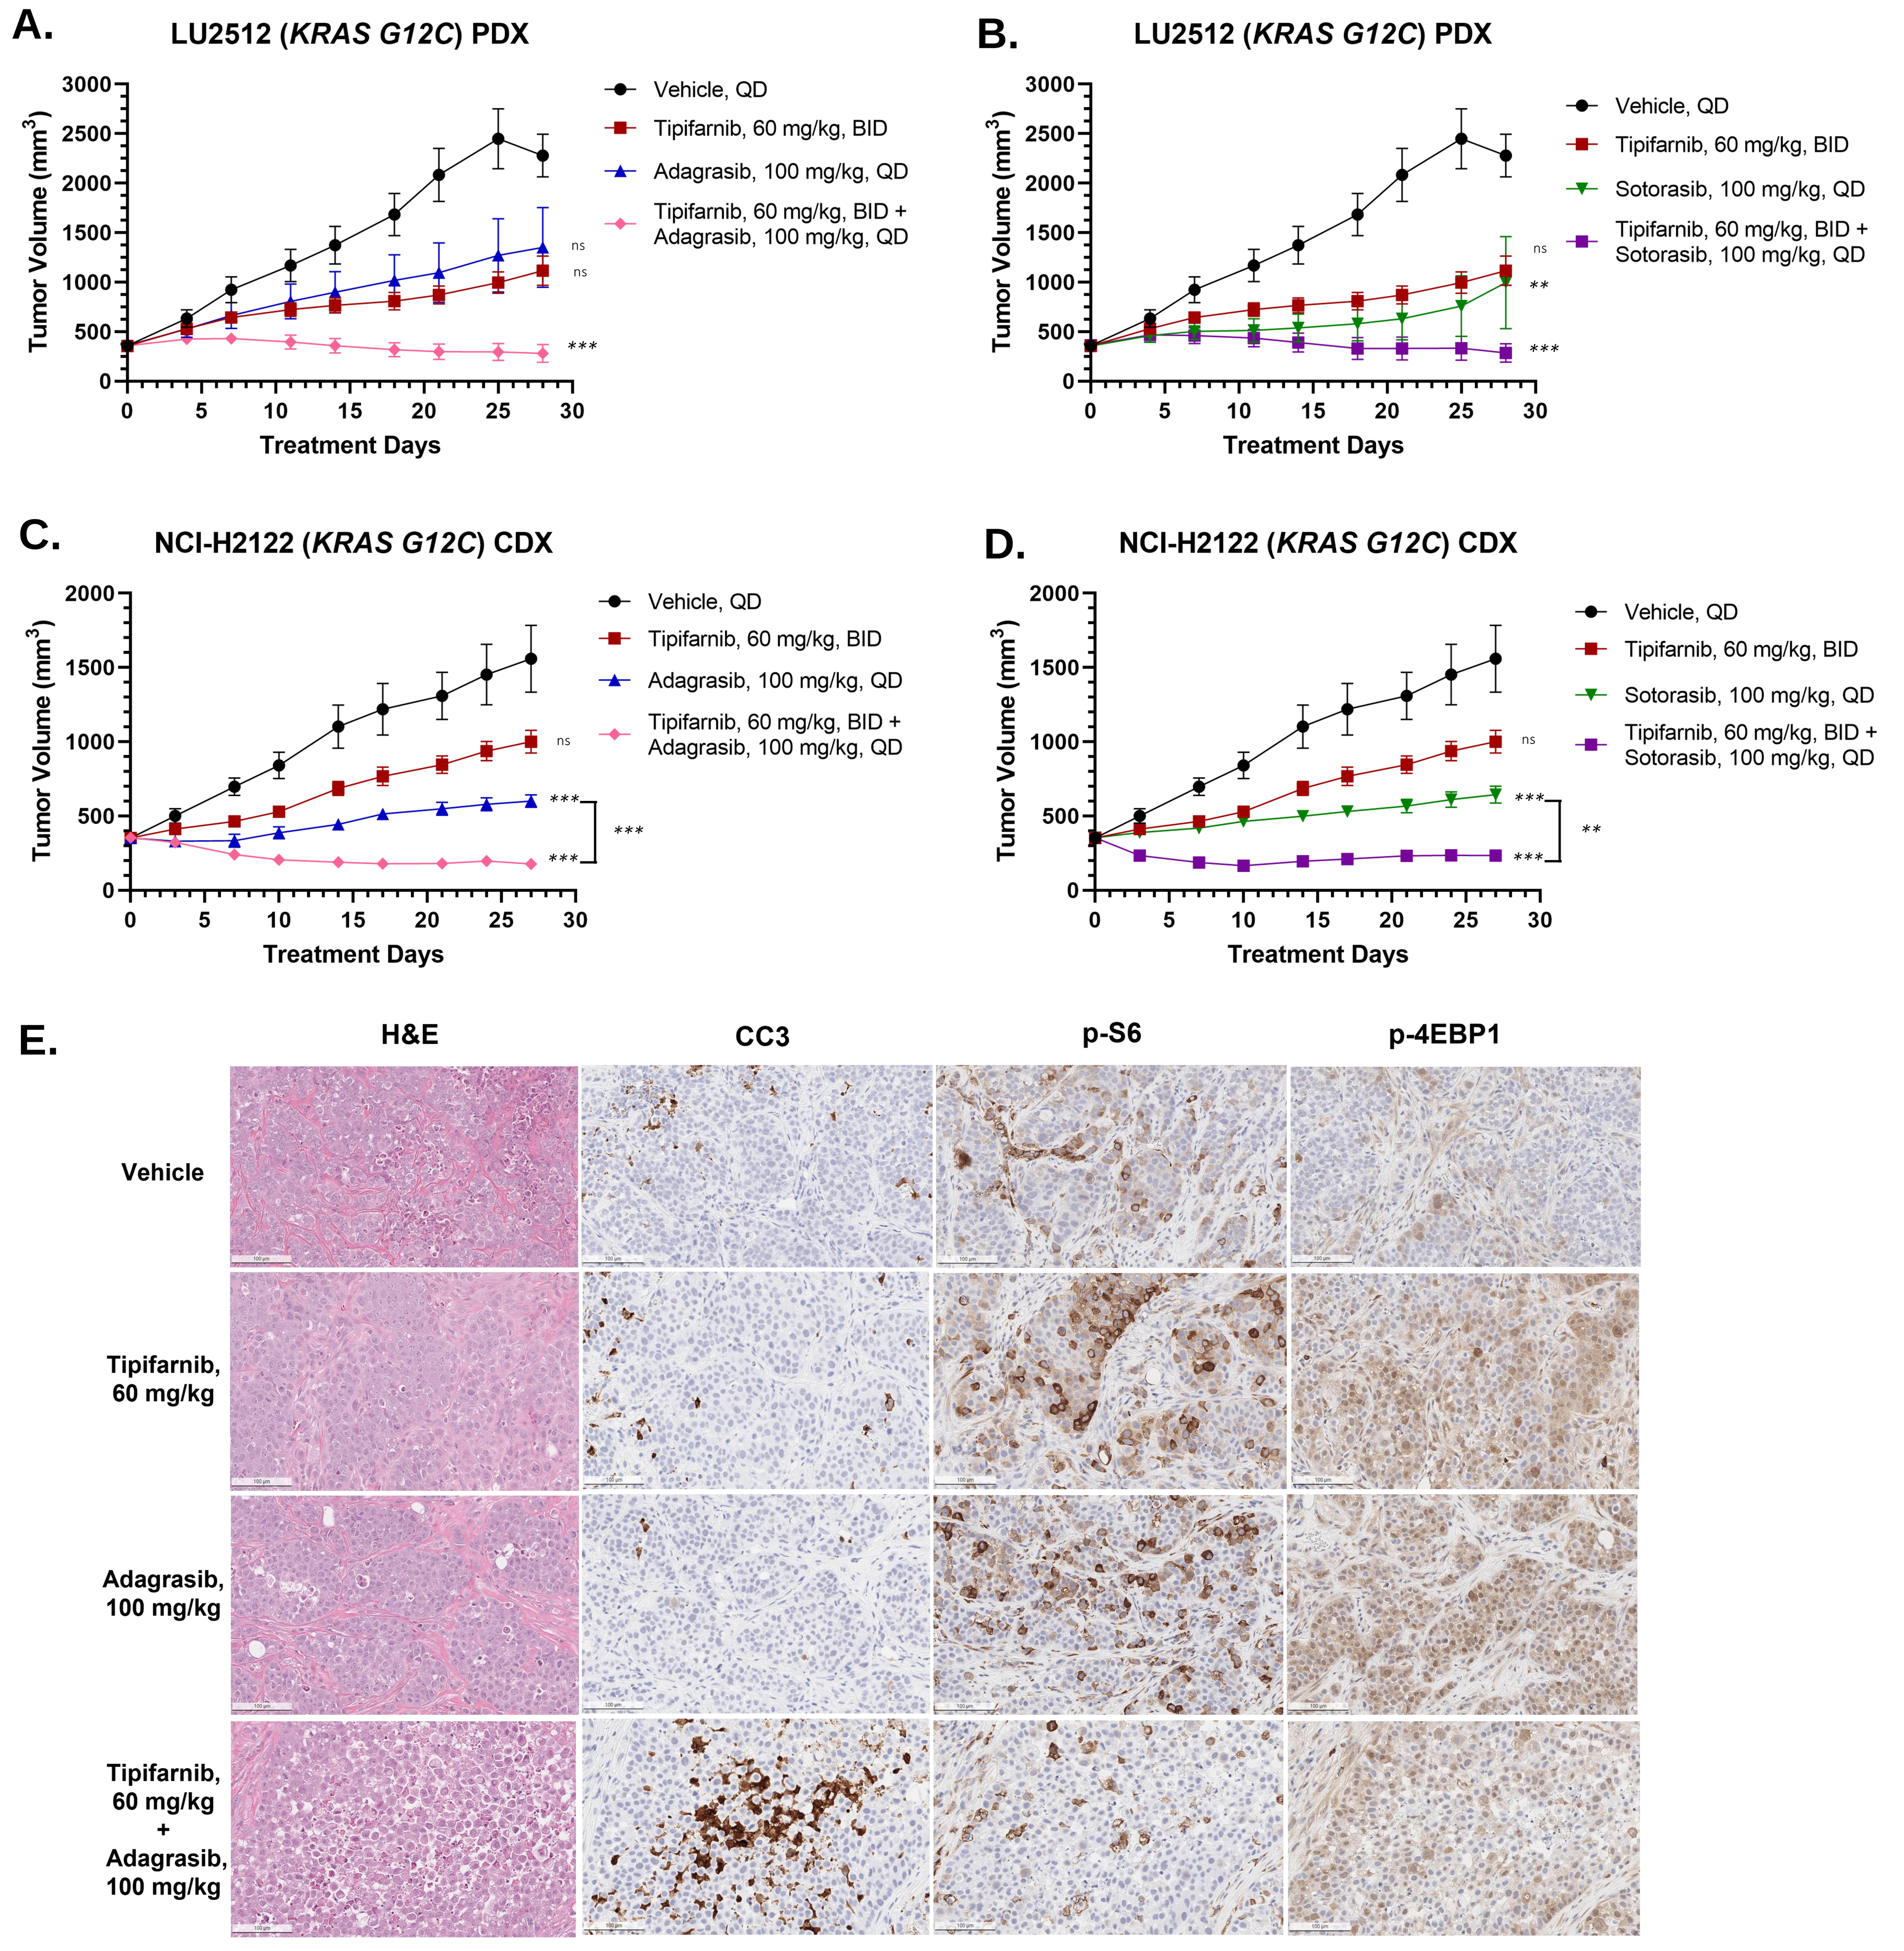
A. LU2512 (KRAS G12C) PDX
B. LU2512 (KRAS G12C) PDX
C. NCI-H2122 (KRAS G12C) CDX
D. NCI-H2122 (KRAS G12C) CDX
Vehicle, QD
Tipifarnib, 60 mg/kg, BID
Adagrasib, 100 mg/kg, QD
Sotorasib, 100 mg/kg, QD
Tipifarnib, 60 mg/kg, BID + Adagrasib, 100 mg/kg, QD
Tipifarnib, 60 mg/kg, BID + Sotorasib, 100 mg/kg, QD
Tumor Volume (mm³)
Treatment Days
E.
H&E
CC3
p-S6
p-4EBP1
Vehicle
Tipifarnib, 60 mg/kg
Adagrasib, 100 mg/kg
Tipifarnib, 60 mg/kg + Adagrasib, 100 mg/kg

Figure 2. Anti-tumor efficacy in *KRAS^{G12C}* NSCLC xenograft models with combination of tipifarnib with KRAS^{G12C} inhibitors. LU2512 *KRAS^{G12C}* NSCLC PDX model had significant tumor regressions with (A) tipifarnib, 60 mg/kg, BID + adagrasib, 100 mg/kg, QD and (B) tipifarnib, 60 mg/kg, BID + sotorasib, 100 mg/kg, QD compared to vehicle control. NCI-H2122 *KRAS^{G12C}* NSCLC CDX model had significant tumor regressions with (C) tipifarnib, 60 mg/kg, BID + adagrasib, 100 mg/kg, QD and (D) tipifarnib, 60 mg/kg, BID + sotorasib, 100 mg/kg, QD compared to vehicle control and single-agent treatment groups. (E) Immunohistochemistry was done for the NCI-H2122 *KRAS^{G12C}* NSCLC CDX tumors. Combination of tipifarnib with adagrasib caused an increase in the apoptotic marker cleaved caspase 3 (CC3) as well as decreases in the mTOR signaling pathway markers, p-S6 and p-4EBP1, compared to single-agent treatments.

p value between each arm vs. vehicle control was performed to determine statistical significance (ns = $p>0.05$; * = $p<0.05$; **= $p<0.01$; ***= $p<0.001$).

Combination of tipifarnib with a KRAS^{G12C} inhibitor suppresses mTOR signaling reactivation and promotes cell apoptosis

Figure 3. Combination of tipifarnib with sotorasib treatment suppresses mTOR signaling more potently and durably than single agent sotorasib. Immunoblots of MAPK/mTOR pathway proteins and apoptotic markers in NCI-H2122 *KRAS^{G12C}* NSCLC cells treated with sotorasib (AMG-510) for 0, 1, 6, 24, and 48 hours in the absence or presence of tipifarnib (72-hour treatment). Combination treatment results in stronger inhibition of mTOR activity (p70 S6K & S6 phosphorylation), induction of cell cycle arrest (Rb phosphorylation), and induction of cell death (cleaved caspase 3).

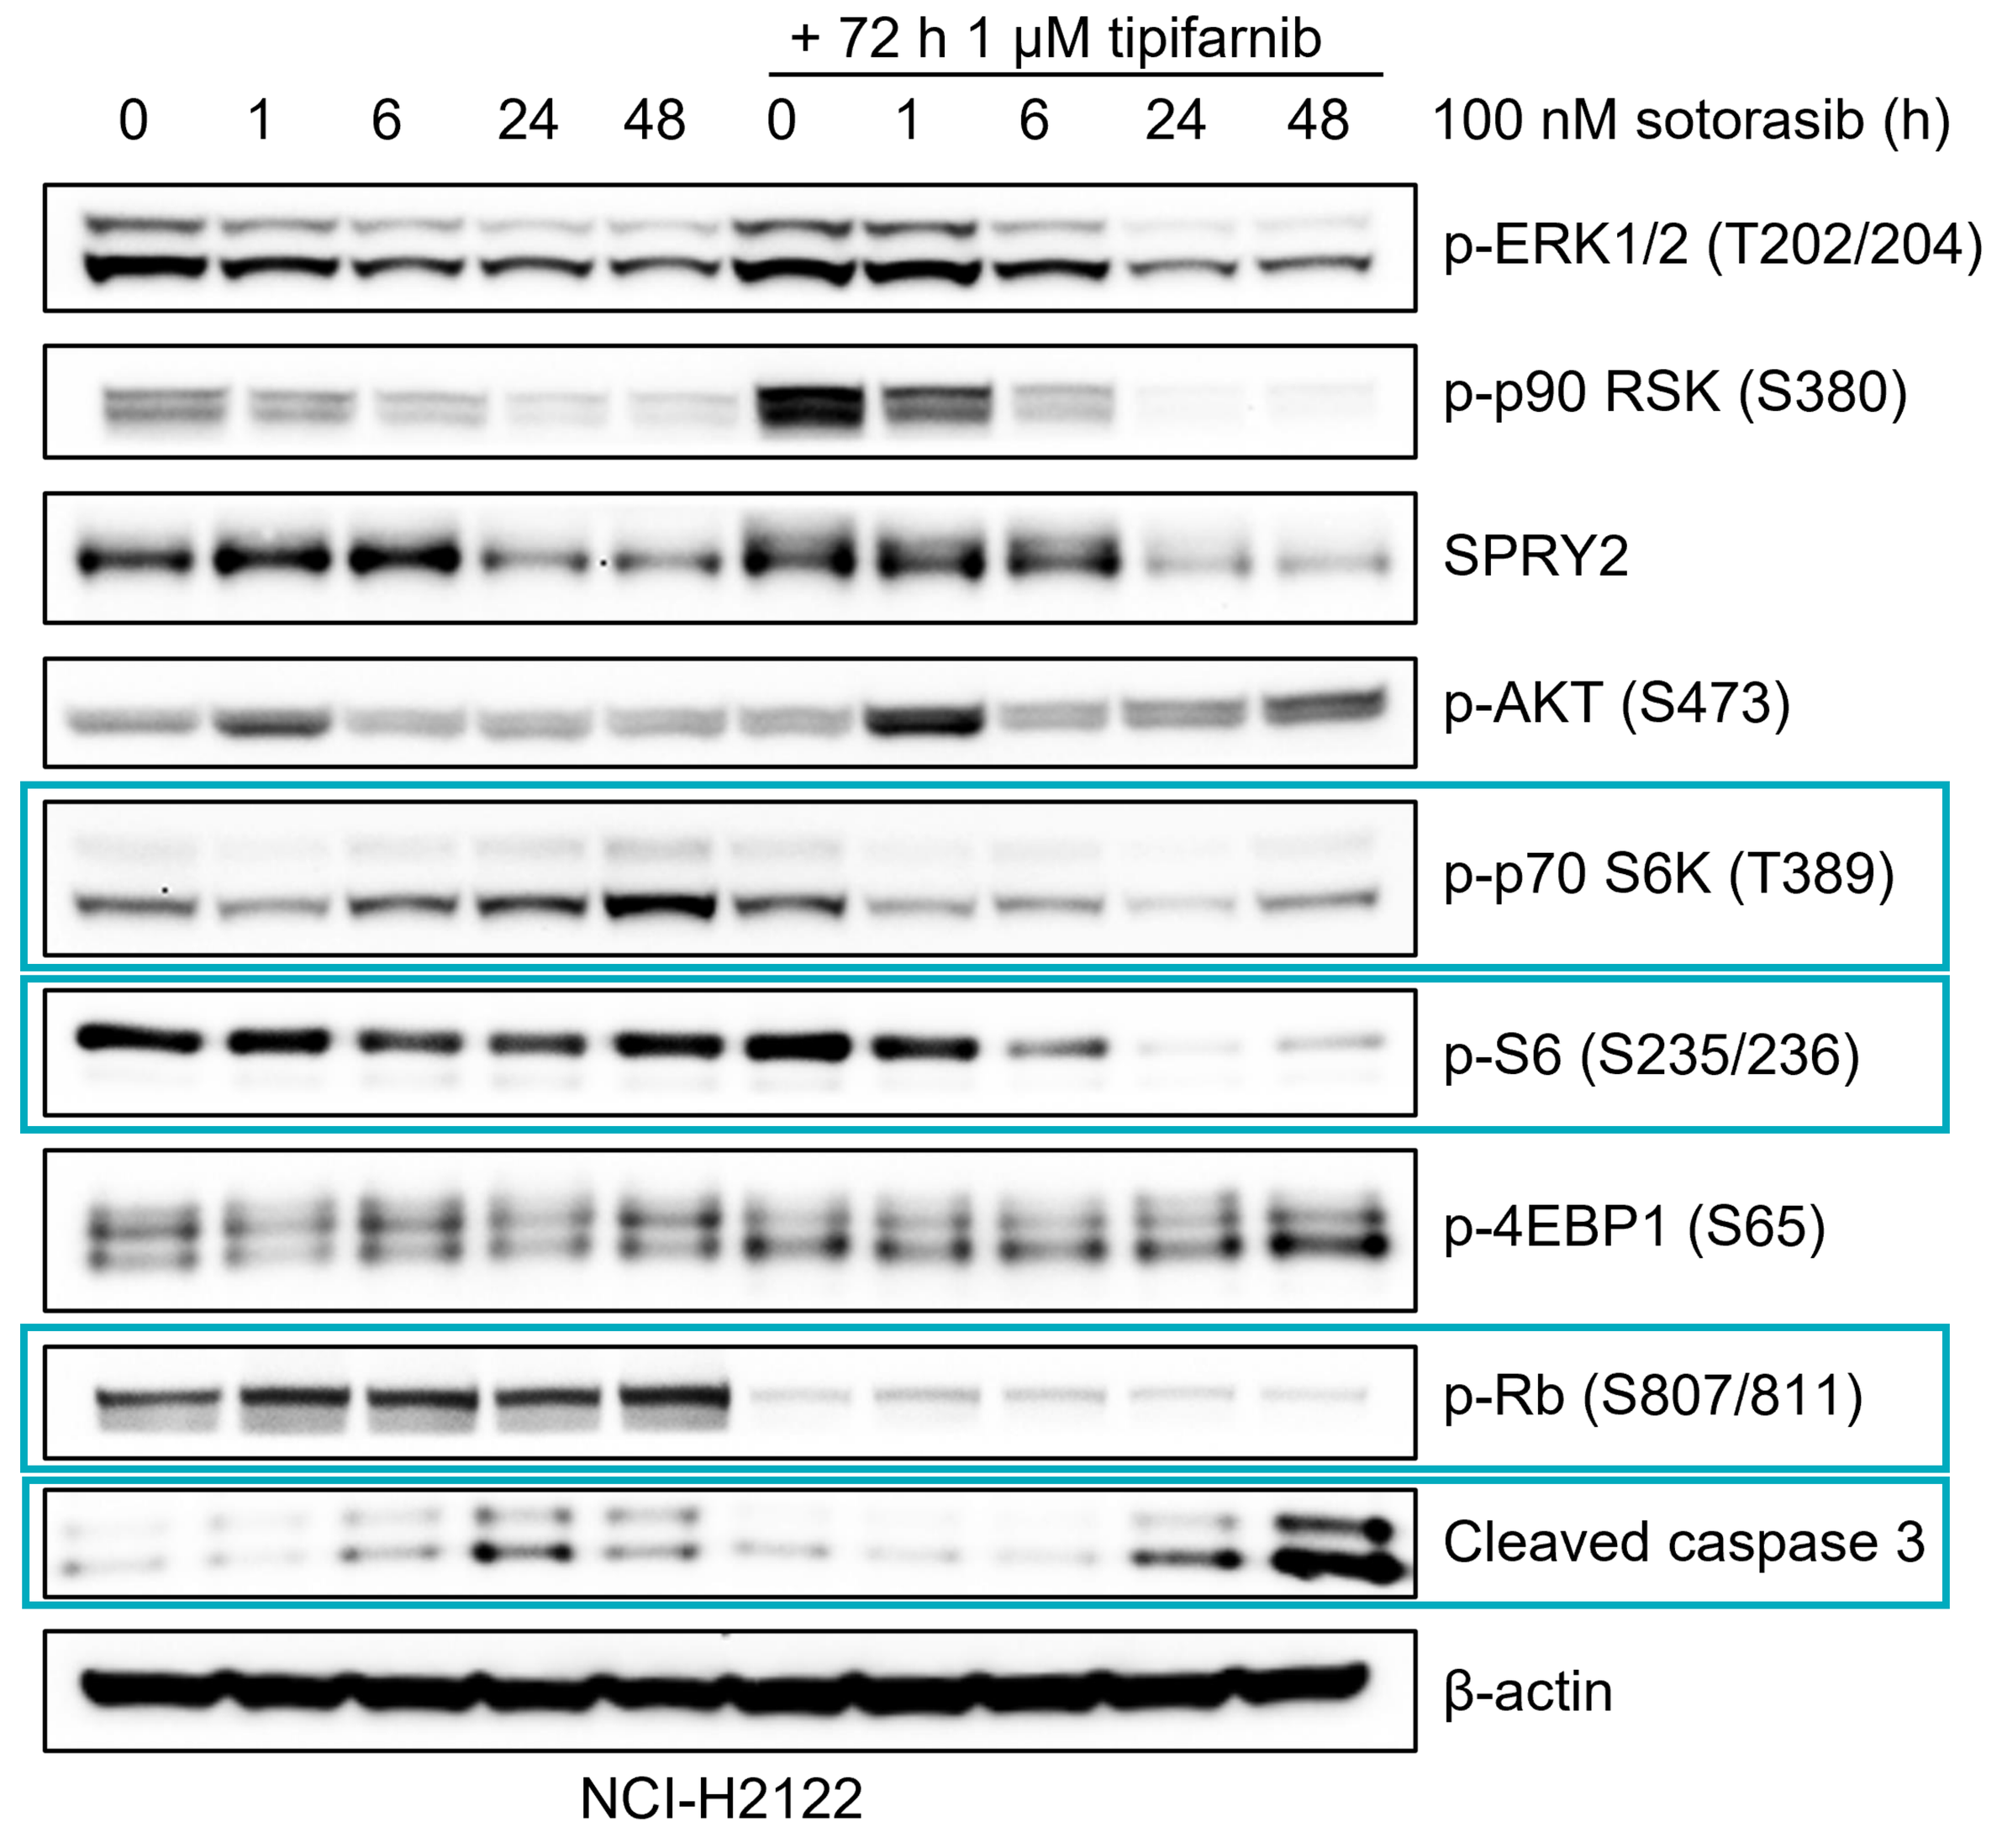
+ 72 h 1 µM tipifarnib
0 1 6 24 48 0 1 6 24 48
100 nM sotorasib (h)
p-ERK1/2 (T202/204)
p-p90 RSK (S380)
SPRY2
p-AKT (S473)
p-p70 S6K (T389)
p-S6 (S235/236)
p-4EBP1 (S65)
p-Rb (S807/811)
Cleaved caspase 3
β-actin
NCI-H2122

CONCLUSIONS

- **Combination of tipifarnib with KRAS^{G12C} inhibitors, such as adagrasib and sotorasib, leads to significant tumor regression in *KRAS^{G12C}* NSCLC CDX and PDX models.**
- **Tipifarnib suppresses the feedback reactivation of mTOR signaling at the level of p-S6 (S235/236) that occurs after single-agent KRAS^{G12C} inhibitor treatment.**
- **Further *in vivo* and *in vitro* mechanistic studies are ongoing to determine MOA of tipifarnib in the context of combination with KRAS^{G12C} inhibitors in *KRAS^{G12C}* NSCLC.**

REFERENCES

1. Palma, G. *et al.* Selective KRAS G12C inhibitors in non-small cell lung cancer: chemistry, concurrent pathway alterations, and clinical outcomes. *npj Precis. Onc.* **5**, 98 (2021).
2. Hallin, J. *et al.* The KRASG12C Inhibitor MRTX849 Provides Insight toward Therapeutic Susceptibility of Kras-Mutant Cancers in Mouse Models and Patients. *Cancer Discov* **10**, 54–71 (2020).
3. Canon, J. *et al.* The clinical KRAS(G12C) inhibitor AMG 510 drives anti-tumour immunity. *Nature* **575**, 217–223 (2019).
4. Patricelli, M. P. *et al.* Selective Inhibition of Oncogenic KRAS Output with Small Molecules Targeting the Inactive State. *Cancer Discov* **6**, 316–329 (2016).
5. Ryan, M. B. *et al.* Vertical Pathway Inhibition Overcomes Adaptive Feedback Resistance to KRASG12C Inhibition. *Clin Cancer Res* **26**, 1633–1643 (2020).
6. Smith, A. E. *et al.* Tipifarnib Potentiates the Antitumor Effects of PI3Kα Inhibition in PIK3CA- and HRAS-Dysregulated HNSCC via Convergent Inhibition of MTOR Activity. *BioRxiv* (2023).