Abstract # 34634

KO-2806, a next-generation farnesyltransferase inhibitor, potentiates the antitumor activity of cabozantinib in clear cell renal cell carcinoma models

Jovylyn Gatchalian, Linda Kessler, Hetika Vora Patel, Stacia Chan, Francis Burrows, and Shivani Malik

Kura Oncology, Inc., San Diego, CA

BACKGROUND

- Conventional treatments for advanced clear cell renal cell carcinoma (ccRCC) include tyrosine kinase inhibitors (TKI) that target angiogenesis, a critical element for ccRCC tumor growth and survival.[1]
- Cabozantinib, a TKI that inhibits VEGFR, AXL, and MET, is an approved therapy in advanced RCC. However, cabozantinib has limited therapeutic benefit; for example, patients with prior VEGFR TKI therapy had an overall response rate of 21% and median progression-free survival of 7.4 months.[2]
- There is an urgent need to identify combination drug partners that can improve the depth and/or durability of clinical responses to cabozantinib.
- KO-2806 is a next generation farnesyltransferase inhibitor (FTI) that potentially blocks hyperactivated growth factor signaling at multiple nodes.[3,4] KO-2806 has increased potency and improved pharmacokinetic properties relative to earlier FTI drug candidates and is poised to enter a first-in-human clinical trial.
- **Here, we utilize cell line and patient-derived xenograft models to explore the scope and mechanistic underpinnings of KO-2806-cabozantinib effects and strengthen the scientific rationale for combining KO-2806 with cabozantinib in the treatment of patients with ccRCC.**

RESULTS

KO-2806 potentiates the antitumor activity of cabozantinib in ccRCC models

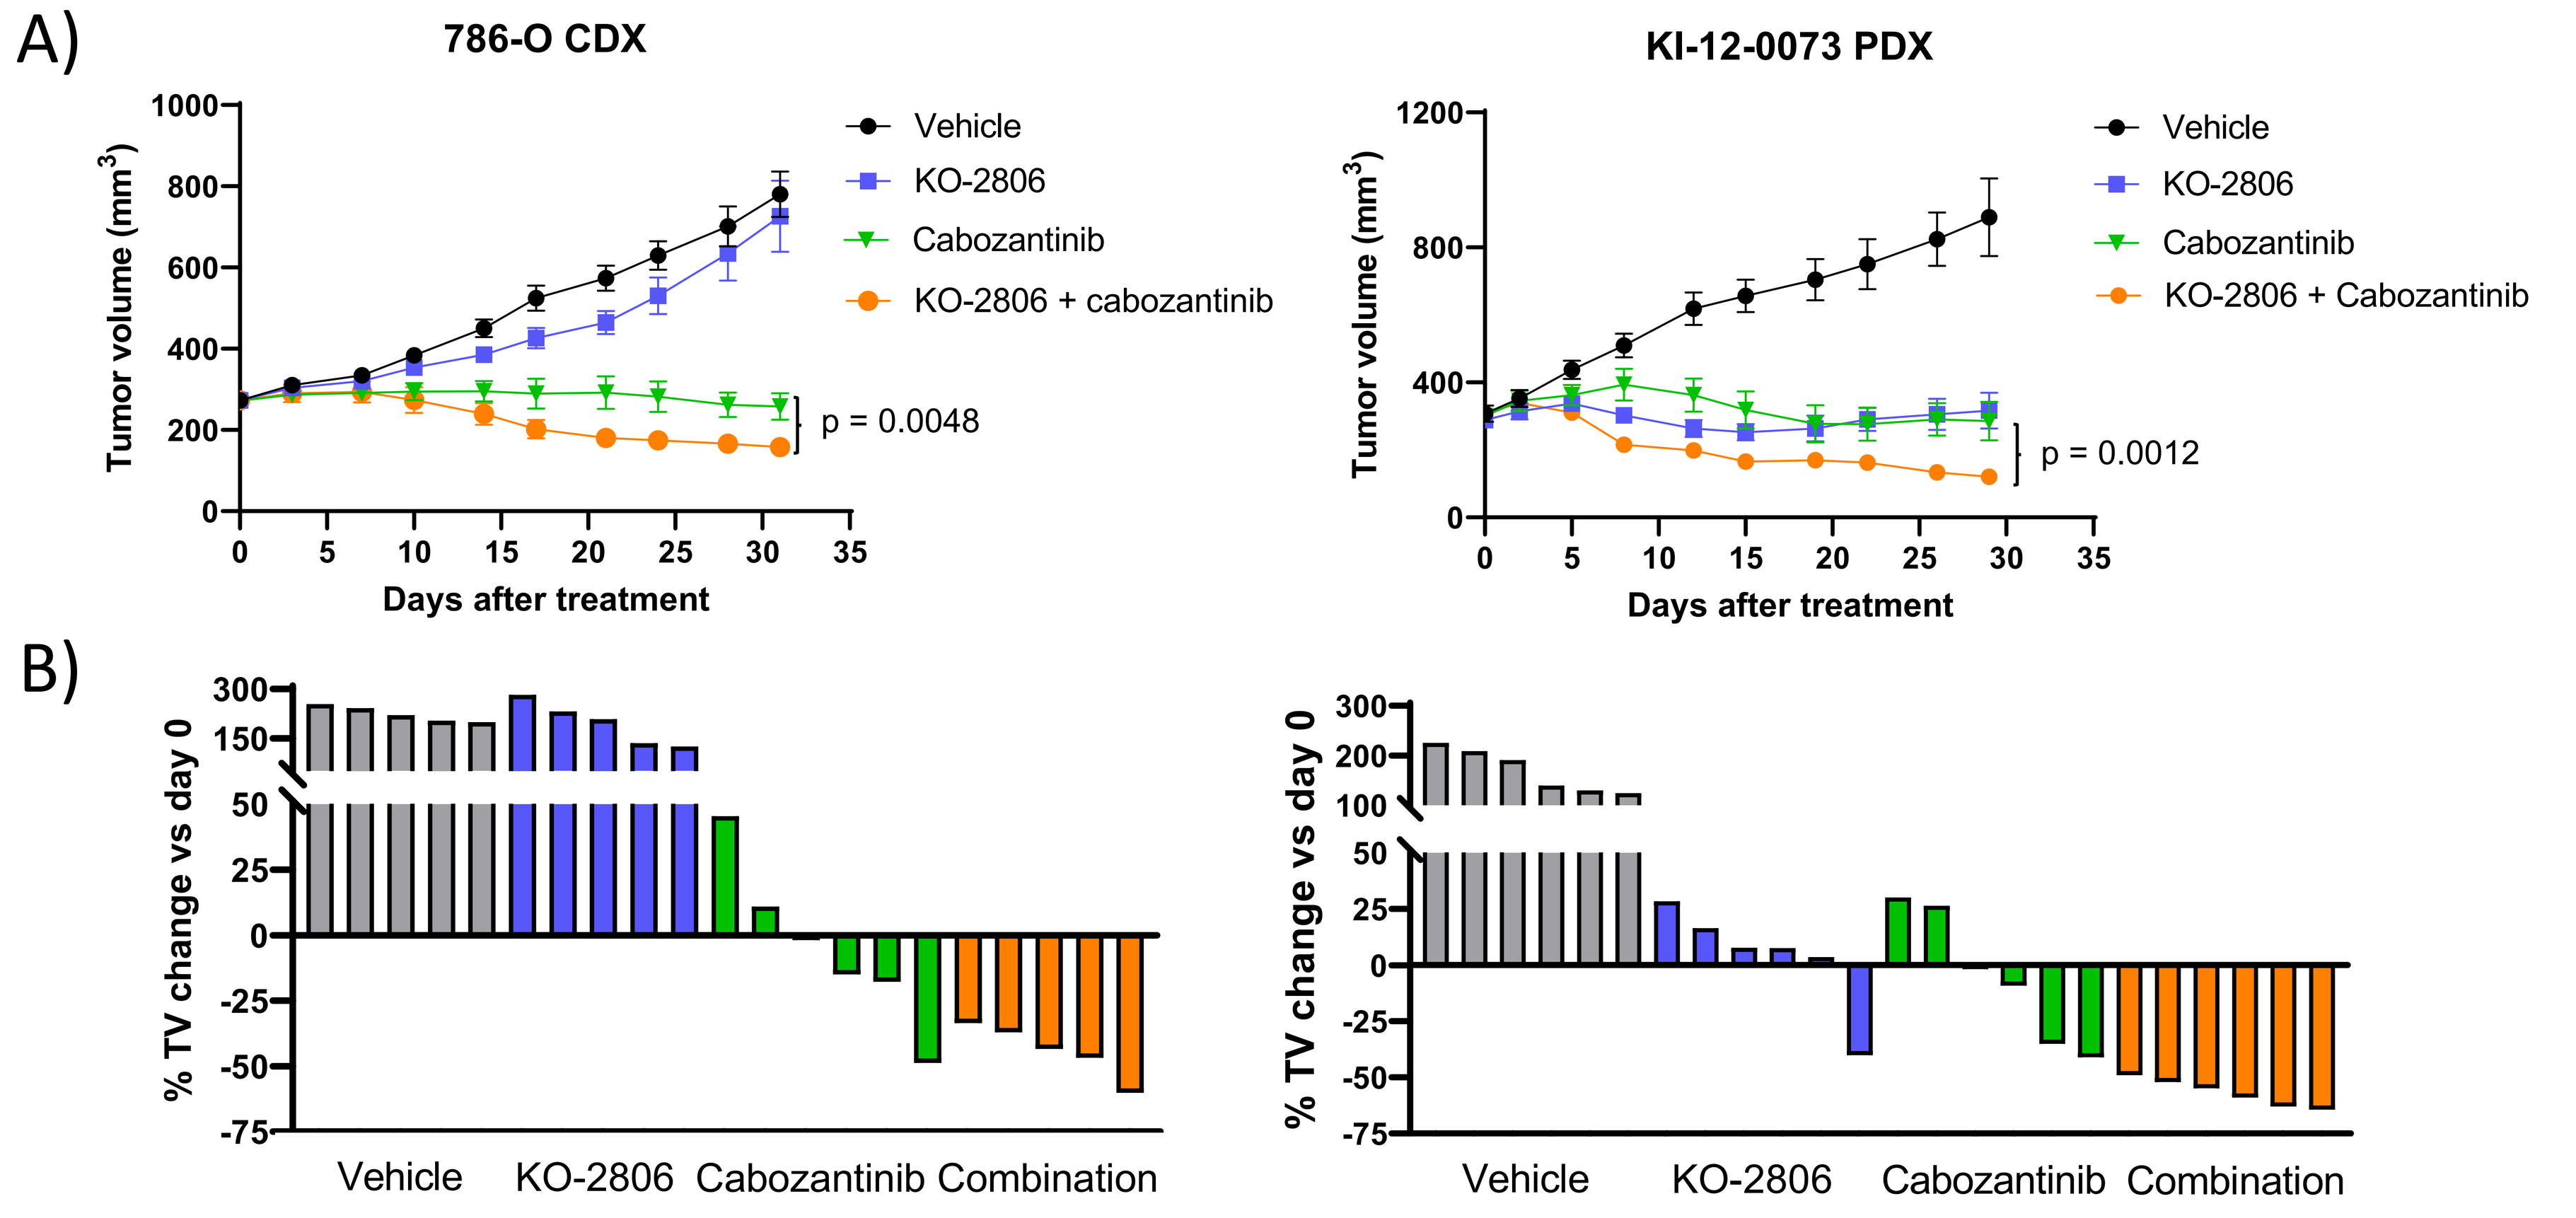
A)
786-O CDX
Tumor volume (mm^3)
Days after treatment
Vehicle
KO-2806
Cabozantinib
KO-2806 + cabozantinib
p = 0.0048
KI-12-0073 PDX
KO-2806 + Cabozantinib
p = 0.0012
B)
% TV change vs day 0
Vehicle
KO-2806
Cabozantinib
Combination

Figure 1. Continuous treatment of KO-2806 plus the anti-angiogenic TKI cabozantinib robustly inhibits the growth of ccRCC cell line and patient derived xenograft models. A) Tumor growth curves of 786-O CDX and KI-12-0073 PDX models, treated with vehicle, KO-2806, cabozantinib (15 or 8 mg/kg QD), or the combination. B) Bar graphs of percent tumor change at end point relative to day 0, for individual 786-O (left) and KI-12-0073 (right) tumors with indicated treatments. 786-O end point is day 32, KI-12-0073 end point is day 28.

KO-2806 plus cabozantinib combination leads to decreased vascular density in 786-O tumors

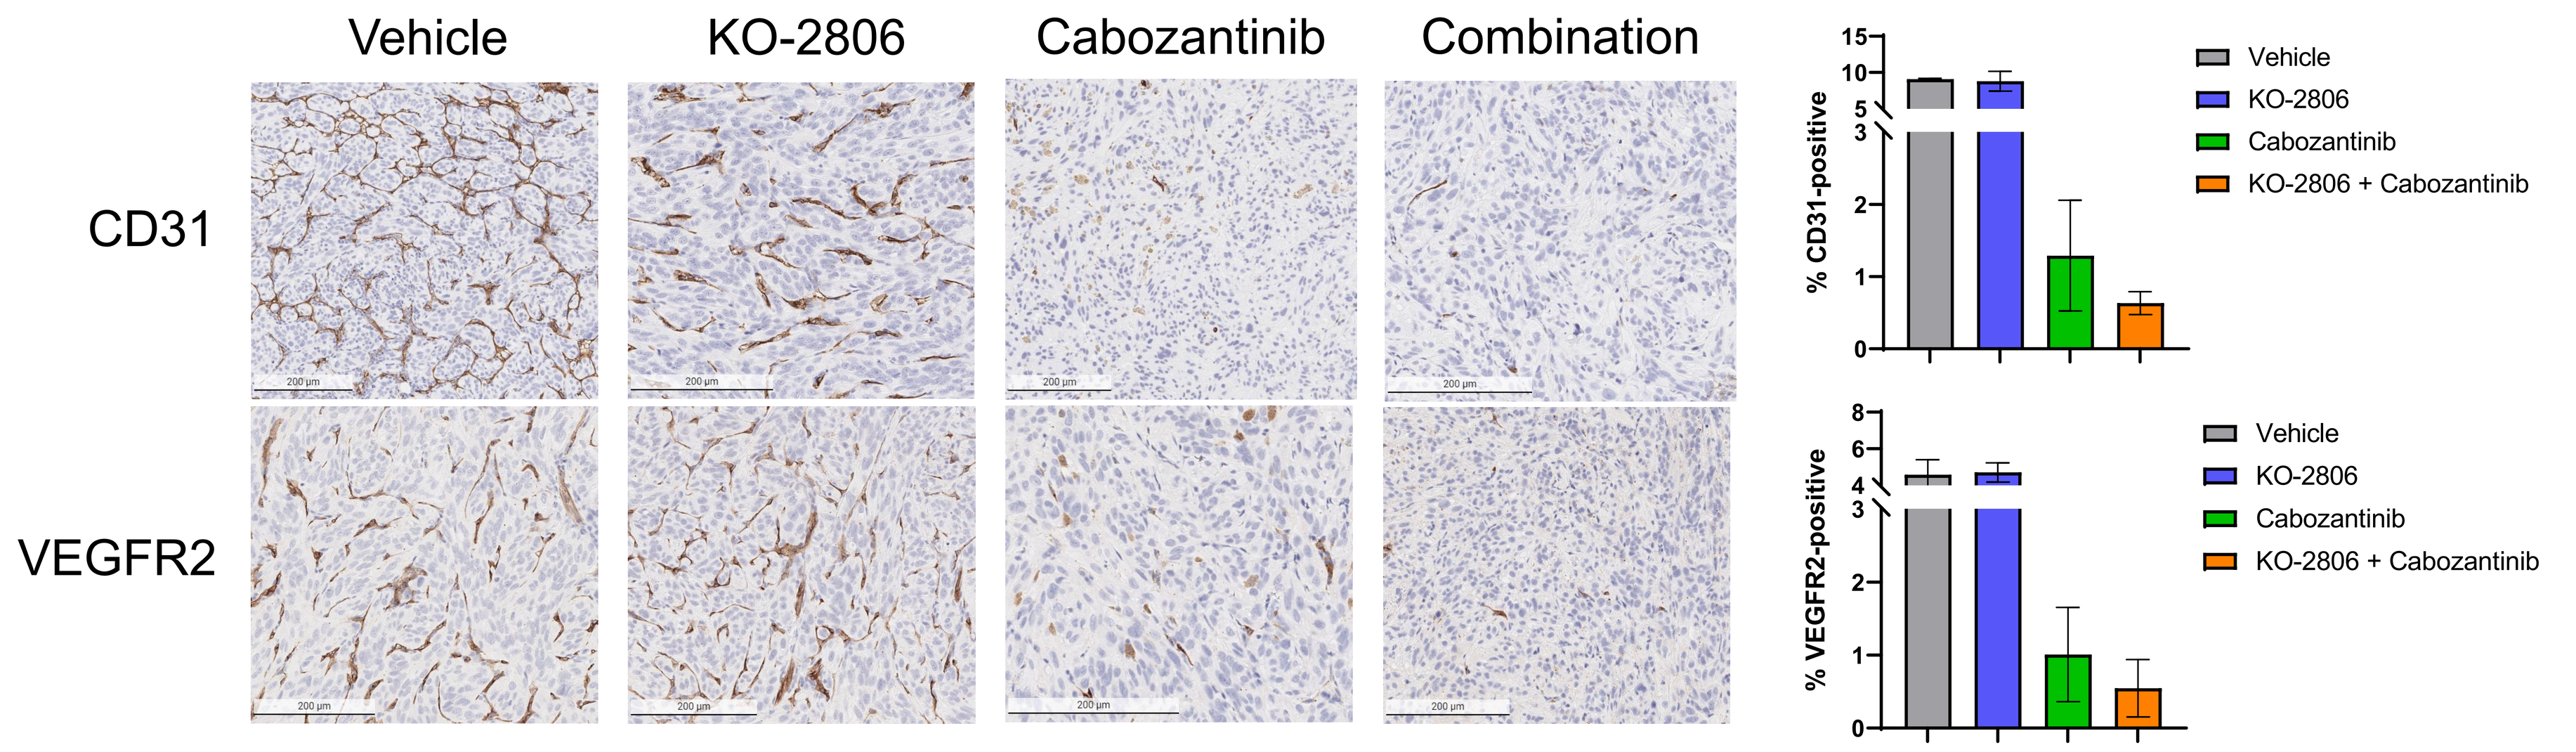
Vehicle
KO-2806
Cabozantinib
Combination
CD31
VEGFR2
% CD31-positive
% VEGFR2-positive
KO-2806 + Cabozantinib

Figure 2. KO-2806 plus cabozantinib combination leads to decreased expression of endothelial cell markers *in vivo*. Representative images of immunohistochemistry analysis for vascular markers CD31 and VEGFR2 in 786-O tumors collected after 14 days of treatment with vehicle, KO-2806, cabozantinib (15 mg/kg QD), or the combination. Right, quantitation of indicated target expression is expressed by mean percent of area with positive stain over the tumor area. Error bars represent standard deviation of the mean, n = 4.

KO-2806 and cabozantinib reduce endothelial cell viability and induce apoptosis *in vitro*

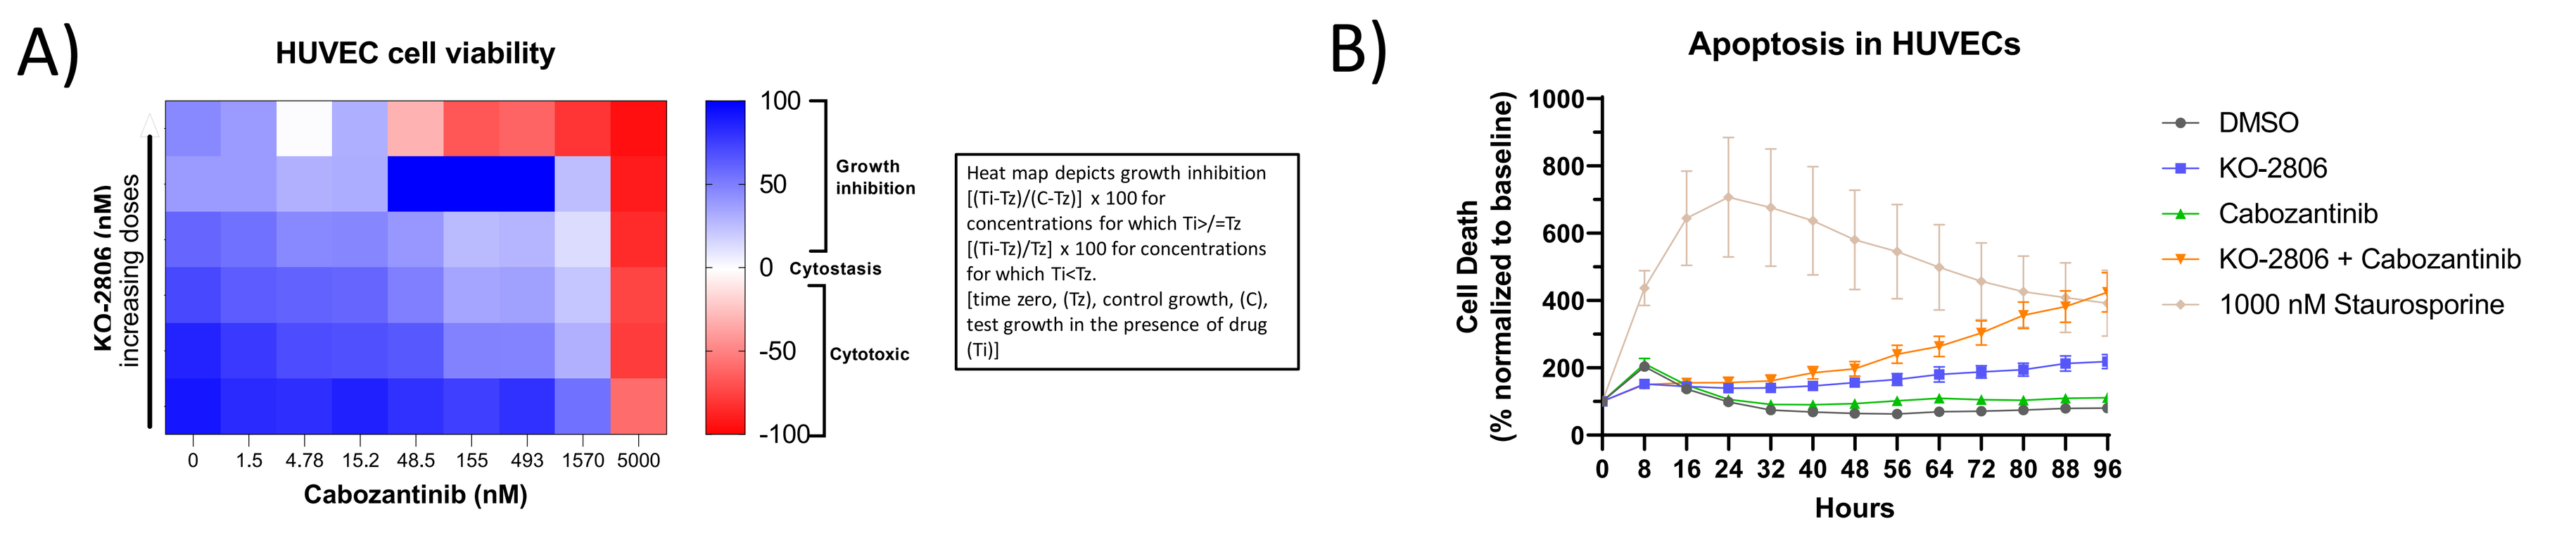
A)
HUVEC cell viability
KO-2806 (nM) increasing doses
Cabozantinib (nM)
0 1.5 4.78 15.2 48.5 155 493 1570 5000
Growth inhibition
Cytostasis
Cytotoxic
Heat map depicts growth inhibition [(Ti-Tz)/(C-Tz)] x 100 for concentrations for which Ti>/=Tz [(Ti-Tz)/Tz] x 100 for concentrations for which Ti<Tz. [time zero, (Tz), control growth, (C), test growth in the presence of drug (Ti)]
B)
Apoptosis in HUVECs
Cell Death (% normalized to baseline)
Hours
DMSO
KO-2806
Cabozantinib
KO-2806 + Cabozantinib
1000 nM Staurosporine

Figure 3. KO-2806 and cabozantinib inhibit human umbilical vein endothelial cell (HUVEC) growth and induce apoptosis *in vitro*. A) Heatmap indicates degree of growth inhibition or cytotoxicity induced by KO-2806, cabozantinib, or the combination in HUVECs after 7 days of treatment. B) Apoptosis in HUVECs, measured by Annexin V signal plotted against time of treatment with KO-2806, 100 nM cabozantinib, or the combination. Staurosporine is a positive control.

KO-2806 does not affect endothelial cell tube formation *in vitro*

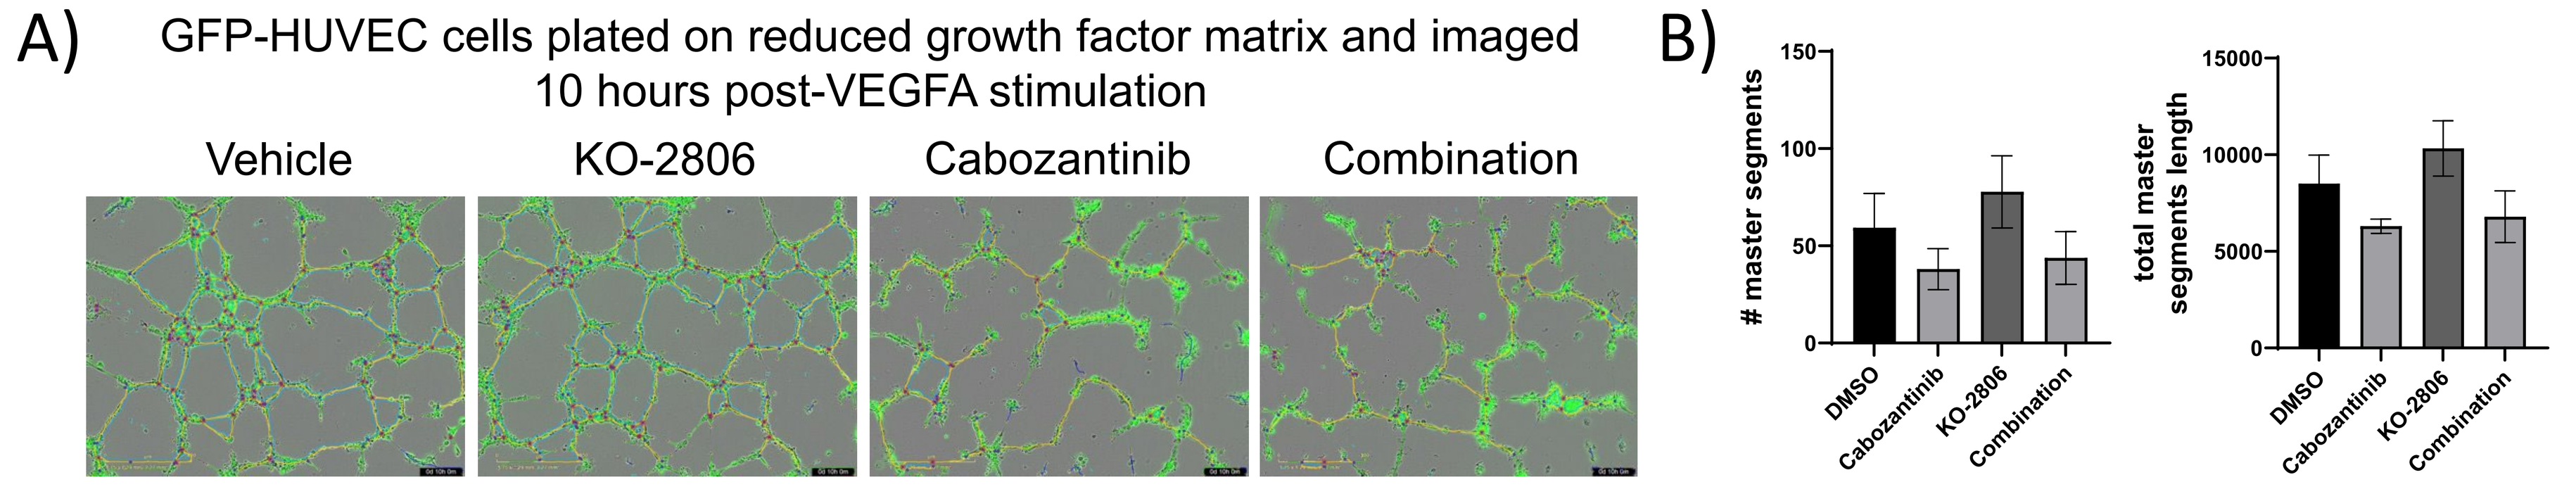
A)
GFP-HUVEC cells plated on reduced growth factor matrix and imaged 10 hours post-VEGFA stimulation
Vehicle
KO-2806
Cabozantinib
Combination
B)
master segments
total master segments length
DMSO
Cabozantinib
KO-2806
Combination

Figure 4. Cabozantinib inhibits HUVEC tube formation *in vitro*. A) Representative images of GFP-HUVEC tubes formed *in vitro* with the indicated treatments. B) Quantitation of tube parameters was performed using ImageJ Angiogenesis Analyzer. Bars represent the mean ± SEM, n = 4.

KO-2806 plus cabozantinib blunts AKT and mTOR signaling and induces cell cycle arrest in 786-O tumors

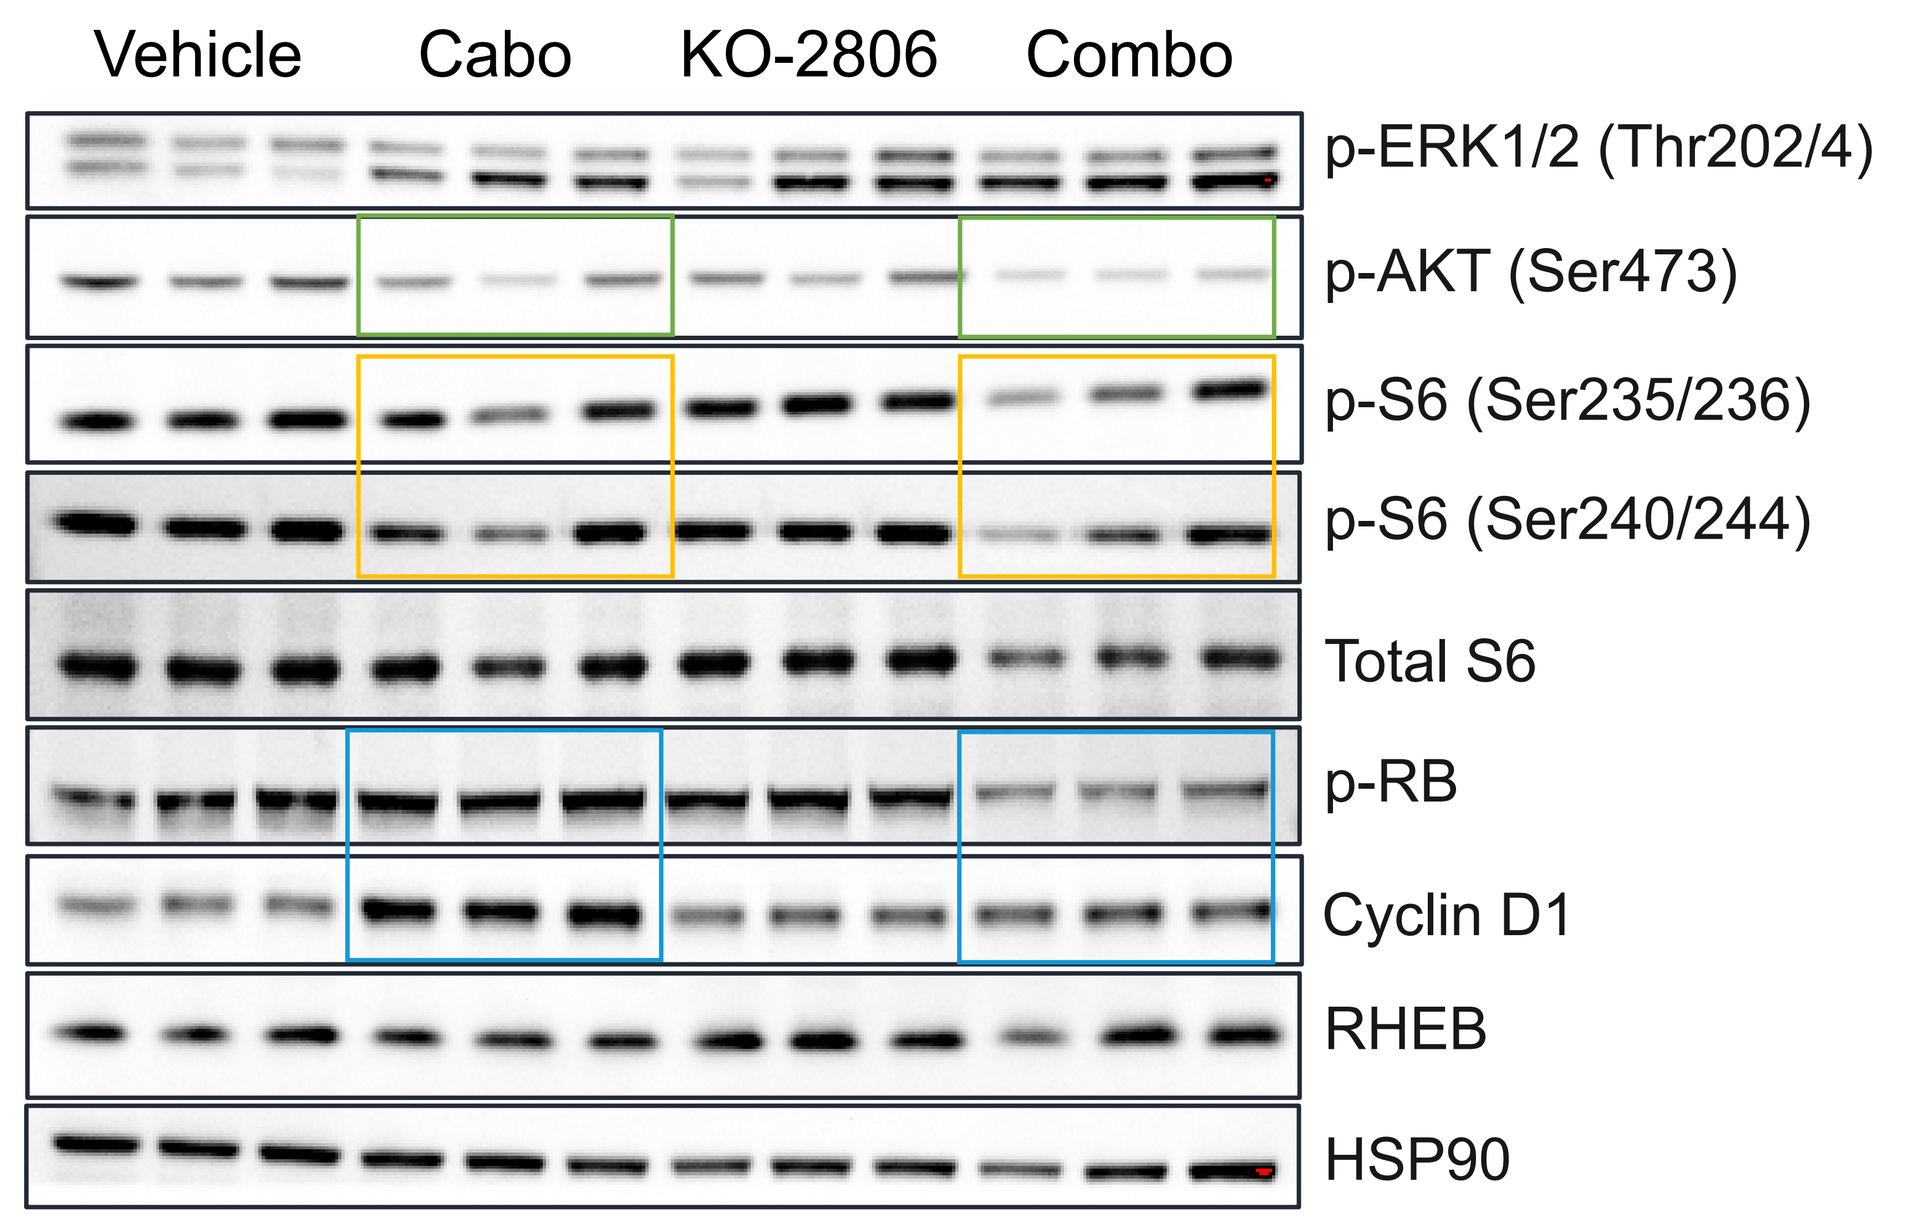
Vehicle
Cabo
KO-2806
Combo
p-ERK1/2 (Thr202/4)
p-AKT (Ser473)
p-S6 (Ser235/236)
p-S6 (Ser240/244)
Total S6
p-RB
Cyclin D1
RHEB
HSP90

Figure 5. Combined KO-2806 and cabozantinib treatment of 786-O tumors inhibits AKT and mTOR signaling more potently than cabozantinib alone. Immunoblots of indicated MAPK/PI3K pathway components and cell cycle arrest markers in 786-O tumors collected after 14 days of treatment with KO-2806, cabozantinib (15 mg/kg QD), or the combination. HSP90 serves as the loading control.

786-O xenografts that progress on axitinib regress after treatment with KO-2806 and cabozantinib

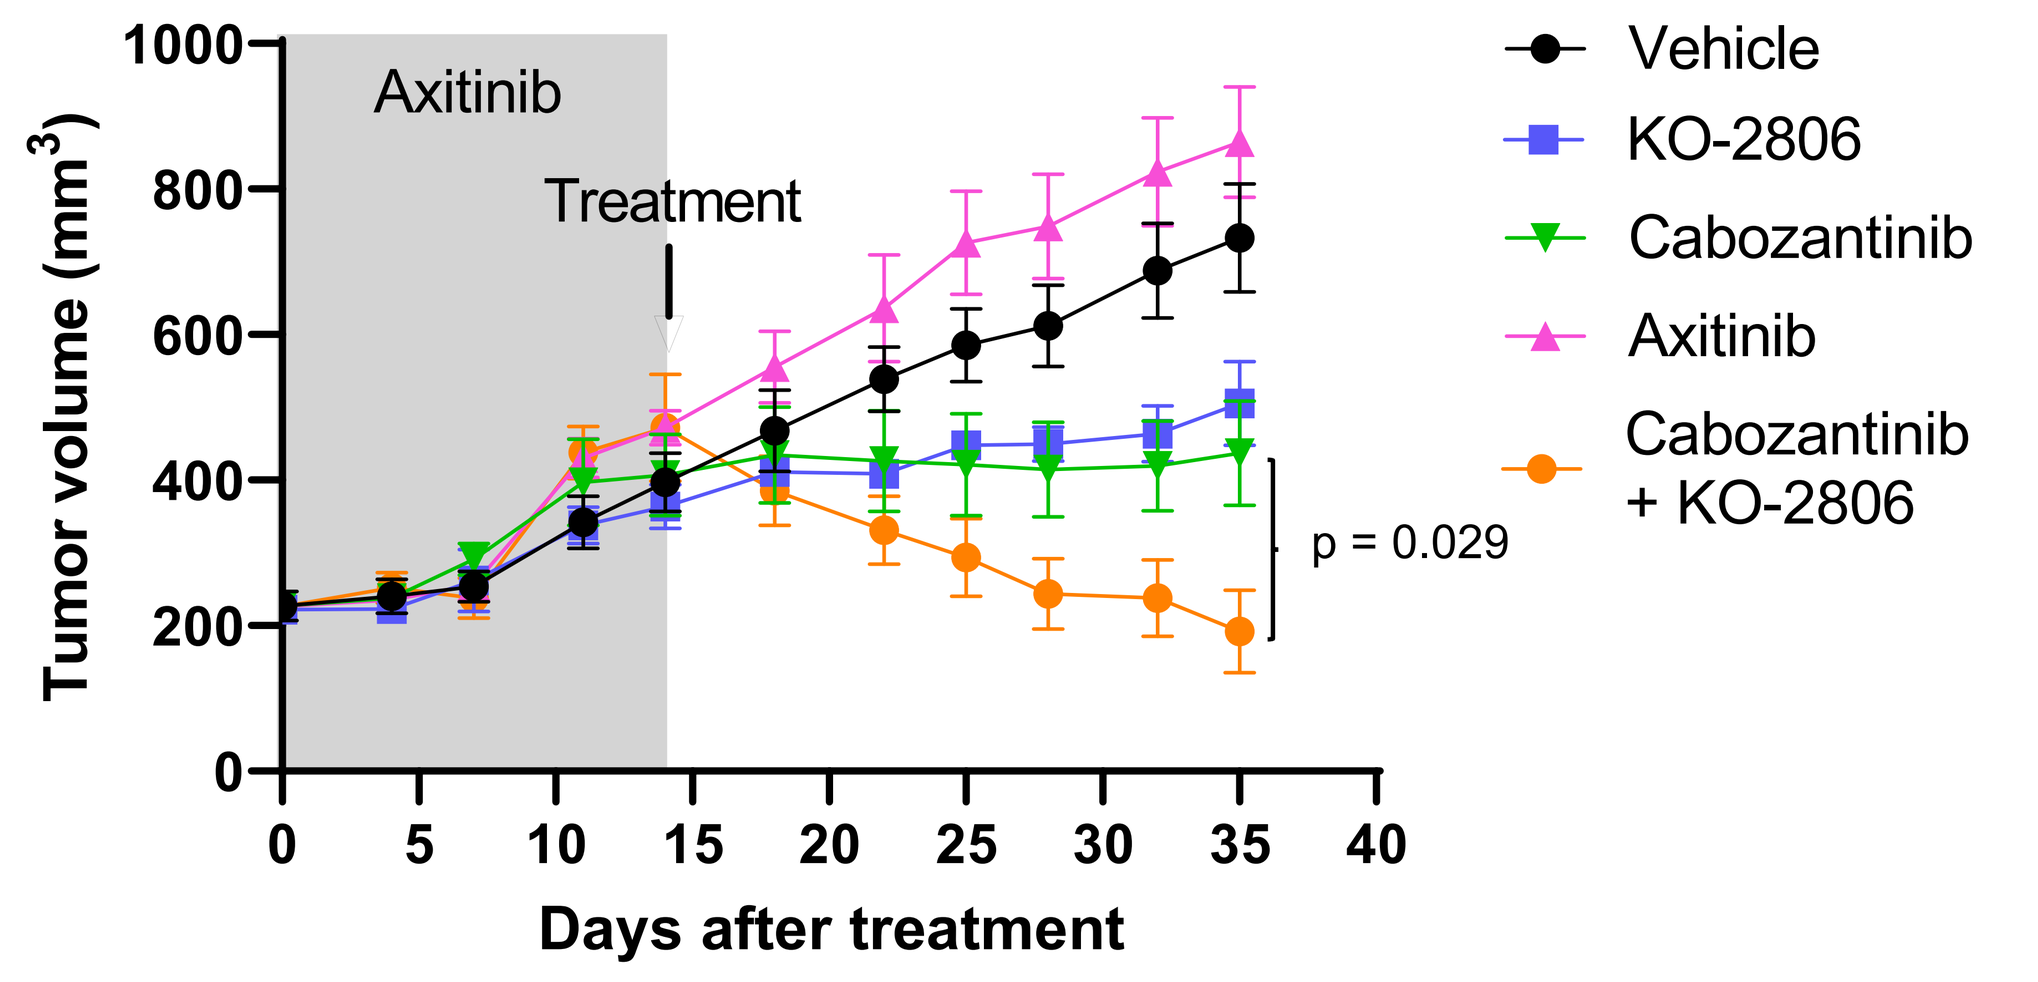
Axitinib
Treatment
Tumor volume (mm^3)
Days after treatment
p = 0.029
Vehicle
KO-2806
Cabozantinib
Axitinib
Cabozantinib + KO-2806

Figure 6. Xenografts that progress on axitinib regress with KO-2806 and cabozantinib combination treatment. Tumor growth curves during the study period. 786-O xenografts were pre-treated with axitinib (36 mg/kg QD) for 14 days prior to switching to treatment with KO-2806, cabozantinib (15 mg/kg QD), or the combination, or axitinib (36 mg/kg QD). Unpaired t-test was performed at Day 35.

CONCLUSIONS

- KO-2806 plus cabozantinib induces tumor regression in the ccRCC models, 786-O and KI-12-0073. In addition, 786-O tumors that advance on an axitinib regimen also regress after treatment with KO-2806 and cabozantinib.
- KO-2806 enhances the anti-angiogenic activity of cabozantinib *in vivo,* as observed by decreased expression of vascular markers in 786-O tumors. In support of this, apoptosis and cell viability experiments in primary human endothelial cells showed that the combination of KO-2806 and cabozantinib was more effective in reducing cell viability and inducing apoptosis than either monotherapy.
- Analysis of the tumor growth signaling pathways shows that the addition of KO-2806 to cabozantinib led to enhanced inhibition of AKT and mTOR signaling and increased cell cycle arrest, compared to either agent alone.
- The combination of KO-2806 and cabozantinib holds potential for the treatment of ccRCC. Ongoing studies aim to further define the mechanistic underpinnings of the synergistic therapeutic activity.

References
1) Huang JJ and Hsieh JJ. "The therapeutic landscape of renal cell carcinoma: from the dark age to the golden age." Semin Nephrol (2020) 40(1):28-41..
2) Choueiri TK, et al. "Cabozantinib versus everolimus in advanced renal cell carcinoma." N Engl J Med (2015) 373(19):1814-23.
3) Gilardi M, et al. "Tipifarnib as a precision therapy for HRAS-mutant head and neck squamous cell carcinomas." Mol Cancer Ther (2020) 19(9):1784-96.
4) Smith AE, et al. "Tipifarnib potentiates the antitumor effects of PI3Kα inhibition in *PIK3CA*- and *HRAS*-dysregulated HNSCC via convergent inhibition of mTOR activity." Cancer Res (2023) doi: 10.1158/0008-5472.CAN-23-0282.