A Novel Small Molecule Menin-MLL Inhibitor for Potential Treatment of MLL-rearranged Leukemias

Tao Wu[1], Linda Kessler[1], Shuangwei Li[1], Trupta Purohit[2], Shisheng Li[1], Hongzhi Miao[2], Brian Linhares[2], Rasmus Hansen[1], Jeff Kucharski[1], Yi Wang[1], Ke Yu[1], Katarzyna Kempinska[2], Tess Ely[1], Szymon Klossowski[2], Ata Zarieh[1], Ulf Peters[1], Jun Feng[1], Yvonne Yao[1], Yuan Liu[1], Bo Wen[2], Francis Burrows[3], Duxin Sun[2], Jingchuan Zhang[1], Levan Darjania[1], Dana Hu-Lowe[1], Patrick Zarrinkar[1], Liansheng Li[1], Tomasz Cierpicki[2], Jolanta Grembecka[2], Pingda Ren[3], Yi Liu[3]*. [1]Wellspring Biosciences, La Jolla, CA; [2]University of Michigan, Ann Arbor, MI; [3]Kura Oncology, La Jolla, CA * Corresponding author: yi@kuraoncology.com

Introduction

Patients with MLL-rearranged leukemia typically have a poor prognosis. With chemotherapy and stem cell transplantation as current standard of care, the 5-year survival rate is estimated to be 40%. As the leukemogenic activity of MLL fusion proteins has been shown to be dependent on their direct interaction with menin, development of small molecules that block the menin-MLL interaction is a promising therapeutic strategy for the treatment of this disease.

We describe KO-539, a menin-MLL inhibitor with optimized drug-like properties that demonstrates potential clinical utility in preclinical models of MLL leukemias. The compound is currently under further preclinical evaluation.

Background

- MLL-r leukemias are an aggressive type of blood cancer, predominantly in children and therapy-related leukemia in adults
- Chromosomal translocations in the MLL gene at 11q23 result in MLL fusions with any of more than 50 partner genes
- MLL fusion proteins drive leukemogenesis through deregulation of *HOX* and *MEIS* genes
- The leukemogenic activity of MLL fusion proteins is critically dependent on menin binding

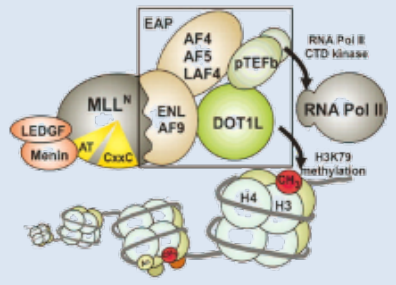
EAP
AF4
AF5
LAF4
pTEFb
RNA Pol II CTD kinase
MLL^N
ENL
AF9
DOT1L
RNA Pol II
LEDGF
Menin
AT
CxxC
H3K79 methylation
H4
H3

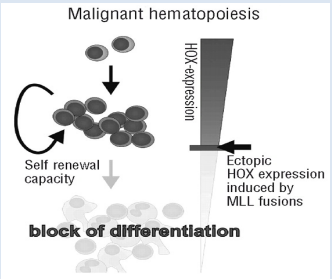
Malignant hematopoiesis
HOX-expression
Self renewal capacity
Ectopic HOX expression induced by MLL fusions
block of differentiation

Characterization of the menin-MLL complex

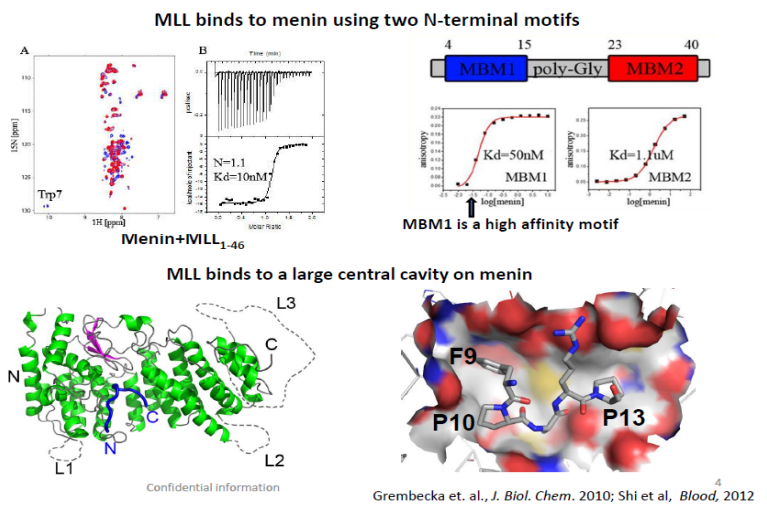
MLL binds to menin using two N-terminal motifs
A
B
Trp7
N=1.1
Kd=10nM
Menin+MLL1-46
4
15
23
40
MBM1
poly-Gly
MBM2
Kd=50nM
MBM1
Kd=1.1uM
MBM2
MBM1 is a high affinity motif
MLL binds to a large central cavity on menin
L3
C
N
C
N
L1
L2
F9
P10
P13
Confidential information

Grembecka et. al., *J. Biol. Chem.* 2010; Shi et al, *Blood,* 2012

KO-539 is a potent and selective menin inhibitor identified through structure-based optimization of a HTS hit

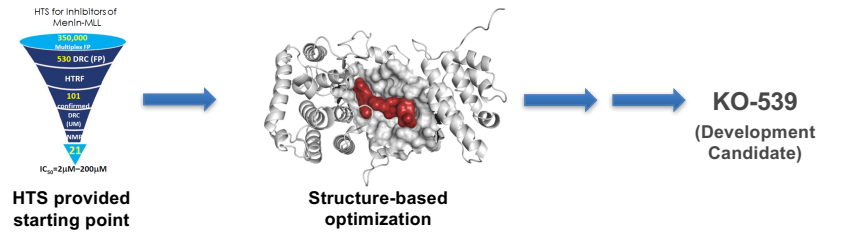
HTS for inhibitors of Menin-MLL
350,000
530 DRC (FP)
HTRF
101
21
HTS provided starting point
Structure-based optimization
KO-539
(Development Candidate)

Cell Lines with MLL Fusions as drivers (GI_{50})		Biochemical Assay (IC_{50})	
Murine BM Cell Line (rMLL-AF9)	7 nM	MLL(4-43)/Menin Binding	22 nM
MV4;11 (MLL-AF4 AML)	15 nM	Control Cell Lines without MLL Fusions (GI_{50})	
MOLM13 (MLL-AF9 AML)	16 nM	REH	1.5 μM
KOPN8 (MLL-ENL AML)	20 nM	U937	> 6 μM
RS4;11 (MLL-AF4 ALL)	23 nM	K562	> 6 μM
SEM (MLL-AF4 ALL)	17 nM	KG-1	> 4.5 μM

- KO-539 shows good biochemical and cellular activity
- KO-539 displays greater than 50-fold selectivity over cell lines without the MLL fusion

KO-539 demonstrates robust efficacy and a durable response in subcutaneous MV4;11 leukemia xenograft model in mice

Subcutaneous MV4;11 model

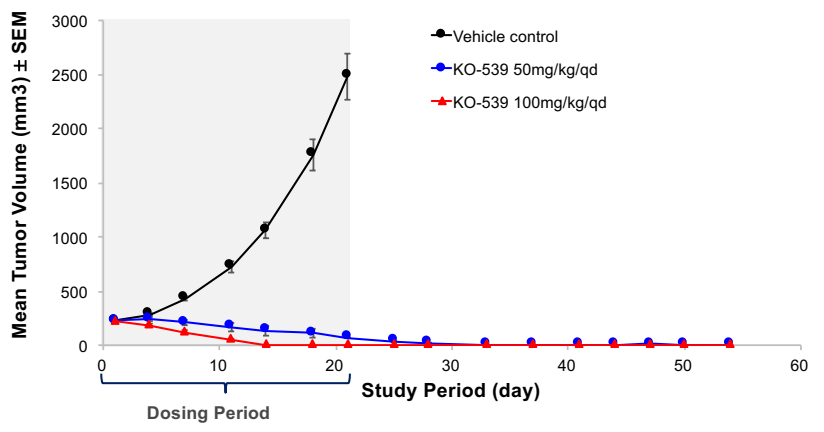
Vehicle control
KO-539 50mg/kg/qd
KO-539 100mg/kg/qd
Mean Tumor Volume (mm3) ± SEM
Study Period (day)
Dosing Period

- Tumor regression was observed at 50 and 100 mpk (oral dosing)
- Durable tumor growth inhibition: tumors were undetectable 30 days after cessation of dosing
- KO-539 is well tolerated at tested dose levels

KO-539 modulates pharmacodynamic (PD) biomarkers in the disseminated MV4;11 leukemia model in mice

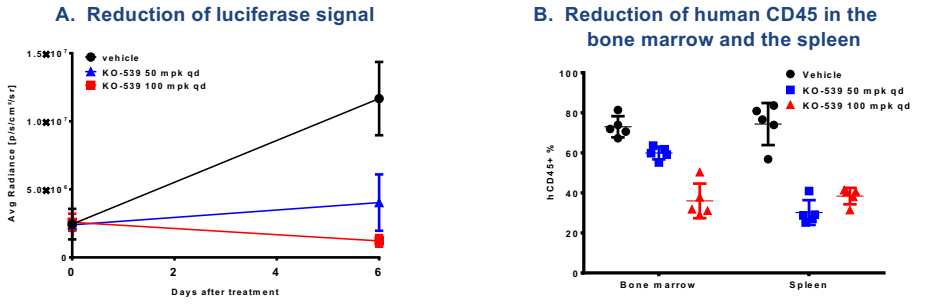
A. Reduction of luciferase signal
vehicle
KO-539 50 mpk qd
KO-539 100 mpk qd
Avg Radiance [p/s/cm²/sr]
Days after treatment
B. Reduction of human CD45 in the bone marrow and the spleen
Vehicle
KO-539 50 mpk qd
KO-539 100 mpk qd
hCD45+ %
Bone marrow
Spleen

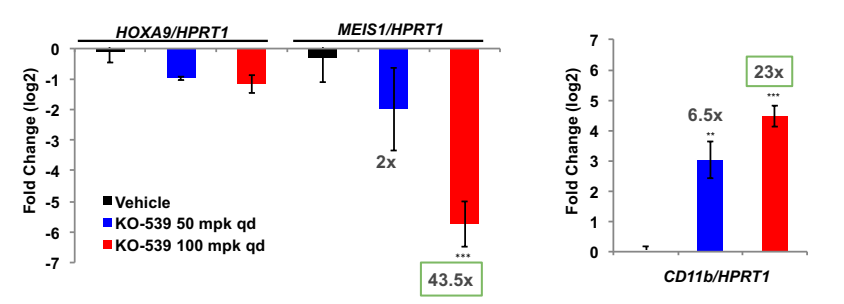
C. Pharmacodynamic activity in the bone marrow (modulation of menin target genes)
HOXA9/HPRT1
MEIS1/HPRT1
Fold Change (log2)
Vehicle
KO-539 50 mpk qd
KO-539 100 mpk qd
2x
43.5x
6.5x
23x
CD11b/HPRT1

- Dosing initiated 19 days post tumor implantation and continued for 6 days
- KO-539 reduces tumor burden relative to the vehicle in the bone marrow and the spleen
- KO-539 dose dependently modulated expression of menin-MLL associated PD markers

KO-539 displays survival benefit in the disseminated MV4;11 leukemia model

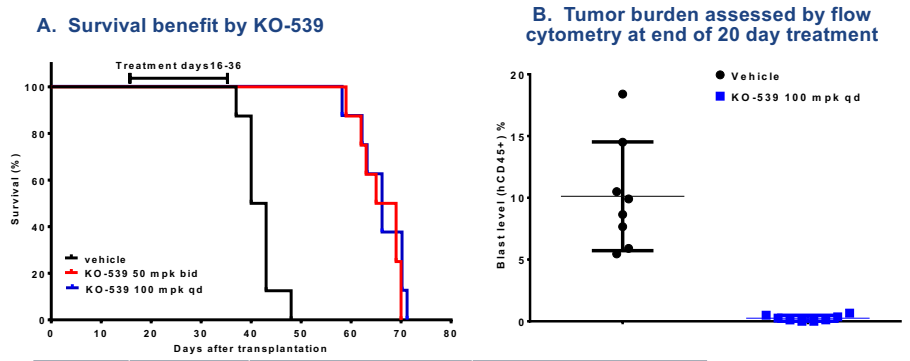
A. Survival benefit by KO-539
Treatment days16-36
vehicle
KO-539 50 mpk bid
KO-539 100 mpk qd
Survival (%)
Days after transplantation
B. Tumor burden assessed by flow cytometry at end of 20 day treatment
Vehicle
KO-539 100 mpk qd
Blast level (hCD45+) %

	Vehicle	KO-539 @ 50 mpk bid	KO-539 @ 100 mpk qd
Mean Survival	41.5 days	67 days	66 days

- KO-539 significantly prolongs mouse survival compared to the vehicle control group
- Survival benefit correlates with reduction of human leukemia blasts (hCD45⁺) in the mice

KO-539 extends survival in the aggressive systemic MOLM-13 leukemia model in mice

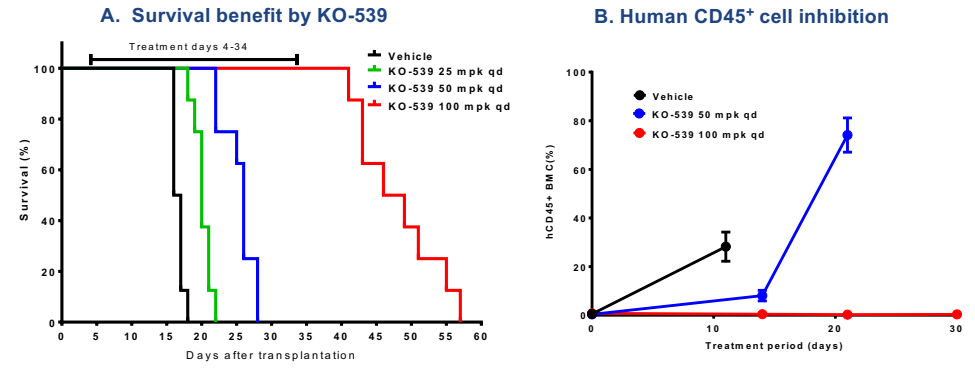
A. Survival benefit by KO-539
Treatment days 4-34
Vehicle
KO-539 25 mpk qd
KO-539 50 mpk qd
KO-539 100 mpk qd
Survival (%)
Days after transplantation
B. Human CD45+ cell inhibition
Vehicle
KO-539 50 mpk qd
KO-539 100 mpk qd
hCD45+ BMC(%)
Treatment period (days)

	Dose	Median Survival	Survival Benefit
Vehicle	-	16.5 days	-
KO-539	25 mpk qd	20 days	3.5 days
	50 mpk qd	26 days	9.5 days
	100 mpk qd	47.5 days	31 days

- KO-539 provides dose-dependent survival benefit in the aggressive disseminated MOLM13 leukemia model
- The observed survival benefit correlates with human blasts reduction

Conclusions

- KO-539 is a highly potent and selective inhibitor of the menin-MLL interaction
- KO-539 demonstrates robust, sustained tumor inhibition in subcutaneous and disseminated models of MLL-r leukemias; *in vivo* activity correlates with PD and target gene expression modulation
- KO-539 demonstrates acceptable tox profiles and drug-like properties
- KO-539 has been selected as a development candidate
- Additional efforts are underway to assess potential utilities of menin-MLL inhibitors in additional hematological malignancies and solid tumor indications

Acknowledgements

- We thank Shanghai Langtze Biomedical Technology Co, LTD Shanghai, China for their support on synthesis of menin-MLL inhibitors
- We thank Dr. Thomas Look, Dana Farber Cancer Institute, for providing MV4;11 cells expressing luciferase to the University of Michigan for studies